Trimodal CAR⁺TCR⁺hnCD16⁺ iPSC-derived T Cells Co-Targeting Surface and Intracellular/Neoantigens Demonstrate Additive Effect on Overcoming Tumor Heterogeneity and Cancer Escape

Bi-Huei Yang, Soheila Shirinbak, Wen-I Yeh, Alma Gutierrez, Mochtar Pribadi, Bjoern Gaertner, Hui-Yi Chu, David Gonzalez, Angela Liao, Tom Lee, Eigen Peralta, Ryan Bjordahl, Martin Hosking, Chia-Wei Chang, and Bahram Valamehr

Fate Therapeutics, Inc., San Diego, CA, USA

ABSTRACT

Background

Antigen escape and tumor heterogeneity remain significant hurdles to the development of curative treatments in many cancers. To address tumor heterogeneity, the introduction of pairs of chimeric antigen receptors (CARs) in donor T cells has been demonstrated, however, this adds complexity to a manufacturing process already challenged by cellular product consistency. To tackle tumor heterogeneity while maintaining product purity, we applied our induced pluripotent stem cell (iPSC)-derived T cell (iT) platform to design an off-the-shelf cell therapy capable of targeting multiple tumor antigens through complementary activation pathways, including targeting of both cell-surface antigens as well as intracellular/neoantigens.

Methods

A CAR construct targeting BCMA or MICA/B, a T-cell receptor (TCR)αβ targeting NYESO1 (1G4) or tumor-associated metabolite presented by MR1 (MC.7.G5), and a high-affinity non-cleavable (hnCD16) to promote antibody-dependent cellular cytotoxicity (ADCC) were engineered into iPSCs for use as a renewable starting source in deriving uniformly-engineered T cells. Resulting multiplexed-engineered CAR⁺TCR⁺hnCD16⁺ (trimodal) iPSCs were differentiated into T cells and the function of each individual edit was evaluated by preclinical models designed to represent tumor heterogeneity. In addition to in vitro mix-culture assays, a mixed cell disseminated in vivo model was used to mimic cancer heterogeneity and to evaluate the in vivo potency of trimodal iT cells at mitigating tumor heterogeneity and antigen escape.

Results

Assessment of individual edits in trimodal iT cells demonstrated independent functionality by exhibiting increased antigen-mediated IFNγ and TNFα production, and degranulation compared to the control group ($p<0.0001$). Using 9-day daily restimulation assay, each edit produced significant tumor reduction compared to tumor only control ($p<0.0001$). By stimulating trimodal iT cells with multiple antigens simultaneously using various solid tumor lines (A549, Caski and MDA-MB-231), we found that co-activation by two or three targeting edits significantly enhanced tumor killing ($p<0.0001$). Furthermore, when challenged with in vivo heterogenous tumor models, we found that the co-activation of all three targeting moieties in trimodal iT cells achieved nearly complete tumor clearance ($p<0.0001$). Ex vivo bone marrow analysis further confirmed antigen-specific target elimination, reenforcing the specificity and potency of the trimodal iT cells.

Conclusions

Our data highlight the potency and broad applicability of trimodal iT cell expressing CAR, TCR, and hnCD16. This consistent and scalable approach to multiplex-engineered T-cell therapy is an ideal strategy to mitigate antigen escape and combat difficult to treat heterogeneous solid tumors.

GRAPHICAL ABSTRACT

Enhance Specificity and Multi-antigen Targeting: Use of synthetic biology to integrate multiple antigen-targeting modalities into iPSC derived iT cells for enhanced anti-tumor activity

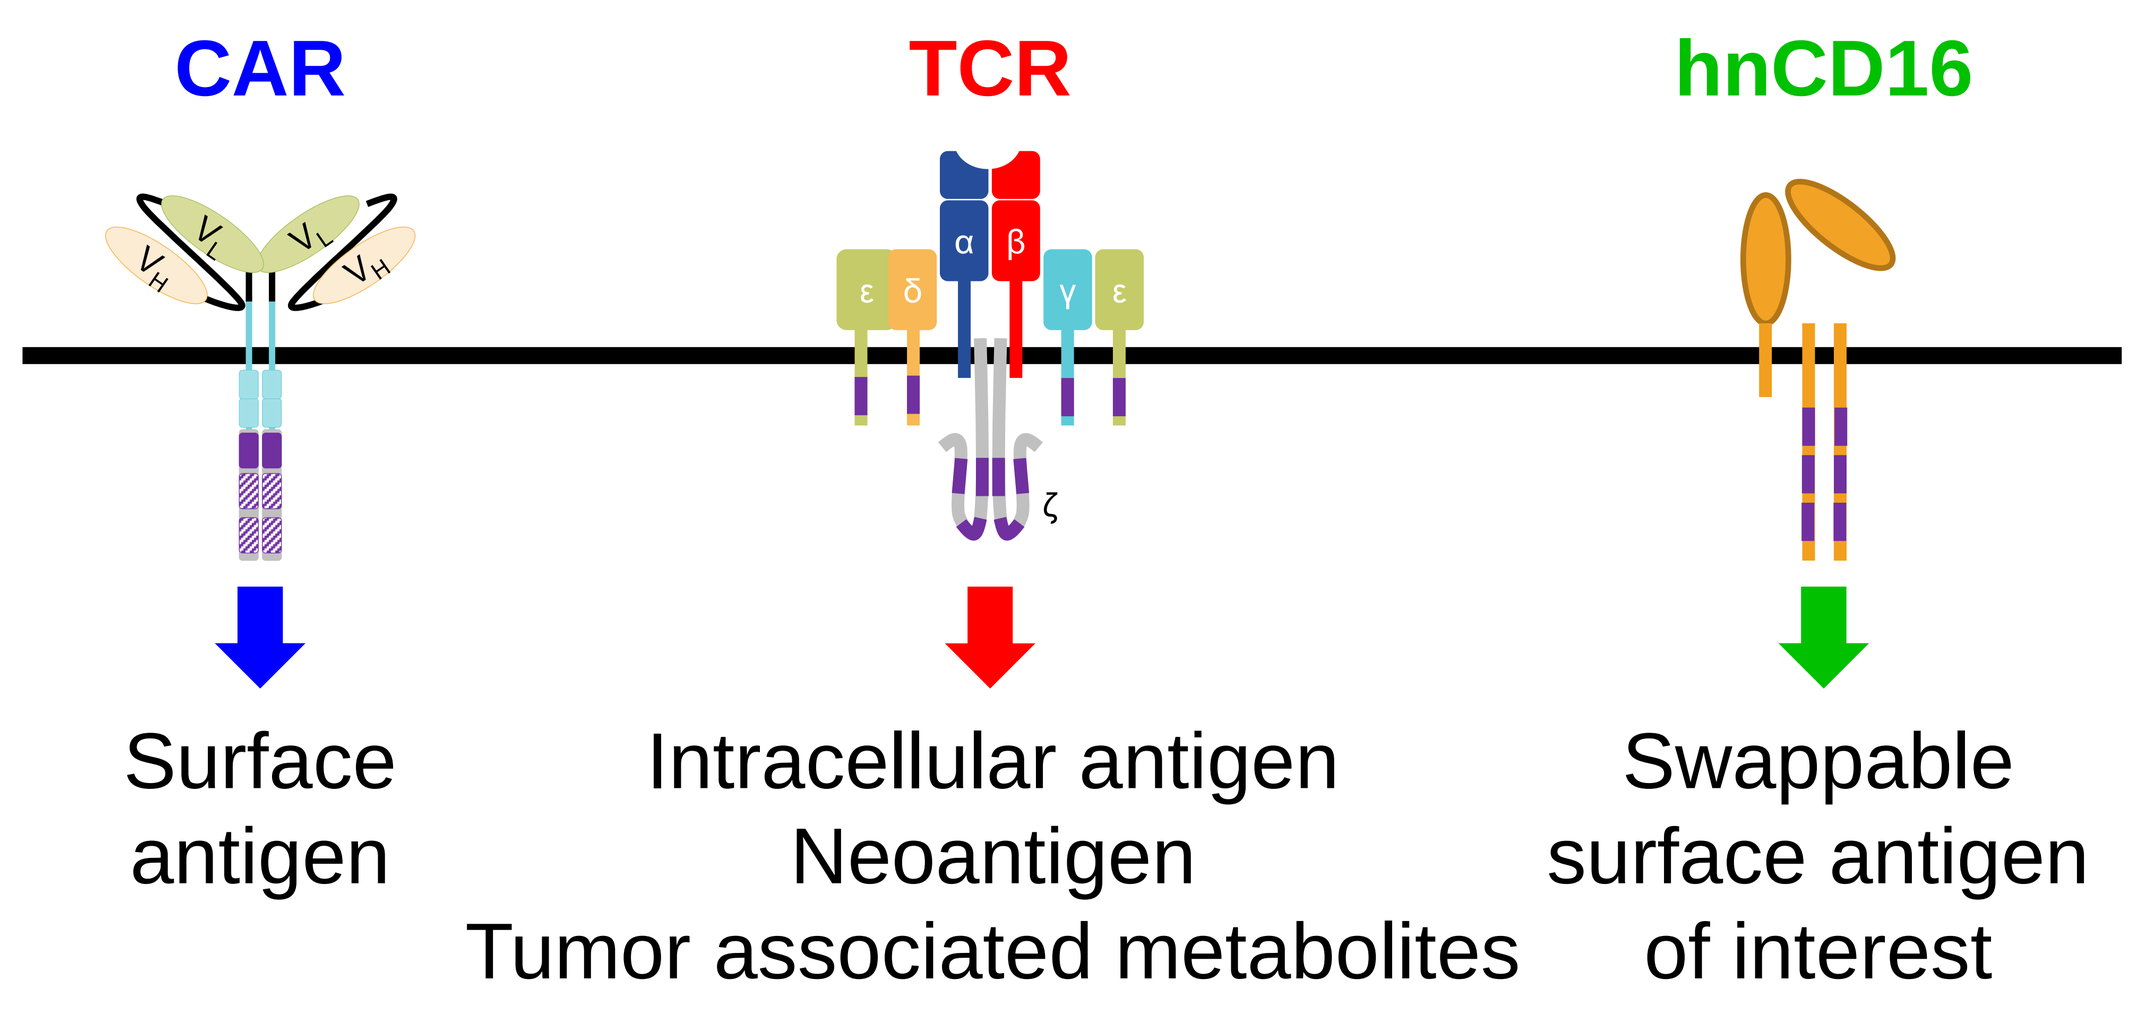
CAR
TCR
hnCD16
Surface antigen
Intracellular antigen
Neoantigen
Tumor associated metabolites
Swappable surface antigen of interest

SUMMARY

- iPSC-derived Trimodal iT cells homogenously express three anti-tumor modalities, CAR, TCR, hnCD16, that are individually functional with specific activity.
- Trimodal iT cells demonstrate compatibility between CAR, TCR, hnCD16 synthetic receptors with additive anti-tumor efficacy.
- Trimodal iT cells represent an ideal strategy to create off-the-shelf cellular immunotherapy for a promising therapeutic approach to combat heterogeneous and difficult to treat solid tumors, including those that are resistant due to antigen escape.

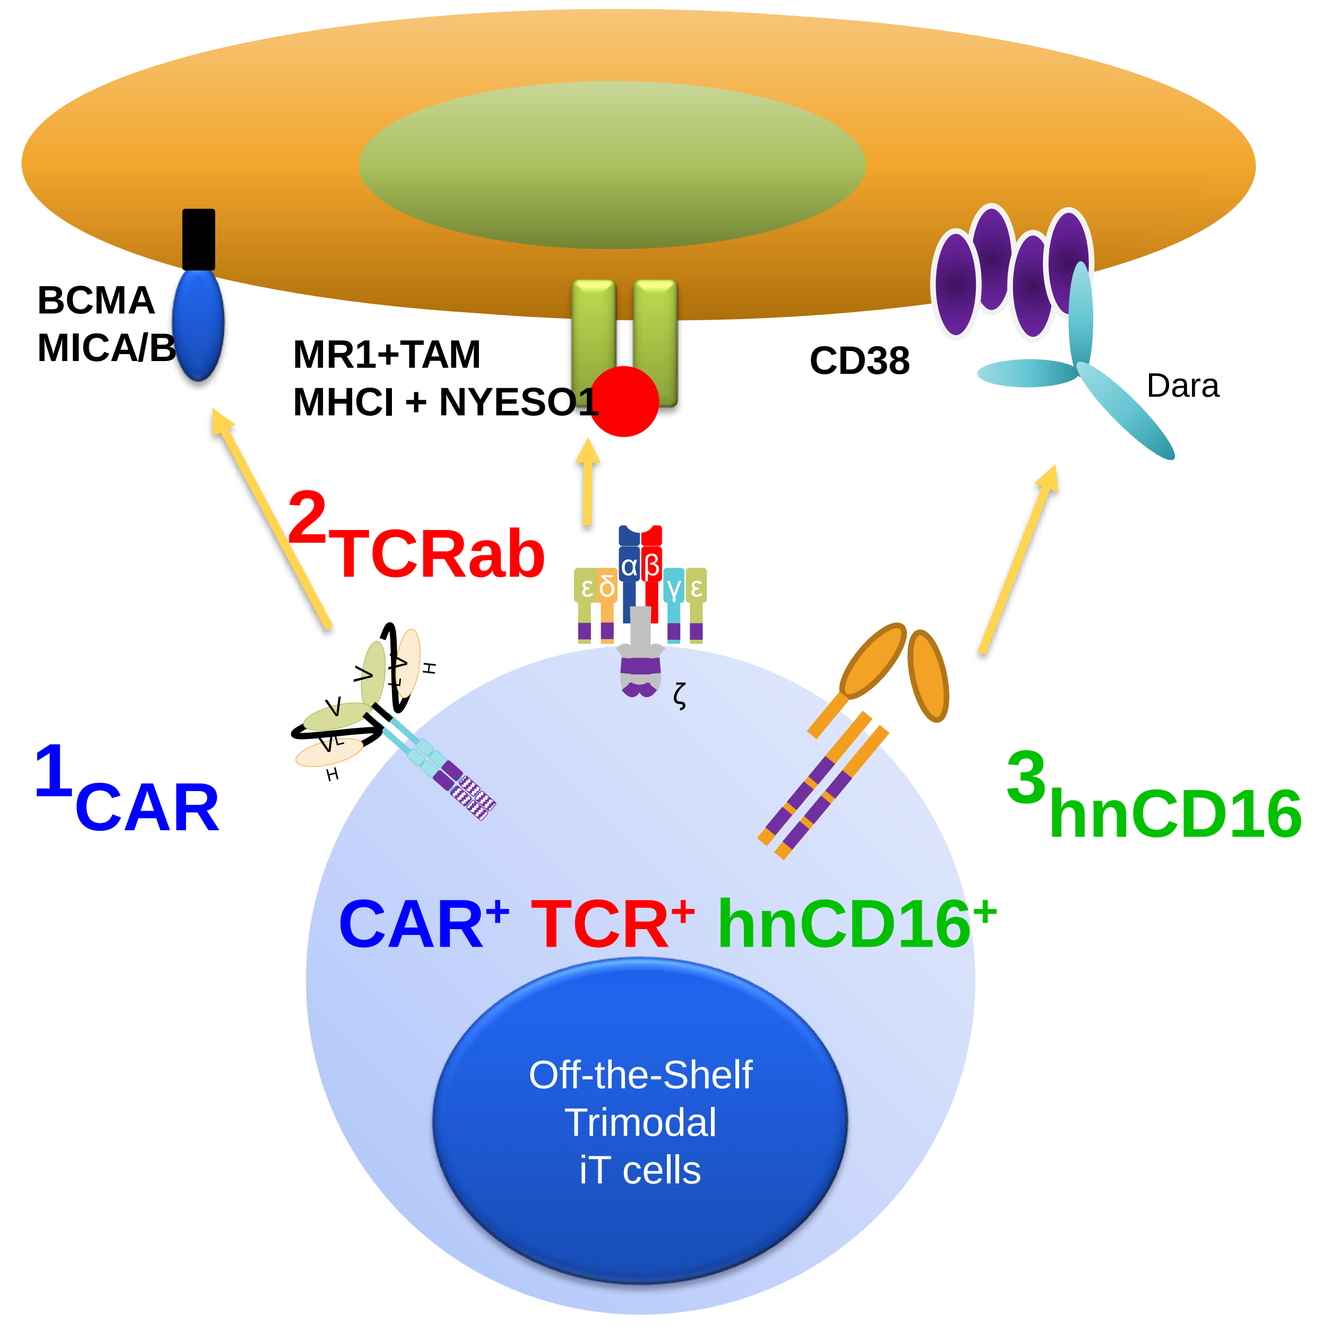
BCMA
MICA/B
MR1+TAM
MHCI + NYESO1
CD38
Dara
2TCRab
1CAR
3hnCD16
CAR⁺ TCR⁺ hnCD16⁺
Off-the-Shelf Trimodal iT cells

RESULTS

iPSC Platform for Mass Production of Universal NK and T-Cell Products

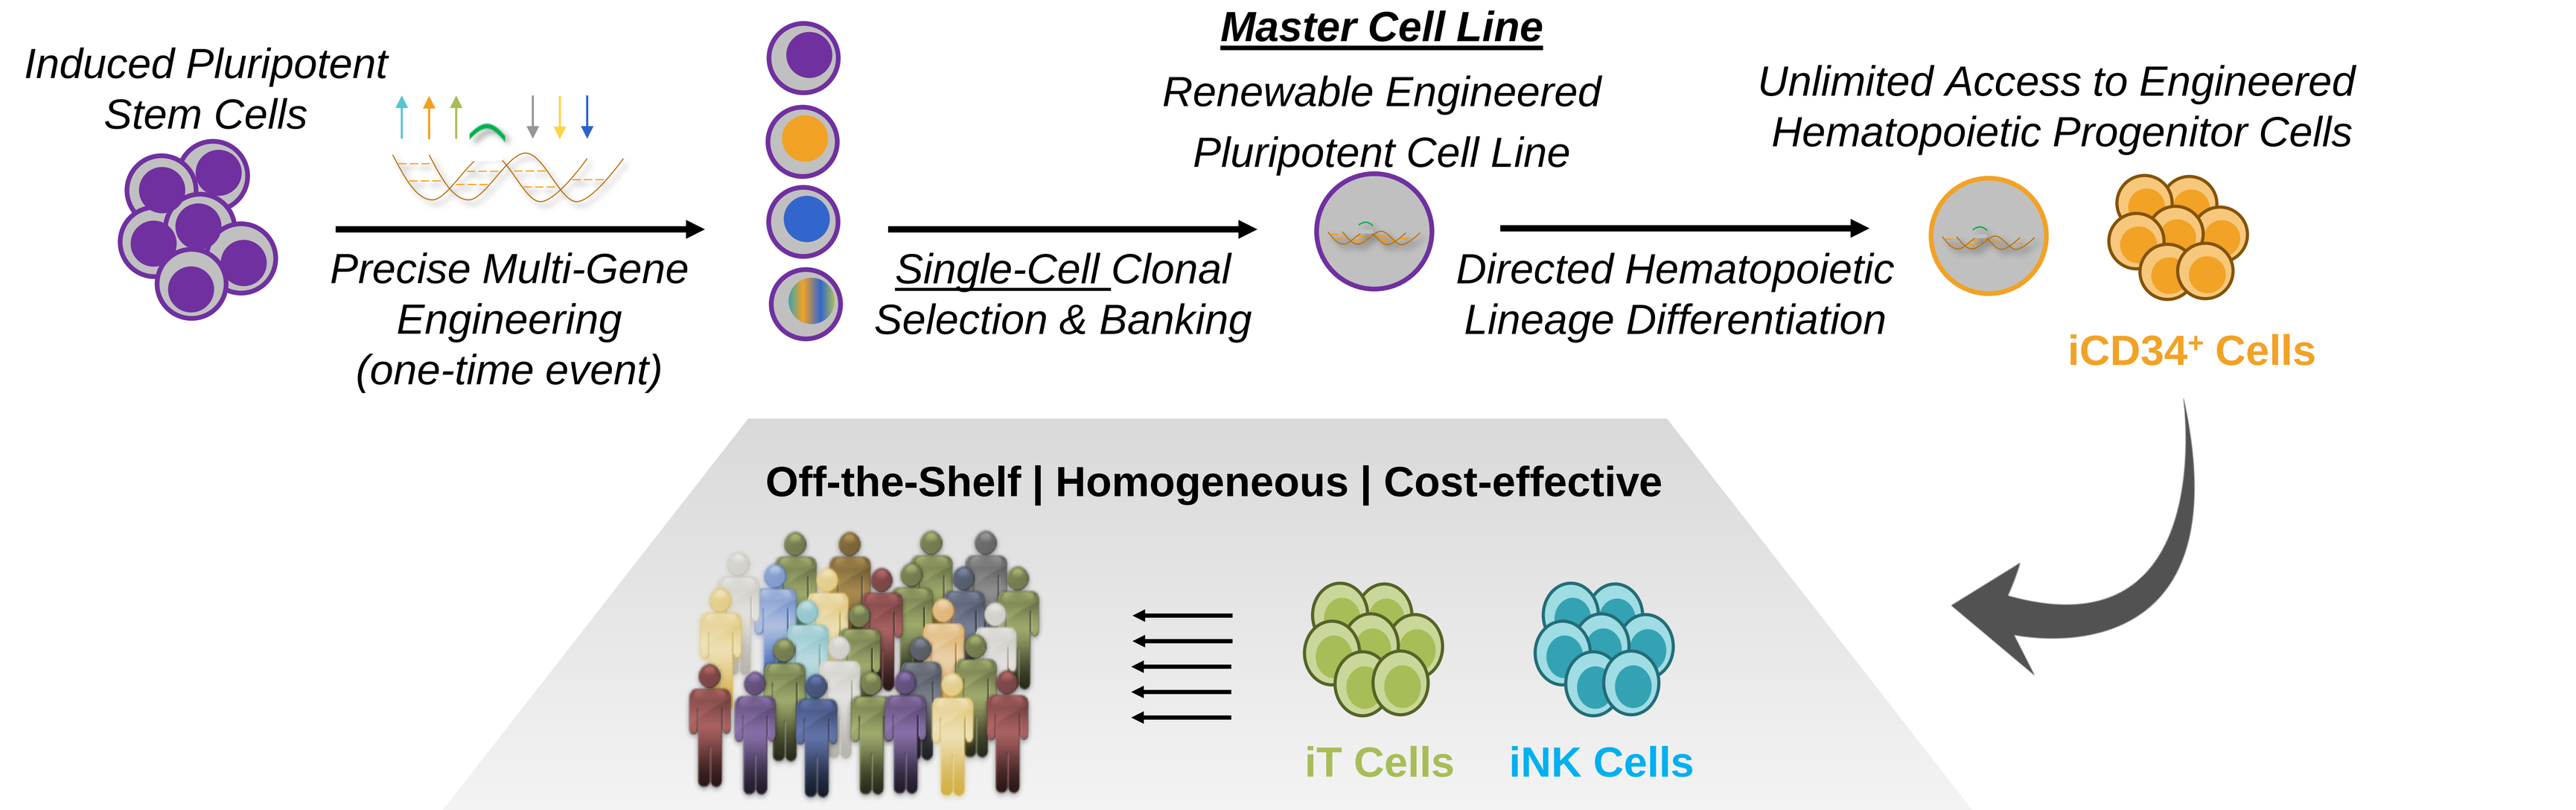
Induced Pluripotent Stem Cells
Precise Multi-Gene Engineering (one-time event)
Single-Cell Clonal Selection & Banking
Master Cell Line
Renewable Engineered Pluripotent Cell Line
Directed Hematopoietic Lineage Differentiation
Unlimited Access to Engineered Hematopoietic Progenitor Cells
iCD34⁺ Cells
Off-the-Shelf | Homogeneous | Cost-effective
iT Cells
iNK Cells

Figure 1. Fate Therapeutics induced pluripotent stem cell (iPSC) product platform enables mass production of off-the-shelf, engineered, homogeneous cell products that can be administered with multiple doses to deliver more effective pharmacologic activity, including in combination with cycles of other cancer treatments. Several programs are currently under clinical investigation.

Trimodal iT Cells Homogeneously Co-Express Three Individually Functional Anti-Tumor Modalities: CAR, TCR, and hnCD16

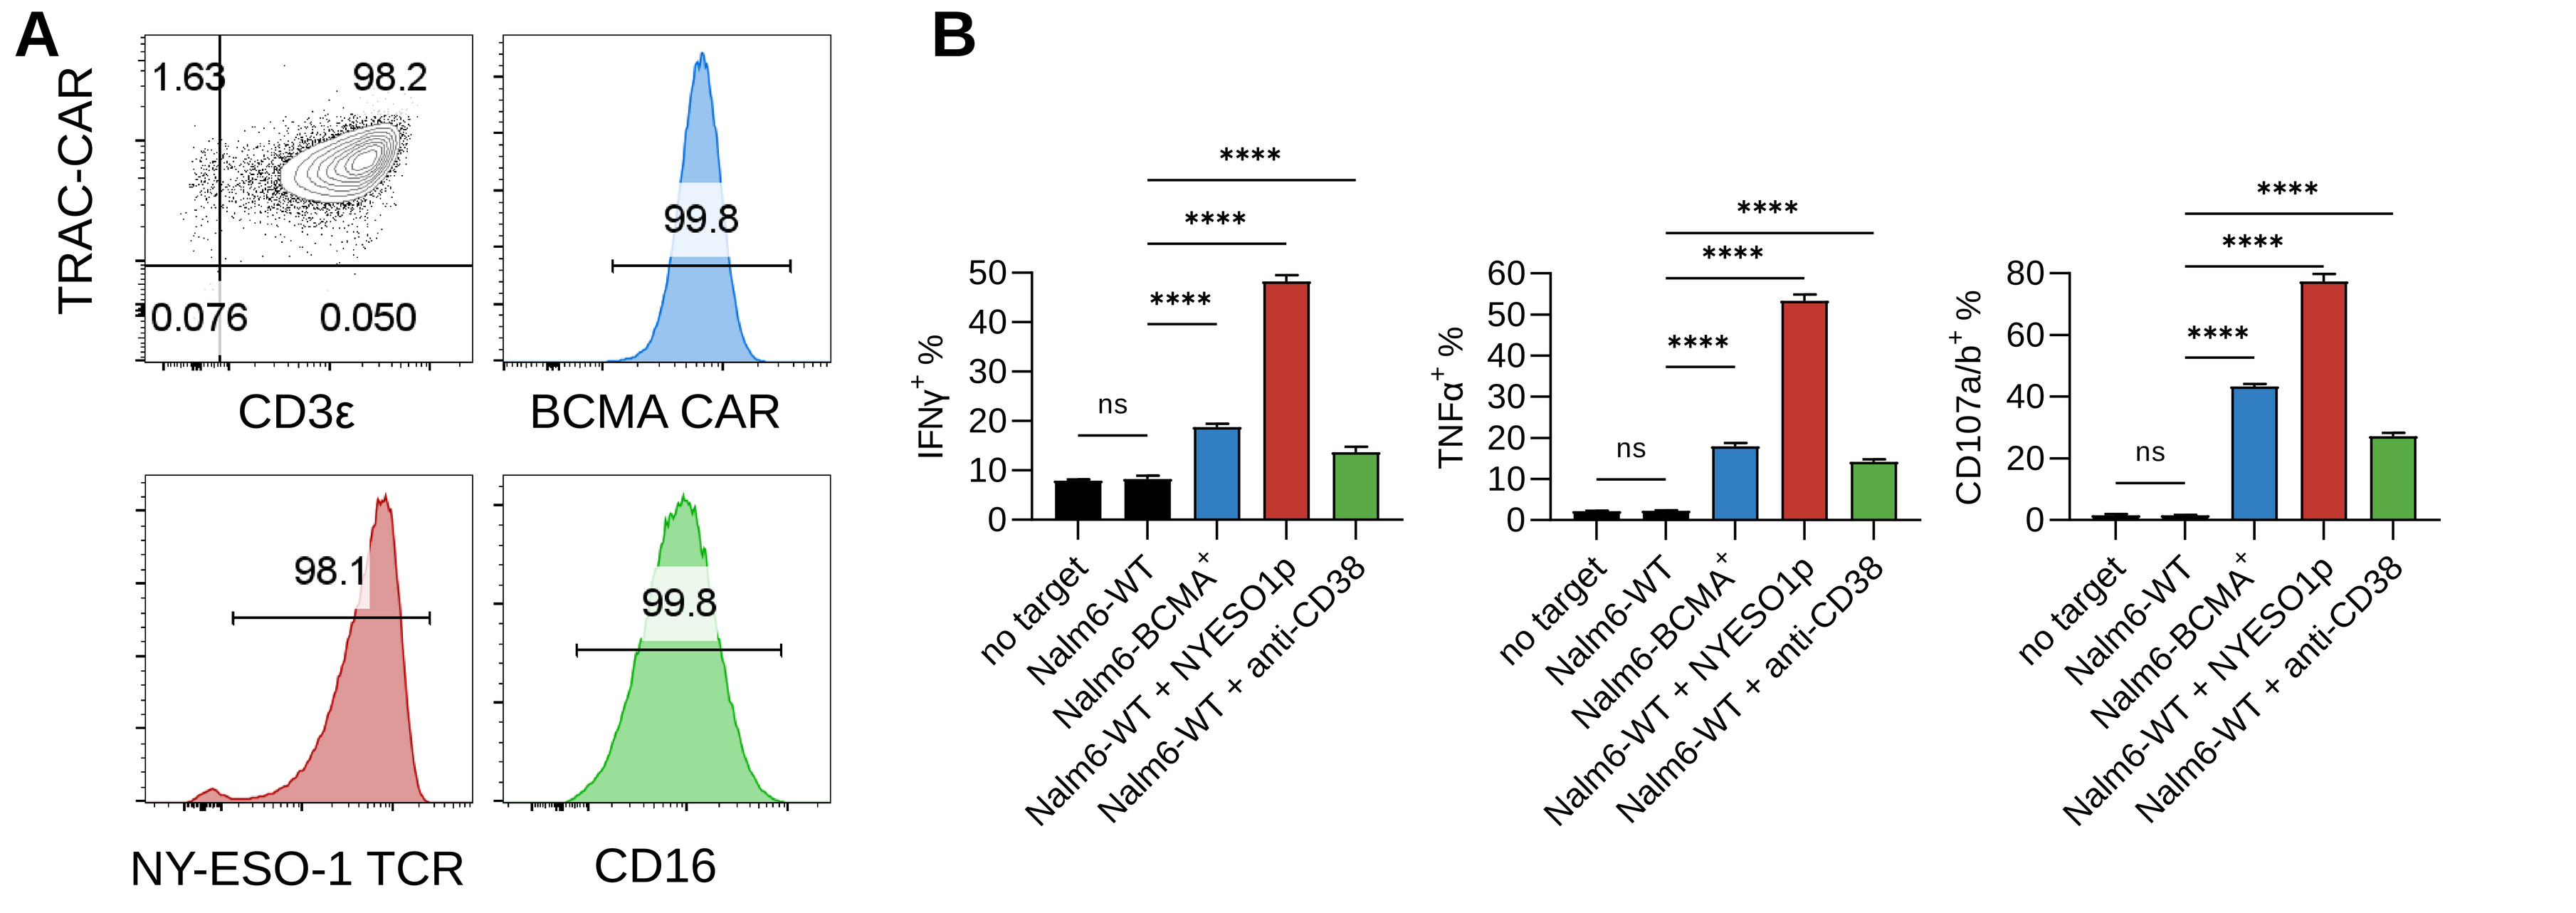
A
TRAC-CAR
1.63
98.2
0.076
0.050
CD3ε
99.8
BCMA CAR
98.1
NY-ESO-1 TCR
99.8
CD16
B
IFNγ⁺ %
TNFα⁺ %
CD107a/b⁺ %
ns

no target
Nalm6-WT
Nalm6-BCMA⁺
Nalm6-WT + NYESO1p
Nalm6-WT + anti-CD38

Figure 2. (A) FACS plots show the co-expression of TRAC-driven CAR and CD3ε to mark T cell identity and the expression of BCMA CAR, NYESO1 TCR, and hnCD16 on trimodal iT cells. (B) Graphs summarize the percentage of IFNγ, TNFα and CD107ab of CAR, TCR, or hnCD16 stimulated iT cells when stimulated with target cells engaging CAR only, TCR only, or hnCD16 only.

Trimodal iT Cells Demonstrate the Compatibility between CAR, TCR and hnCD16 to Enhance Anti-Tumor Efficacy

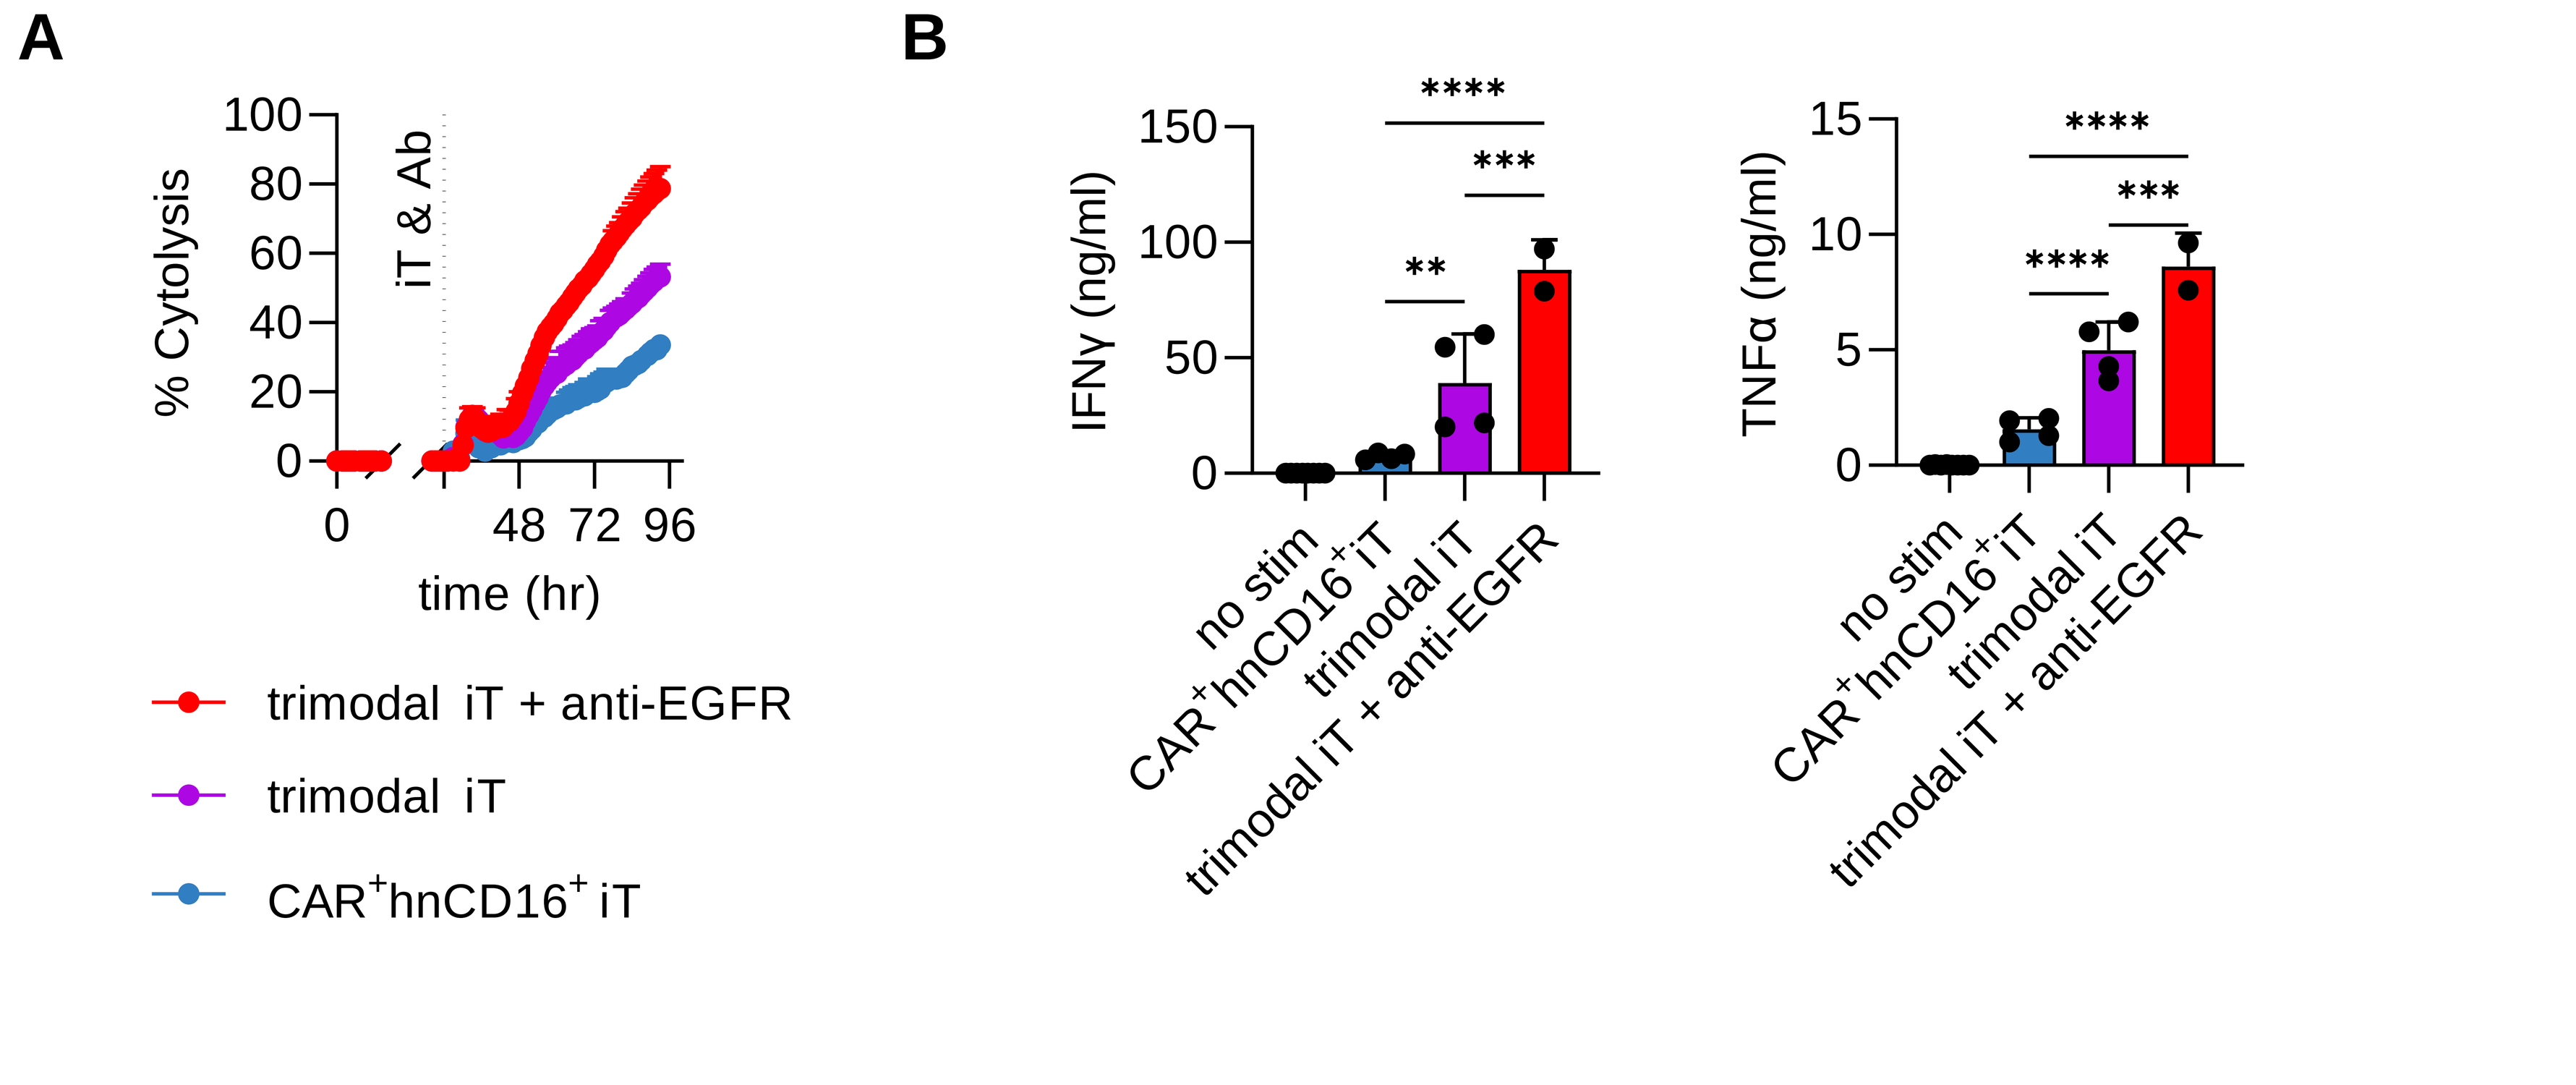
A
% Cytolysis
iT & Ab
time (hr)
trimodal iT + anti-EGFR
trimodal iT
CAR⁺hnCD16⁺ iT
B
IFNγ (ng/ml)
TNFα (ng/ml)
no stim
CAR⁺hnCD16⁺iT
trimodal iT
trimodal iT + anti-EGFR

Figure 3. (A) Percentage of cytolysis of 3 independent replicates using A549 treated with indicated conditions. (B) IFNγ and TNFα production were evaluated 6 hours post tumor stimulation at 1:1 effector to tumor ratio and quantified by MSD assay.

Trimodal iT Cells (BCMA CAR⁺, NYESO TCR⁺, hnCD16⁺) Demonstrate Control of a Heterogenous Tumor Cell Population In Vivo Through Engagement of Three Antigen Targeting Strategies

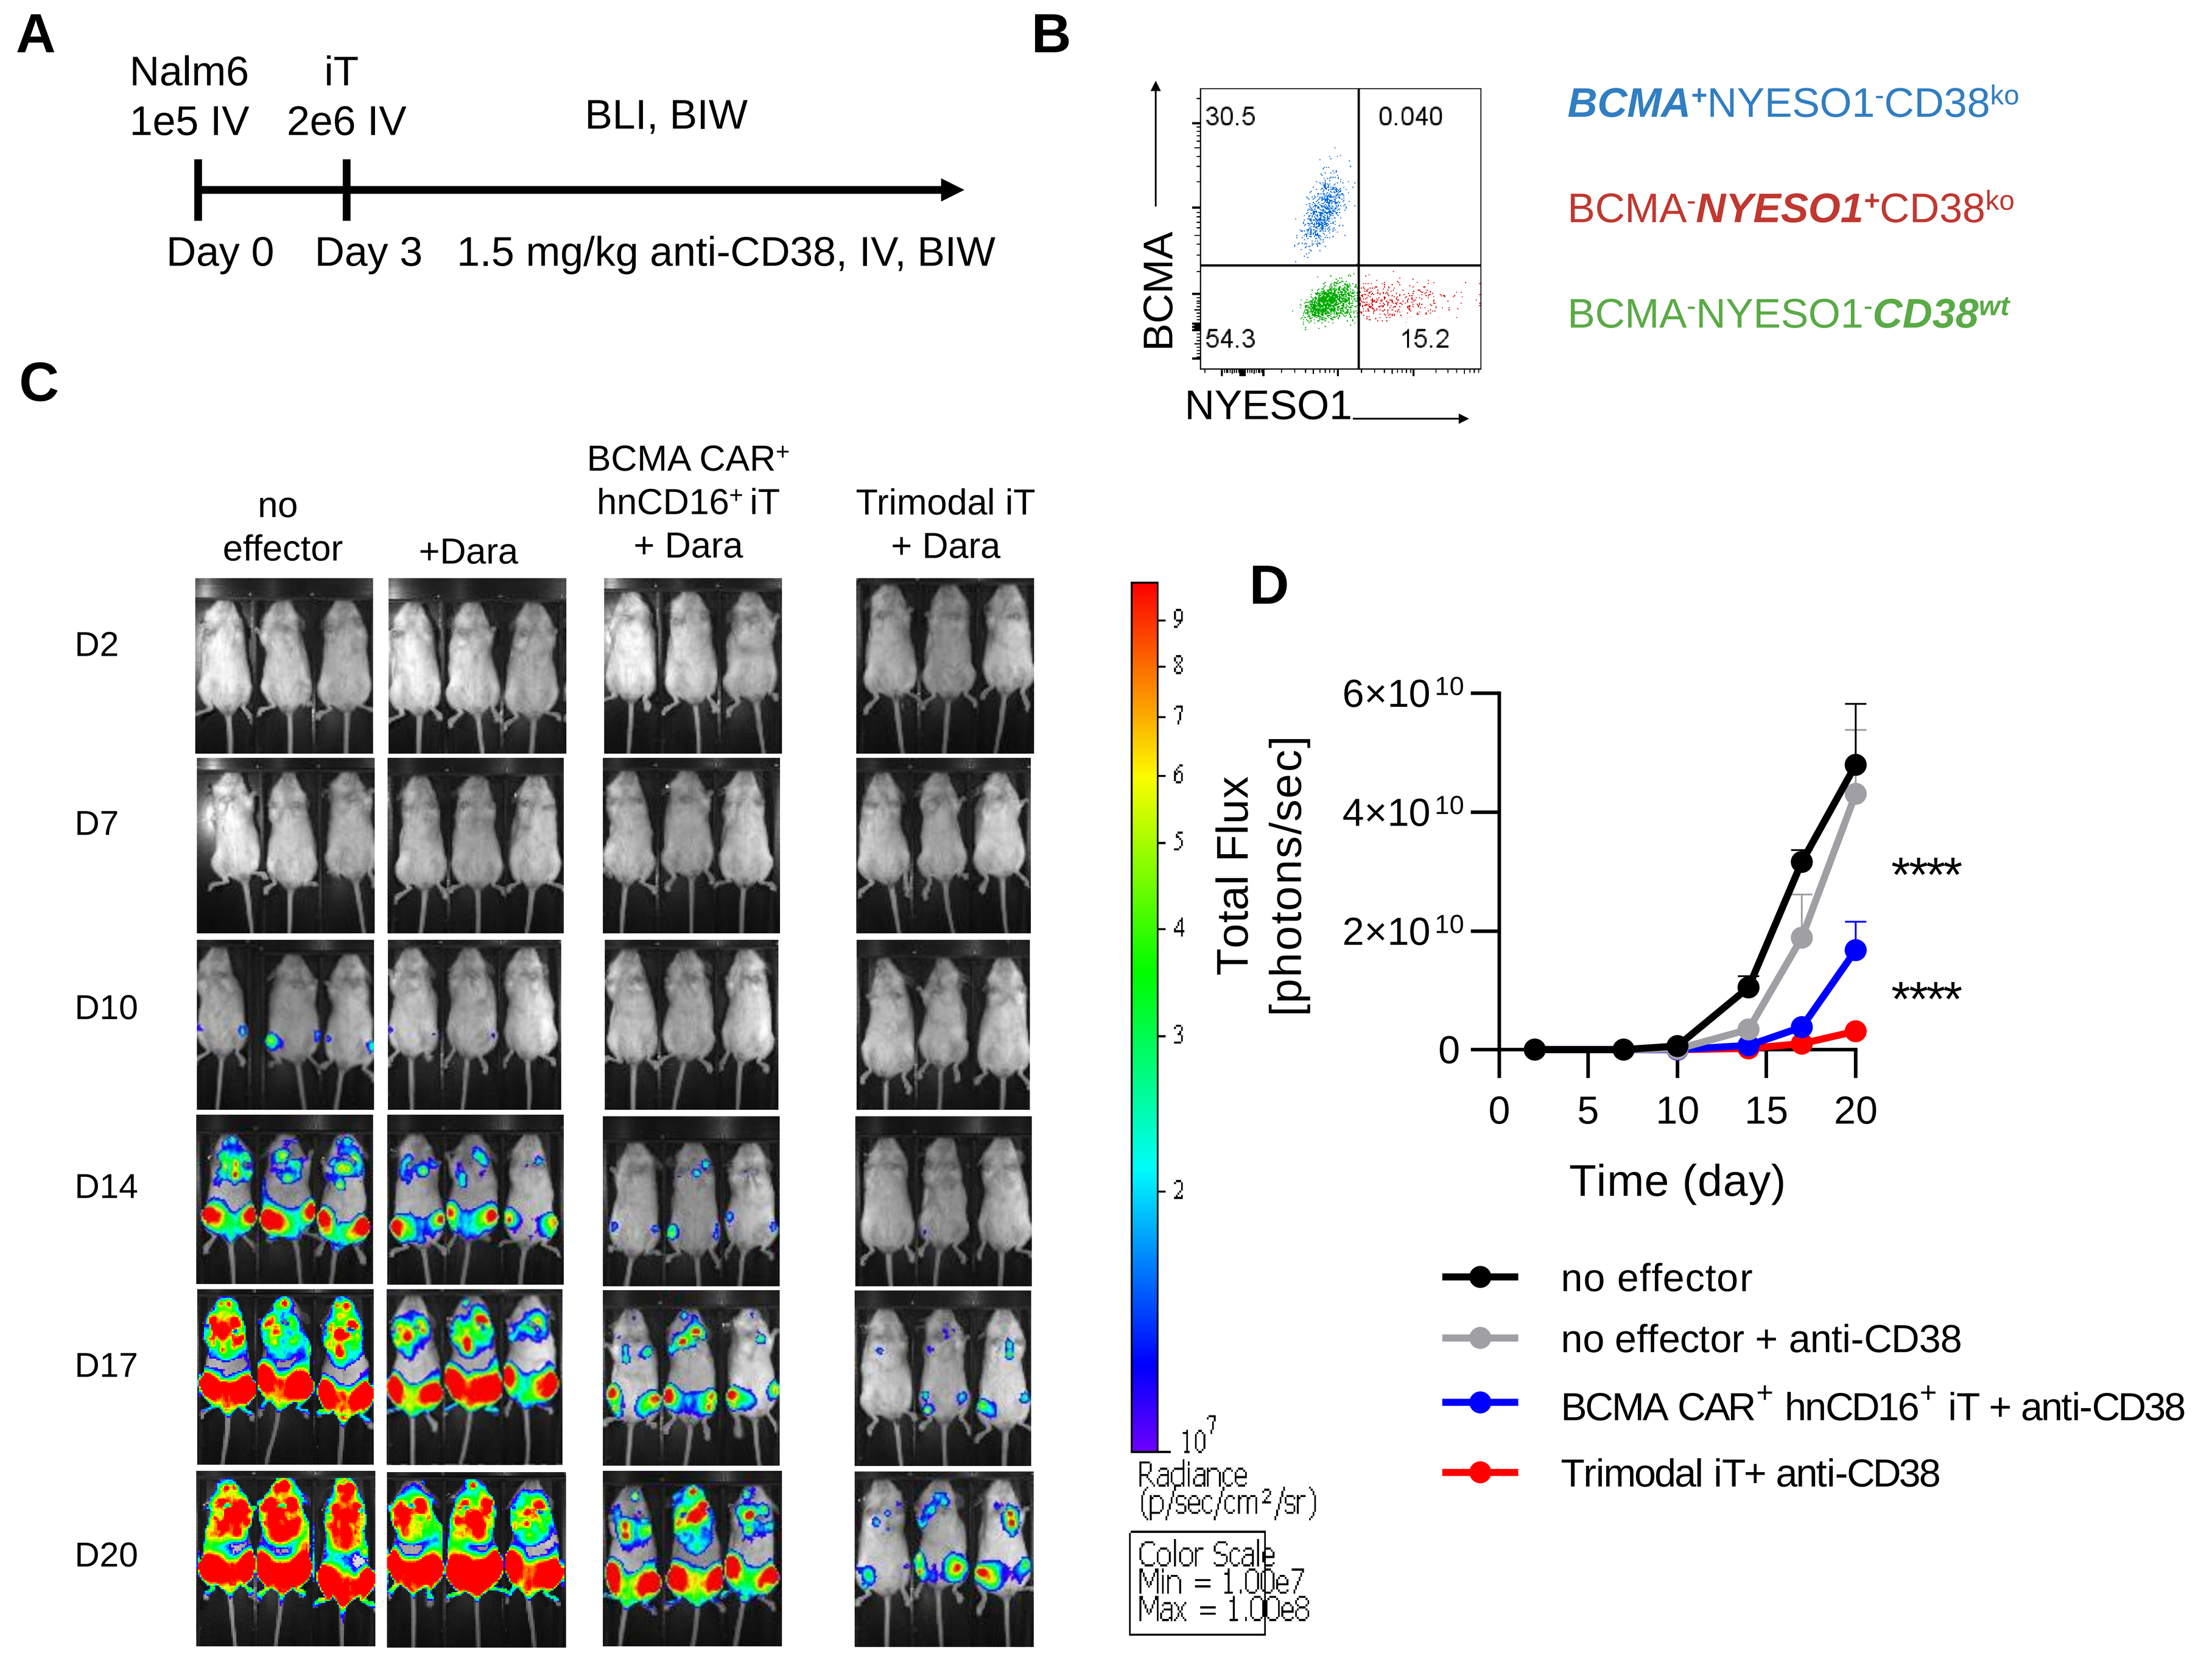
A
Nalm6 1e5 IV
iT 2e6 IV
BLI, BIW
Day 0
Day 3
1.5 mg/kg anti-CD38, IV, BIW
B
BCMA
NYESO1
30.5
0.040
54.3
15.2
BCMA⁺NYESO1⁻CD38ᵏᵒ
BCMA⁻NYESO1⁺CD38ᵏᵒ
BCMA⁻NYESO1⁻CD38ʷᵗ
C
no effector
+Dara
BCMA CAR⁺ hnCD16⁺ iT + Dara
Trimodal iT + Dara
D2
D7
D10
D14
D17
D20
Radiance (p/sec/cm²/sr)
Color Scale Min = 1.00e7 Max = 1.00e8
D
Total Flux [photons/sec]
Time (day)
no effector
no effector + anti-CD38
BCMA CAR⁺ hnCD16⁺ iT + anti-CD38
Trimodal iT+ anti-CD38

Figure 4. (A) Scheme illustrates Nalm6 model used in the study. (B) The FACS analysis of day 0 tumor input gated on CD10⁺ cells for BCMA-mCherry and NYESO1-GFP signals. (C) IVIS images demonstrate the tumor burden and location from D2 to D20. (D) Graph summary of BLI of indicated groups.

Ex vivo Bone Marrow Analysis from Xenograft Tumor Model Demonstrates Specific Targeting of Multiple Heterogenous Tumor Antigens by Trimodal iT Cells

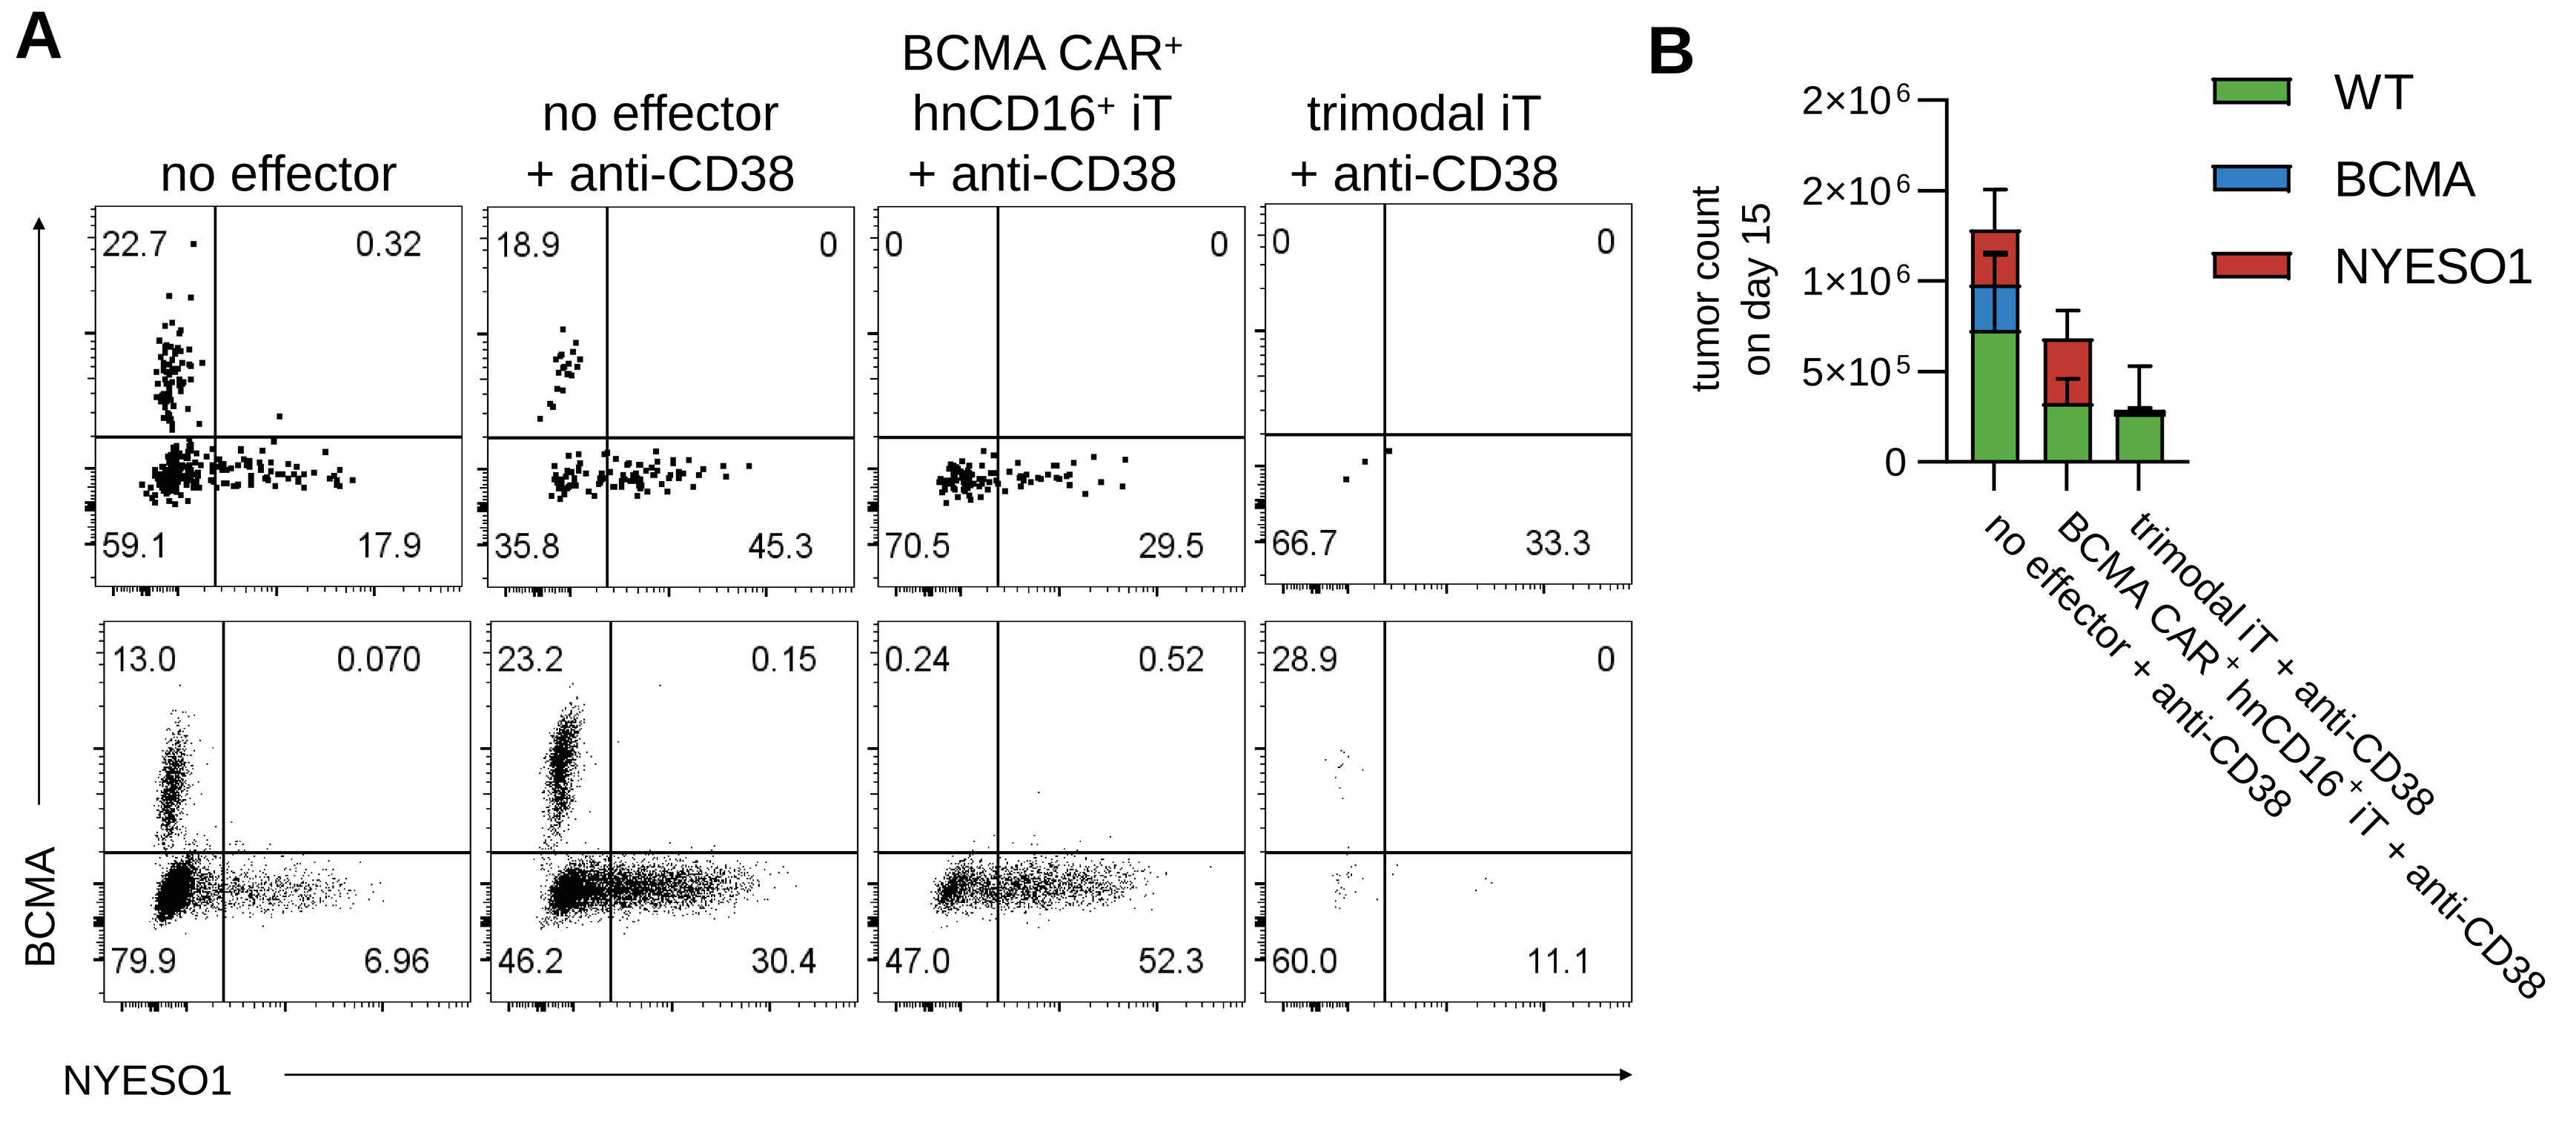
A
no effector
no effector + anti-CD38
BCMA CAR⁺ hnCD16⁺ iT + anti-CD38
trimodal iT + anti-CD38
Day 8
Day 15
BCMA
NYESO1
B
tumor count on day 15
WT
BCMA
NYESO1

Figure 5. (A) The FACS analysis of *ex vivo* CD10⁺ tumor cells from bone marrow (BM) for BCMA-mCherry and NYESO1-GFP signals. (B) The absolute tumor count in each bone marrow sample for indicated tumor lines are plotted for all groups on experiment day 15.