A CD3 Fusion Receptor (CD3-FR) Uniquely Enables Compatibility of Allogeneic CAR-T and -NK cells with T Cell Engagers to Enhance Antitumor Function and Limit Antigen Escape

Eigen Peralta*, Dan Lu*, Mark Landon, Hui-Yi Chu, Soo Park, Masanao Tsuda, Trevor Zarecki, Elena Demeester, Matthew Denholtz, Amit Mehta, Earl Avramis, Philip Chu, Jeffrey Chen, Eric Sung, Alec Witty, Tom Lee, and Bahram Valamehr

Fate Therapeutics, Inc., San Diego, CA, USA

Introduction

Chimeric antigen receptor (CAR) T cells have shown remarkable clinical success in the treatment of many malignancies. However, certain challenges remain, including broad patient access and durability of response. We have shown that a scalable manufacturing platform, where genetically edited induced pluripotent stem cell (iPSC) master cell lines are created to serve as a renewable starting material for the derivation of uniformly engineered CAR T or NK cells, can facilitate broad patient access in an off-the-shelf manner. To improve durability of response, often plagued by antigen heterogeneity found in cancer, we are pursuing several combination strategies, merging cell therapy with therapeutic agents such as checkpoint blockade therapy and monoclonal antibodies to increase anti-tumor activity and multi-antigen targeting of cancer.

Bispecific T cell engagers (BiTEs) have also shown remarkable advancements in the treatment of various cancer types, but they are also afflicted by their own challenges and would equally benefit from combination strategies. Unfortunately, T cell engagers that bind CD3 on effector cells are not compatible with allogeneic adoptive cell therapy, since the T cell receptor (TCR) that supports CD3 surface expression has been ablated to prevent graft-versus-host disease in the case of T cells, or not expressed, which is the case with NK cells. To investigate the anti-tumor synergy between off-the-shelf cell therapy and T cell engagers, we developed a novel CD3ε fusion receptor (CD3-FR) to uniquely support the expression of a functional CD3 on TCR-less allogeneic T and NK cells and to enable compatibility between allogeneic cell therapy and T cell engagers.

Our multiplexed-engineering iPSC platform overcomes multiple challenges associated with existing approaches to cellular therapy

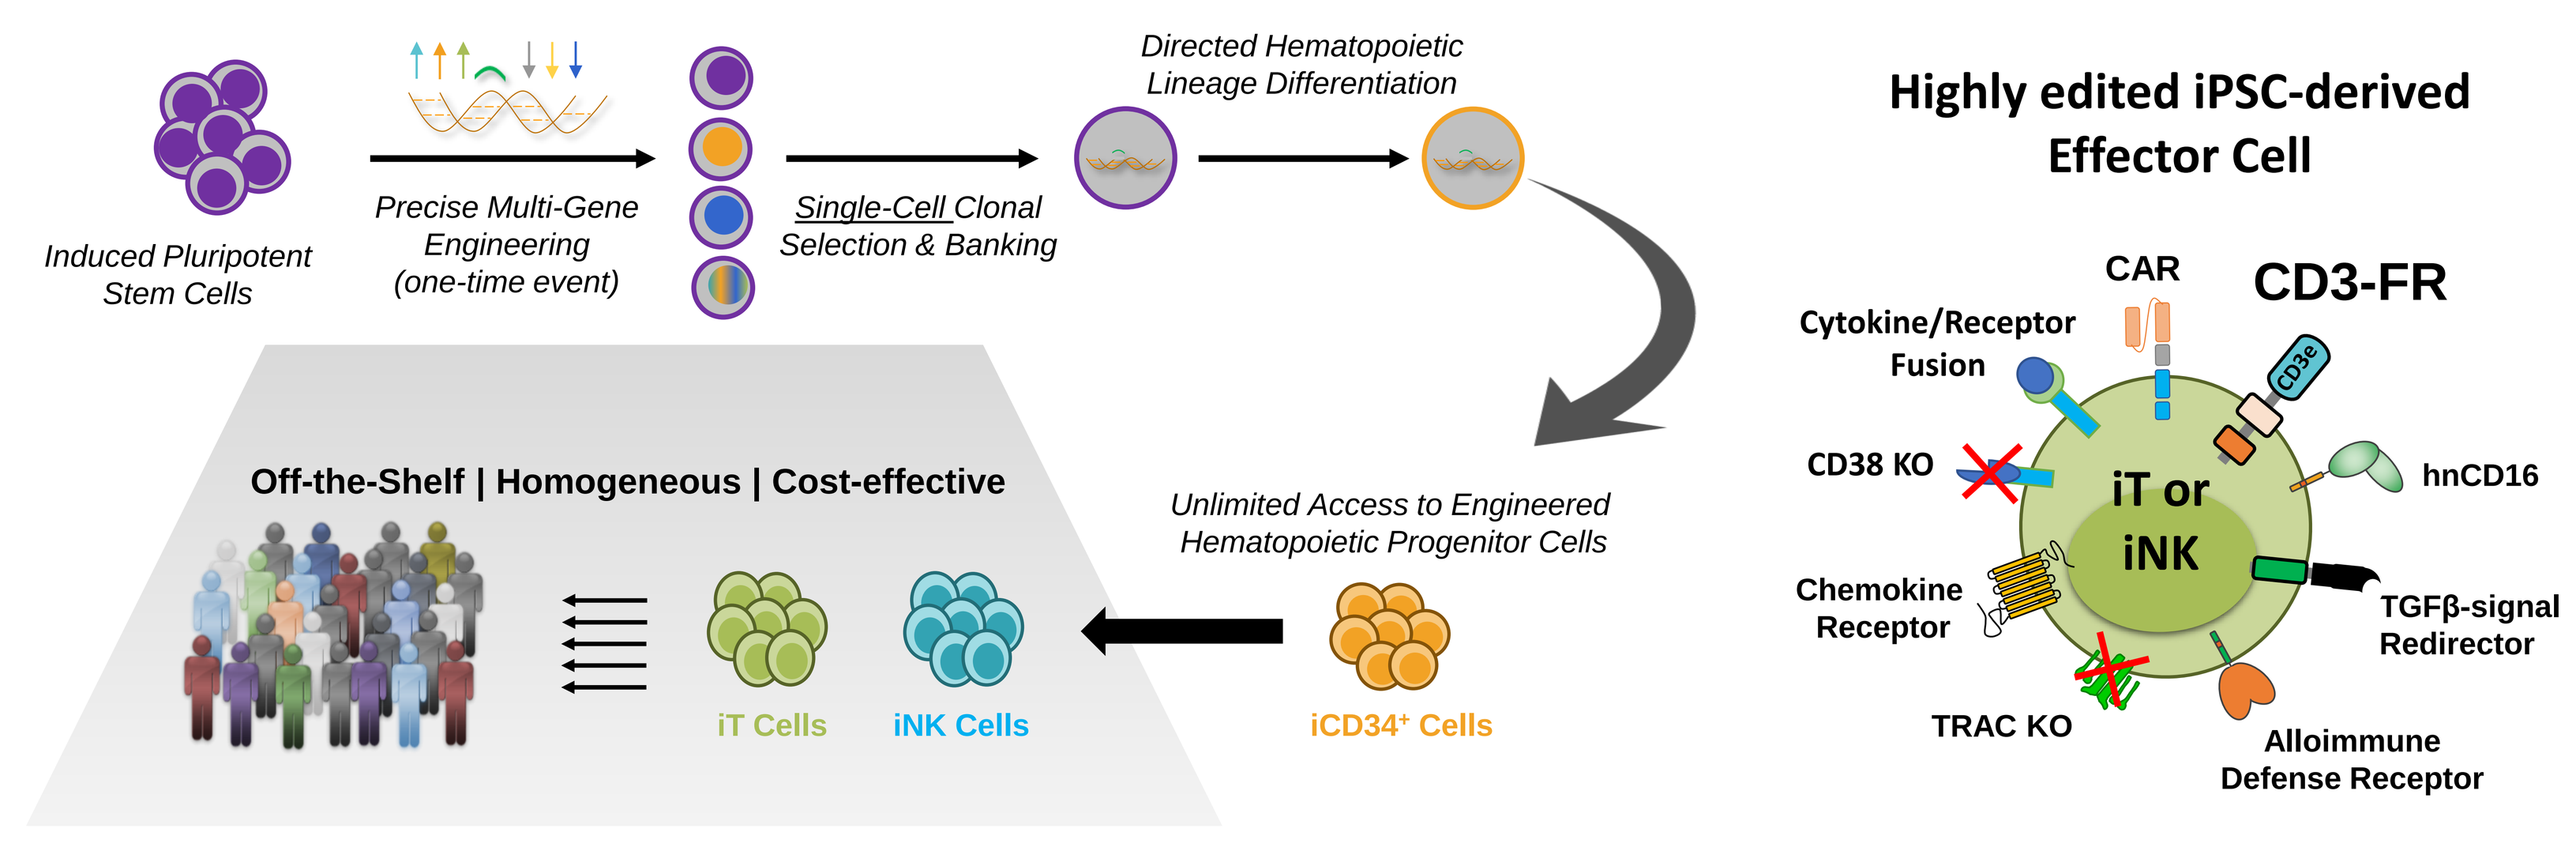
Induced Pluripotent Stem Cells
Precise Multi-Gene Engineering (one-time event)
Single-Cell Clonal Selection & Banking
Directed Hematopoietic Lineage Differentiation
Unlimited Access to Engineered Hematopoietic Progenitor Cells
iCD34⁺ Cells
iT Cells
iNK Cells
Off-the-Shelf | Homogeneous | Cost-effective
Highly edited iPSC-derived Effector Cell
CAR
CD3-FR
Cytokine/Receptor Fusion
CD38 KO
hnCD16
iT or iNK
Chemokine Receptor
TGFβ-signal Redirector
TRAC KO
Alloimmune Defense Receptor

Results

A novel CD3 Fusion Receptor (CD3-FR) designed to enable surface expression on allogeneic TCR-less effector cells can uniquely transduce a signal in response to BiTE-dependent target cell engagement

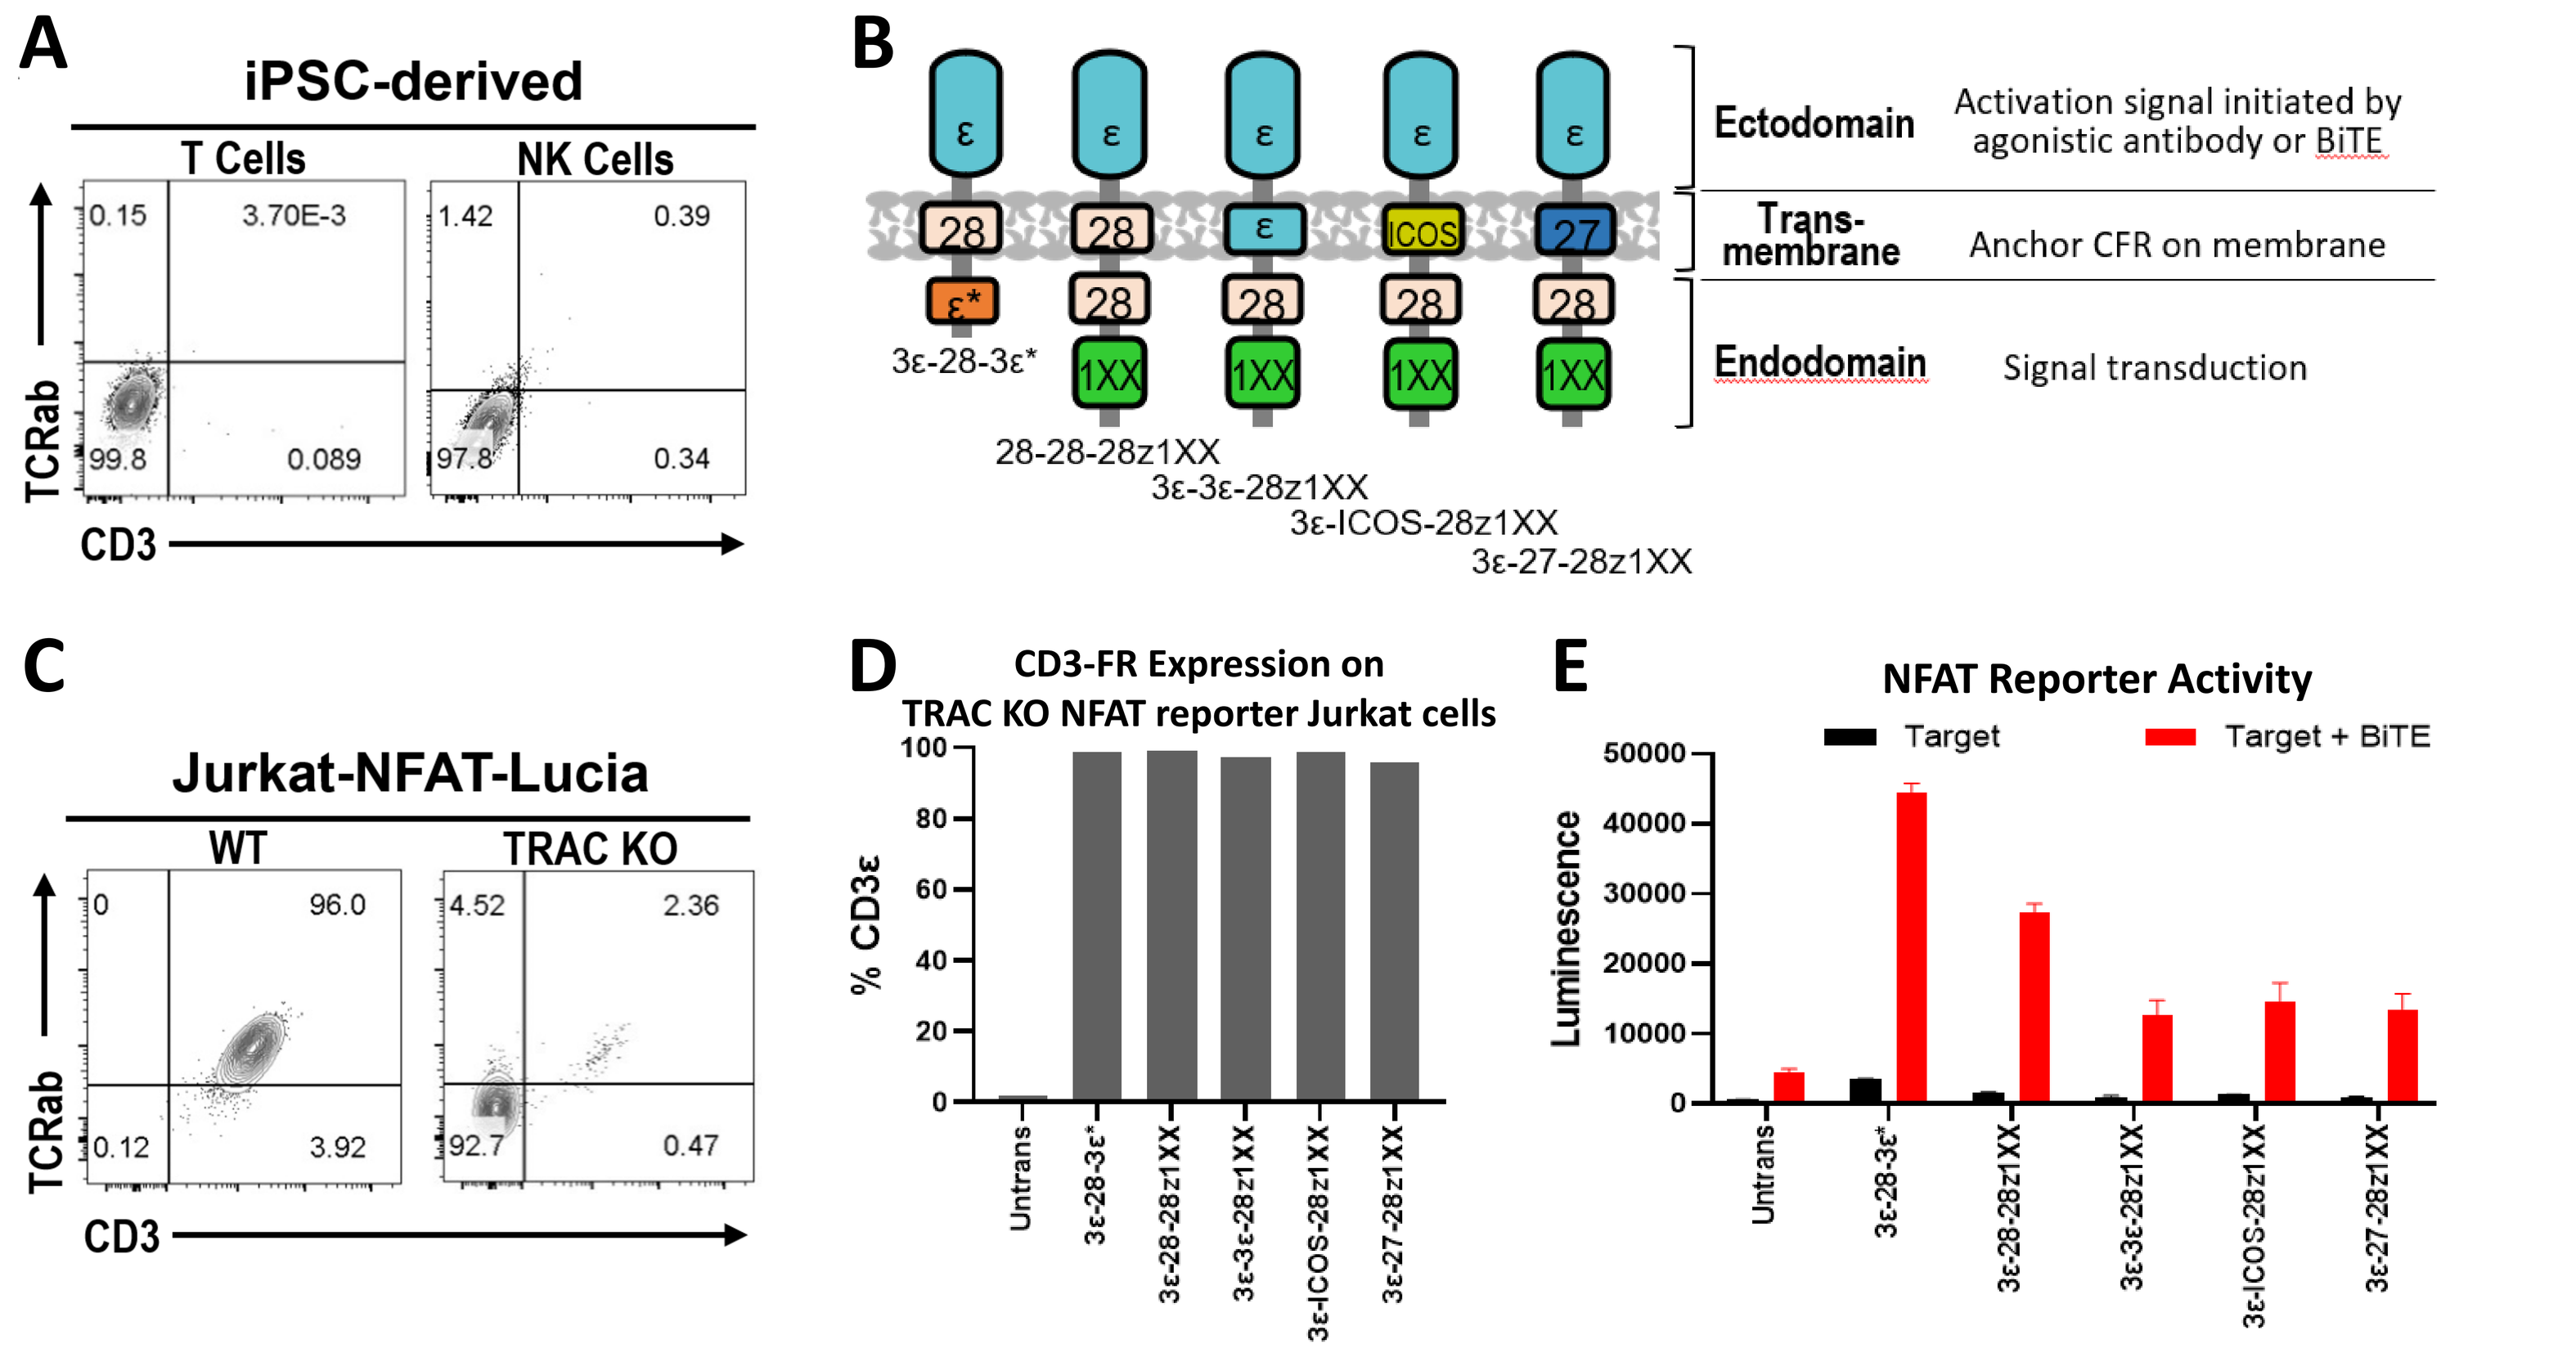
A
iPSC-derived
T Cells
NK Cells
TCRab
CD3
B
Ectodomain
Activation signal initiated by agonistic antibody or BiTE
Trans-membrane
Anchor CFR on membrane
Endodomain
Signal transduction
3ε-28-3ε*
28-28-28z1XX
3ε-3ε-28z1XX
3ε-ICOS-28z1XX
3ε-27-28z1XX
C
Jurkat-NFAT-Lucia
WT
TRAC KO
D
CD3-FR Expression on TRAC KO NFAT reporter Jurkat cells
% CD3ε
E
NFAT Reporter Activity
Target
Target + BiTE
Luminescence
Untrans

Figure 1. **(A)** Flow cytometry data showing absence of surface-expressed TCRab and CD3 molecules on allogeneic iPSC-derived T and NK cells. **(B)** Examples of CD3 Fusion Receptor (CD3-FR) construct designs with modular transmembrane and endodomains. **(C)** Flow cytometry data showing TCRab x CD3 surface staining on TRAC KO Jurkat cells that also express an NFAT-Lucia reporter. **(D)** Flow cytometric staining for surface CD3e on TRAC KO cells from (C) that were transduced with indicated CD3-FR constructs. **(E)** Luciferase assay measuring signal transduction from CD3-FR constructs in response to BiTE-dependent target engagement.

TRAC-CAR iPSCs can be sequentially engineered by CRISPR-editing to express a CD3 fusion receptor and differentiated into CD3-FR⁺ CAR-iT cells with potent CAR function

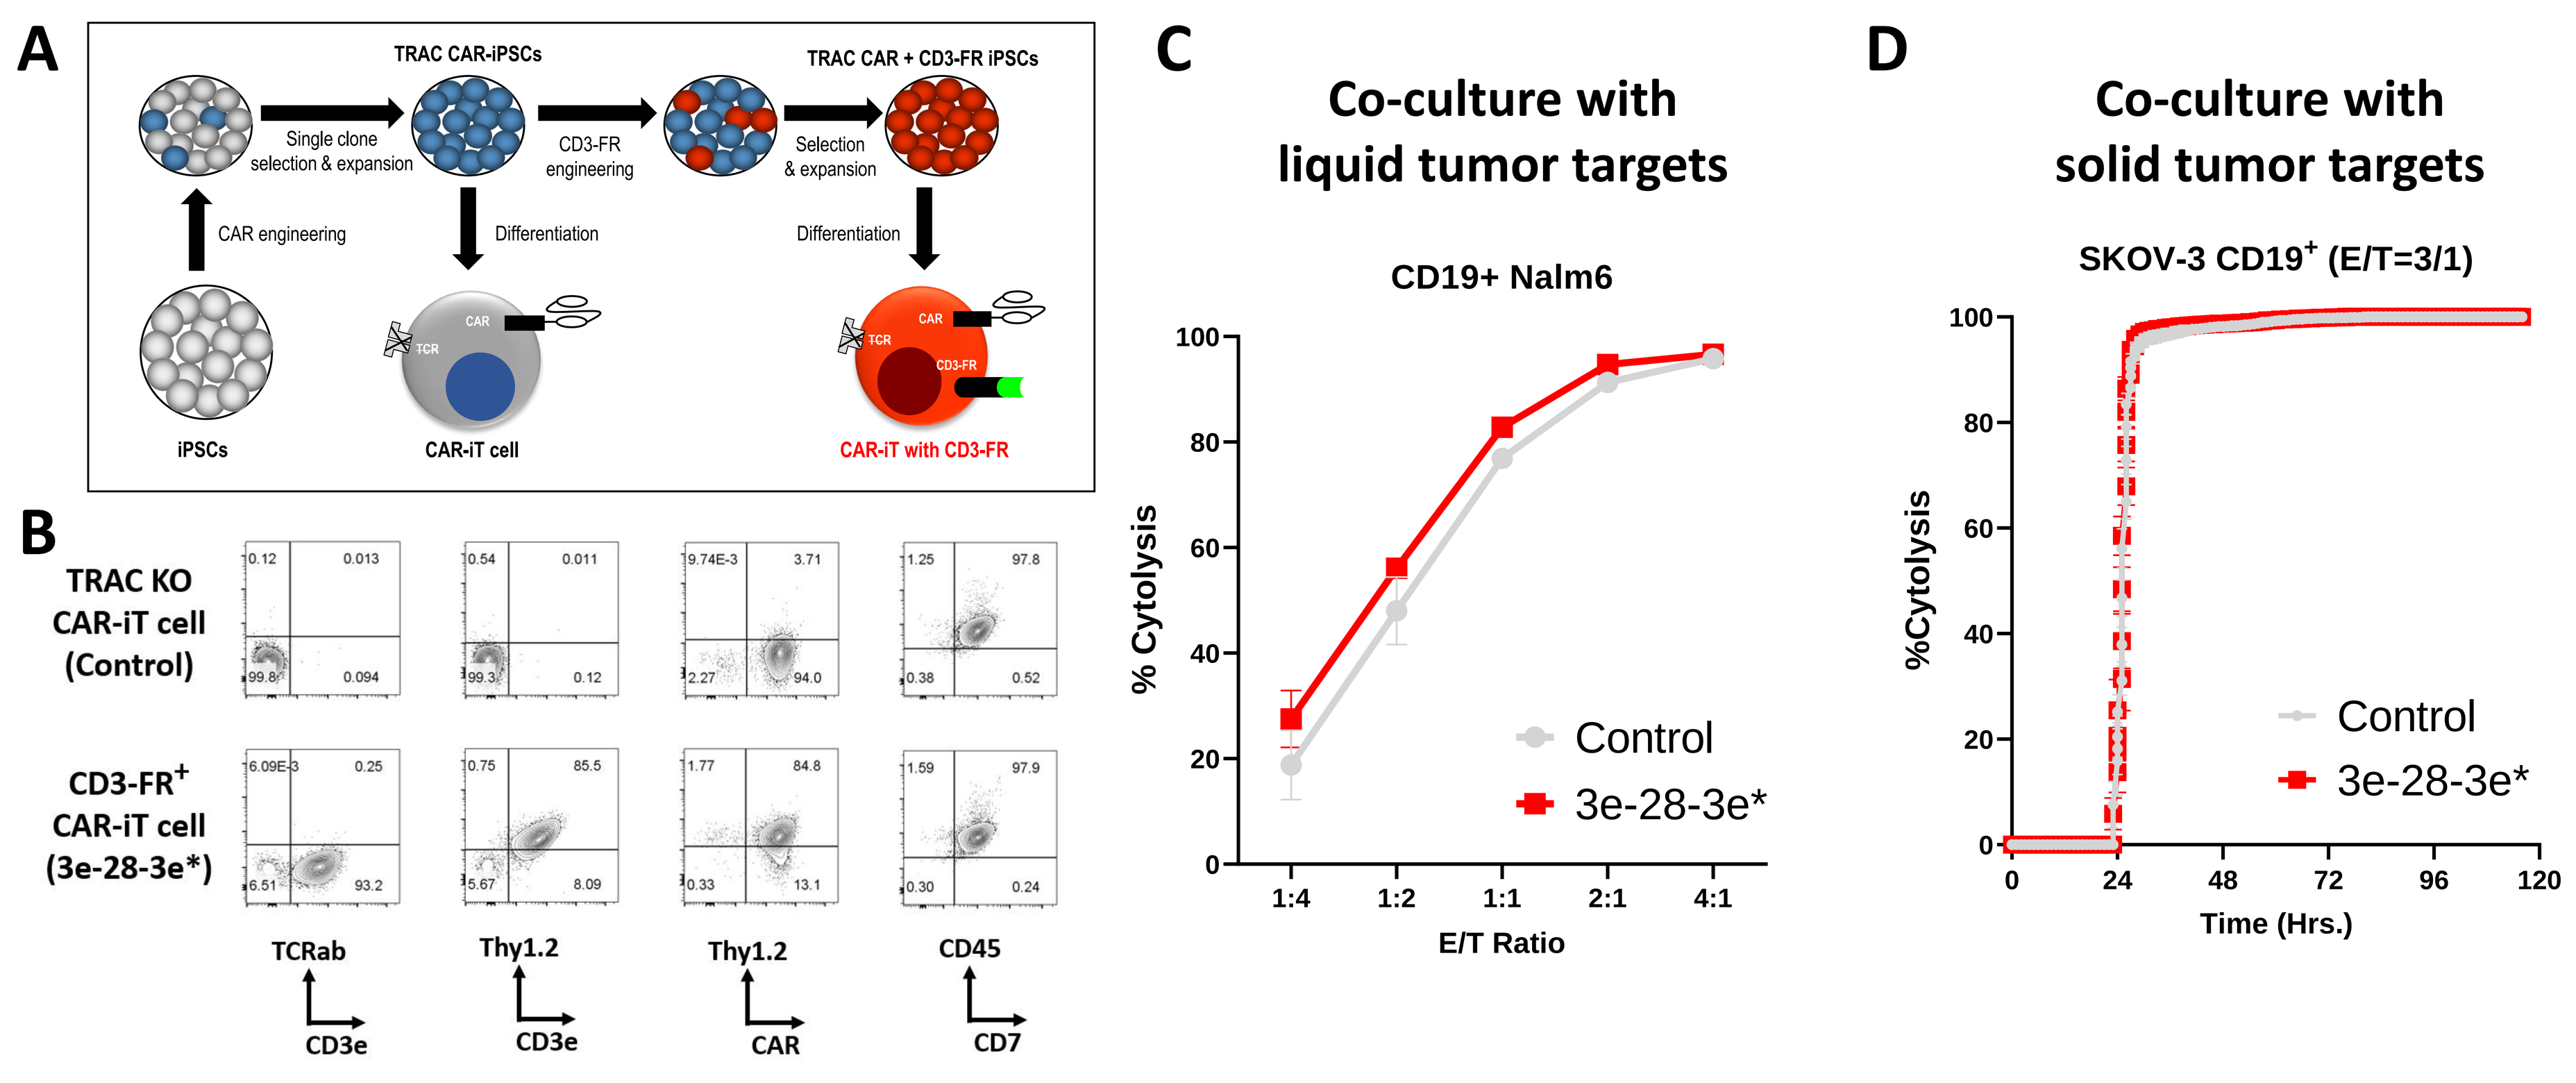
A
TRAC CAR-iPSCs
TRAC CAR + CD3-FR iPSCs
Single clone selection & expansion
CD3-FR engineering
Selection & expansion
CAR engineering
Differentiation
iPSCs
CAR-iT cell
CAR-iT with CD3-FR
B
TRAC KO CAR-iT cell (Control)
CD3-FR+ CAR-iT cell (3e-28-3e*)
C
Co-culture with liquid tumor targets
CD19+ Nalm6
% Cytolysis
E/T Ratio
Control
3e-28-3e*
D
Co-culture with solid tumor targets
SKOV-3 CD19+ (E/T=3/1)
%Cytolysis
Time (Hrs.)

Figure 2. **(A)** Schematic showing a strategy where TRAC-CAR iPSCs undergo a second round of CRISPR engineering to introduce a CD3-FR modality. **(B)** Flow cytometry data showing normal phenotypic profile of CD3-FR+ CAR-iT cells. **(C-D)** *In vitro* killing assays in co-cultures with indicated target cells demonstrating that the CD3-FR modality does not interfere with CAR function.

CD3-FR⁺ CAR-iT cells show directed *in vitro* and *in vivo* activity toward CAR antigen negative targets via BiTE-dependent targeting of a second tumor-associated antigen

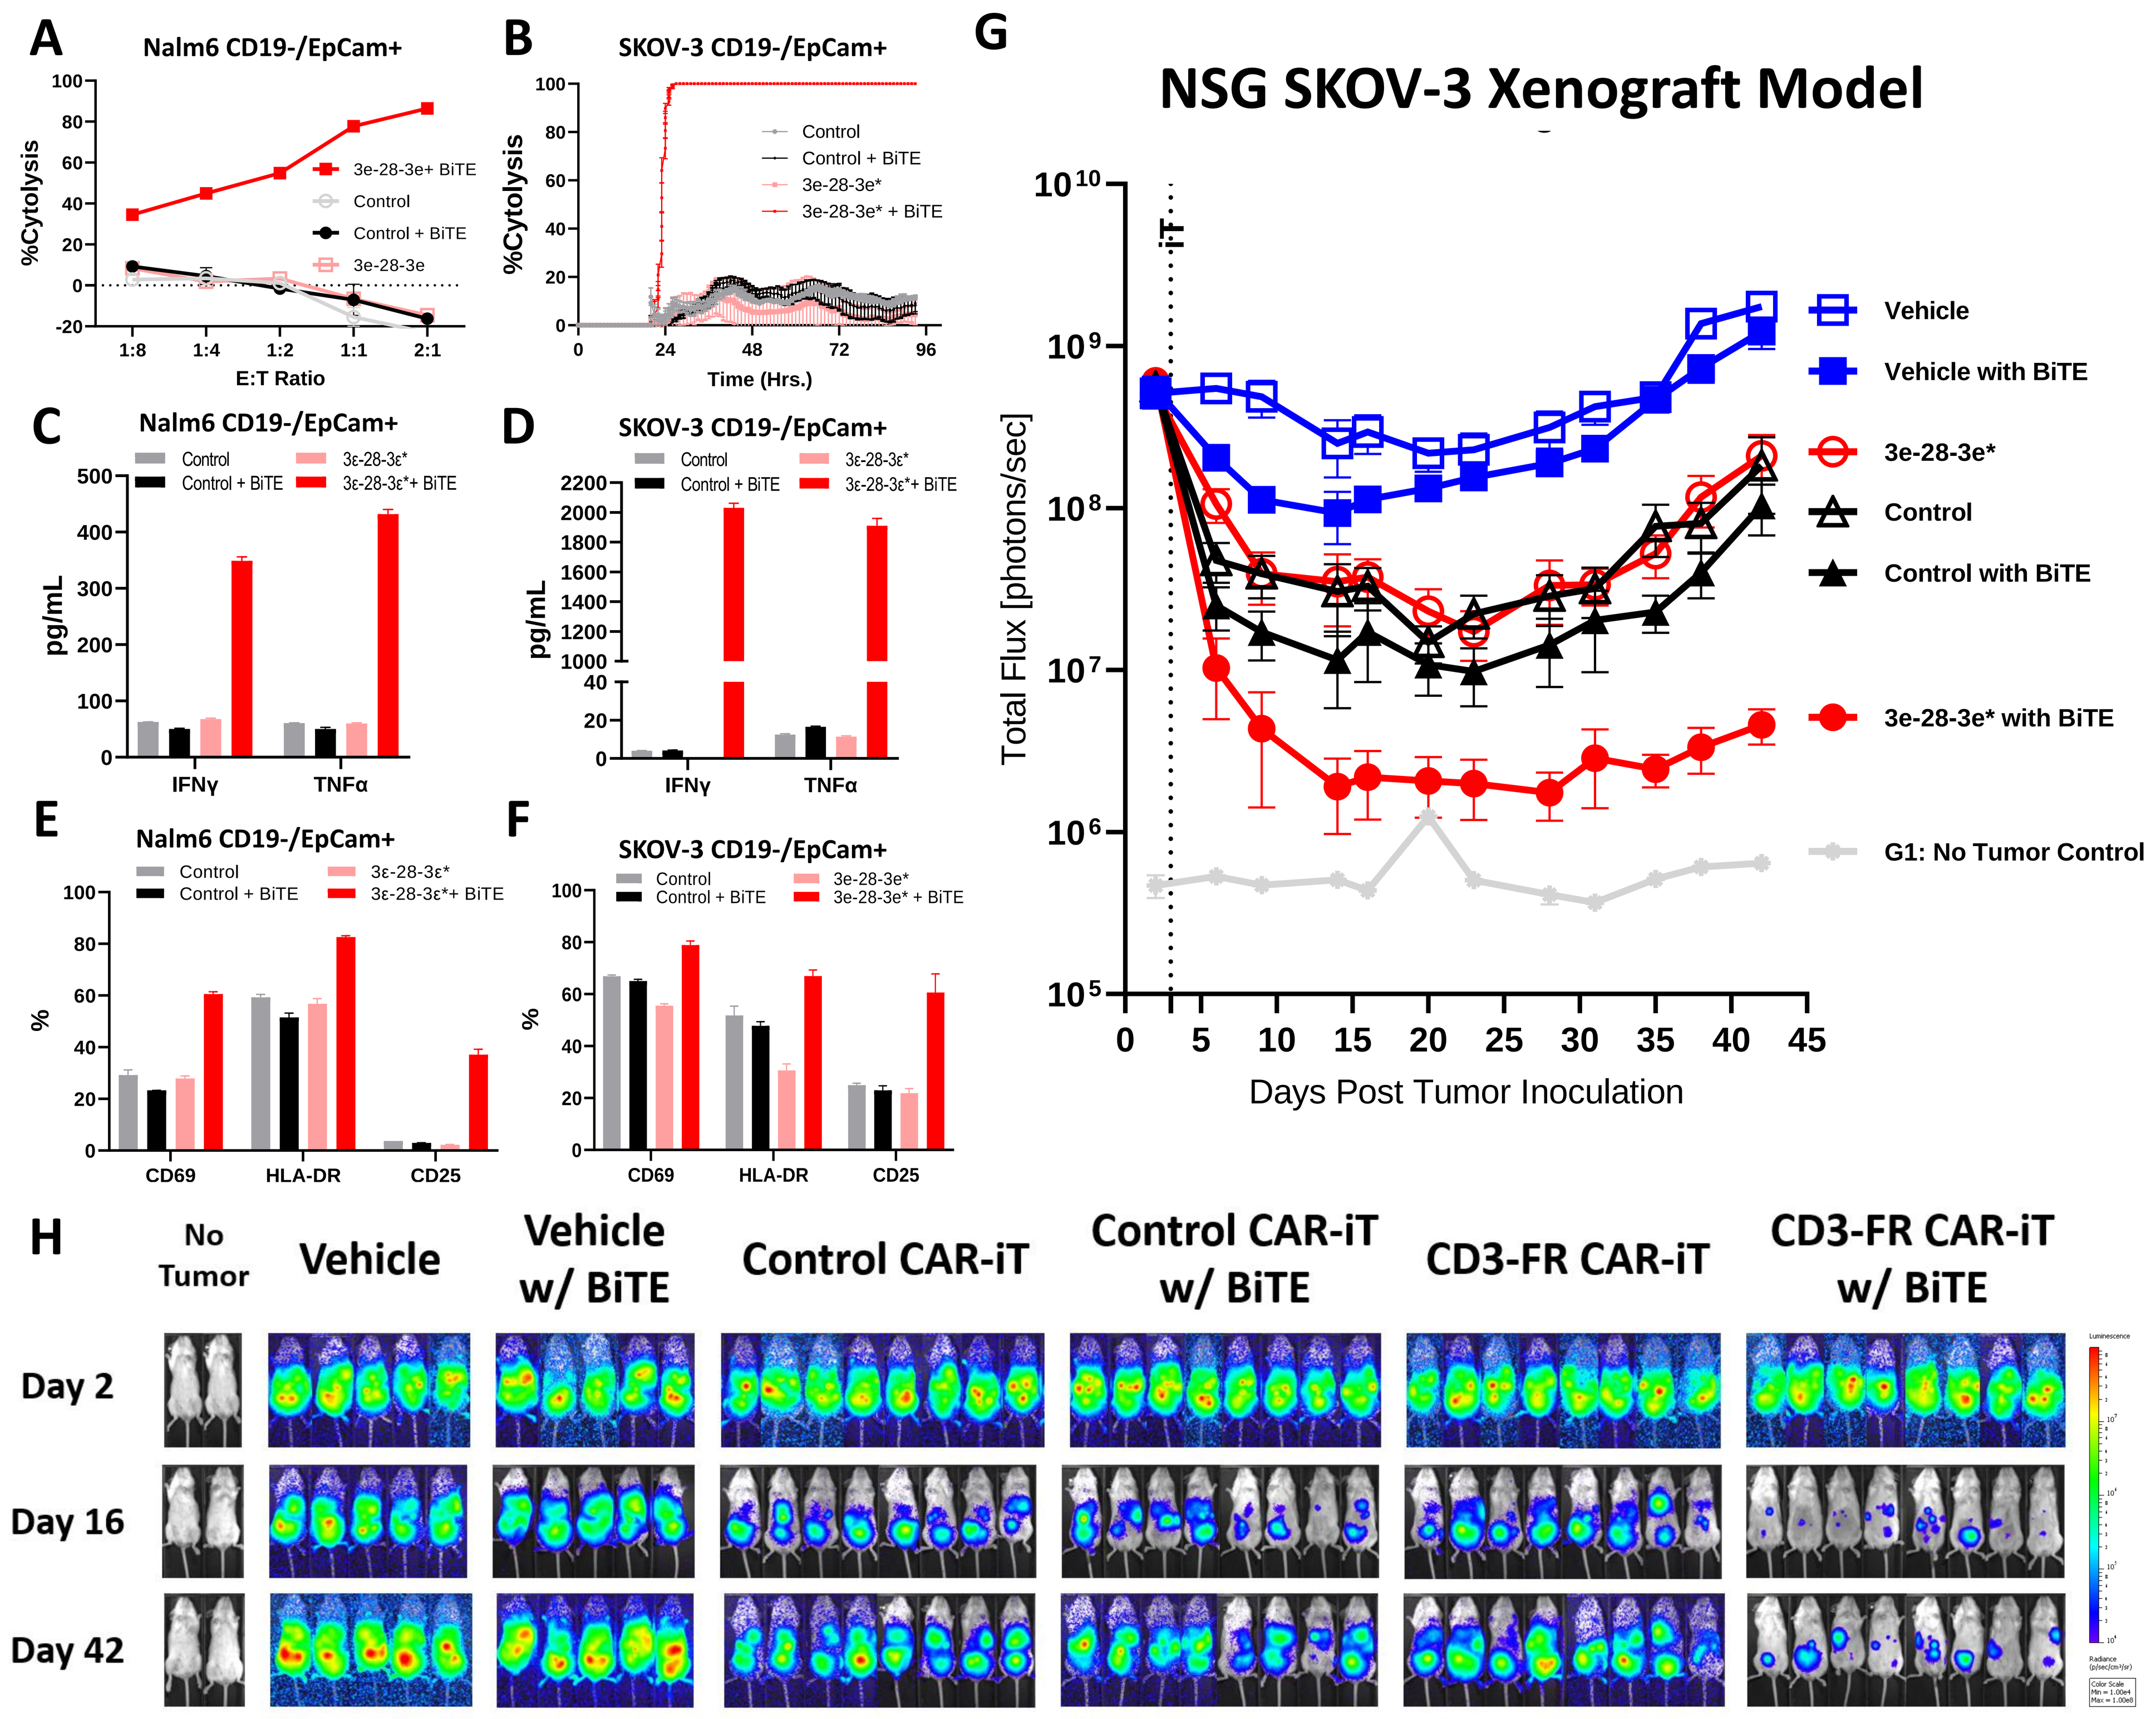
A
Nalm6 CD19-/EpCam+
B
SKOV-3 CD19-/EpCam+
C
D
E
F
G
NSG SKOV-3 Xenograft Model
Total Flux [photons/sec]
Days Post Tumor Inoculation
Vehicle
Vehicle with BiTE
3e-28-3e*
Control
Control with BiTE
3e-28-3e* with BiTE
G1: No Tumor Control
H
No Tumor
Vehicle
Vehicle w/ BiTE
Control CAR-iT
Control CAR-iT w/ BiTE
CD3-FR CAR-iT
CD3-FR CAR-iT w/ BiTE
Day 2
Day 16
Day 42

Figure 3. **(A-B)** Cytolytic function, **(C-D)** cytokine production, and **(E-F)** activation marker expression from CD3-FR+ iT cells *in vitro* in the presence or absence of anti-EpCam BiTE. **(G)** BLI measurement of tumor burden over time in NSG mice treated with a single dose of indicated effectors +/- bi-weekly 20 mg/kg EpCam BiTE injection. **(H)** IVIS images from indicated timepoints from the in vivo study in (G).

CD3-FR⁺ CAR-iT cells can uniquely control growth of a heterogenous tumor population with the combination of CAR and CD3-FR modality targeting

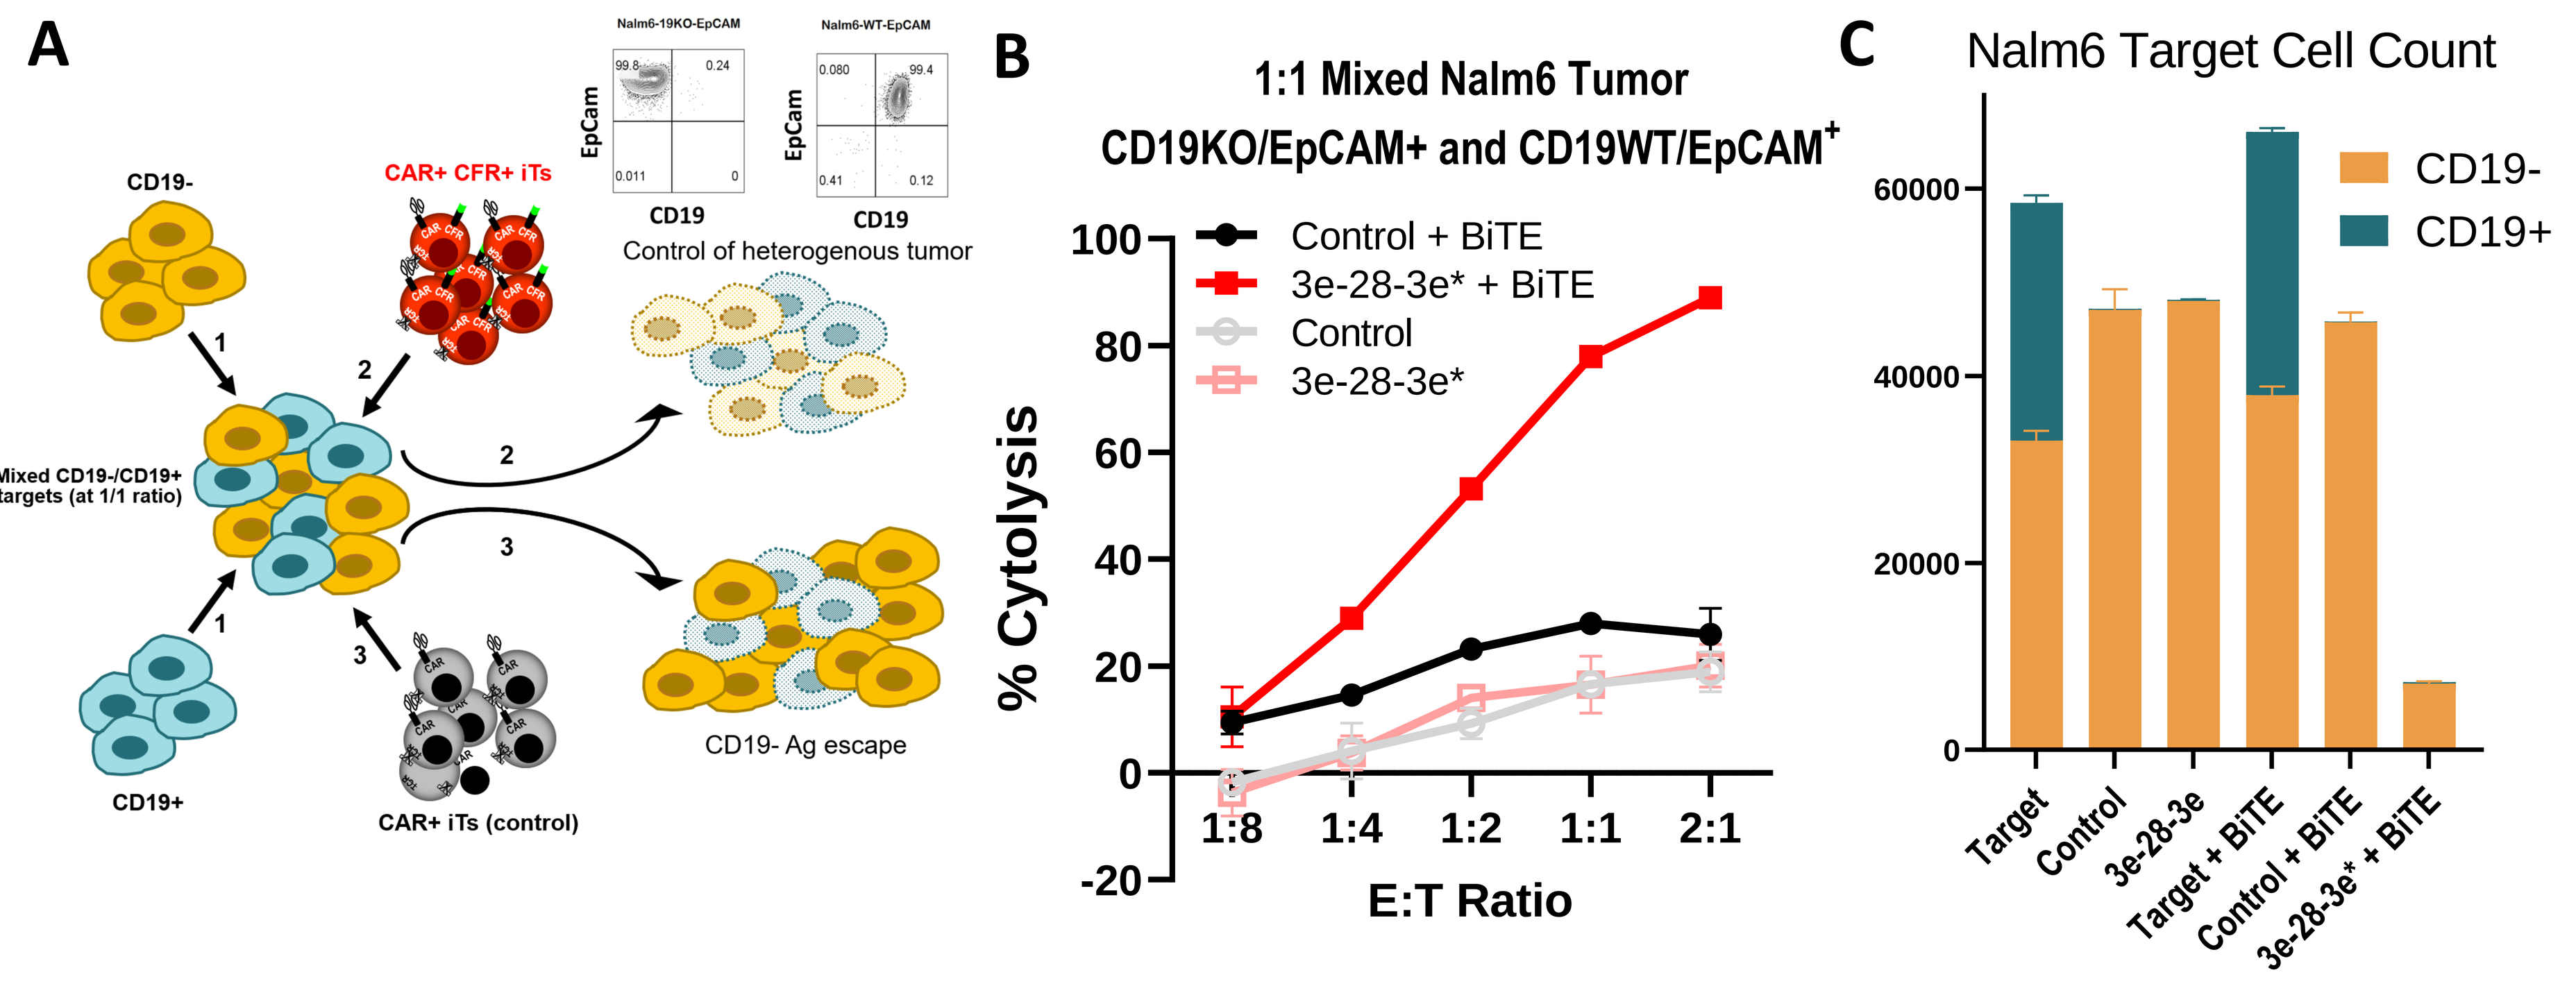
A
CD19-
CAR+ CFR+ iTs
Mixed CD19-/CD19+ targets (at 1/1 ratio)
CD19+
CAR+ iTs (control)
Control of heterogenous tumor
CD19- Ag escape
B
1:1 Mixed Nalm6 Tumor CD19KO/EpCAM+ and CD19WT/EpCAM+
% Cytolysis
E:T Ratio
Control + BiTE
3e-28-3e* + BiTE
Control
3e-28-3e*
C
Nalm6 Target Cell Count
CD19-
CD19+

Figure 4. **(A)** CD19⁺ EpCam⁺ and CD19⁻ EpCam⁺ target cells were mixed at 1:1 ratio and co-cultured with Control (CD3-FR negative) or CD3-FR⁺ CAR-iT cells in the presence or absence of BiTE targeting EpCam. **(B)** Flow-based killing assay results. **(C)** Flow-based counting of viable target cells remaining in the co-culture where E:T is at 1:1.

Expression of a CD3-FR on iPSC-derived NK cells (iNK) enables compatibility with T cell engagers, a first-time demonstration

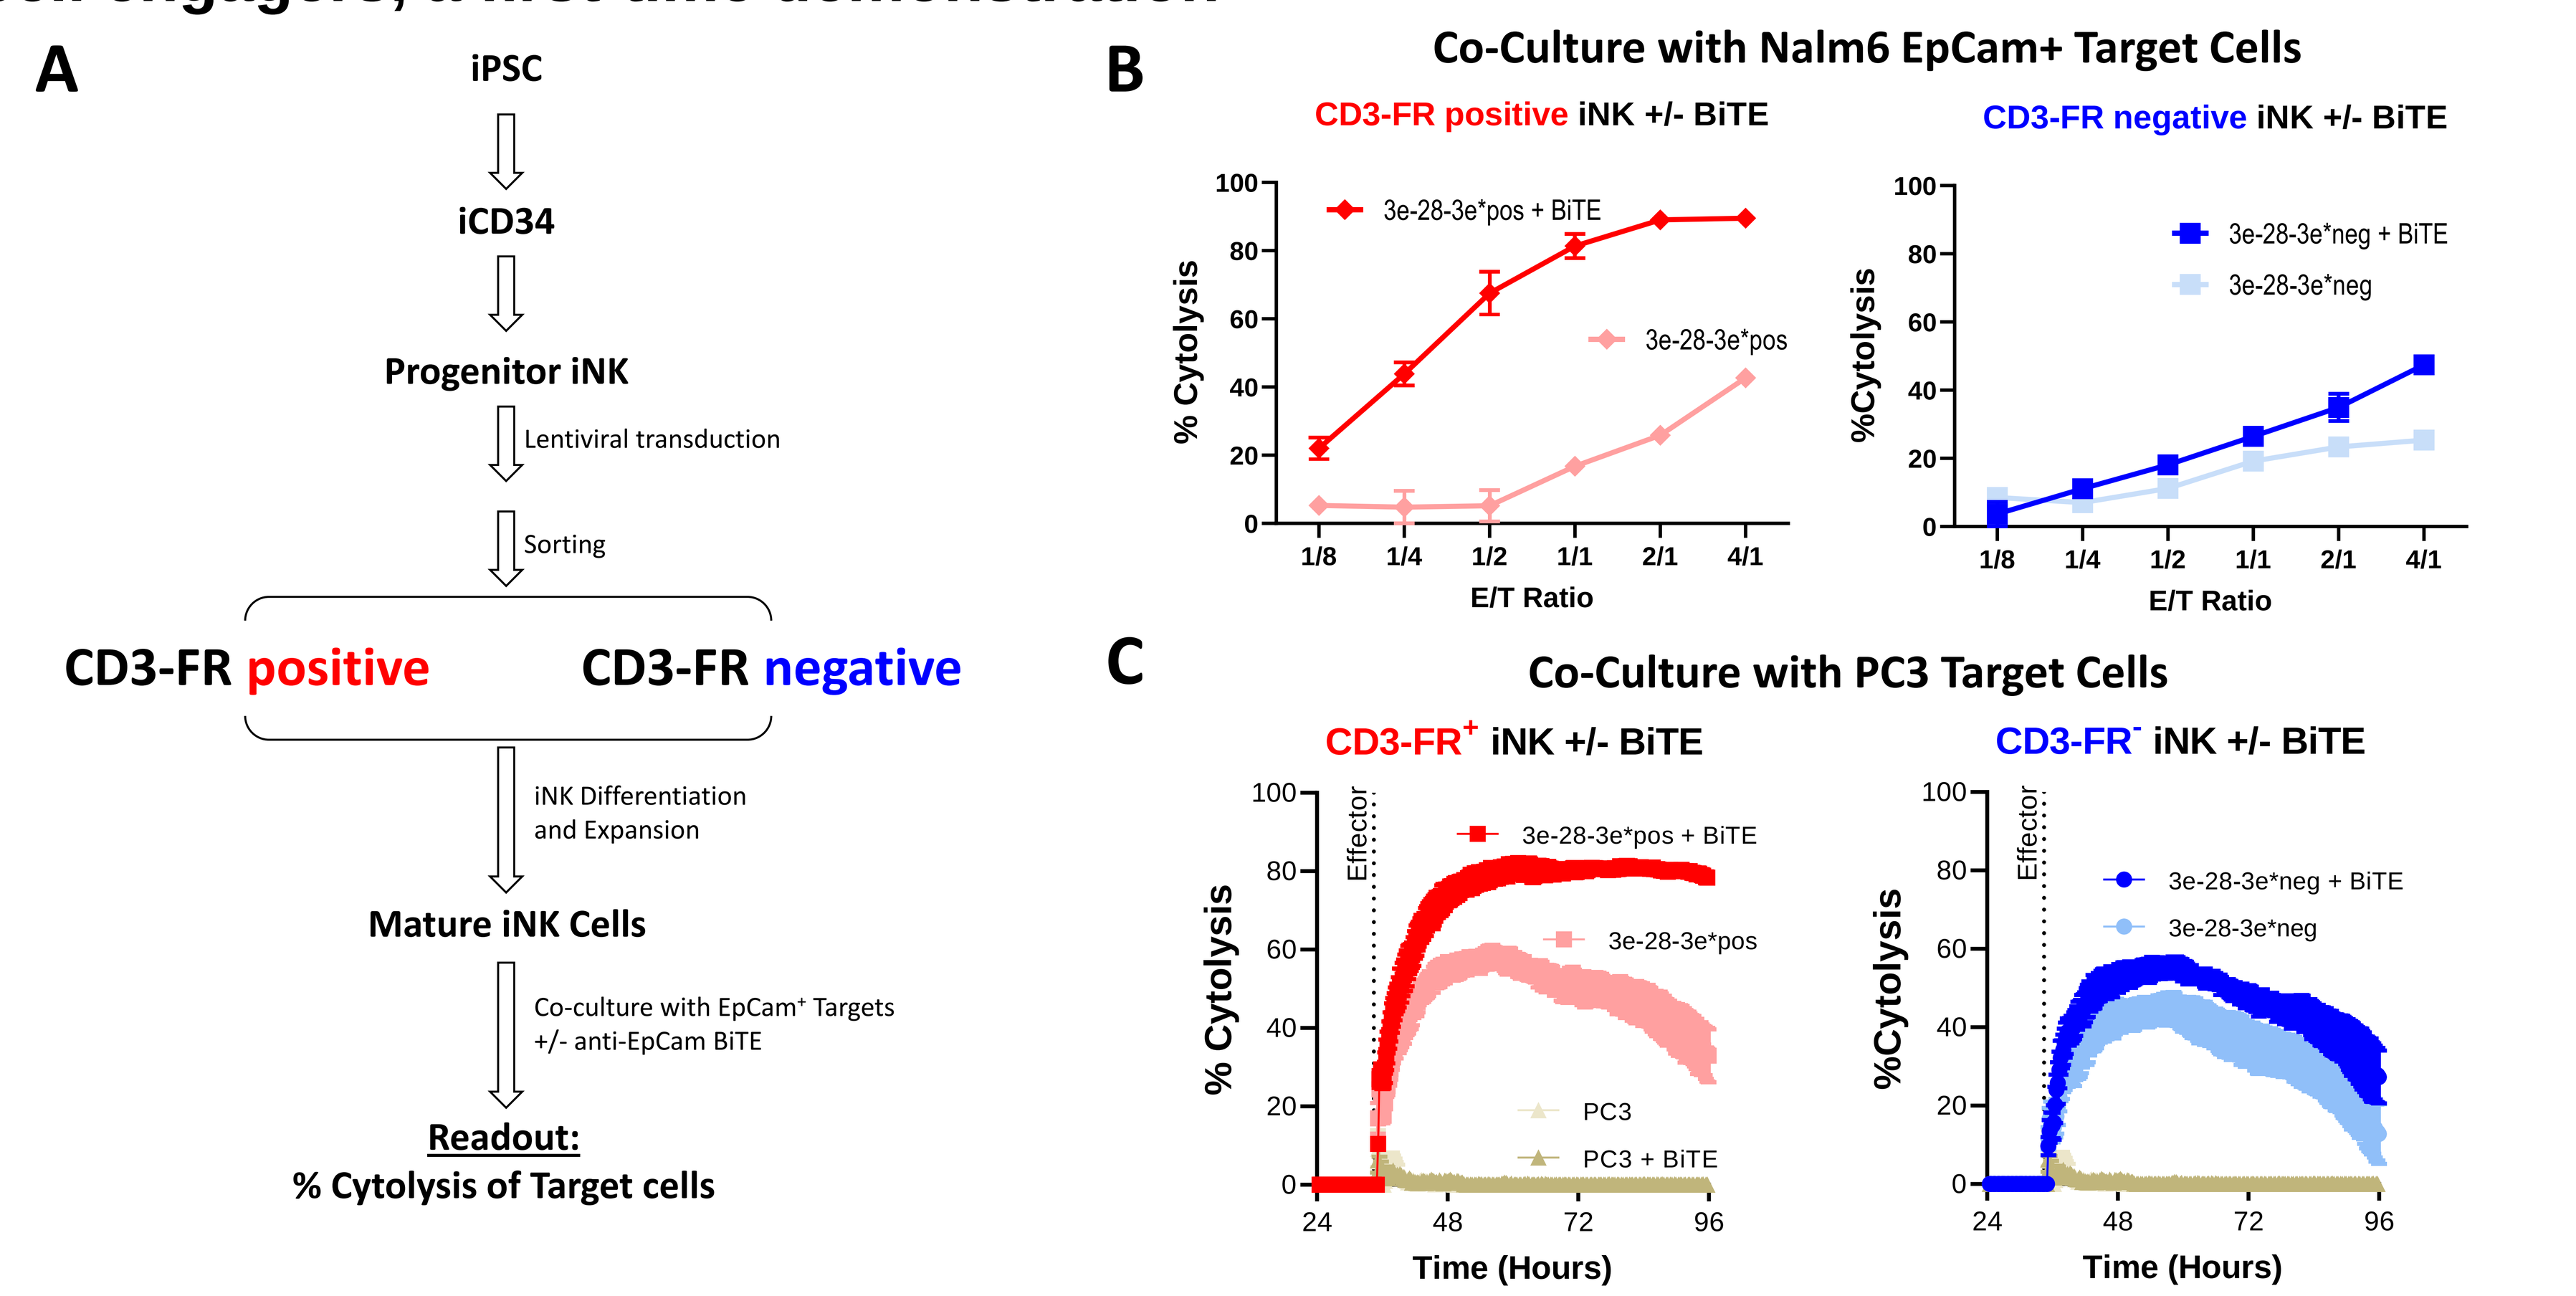
A
iPSC
iCD34
Progenitor iNK
Lentiviral transduction
Sorting
CD3-FR positive
CD3-FR negative
iNK Differentiation and Expansion
Mature iNK Cells
Co-culture with EpCam⁺ Targets +/- anti-EpCam BiTE
Readout:
% Cytolysis of Target cells
B
Co-Culture with Nalm6 EpCam+ Target Cells
CD3-FR positive iNK +/- BiTE
CD3-FR negative iNK +/- BiTE
E/T Ratio
C
Co-Culture with PC3 Target Cells
CD3-FR+ iNK +/- BiTE
CD3-FR- iNK +/- BiTE
Time (Hours)

Figure 5. **(A)** Experimental design describing the generation of CD3-FR positive or negative iNK cells that were tested for BiTE-dependent killing of a **(B)** Nalm6 EpCam+ liquid tumor line or **(C)** EpCam+ PC3 targets solid tumor line.

Summary and Conclusions

- Novel CD3 Fusion Receptors (CD3-FR) can be uniquely expressed on the surface of TCR-less allogeneic effector cells
- The endodomain of a CD3-FR is modular and can be tailored to provide an additional costimulatory signal to customize anti-tumor response
- Signaling from the CD3-FR is induced in the presence of target cells and a T cell engager
- iPSCs engineered to express a CD3-FR can be successfully differentiated into mature CAR-iT cells that show normal CAR-dependent function
- CD3-FR⁺ CAR-iT cells show robust *in vitro* and *in vivo* antitumor activity toward CAR-antigen negative tumor cells by targeting a second tumor associated antigen in a BiTE-dependent manner
- Dual targeting via the CAR and CD3-FR modalities enhance potency of anti-tumor activity and can mitigate antigen escape
- NK cells expressing the CD3-FR can be made compatible with T cell engagers
- CFR⁺ CAR⁺ effector cells can synergize with BiTEs to limit antigen escape in heterogenous tumors and elicit durable responses in challenging tumor settings.