Engineering of Synthetic Chemokine Receptors into iPSC-derived CAR-T cells to Increase Homing and Enhance Trafficking into Solid Tumors

Soheila Shirinbak, Joy Grant, Angela Gentile, Lexe Linderhof, Bishwas Shrestha, Amit Mehta, Philip Chu, Bjoern Gaertner, Yijia Pan, Mochtar Pribadi, Layton Smith, Matthew Denholtz, Alec Witty, Eigen Peralta, Tom Lee, Martin Hosking, Bahram Valamehr

Fate Therapeutics, Inc., San Diego, CA, USA

Introduction

Despite the success of chimeric antigen receptor (CAR)-T cells in treating hematological malignancies, the efficacious treatment of solid tumors has been hampered by lack of CAR-T cell persistence, tumor-associated antigen heterogeneity, and the immuno-suppressive tumor microenvironment (TME). In addition, trafficking of CAR-T cells to solid tumors, potentially due to a chemokine-chemokine receptor mismatch between the tumor and the CAR-T cells, has proven ineffective. Early and sustained detection of T cells within a solid tumor has been associated with better outcomes across several clinical trials, suggesting that strategies focused on enabling CAR-T cell homing and trafficking can generate significant therapeutic benefit.

Summary of results:

I. The CXCR2 ligand CXCL8 is expressed by various tumor cell lines and is specifically upregulated following radio- and chemo-therapy preconditioning (**Fig 1**).

II. Specific engineering of CXCR2 into iPSC derived CAR-T cells enables their ability to specifically migrate to the CXCR2 ligand CXCL8 without impacting their differentiation, phenotype, or anti-tumor efficacy **(Fig 2 & 3)**.

III. CXCR2 expression by CAR iTs enhances their homing to and retention within multiple tumor models expressing CXCL8, either naturally or chemotherapy-induced, leading to enhanced tumor control *in vivo* (**Fig 4 & 5**).

CXCL8 (IL-8) is upregulated following radio- and chemo-therapy treatment *in vitro*

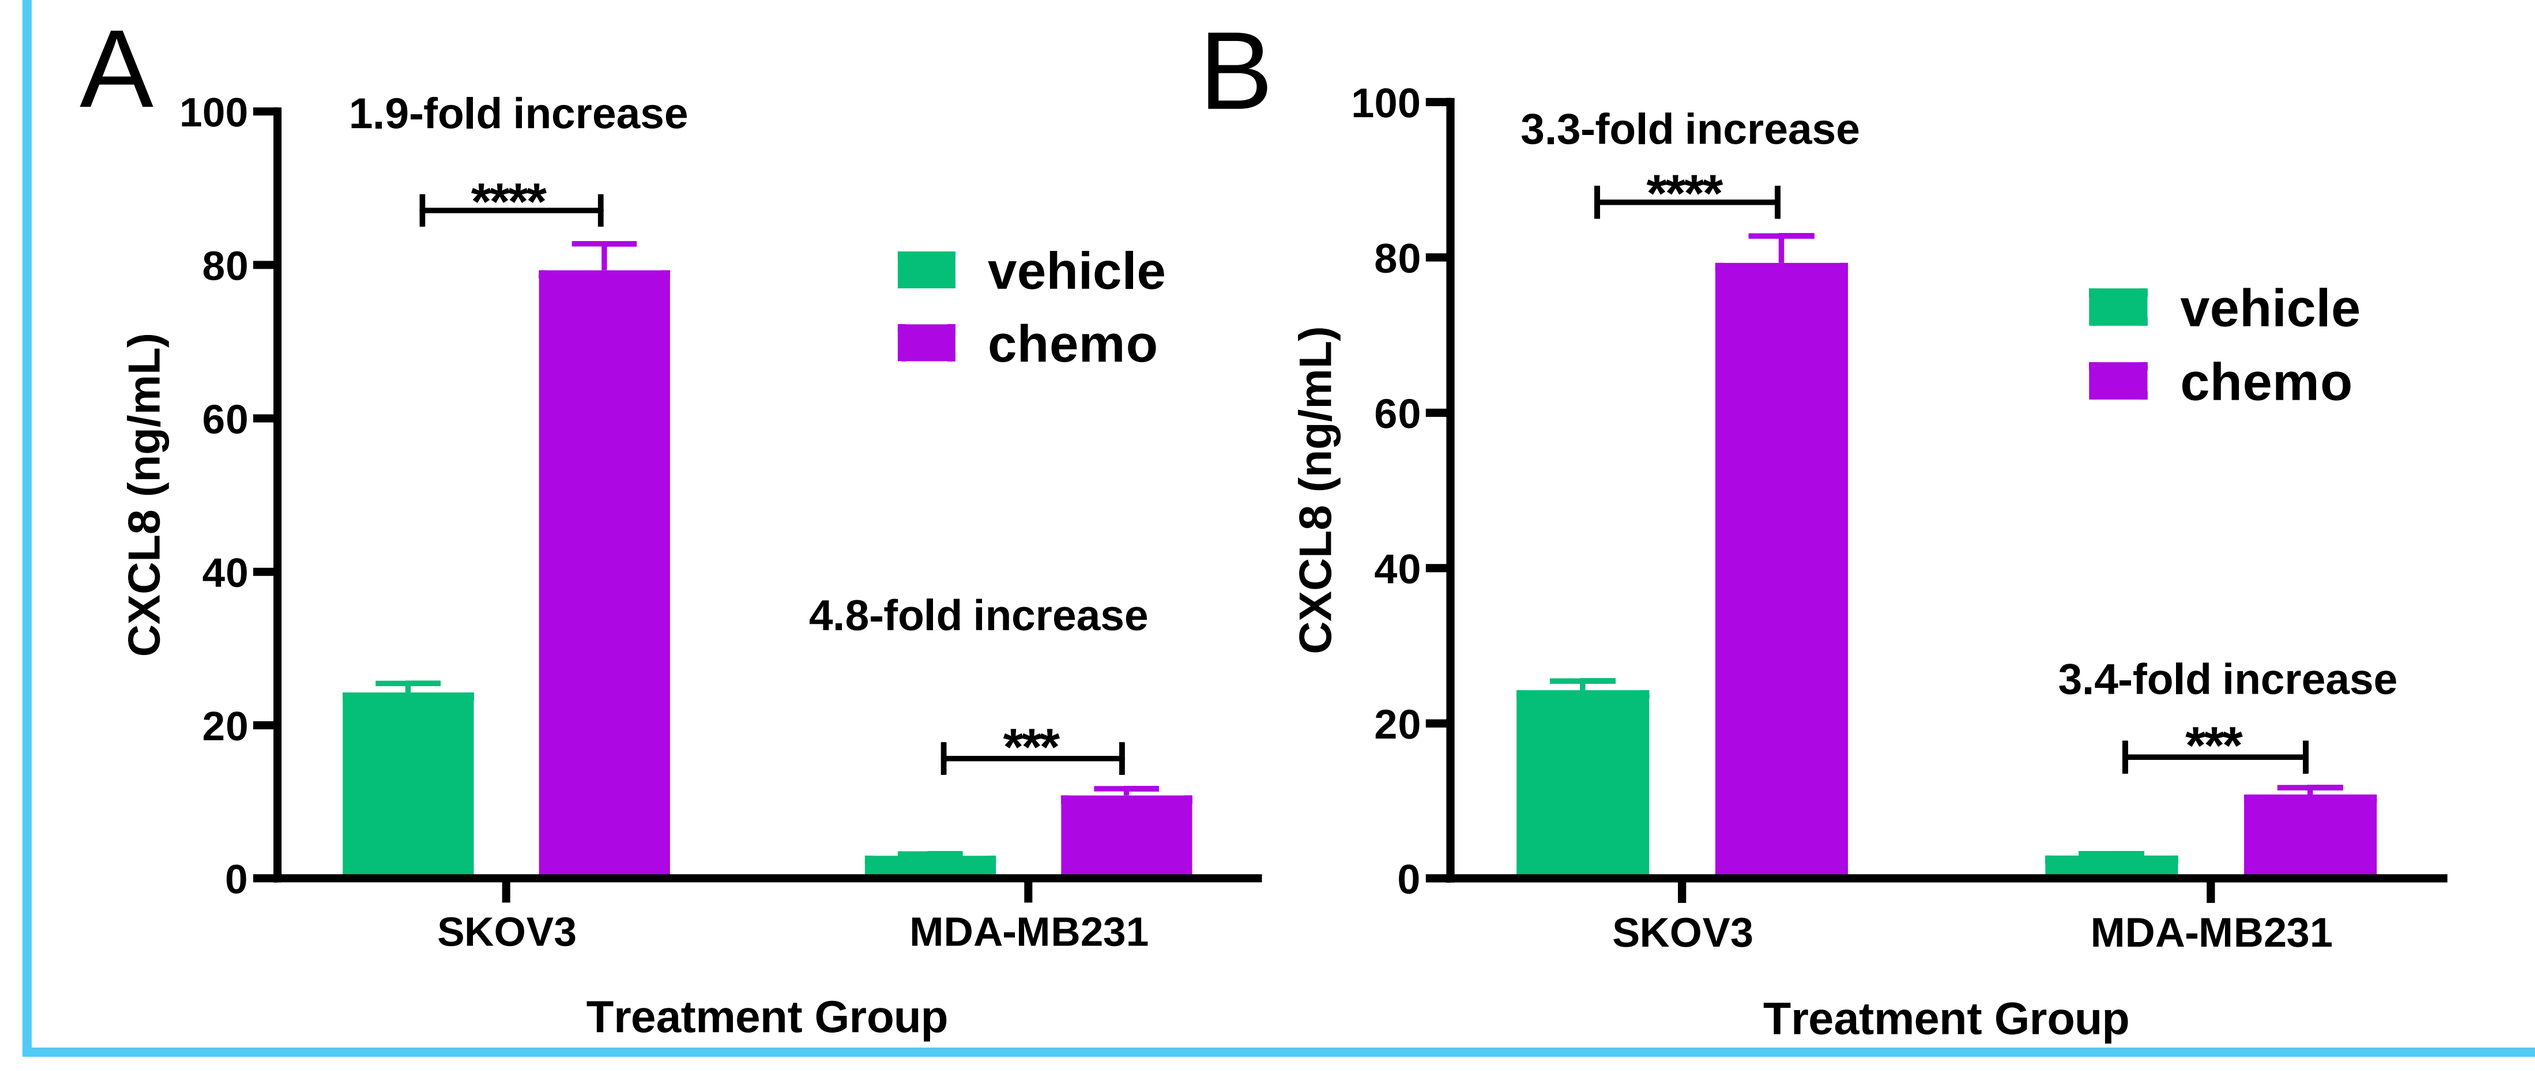
A
1.9-fold increase
4.8-fold increase
CXCL8 (ng/mL)
vehicle
chemo
SKOV3
MDA-MB231
Treatment Group
B
3.3-fold increase
3.4-fold increase

Figure 1. *In-vitro* analysis of CXCL8 levels following (A) radiation and (B) chemotherapy treatment. Ovarian (SKOV3) and Triple negative breast cancer lines (MDA-MB-231) were treated either with radiation (21Gy) or chemotherapy (Cisplatin, IC_{50}). Supernatants were collected 48hrs post treatment and IL-8 levels were determined by ELISA. (**** P<0.0001,***P<0.001**P<0.01).

Generation of CXCR2 engineered off-the shelf iPSC derived CAR-T cells

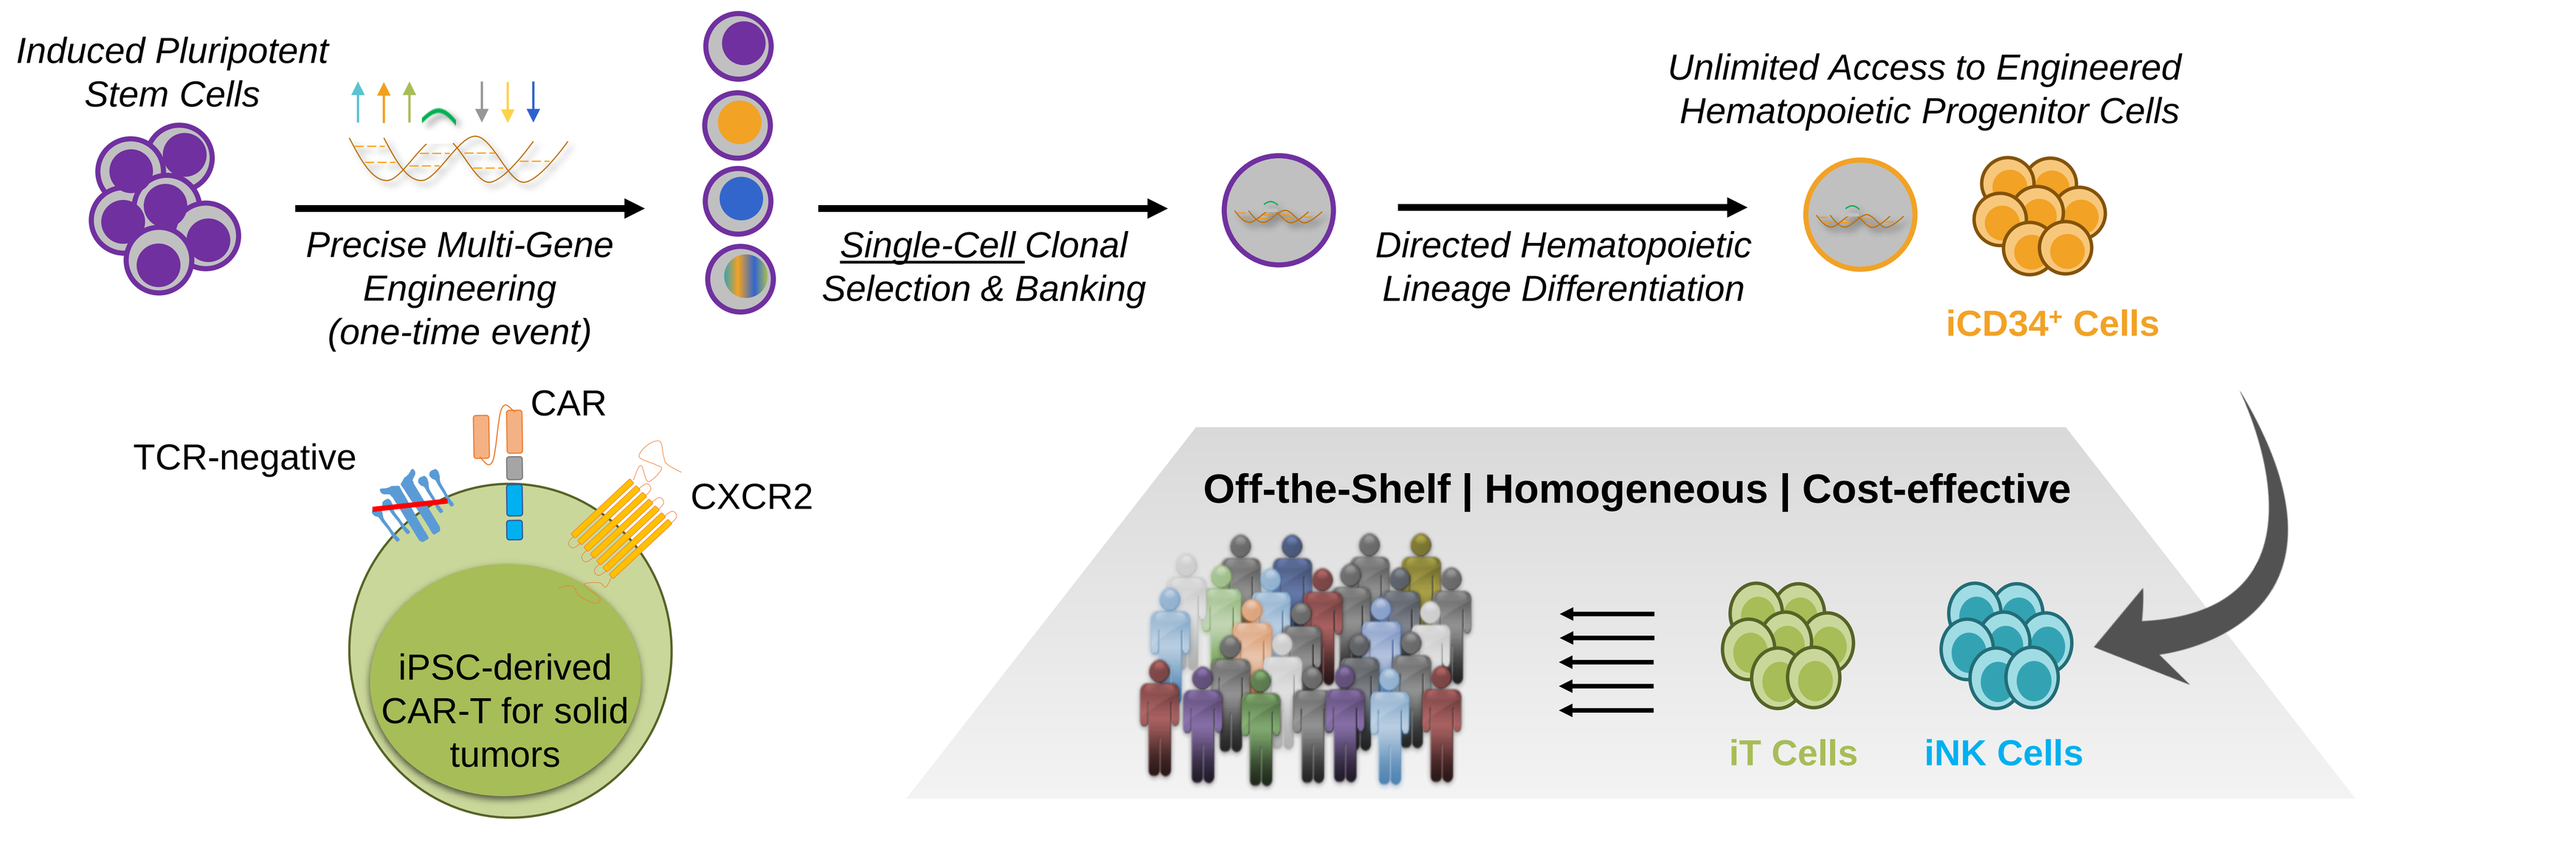
Induced Pluripotent Stem Cells
Precise Multi-Gene Engineering (one-time event)
Single-Cell Clonal Selection & Banking
Directed Hematopoietic Lineage Differentiation
Unlimited Access to Engineered Hematopoietic Progenitor Cells
iCD34+ Cells
TCR-negative
CAR
CXCR2
iPSC-derived CAR-T for solid tumors
Off-the-Shelf | Homogeneous | Cost-effective
iT Cells
iNK Cells

Figure 2. Overview of Fate's proprietary IPSC platform for the generation of precisely and uniformly engineered effector NK and T cells to express chemokine receptors such as CXCR2.

CXCR2 engineering enables specific migration to the CXCR2 ligand CXCL8 without altering CAR iT differentiation or anti-tumor efficacy

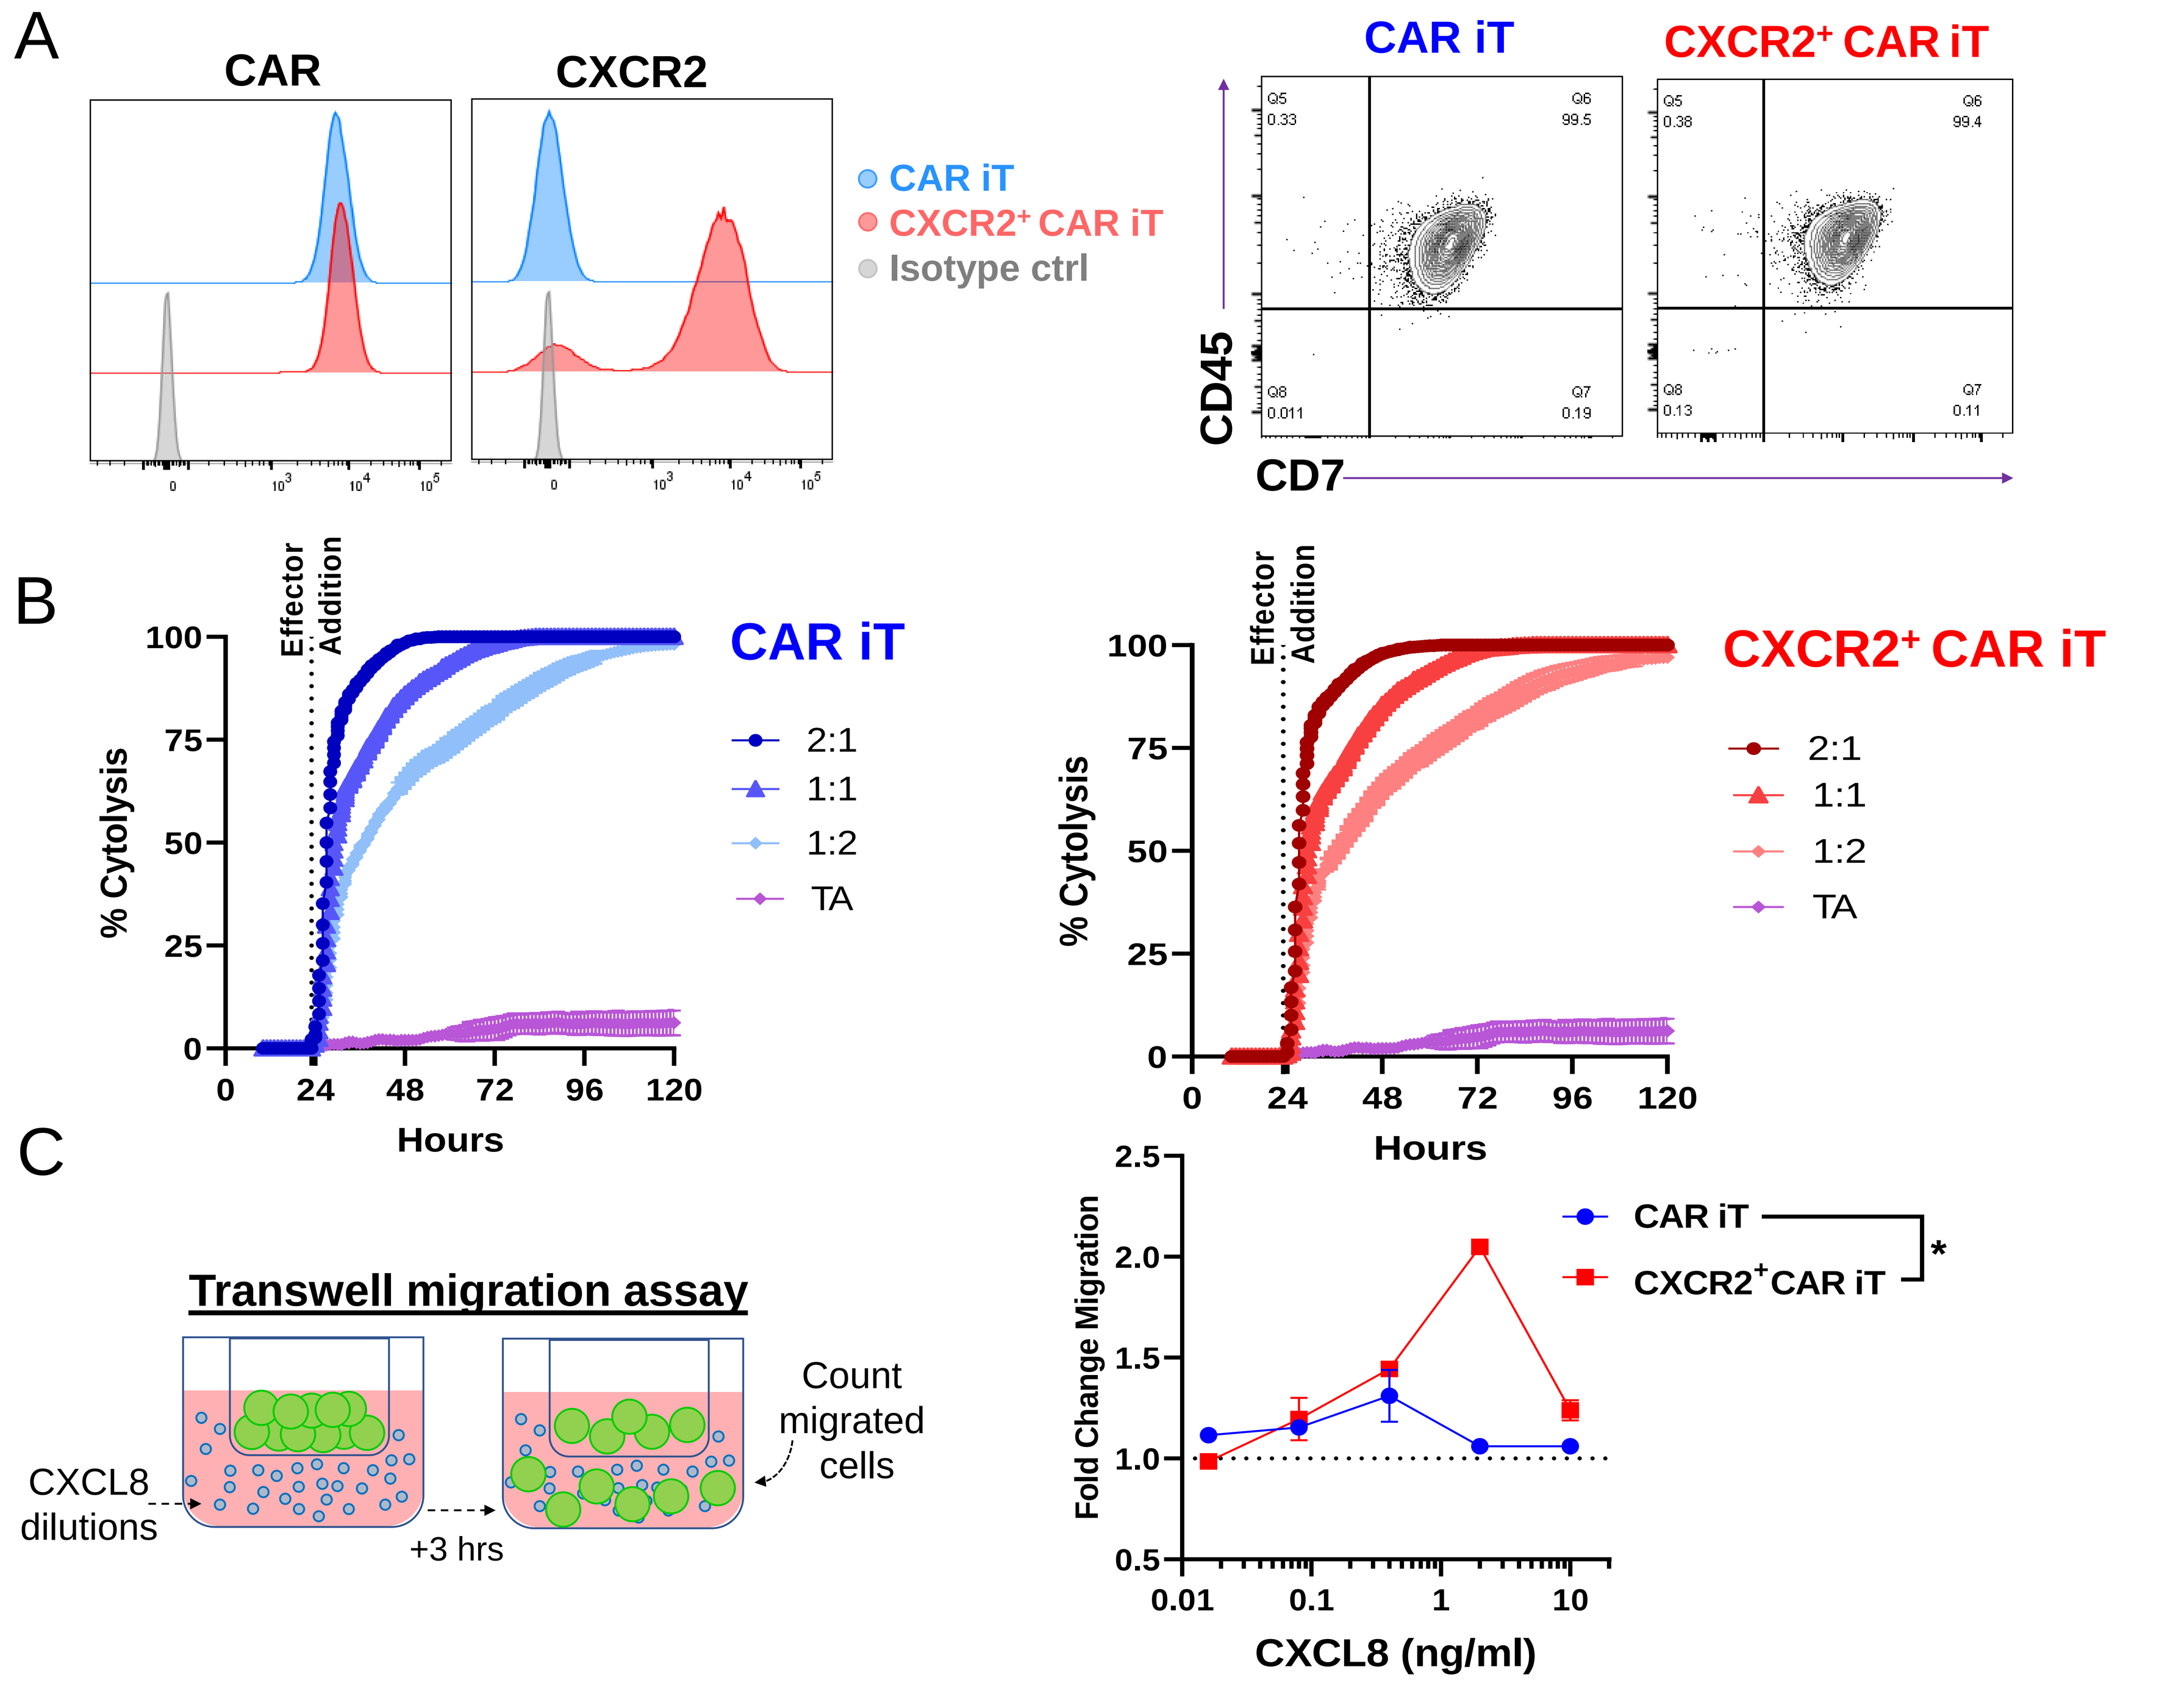
A
CAR
CXCR2
CAR iT
CXCR2+ CAR iT
Isotype ctrl
CD45
CD7
B
Effector Addition
% Cytolysis
Hours
2:1
1:1
1:2
TA
C
Transwell migration assay
CXCL8 dilutions
+3 hrs
Count migrated cells
Fold Change Migration
CXCL8 (ng/ml)

Figure 3. (A) Phenotypic evaluation of iPSC derived CAR iT cells. All CAR iT cells express uniform levels of CAR (99%) along with high levels of CXCR2 (90%) and are more than 99% positive for CD45 and CD7. (B) Both CAR iT cells with/without CXCR2 exerted robust cytolytic targeting of tumor targets at the indicated [Effector:Target] ratios, compared to tumor alone (TA) controls as evaluated in an xCELLigence assay. (C) Transwell migration assays demonstrated that CXCR2 expressing CAR iTs were chemotactic to the CXCR2 ligand CXCL8.* P<0.05

Enhanced intratumoral infiltration/retention and tumor control by CXCR2+ CAR iT cells in CXCL8 rich tumors

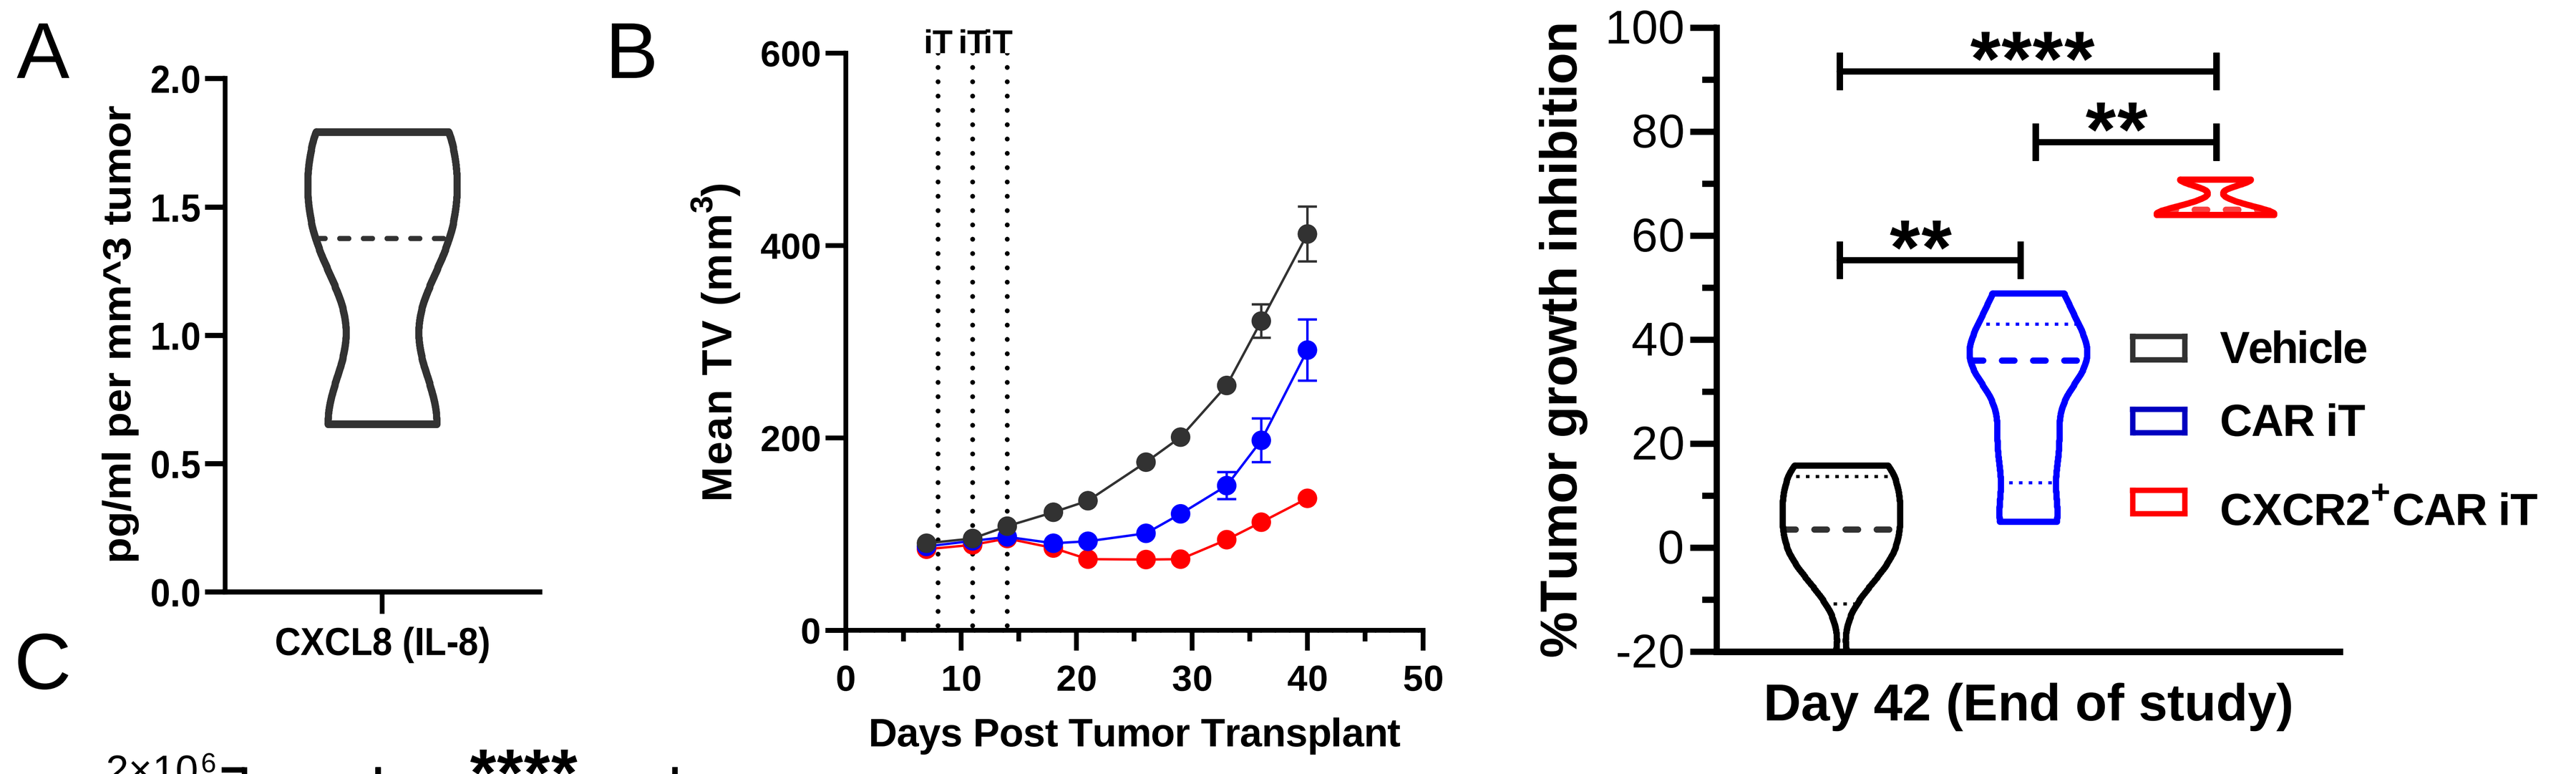
A
pg/ml per mm^3 tumor
CXCL8 (IL-8)
B
Mean TV (mm3)
Days Post Tumor Transplant
%Tumor growth inhibition
Vehicle
CAR iT
CXCR2+CAR iT
Day 42 (End of study)

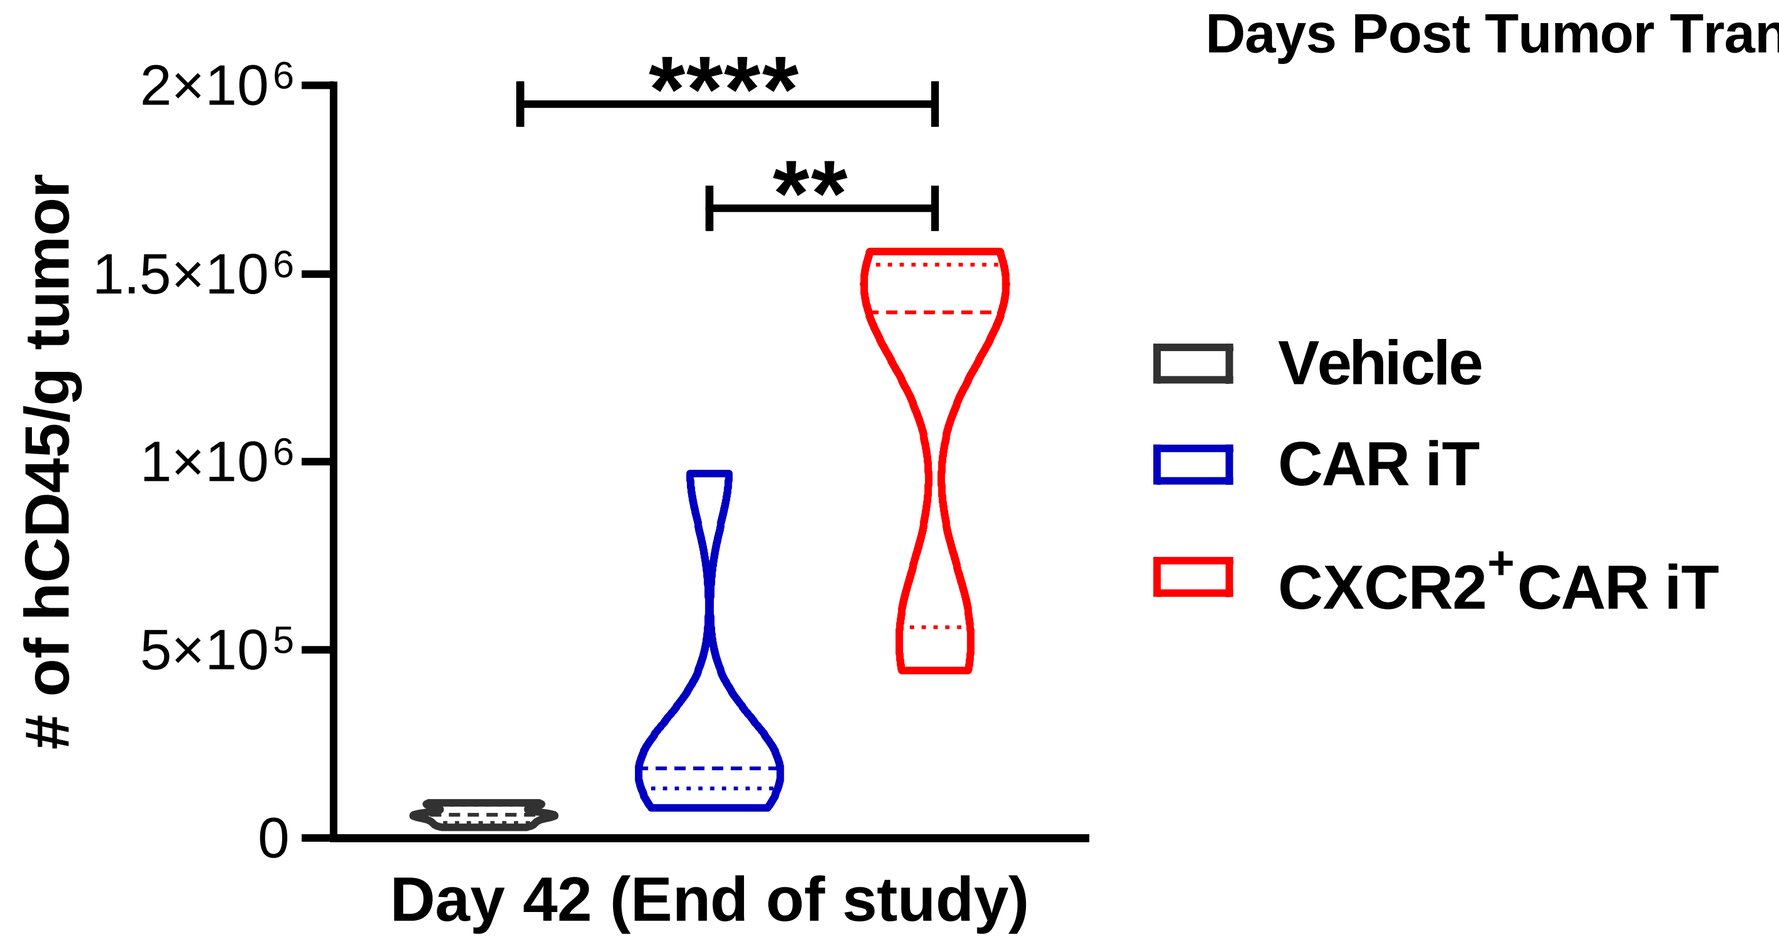
C
of hCD45/g tumor
Vehicle
CAR iT
CXCR2+CAR iT
Day 42 (End of study)

Figure 4. Higher tumor growth inhibition and enhanced CAR iT infiltration to tumors with CXCR2+CAR iTs. 2e6 ovarian tumor cells were injected s.c. to the NSG mice on day 0. CAR iTs were administered (i.v.) at 3 doses of 10e6 every 3 days on day 8. (A) CXCL8 levels in serum collected prior to effector dosing. (B) Tumors growth evaluation during the course of experiment. (C) Tumors were harvested on day 42 and number of CAR iTs were determined via flowcytometry analysis. N=5 mice per group, **** P<0.0001, ***P<0.001, **P<0.01, *P<0.05 *

CXCL8 upregulation following chemotherapy, specifically enhances intratumoral infiltration and retention of CXCR2+ CAR iT cells leading to improved tumor control in a CAR T cell resistant tumor xenograft model, consisting of aggressive tumor growth

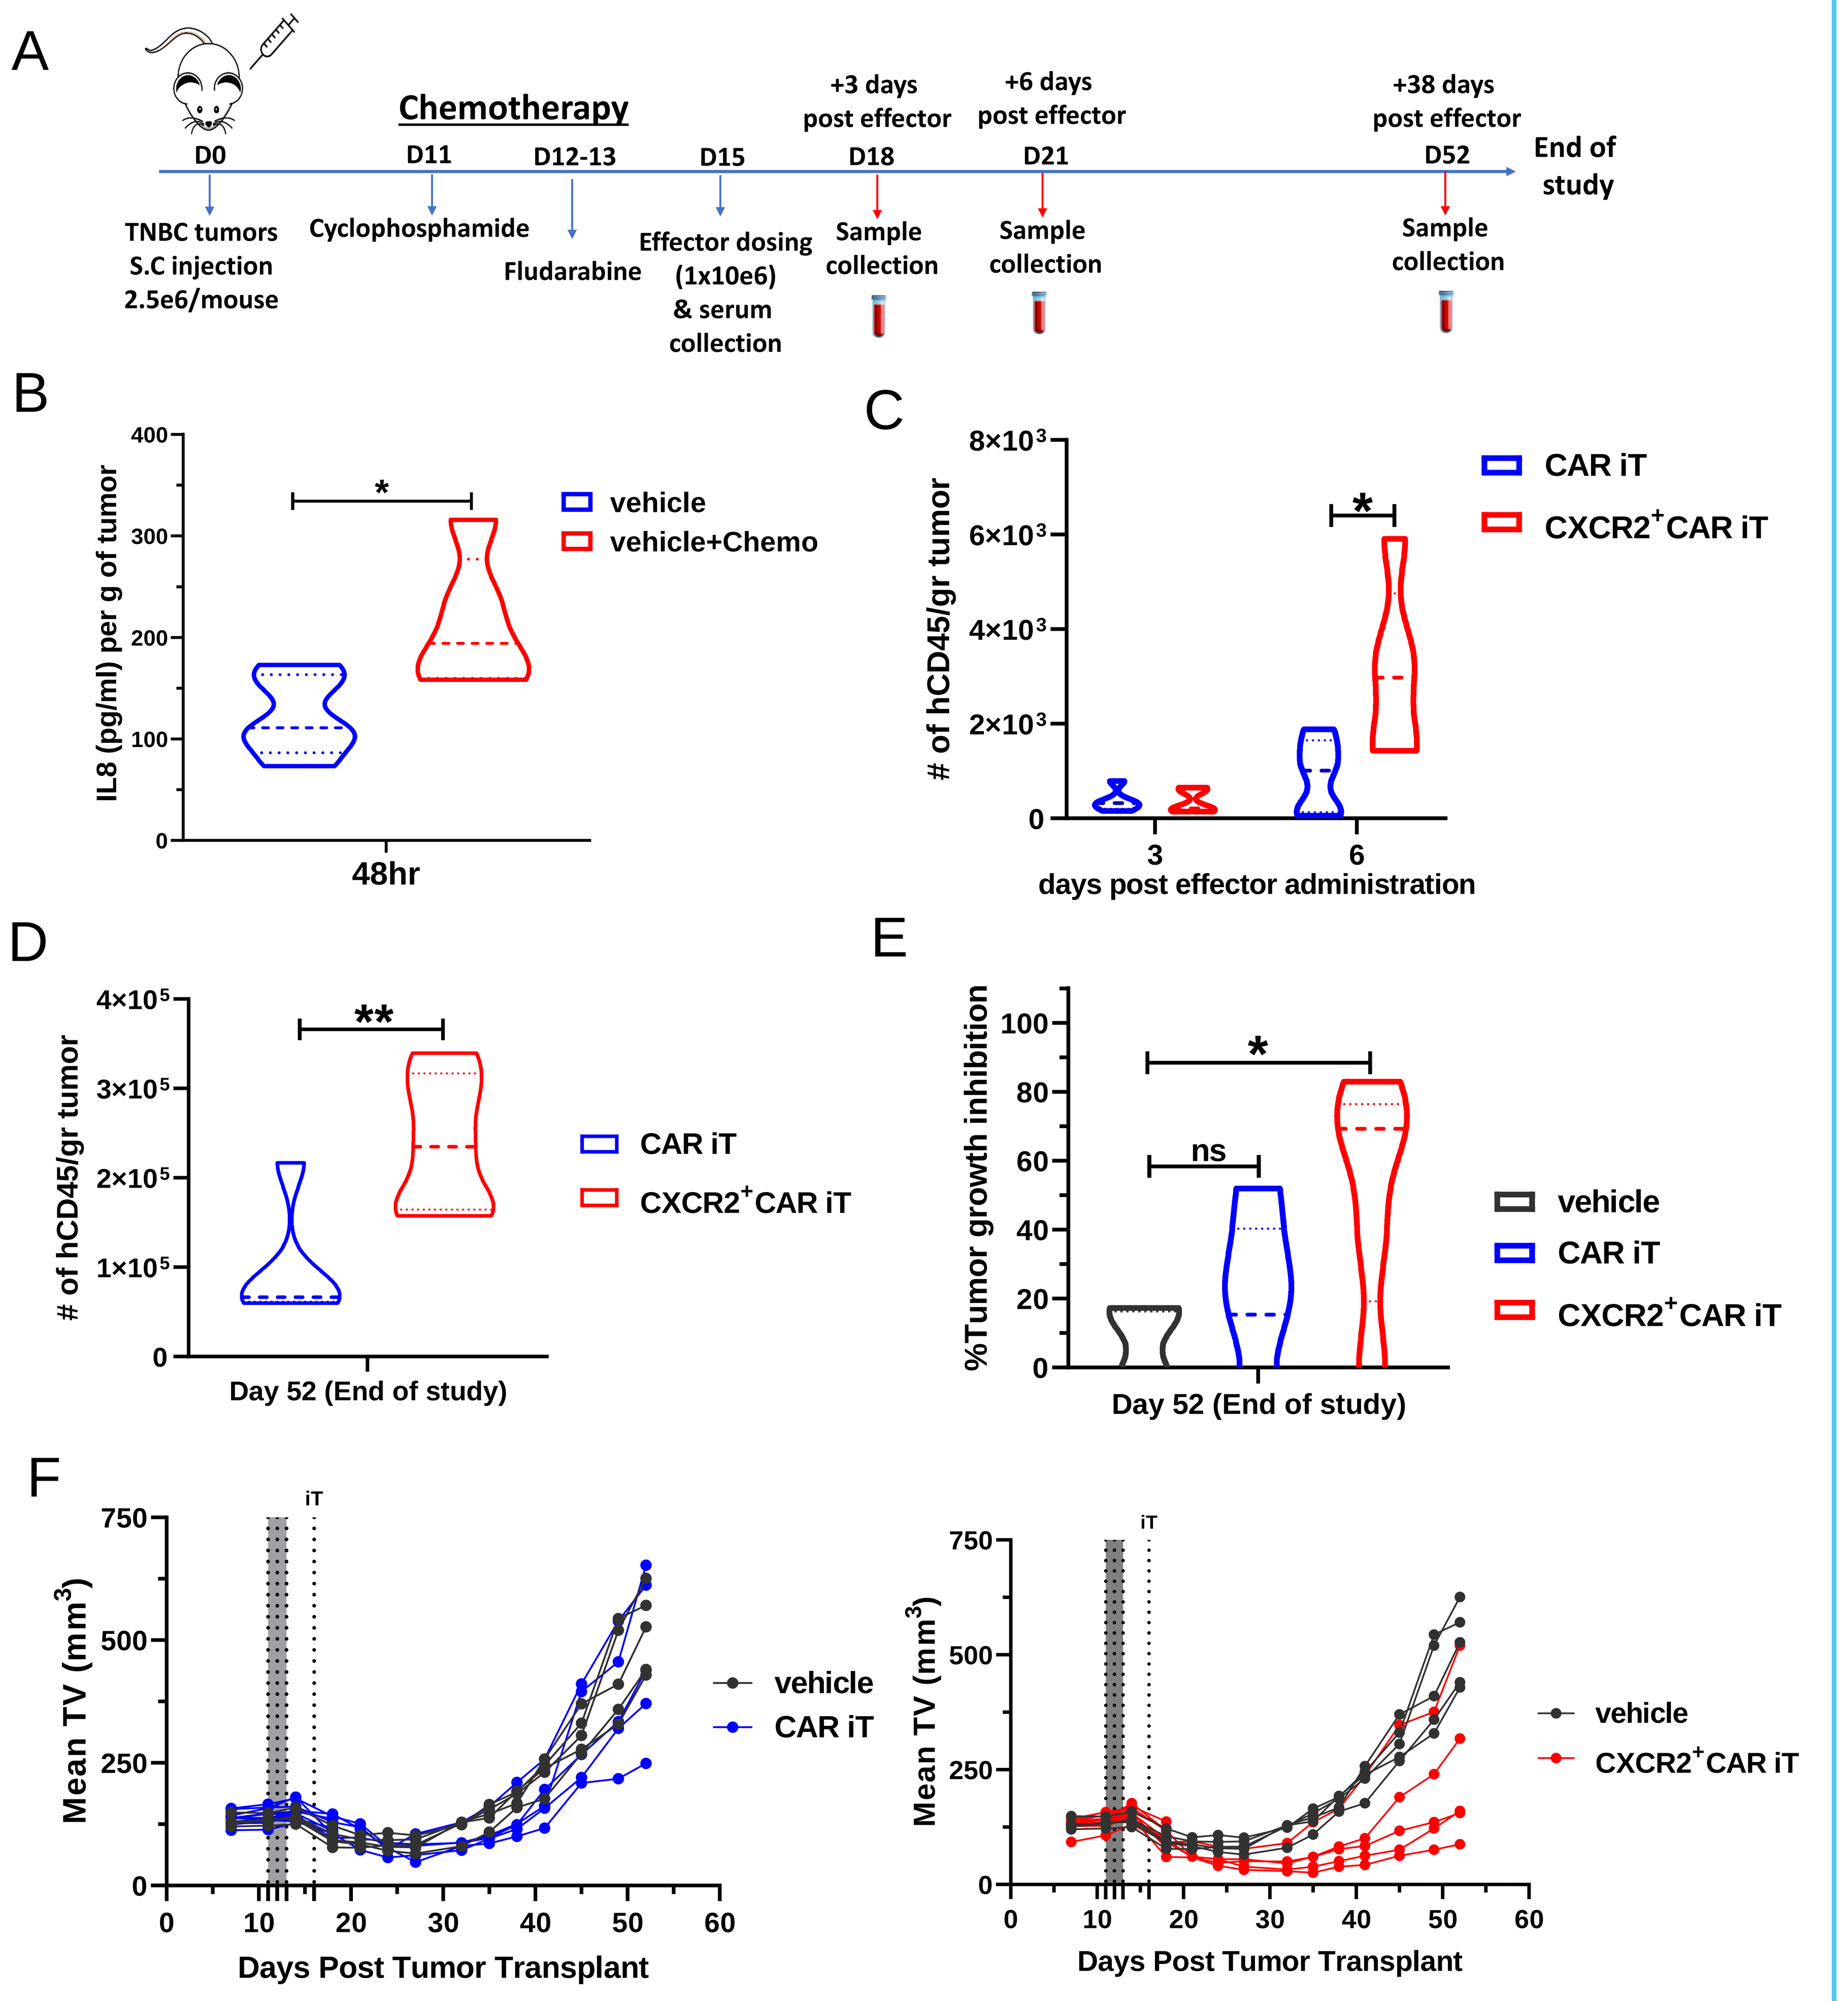
A
Chemotherapy
D0
TNBC tumors S.C injection 2.5e6/mouse
D11
Cyclophosphamide
D12-13
Fludarabine
D15
Effector dosing (1x10e6) & serum collection
+3 days post effector
D18
Sample collection
+6 days post effector
D21
Sample collection
+38 days post effector
D52
Sample collection
End of study
B
IL8 (pg/ml) per g of tumor
vehicle
vehicle+Chemo
48hr
C
of hCD45/gr tumor
CAR iT
CXCR2+CAR iT
days post effector administration
D
Day 52 (End of study)
E
%Tumor growth inhibition
vehicle
F
Mean TV (mm3)
Days Post Tumor Transplant

Figure 5 (A) Experimental design for *in vivo* evaluation of $CXCR2^-$ and $CXCR2^+$ CAR iT cells with and without (data are not shown) chemotherapy (Cyclophosphamide/Fludarabine) preconditioning. (B) CXCL8 levels in serum collected 48hrs post chemo-preconditioning. (C-D) Intratumoral CAR iT infiltration & retention was evaluated by flow cytometry in tumors collected at 3, 6 and 38 days post CAR iT administration. (E-F)Tumor measurements (avg +/- SEM) for CAR iT and $CXCR2^+$ CAR iT with chemo preconditioning. **P<0.01, *P<0.05.

Conclusion

Collectively, the preclinical data demonstrate that the engineering of synthetic chemokine receptors to further direct off-the-shelf CAR iT cells to the tumor site is a viable strategy to improve anti-tumor activity, including as part of a multiplexed-engineering platform for overcoming challenges in treating solid tumors.