A novel synthetic stealth receptor that redirects host immune cell alloreactivity and potentiates functional persistence of adoptively transferred off-the-shelf cell-based cancer therapy

2828

A.M. Williams[1], K.L Hayama[1], Y. Pan[1], B. Groff[1], R. Mbofung[1], L. Fong[1], N. Brookhouser[1], B. Mandefro[1], R. Abujarour[1], T. Lee[1], Q. Hammer[2], KJ. Malmberg[2], M. Mamonkin[3], R. Bjordahl[1], J.P. Goodridge[1], B. Valamehr[1]

1. Fate Therapeutics, Inc., San Diego, CA, USA 2. University of Oslo, Norway 3. Baylor University, Waco, TX, USA

INTRODUCTION

Chimeric antigen receptor (CAR) T-cell therapies have revolutionized the treatment of hematologic malignancies and have shown significant potential in solid tumor indications. However, complexities associated with patient-specific CAR T-cell therapies often limit broad patient accessibility. Many of these challenges can be overcome with an allogeneic cellular product.

Both autologous and allogeneic cell therapies currently rely on lympho-conditioning of the patient, which induces a cytokine-rich environment for potentiation of adoptively-transferred cells and modulates the host immune system. Lympho-conditioning, however, has been associated with hematologic toxicities, including increased susceptibility to severe infections. Therefore, next-generation cell therapies should seek to significantly reduce the requirement for chemotherapy conditioning while maintaining the anti-tumor activity of adoptively-transferred cells.

To address these challenges, we developed a novel alloimmune defense receptor (ADR) that selectively targets host immune cells and significantly boosts the functional activity of adoptively-transferred cells. In preclinical studies, off-the-shelf iPSC-derived NK (CAR-iNK) cells armed with a novel ADR exhibited functional persistence and maintained anti-tumor activity in an immuno-competent host system.

STRATEGIES TO REDUCE LYMPHO-CONDITIONING

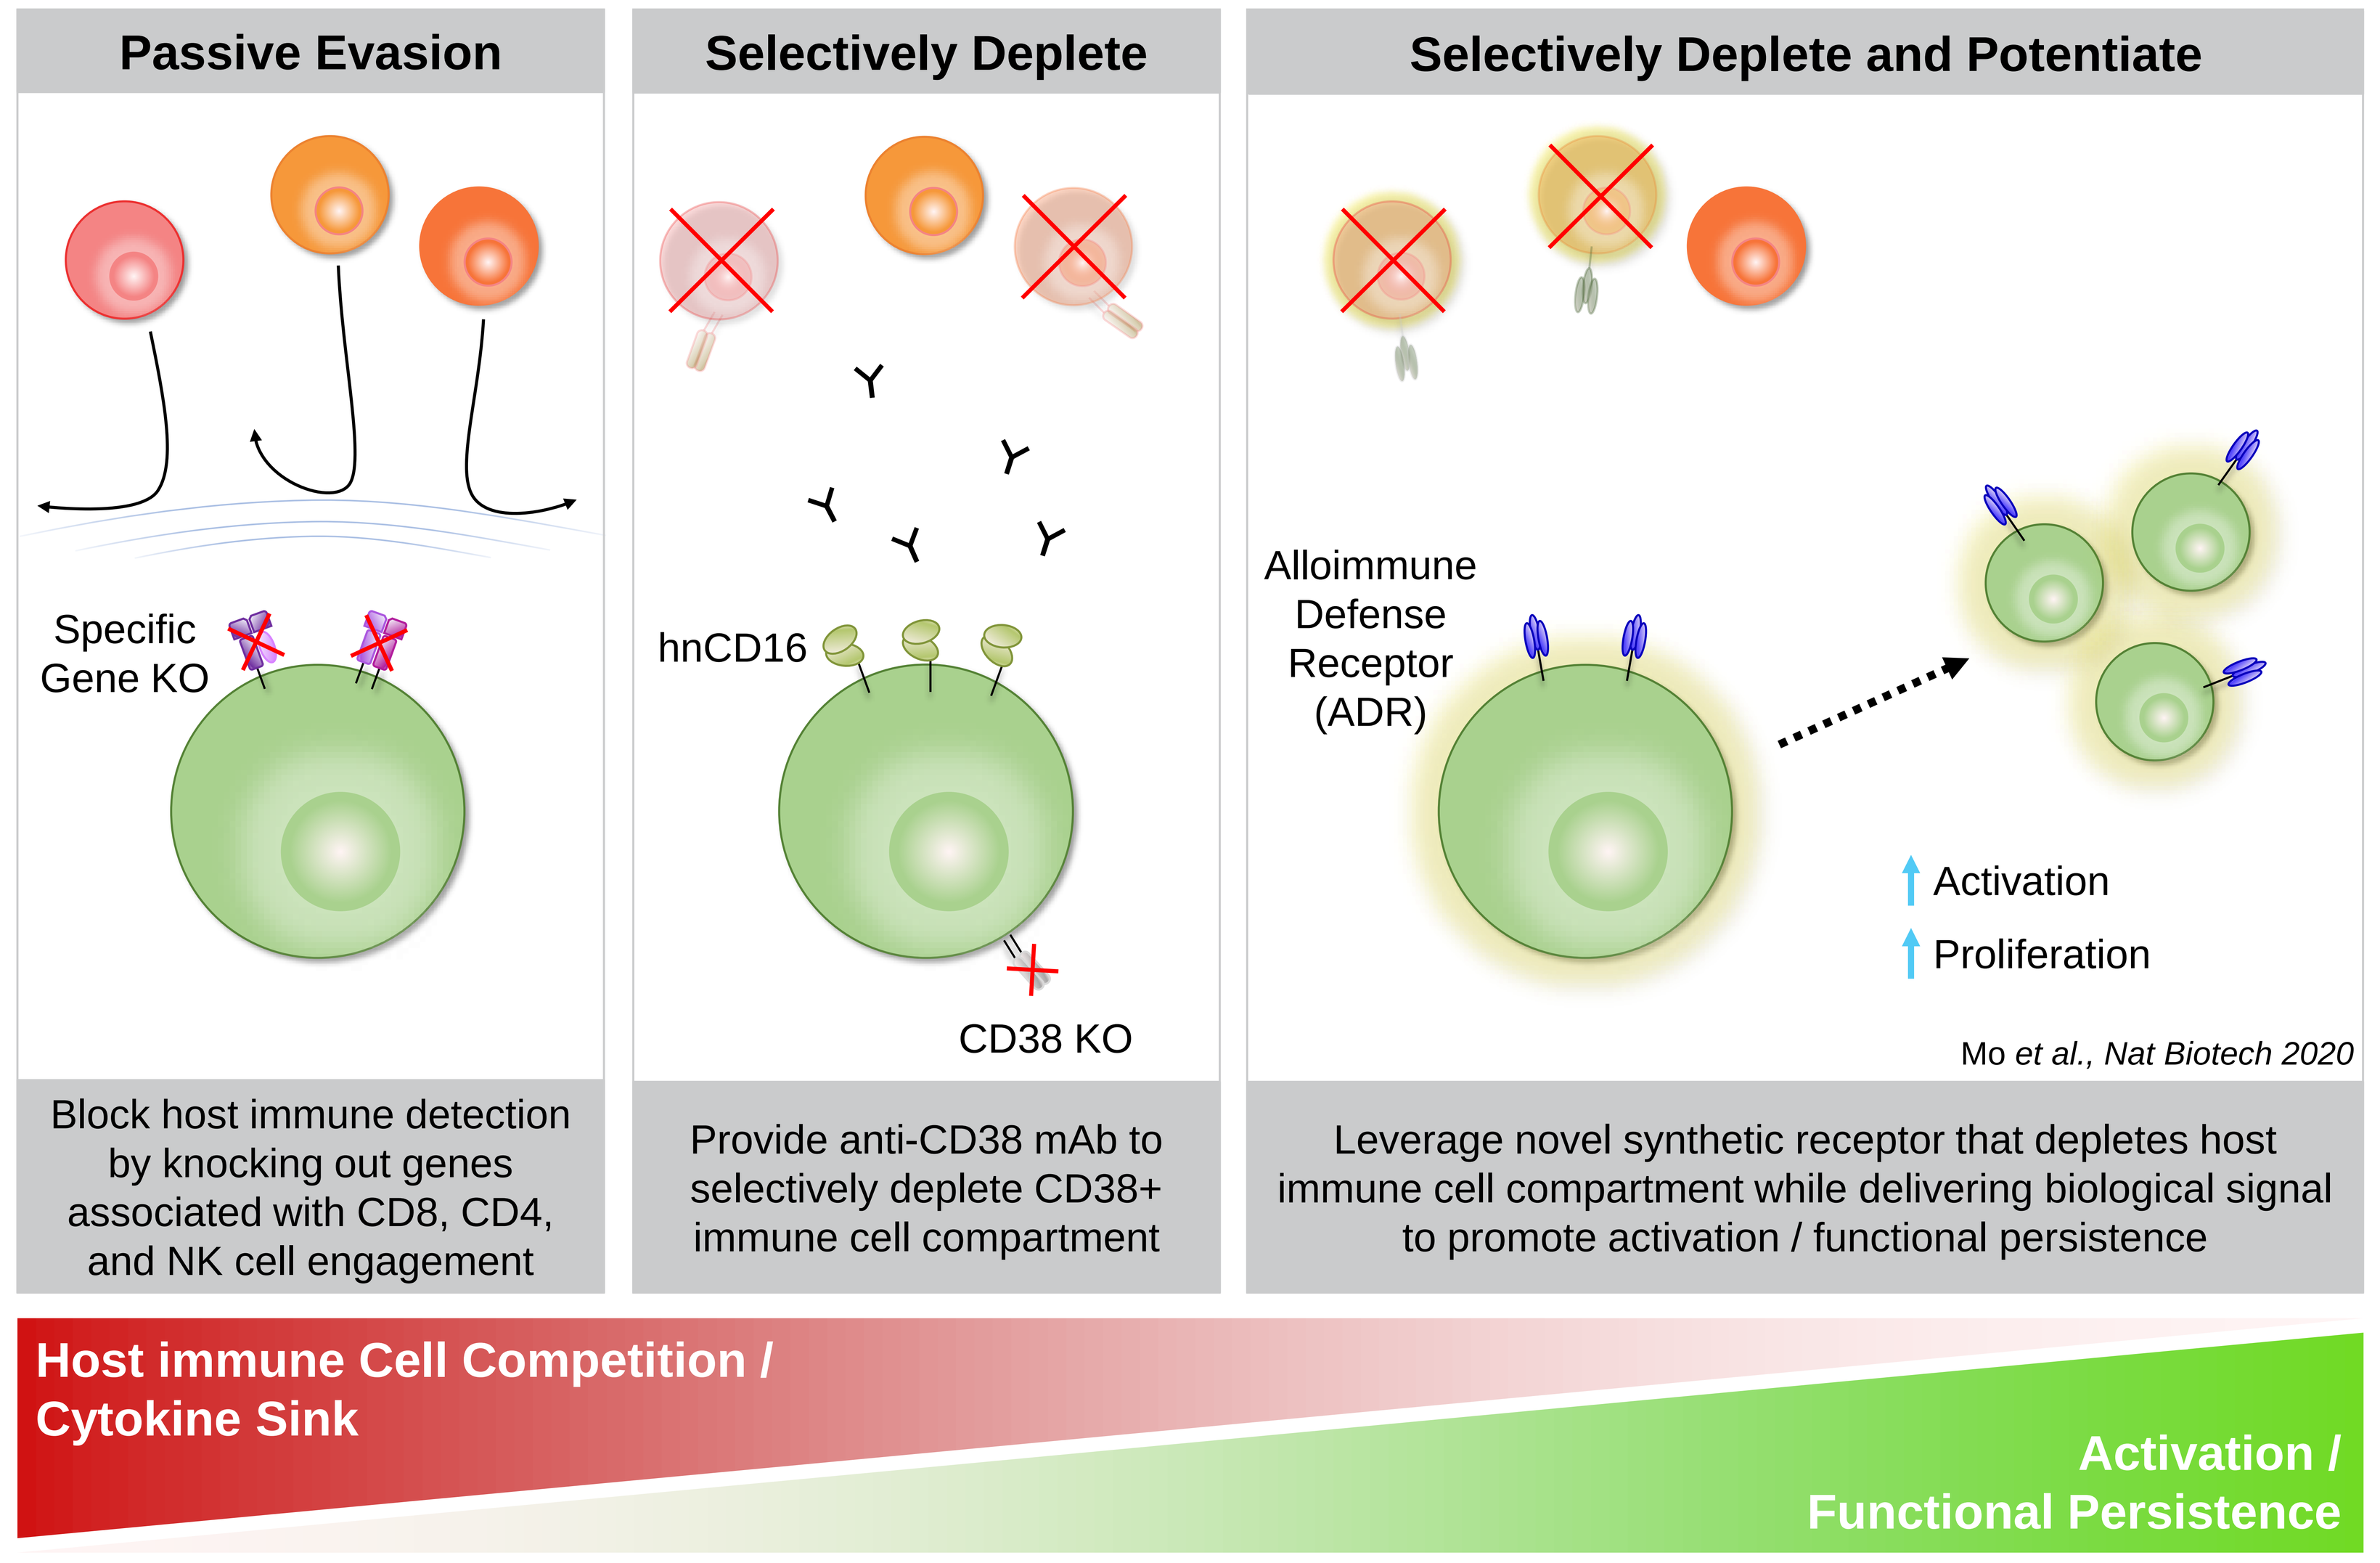
Passive Evasion
Selectively Deplete
Selectively Deplete and Potentiate
Specific Gene KO
hnCD16
CD38 KO
Alloimmune Defense Receptor (ADR)
Activation
Proliferation
Mo et al., Nat Biotech 2020
Block host immune detection by knocking out genes associated with CD8, CD4, and NK cell engagement
Provide anti-CD38 mAb to selectively deplete CD38+ immune cell compartment
Leverage novel synthetic receptor that depletes host immune cell compartment while delivering biological signal to promote activation / functional persistence
Host immune Cell Competition / Cytokine Sink
Activation / Functional Persistence

Mo *et al.*, Engineered off-the-shelf therapeutic T cells resist host immune rejection. Nature Biotechnology 2020
doi: 10.1038/s41587-020-0601-5

CONCLUSIONS

Our preclinical work provides a framework to eliminate the need for patient lympho-conditioning in the field of cell-based cancer immunotherapy by arming adoptively-transferred cells with a novel alloimmune defense receptor (ADR) targeting 4-1BB expressed on host NK and T cells.

- 4-1BB is upregulated on activated effector cells, including allo-reactive NK and T cells.
- ADR+ CAR-iNK cells uniquely demonstrate high levels of functional persistence *in vitro* in the presence of allo-reactive lymphocytes.
- Unlike ADR- controls, anti-tumor efficacy of ADR+ CAR-iNK cells is uncompromised *in vivo* in the presence of a competent allo-reactive immune system.

These data support the development of off-the-shelf, iPSC-derived CAR NK cell therapies which, when armed with a novel ADR targeting 4-1BB, have the potential to maintain potent anti-tumor activity without requiring chemotherapy conditioning.

RESULTS

Off-the-shelf, iPSC-derived CAR NK cells armed with a novel synthetic alloimmune defense receptor (ADR) targeting 4-1BB expressed on allo-reactive immune cells

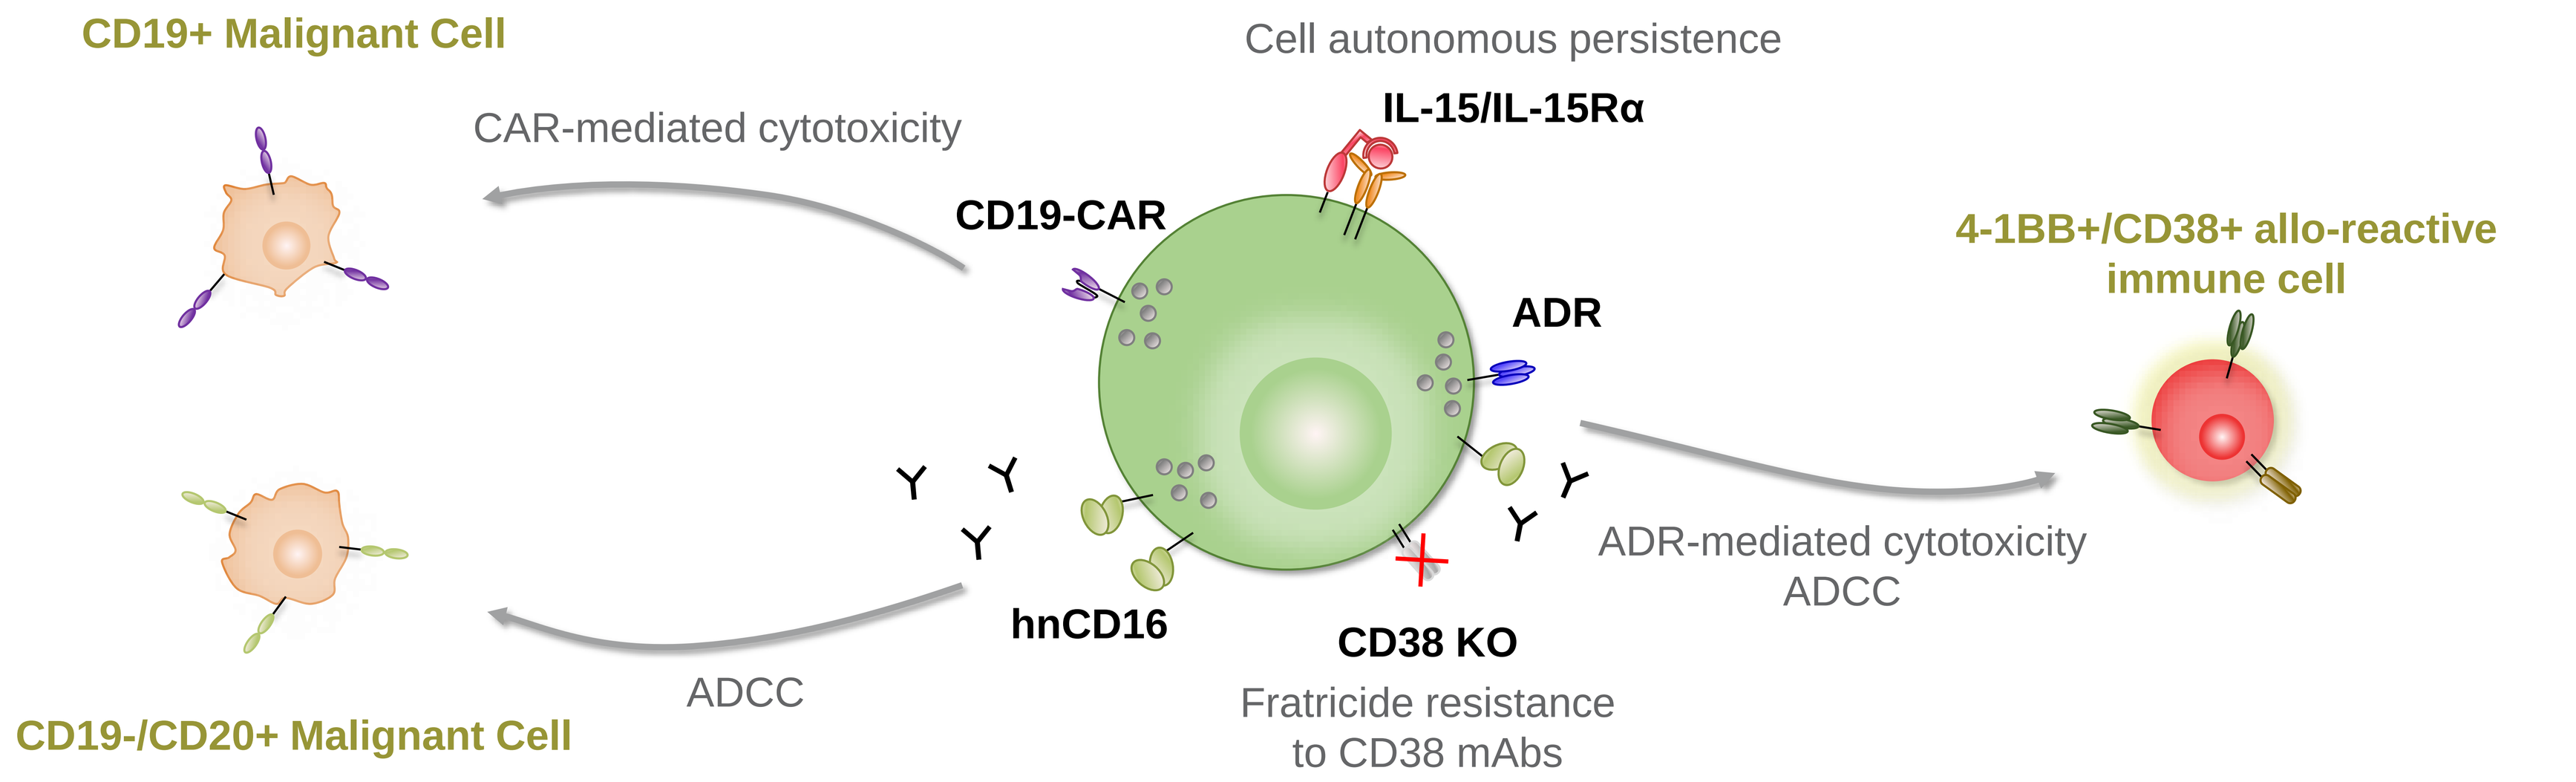
CD19+ Malignant Cell
CAR-mediated cytotoxicity
Cell autonomous persistence
IL-15/IL-15Rα
CD19-CAR
ADR
4-1BB+/CD38+ allo-reactive immune cell
ADR-mediated cytotoxicity
ADCC
hnCD16
CD38 KO
Fratricide resistance to CD38 mAbs
ADCC
CD19-/CD20+ Malignant Cell

Figure 1 iNK cells are engineered to express: (1) high-affinity, non-cleavable CD16 (hnCD16) to augment antibody dependent cellular cytotoxicity (ADCC); (2) an IL-15/IL-15Rα fusion to enhance NK functionality and persistence; (3) CD38 knockout to eliminate anti-CD38 monoclonal antibody-induced fratricide; (4) CD19-CAR to selectively target CD19 antigen expressed on B-cell lymphomas; (5) ADR to selectively target 4-1BB expressed on allo-reactive NK and T cells.

ADR-armed CAR-iNK cells demonstrate functional persistence *in vitro* in the presence of allo-reactive T and NK cells

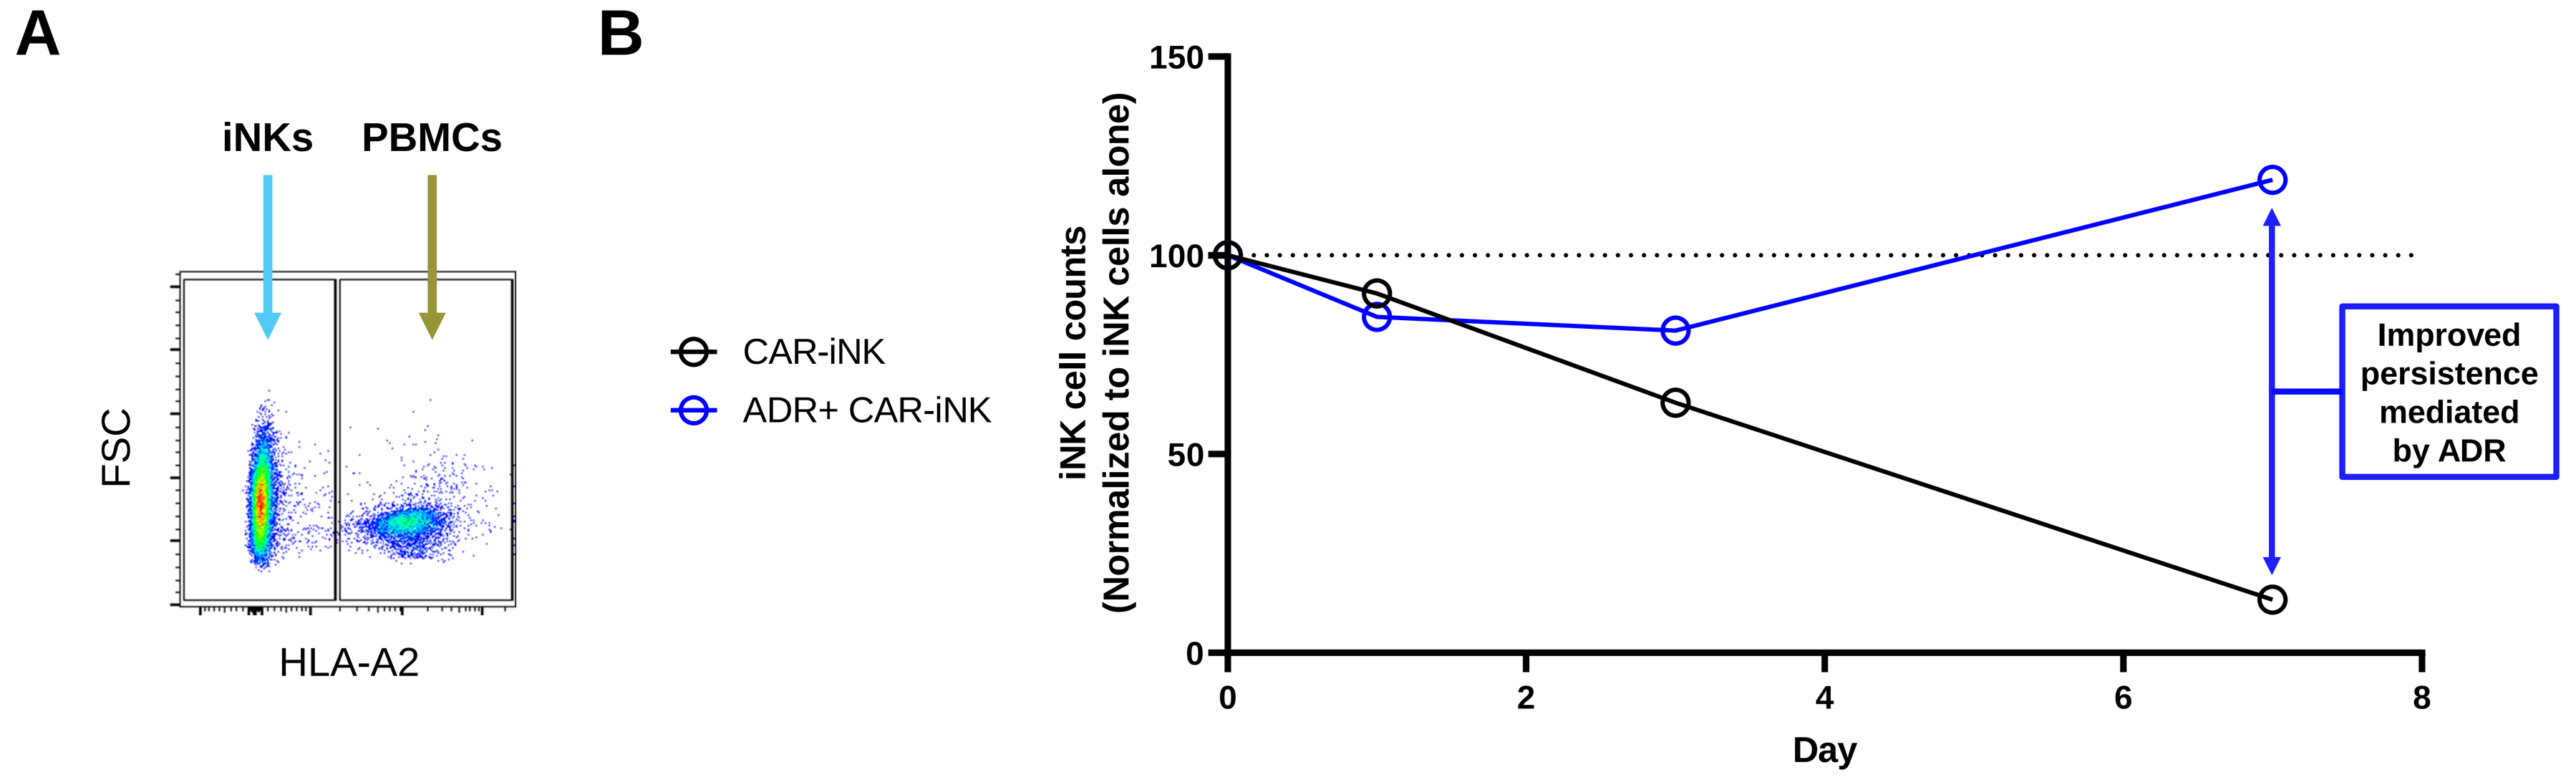
A
B
iNKs
PBMCs
FSC
HLA-A2
CAR-iNK
ADR+ CAR-iNK
iNK cell counts (Normalized to iNK cells alone)
Improved persistence mediated by ADR
Day

Figure 2 ADR+ CAR-iNK cells demonstrate enhanced functional persistence in mixed lymphocyte reactions (MLR). (A) Representative FACS plot depicting gating schematic to quantify iNK cells against HLA-A2+ PBMCs. (B) CAR-iNK ± ADR cells were co-cultured at a 4:1 iNK cell to PBMC ratio. iNK cell counts from the co-cultures were plotted at the indicated timepoints. Data are normalized to iNK cells cultured without PBMCs.

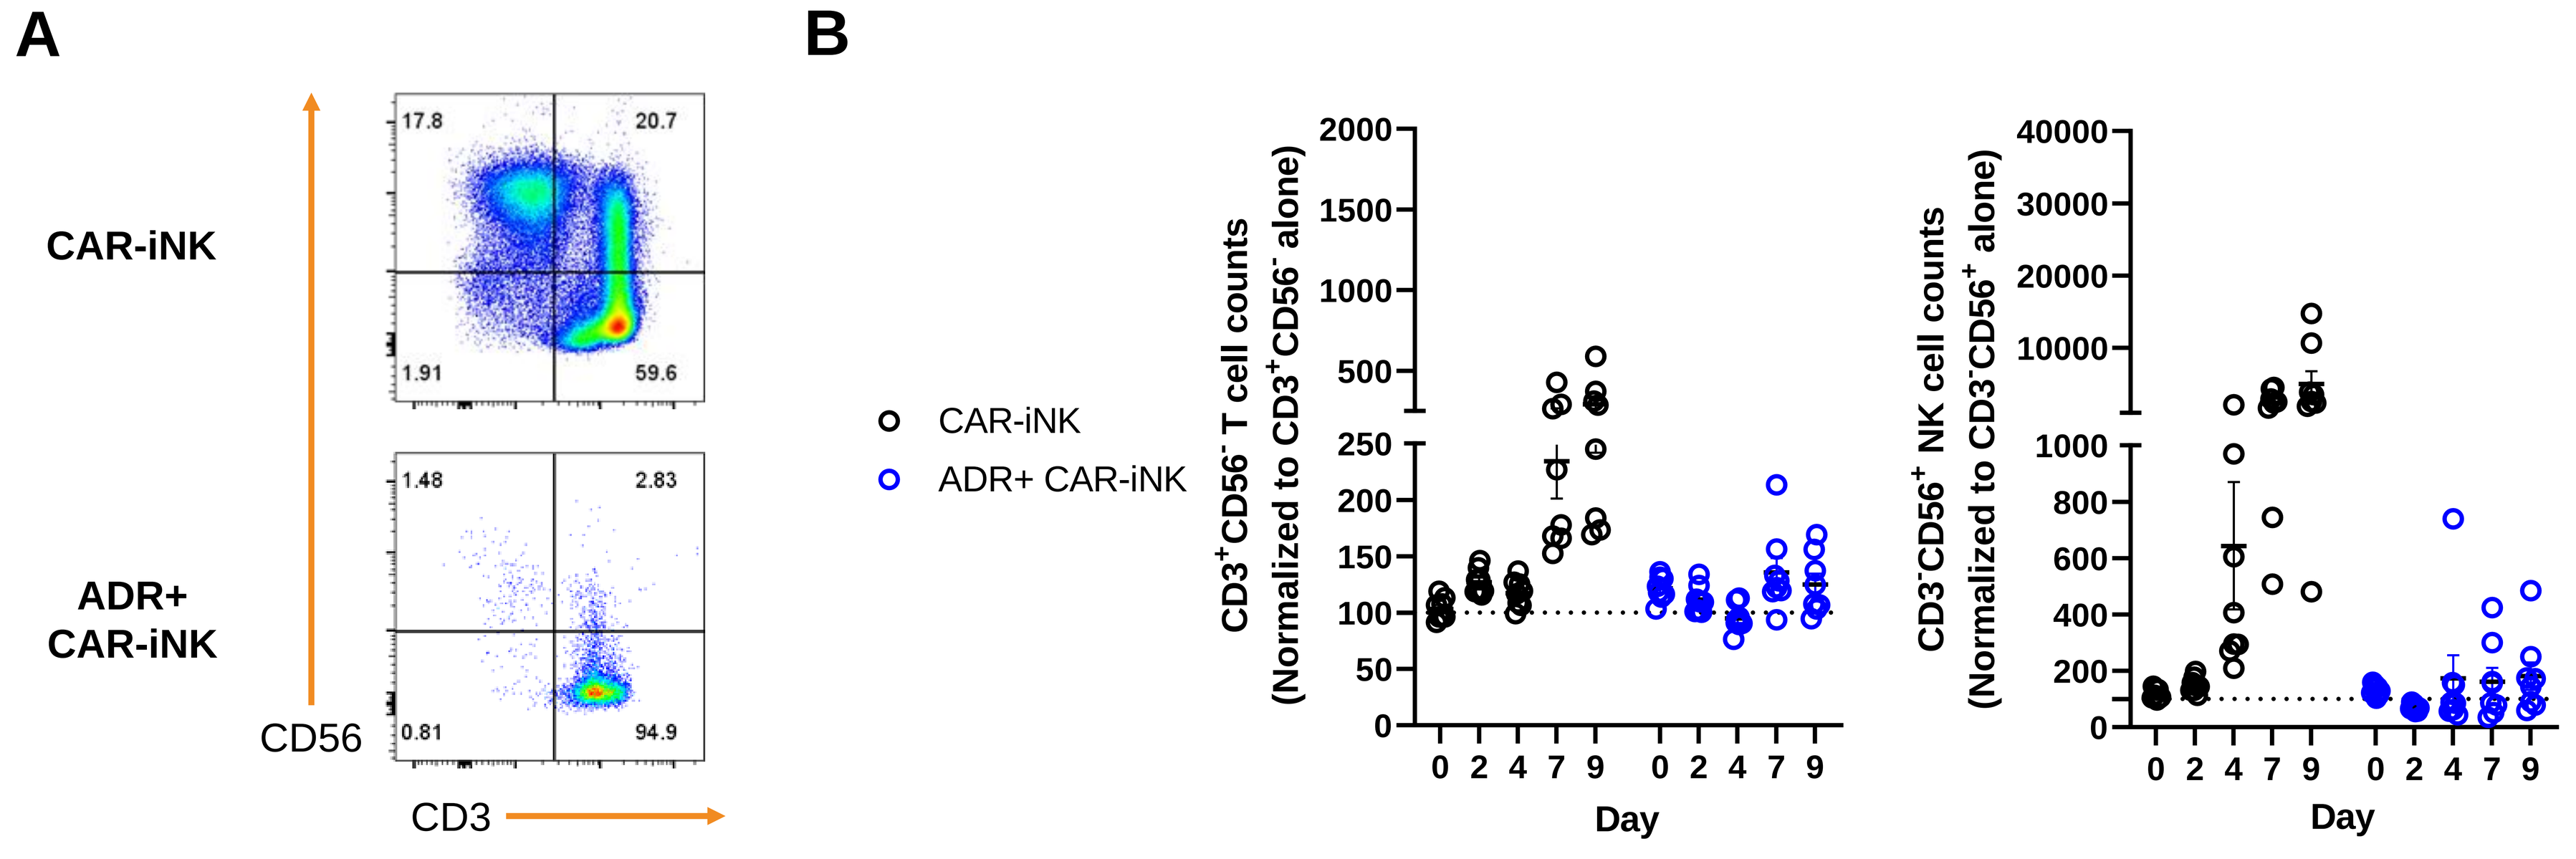
A
B
CAR-iNK
ADR+ CAR-iNK
CD56
CD3
17.8
20.7
1.91
59.6
1.48
2.83
0.81
94.9
CAR-iNK
ADR+ CAR-iNK
CD3+CD56- T cell counts (Normalized to CD3+CD56- alone)
CD3-CD56+ NK cell counts (Normalized to CD3-CD56+ alone)
Day

Figure 3 ADR+ CAR-iNK cells stifle expansion of allo-reactive T and NK cells. (A) Representative FACS plots depicting gating schematic of CD3+ T and CD56+ NK cells after nine days of co-culture with iNK cells. (B) Quantification of T- and NK cell counts across eight donors co-cultured with CAR-iNK ± ADR cells at a 2:1 iNK cell to PBMC ratio. Data are normalized to T- and NK cell counts from cultures of eight donor PBMCs maintained in the absence of iNK cells.

ADR-armed CAR-iNK cells selectively target and deplete allo-reactive CD8+ and CD4+ T-cell compartments *in vitro*

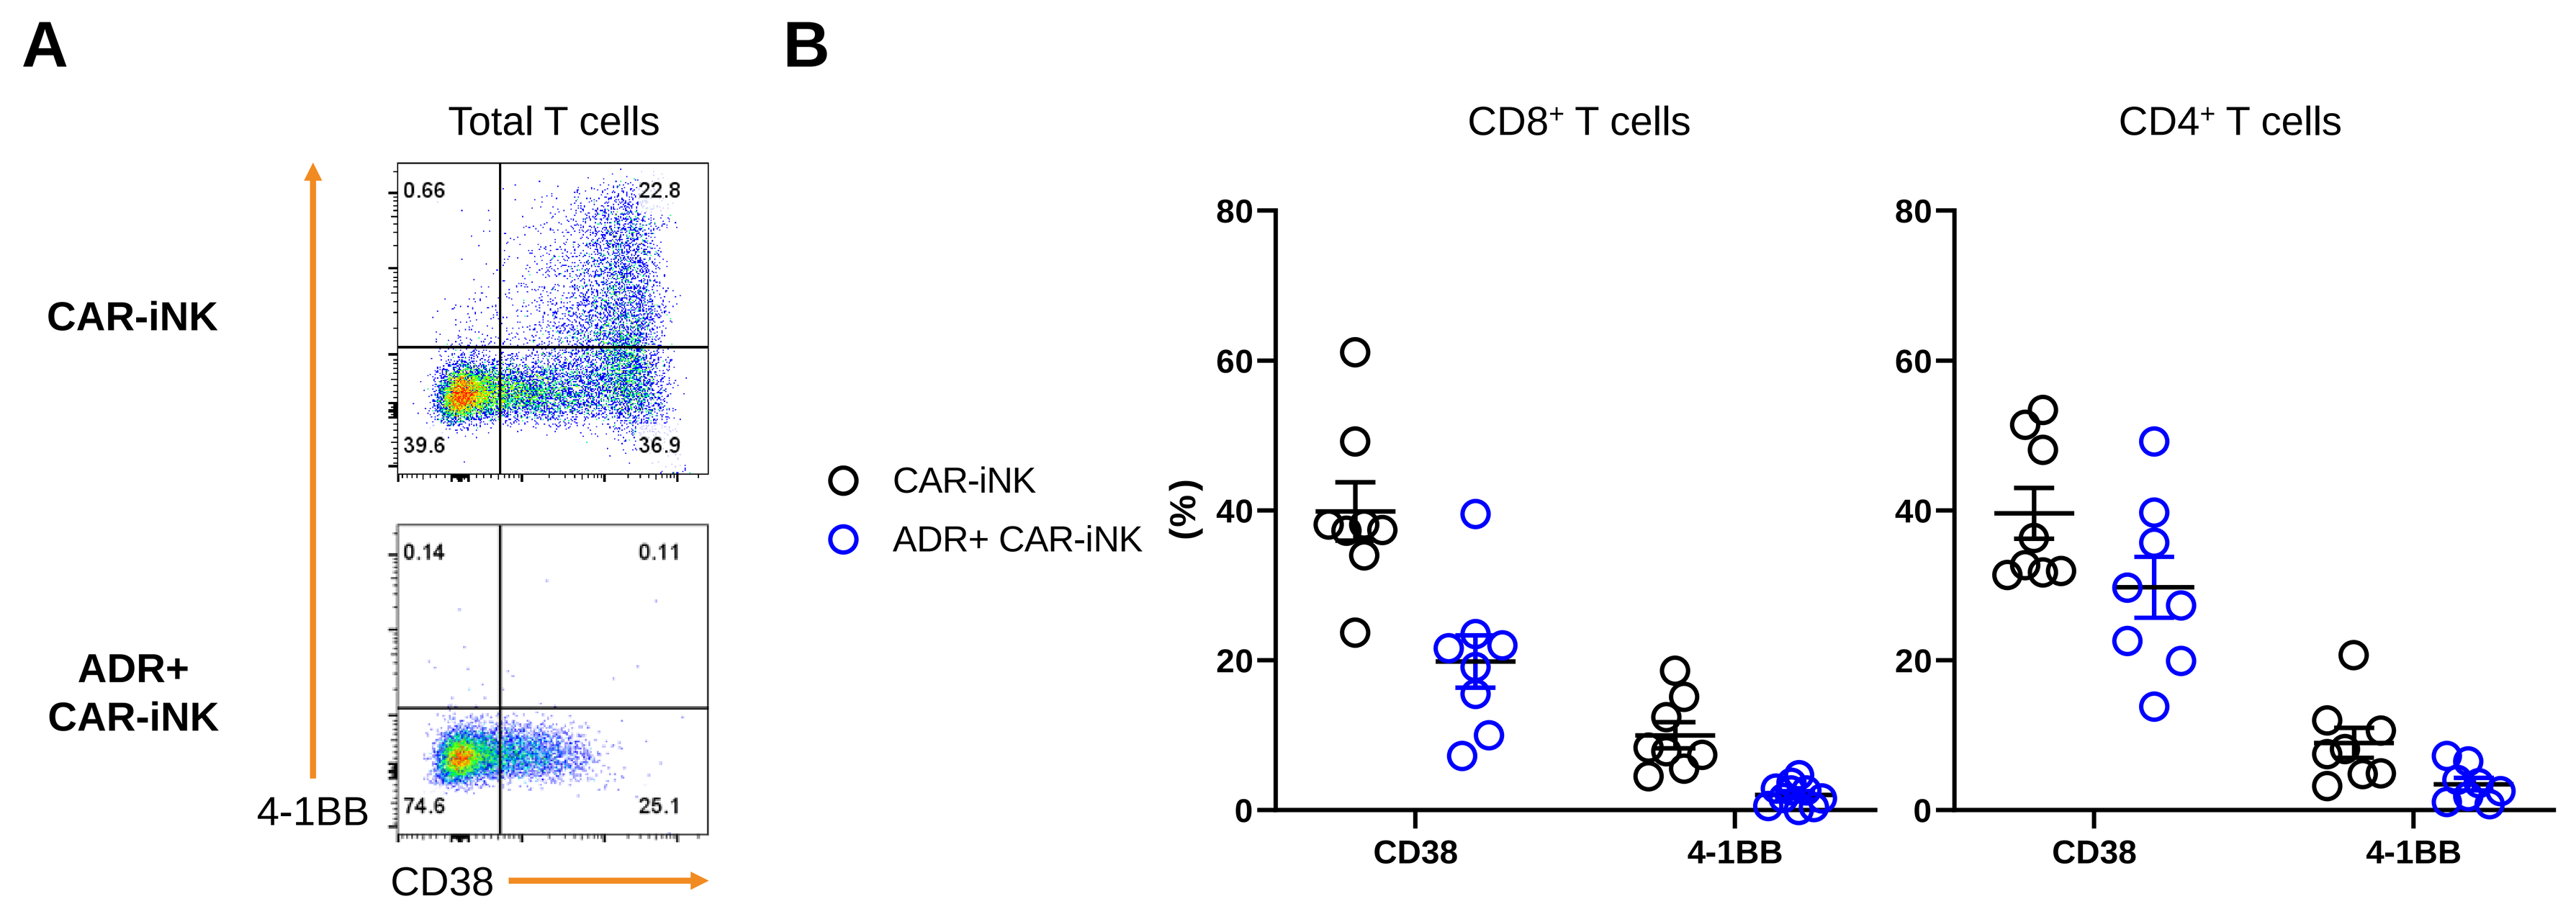
A
B
Total T cells
CAR-iNK
ADR+ CAR-iNK
4-1BB
CD38
0.66
22.8
39.6
36.9
0.14
0.11
74.6
25.1
CAR-iNK
ADR+ CAR-iNK
CD8+ T cells
CD4+ T cells
(%)
CD38
4-1BB

Figure 4 ADR+ CAR-iNK cells selectively target both CD4+ and CD8+ allo-reactive T-cell subsets. (A) Representative FACS plots of T cells after nine days of co-culture with iNK cells and analyzed for expression of CD38 and 4-1BB among CD3+ T cells as depicted in Figure 2. (B) Compilation of %CD38 and %4-1BB expression among CD4+ and CD8+ T cells from the eight donors in CAR-iNK ± ADR co-cultures.

ADR-armed CAR-iNK cells exhibit uncompromised tumor control *in vivo* in the presence of host allo-reactive T-cell system

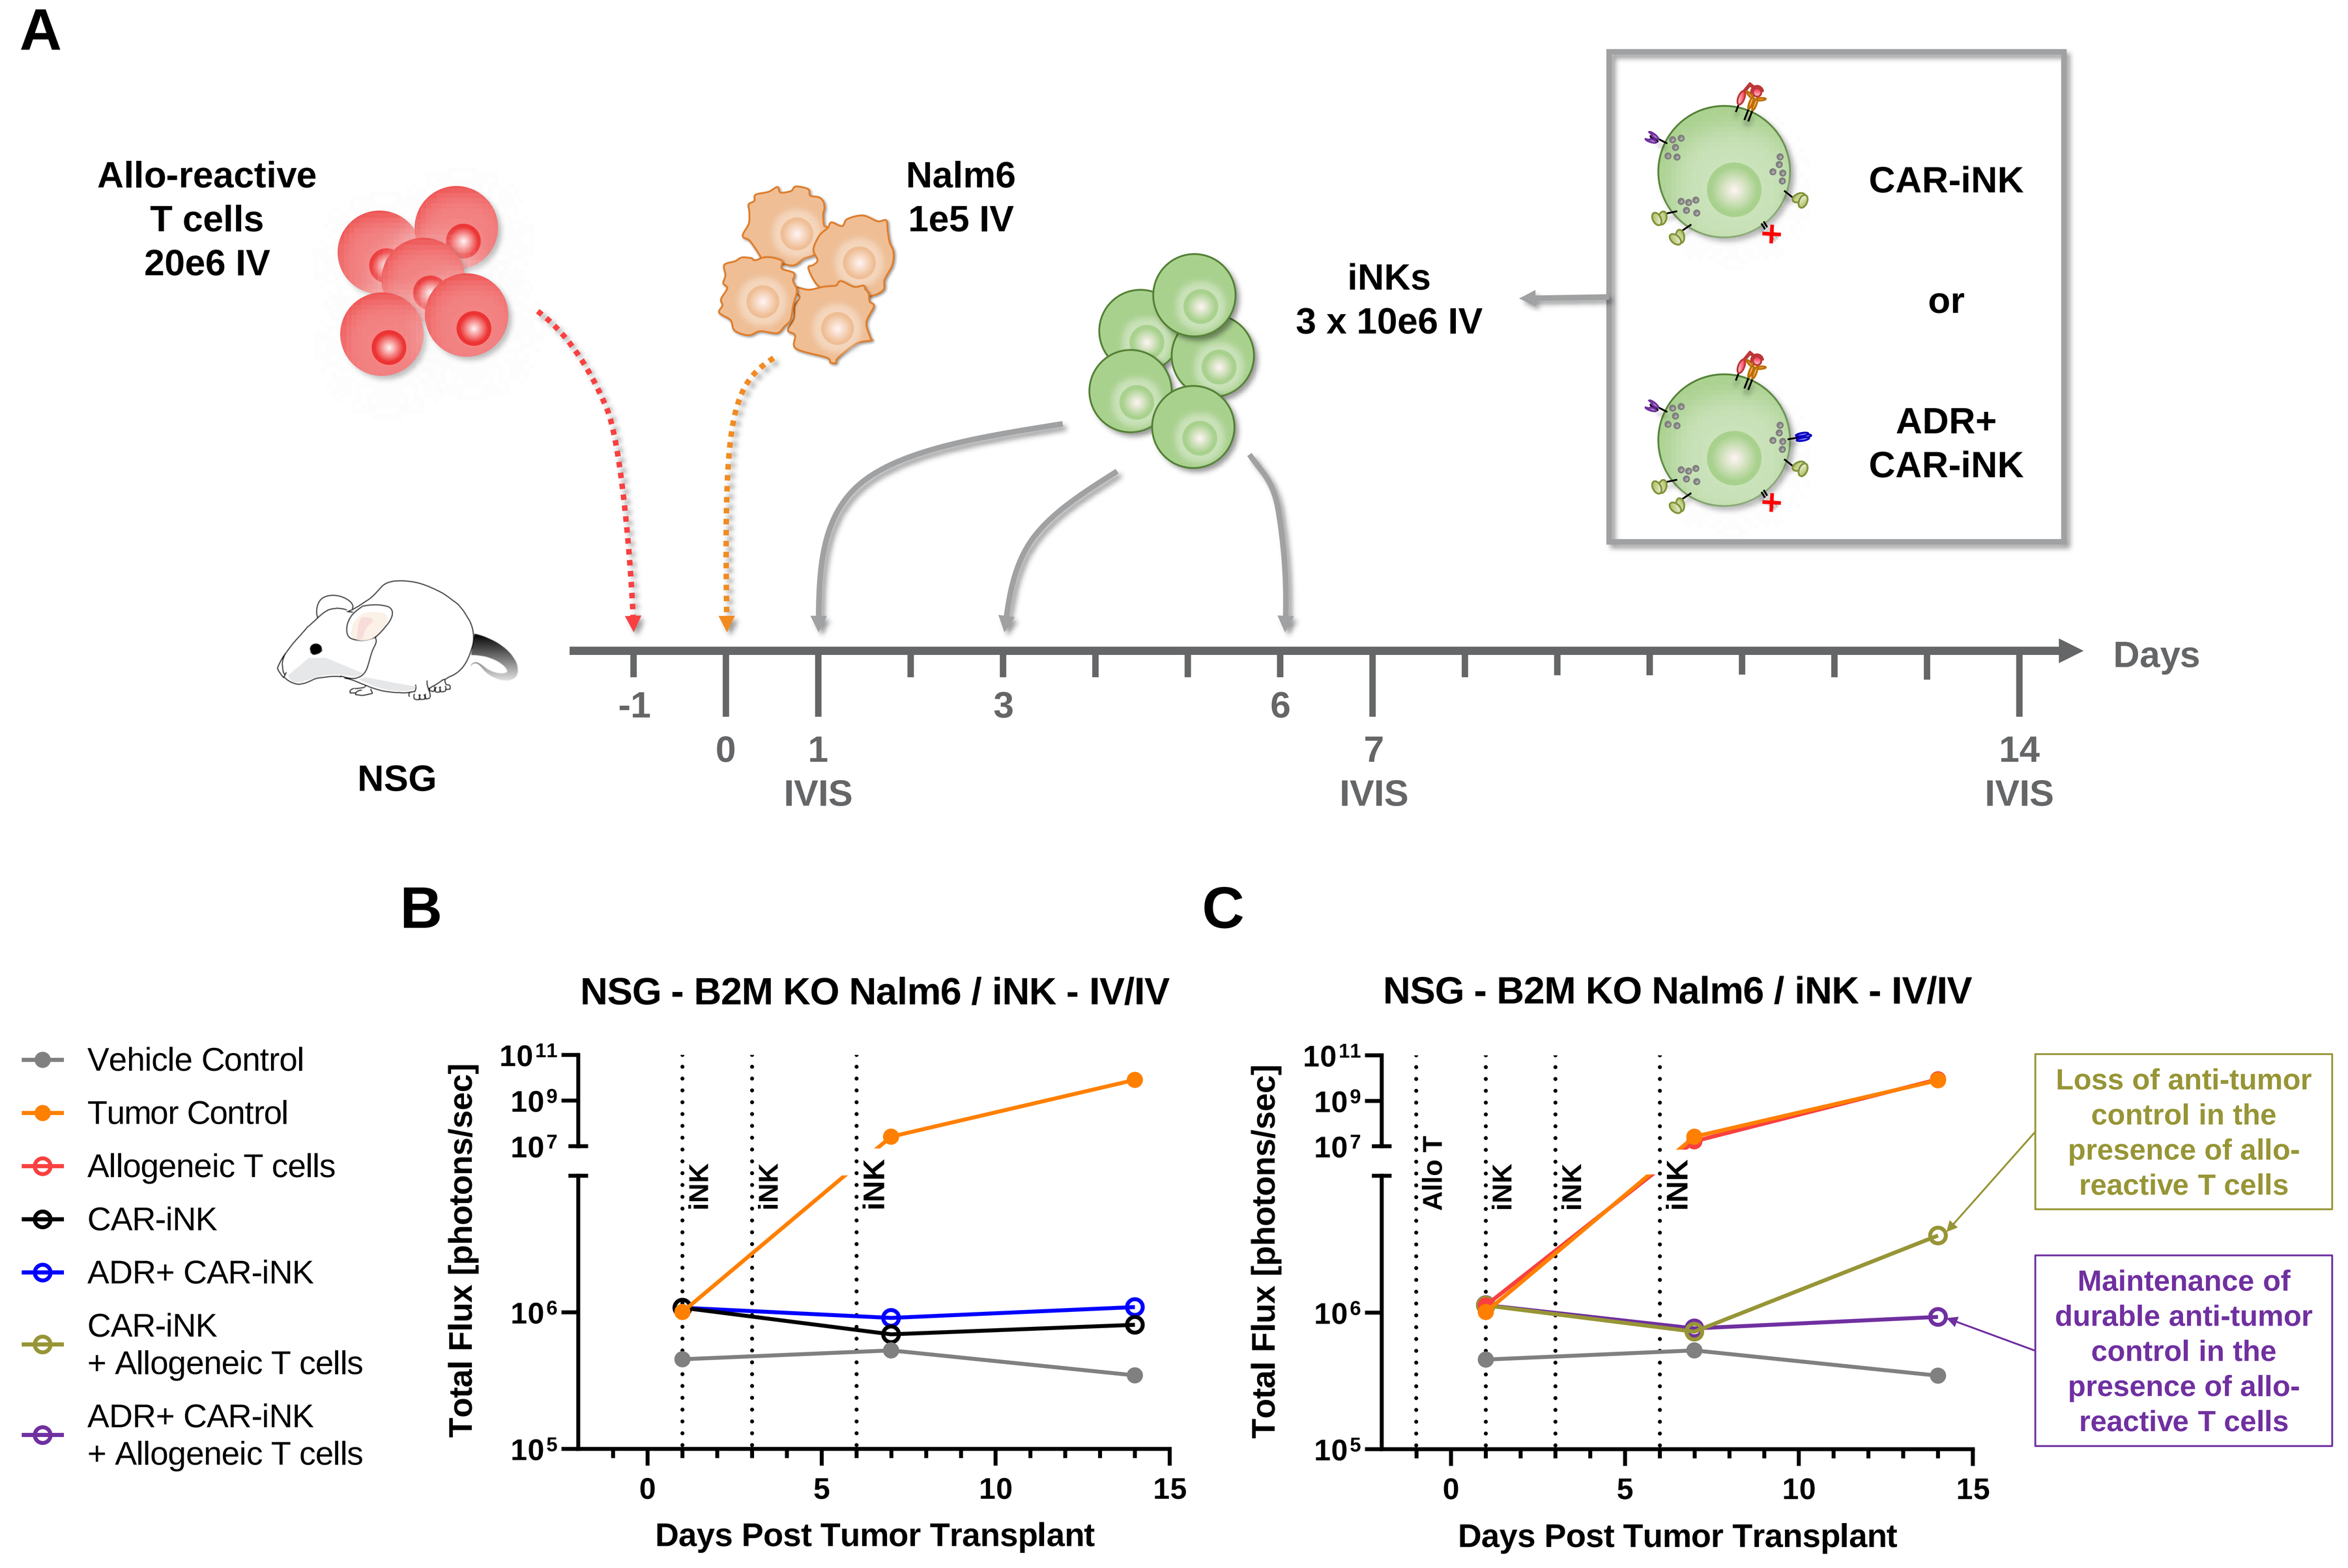
A
Allo-reactive T cells 20e6 IV
Nalm6 1e5 IV
iNKs 3 x 10e6 IV
CAR-iNK
or
ADR+ CAR-iNK
NSG
Days
IVIS
B
C
NSG - B2M KO Nalm6 / iNK - IV/IV
Vehicle Control
Tumor Control
Allogeneic T cells
CAR-iNK
ADR+ CAR-iNK
CAR-iNK + Allogeneic T cells
ADR+ CAR-iNK + Allogeneic T cells
Total Flux [photons/sec]
Days Post Tumor Transplant
iNK
Allo T
Loss of anti-tumor control in the presence of allo-reactive T cells
Maintenance of durable anti-tumor control in the presence of allo-reactive T cells

Figure 5 *In vivo* schematics. (A) NSG mice were injected with iNK cells, luciferized B2M KO Nalm6 and allo-reactive T cells. Allo-reactive T cells were previously expanded *in vitro* for 10 days with 100IU/ml IL-2 and Dynabeads™ Human T-Activator CD3/CD28 as indicated by manufacturer protocol. (B) Bioluminescence-based tumor quantification by total flux was measured to monitor tumor growth in mice. (C) Total flux analysis of mice injected with allo-reactive T cells.