A Novel Dual-Antigen Targeting Approach Enables Off-the-Shelf CAR NK Cells to Effectively Recognize and Eliminate the Heterogeneous Population Associated with AML

Zachary Davis, PhD, Frank Cichocki, PhD, Martin Felices, PhD, Hongbo Wang, Peter Hinderlie, Mark Juckett, MD, Joseph Maakaron, MD, Gregory I. Berk, MD, Bryan Hancock, PhD, John Goulding, PhD, Ryan Bjordahl, PhD, Bahram Valamehr, PhD and Jeffrey S. Miller, MD

Masonic Cancer Center, University of Minnesota, Minneapolis, MN; GT BioPharma, Brisbane, CA; Fate Therapeutics Inc., San Diego, CA

Abstract

While acute myeloid leukemia (AML) treatment has improved, relapse is still common, even after reduced intensity conditioning allogeneic transplantation. Natural killer (NK) cells are a promising allogeneic cell immunotherapy without the risk of graft-versus-host disease or cytokine release syndrome. We and others have reported that haploidentical NK cells can safely result in 30-50% complete remissions in advanced AML patients when given with high-dose lymphodepleting chemotherapy and exogenous cytokine support. However, while NK cell natural cytotoxicity has a clinical signal in AML, increased specificity and multi-dosing are likely to enhance the efficacy and durability of this treatment strategy. A number of stress ligands, including the MHC-I polypeptide-related sequence A and B (MICA/B), bind to the NK cell activating receptor NKG2D and initiate NK cell killing of transformed and infected cells, and several groups have shown promising results in clinical trials by developing NK cells that carry a NKG2D chimeric antigen receptor (CAR). However, the distal α1 and α2 domains of MICA/B that are recognized by NKG2D can be proteolytically cleaved from the surface of the tumor by metalloproteases, allowing for escape from NK cell-mediated detection and elimination. Wucherpfennig and colleagues have described that the membrane proximal α3 domain remains on the cell surface and may be a potential target for immunotherapy across many cancers, including AML. However, since AML is heterogenous, we hypothesized that clinical success would require dual-targeting and selected CD33 as the ideal complementary targeting approach since CD33 is a validated marker expressed on greater than 80% of AML blasts. To this end, our lab has developed a Tri-specific Killer Engager (TriKE) capable of agonistically ligating theCD16 Fc receptor to CD33 found on AML along with IL-15 co-stimulation to further activate the NK cell response. Here, we demonstrate the efficacy of a multiplexed-gene edited induced pluripotent stem-cell (iPSC) derived NK cell (iNK) product that expresses a high-affinity, non-cleavable version of CD16 (hnCD16), a membrane-bound IL-15 fusion receptor, *CD38* knockout to enhance metabolic fitness, and a CAR against the α3 domain of MICA/B (α3 MICA/B) to drive a potent response against AML alone and when combined with anti-CD33 TriKE. In the initial study, the iNK cell backbone (iNK cells without the α3 MICA/B CAR) induced potent activity against the AML cell line HL60 and displayed further enhancement of activity with the addition of anti-CD33 TriKE (GTB-3650), representing combined effects of natural cytotoxicity and antibody-dependent cellular cytotoxicity. To show specificity for α3 MICA/B targeting, the AML cell line THP-1 was stained for the presence of MICA/B by flowcytometry using the 6D4 clone that recognizes the α1/2 domains of MICA/B or the7C6 clone that uniquely recognizes the α3 domain +/- proteolytic cleavage with trypsin. Significant expression of the α3 domain was observed on THP-1cells using the 7C6 antibody, which was highly expressed even after protease treatment. On the other hand, the α1/2 domains of MICA/B were detectable at lower levels using the 6D4 antibody and were undetectable after protease treatment. To assess a dual targeting approach, THP-1 cells were used as targets in live imaging functional assays under standard conditions (Effector:Target [E:T] 2:1). The iNK cell backbone had modest natural cytotoxicity at 3 hours that was significantly enhanced by the addition of α3 MICA/B CAR. The effect was further improved with more rapid killing kinetics when combined with the anti-CD33 TriKE. To evaluate the efficacy of dual-targeting in a more stringent physiologic manner, we mimicked "stress" conditions by minimizing the E:T ratio to 0.25:1. While all effector conditions induced immediate killing in 4 hours at the low E:T ratio, sustained tumor control was only observed with dual-antigen targeting. Studies with primary AML targets, +/- preincubation of decitabine and all-trans retinoic acid, known to upregulate NKG2D ligands in AML, are in progress and will be discussed. In summary, dual-targeting strategies using off-the-shelf CAR NK cells targeting α3 MICA/B in combination with antigen-specific TriKE targeting CD33 represent an ideal clinical strategy to enhance efficacy and durability of treatment in advanced AML.

Off-the-shelf Multiplex-Engineered Cell Therapy Platform

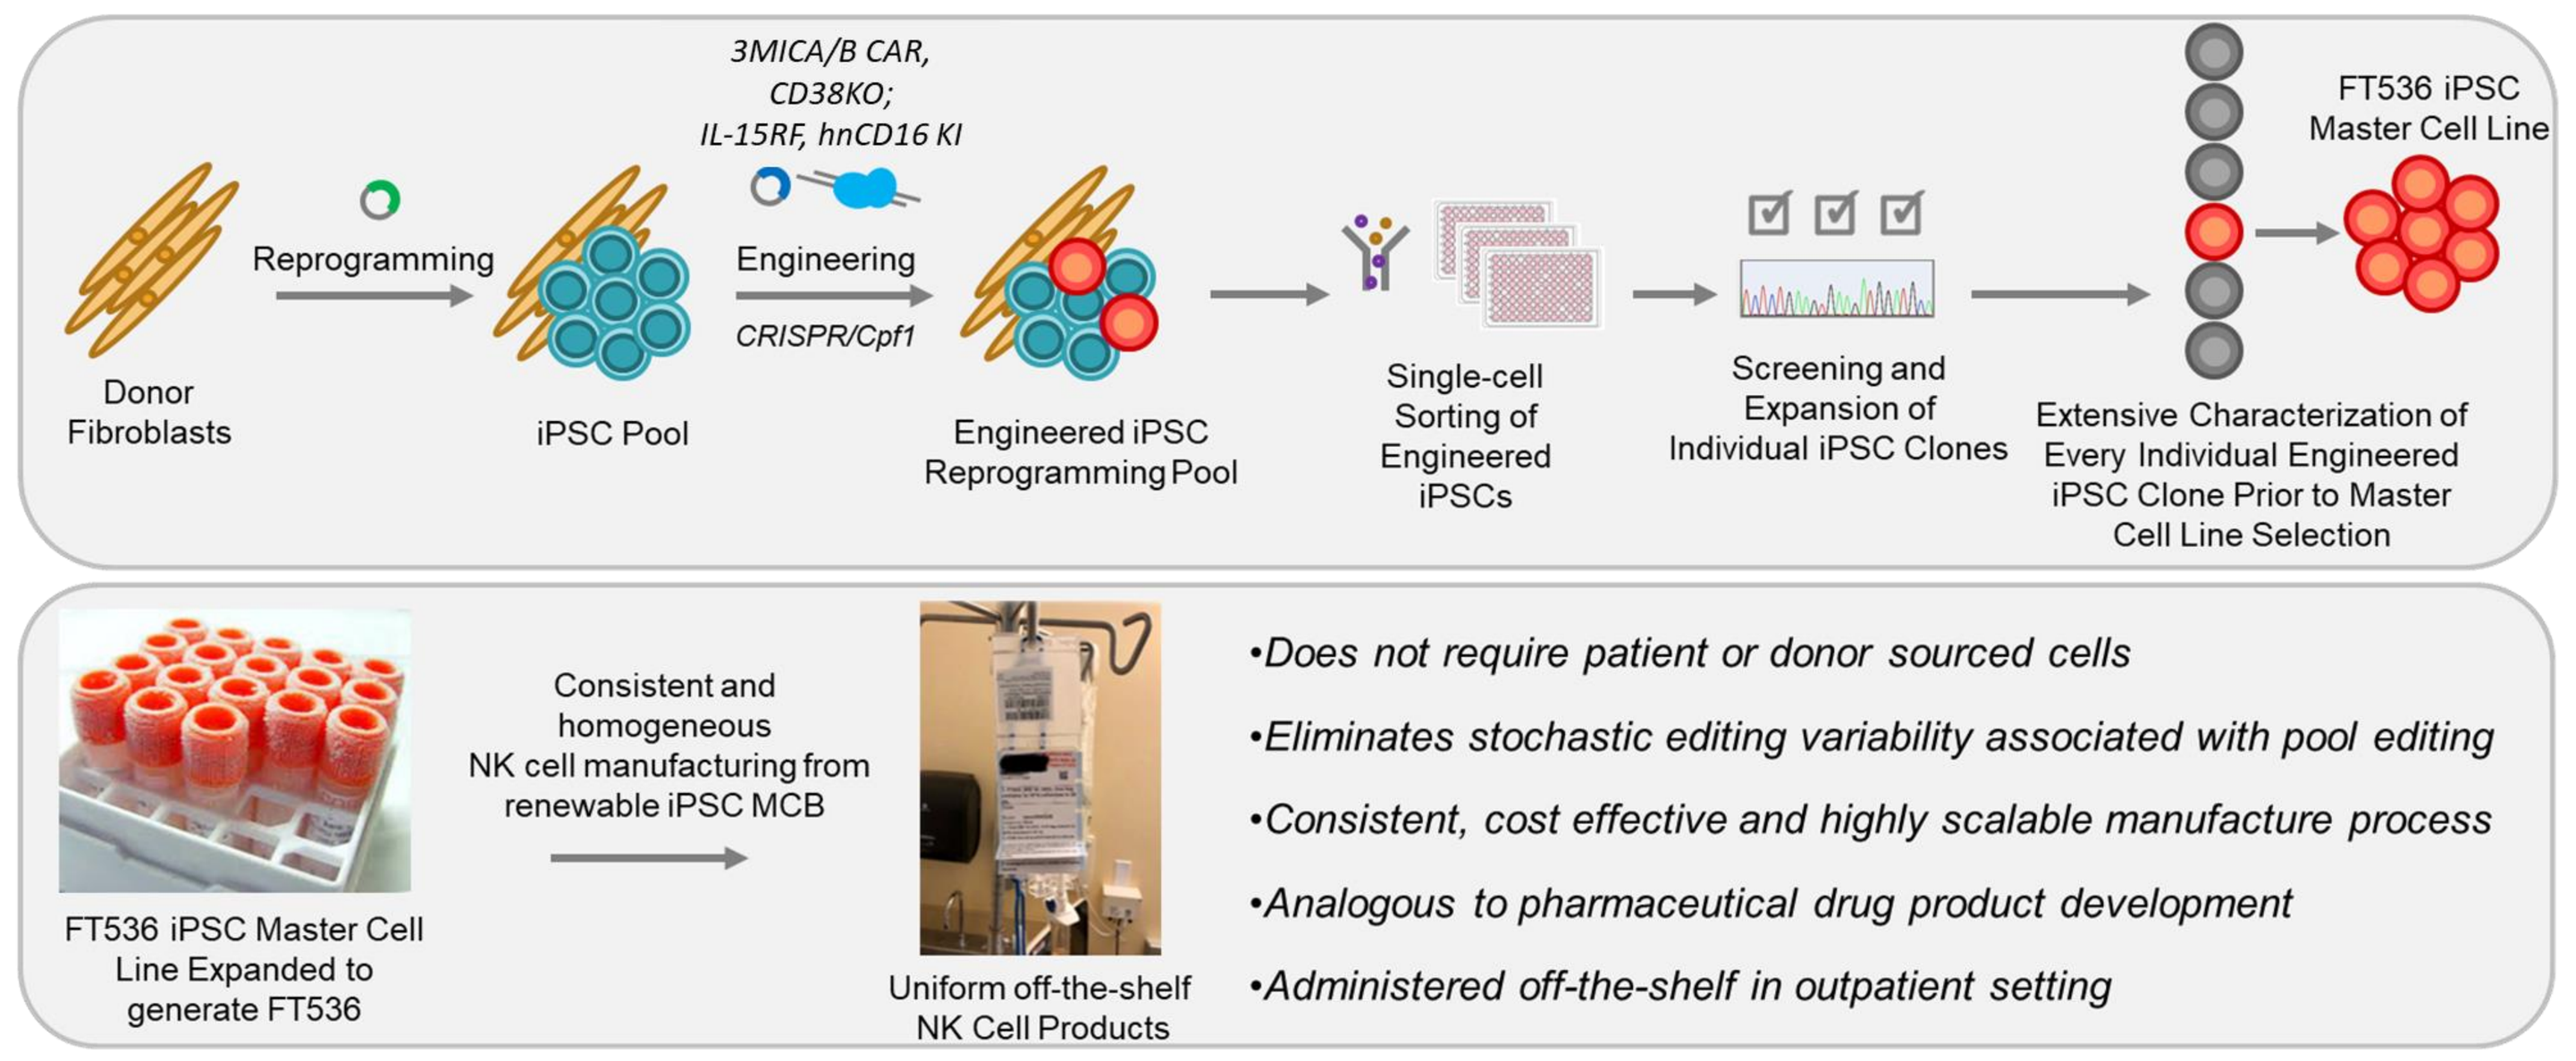

Donor Fibroblasts
Reprogramming
iPSC Pool
3MICA/B CAR, CD38KO; IL-15RF, hnCD16 KI
Engineering
CRISPR/Cpf1
Engineered iPSC Reprogramming Pool
Single-cell Sorting of Engineered iPSCs
Screening and Expansion of Individual iPSC Clones
Extensive Characterization of Every Individual Engineered iPSC Clone Prior to Master Cell Line Selection
FT536 iPSC Master Cell Line
FT536 iPSC Master Cell Line Expanded to generate FT536
Consistent and homogeneous NK cell manufacturing from renewable iPSC MCB
Uniform off-the-shelf NK Cell Products
•Does not require patient or donor sourced cells
•Eliminates stochastic editing variability associated with pool editing
•Consistent, cost effective and highly scalable manufacture process
•Analogous to pharmaceutical drug product development
•Administered off-the-shelf in outpatient setting

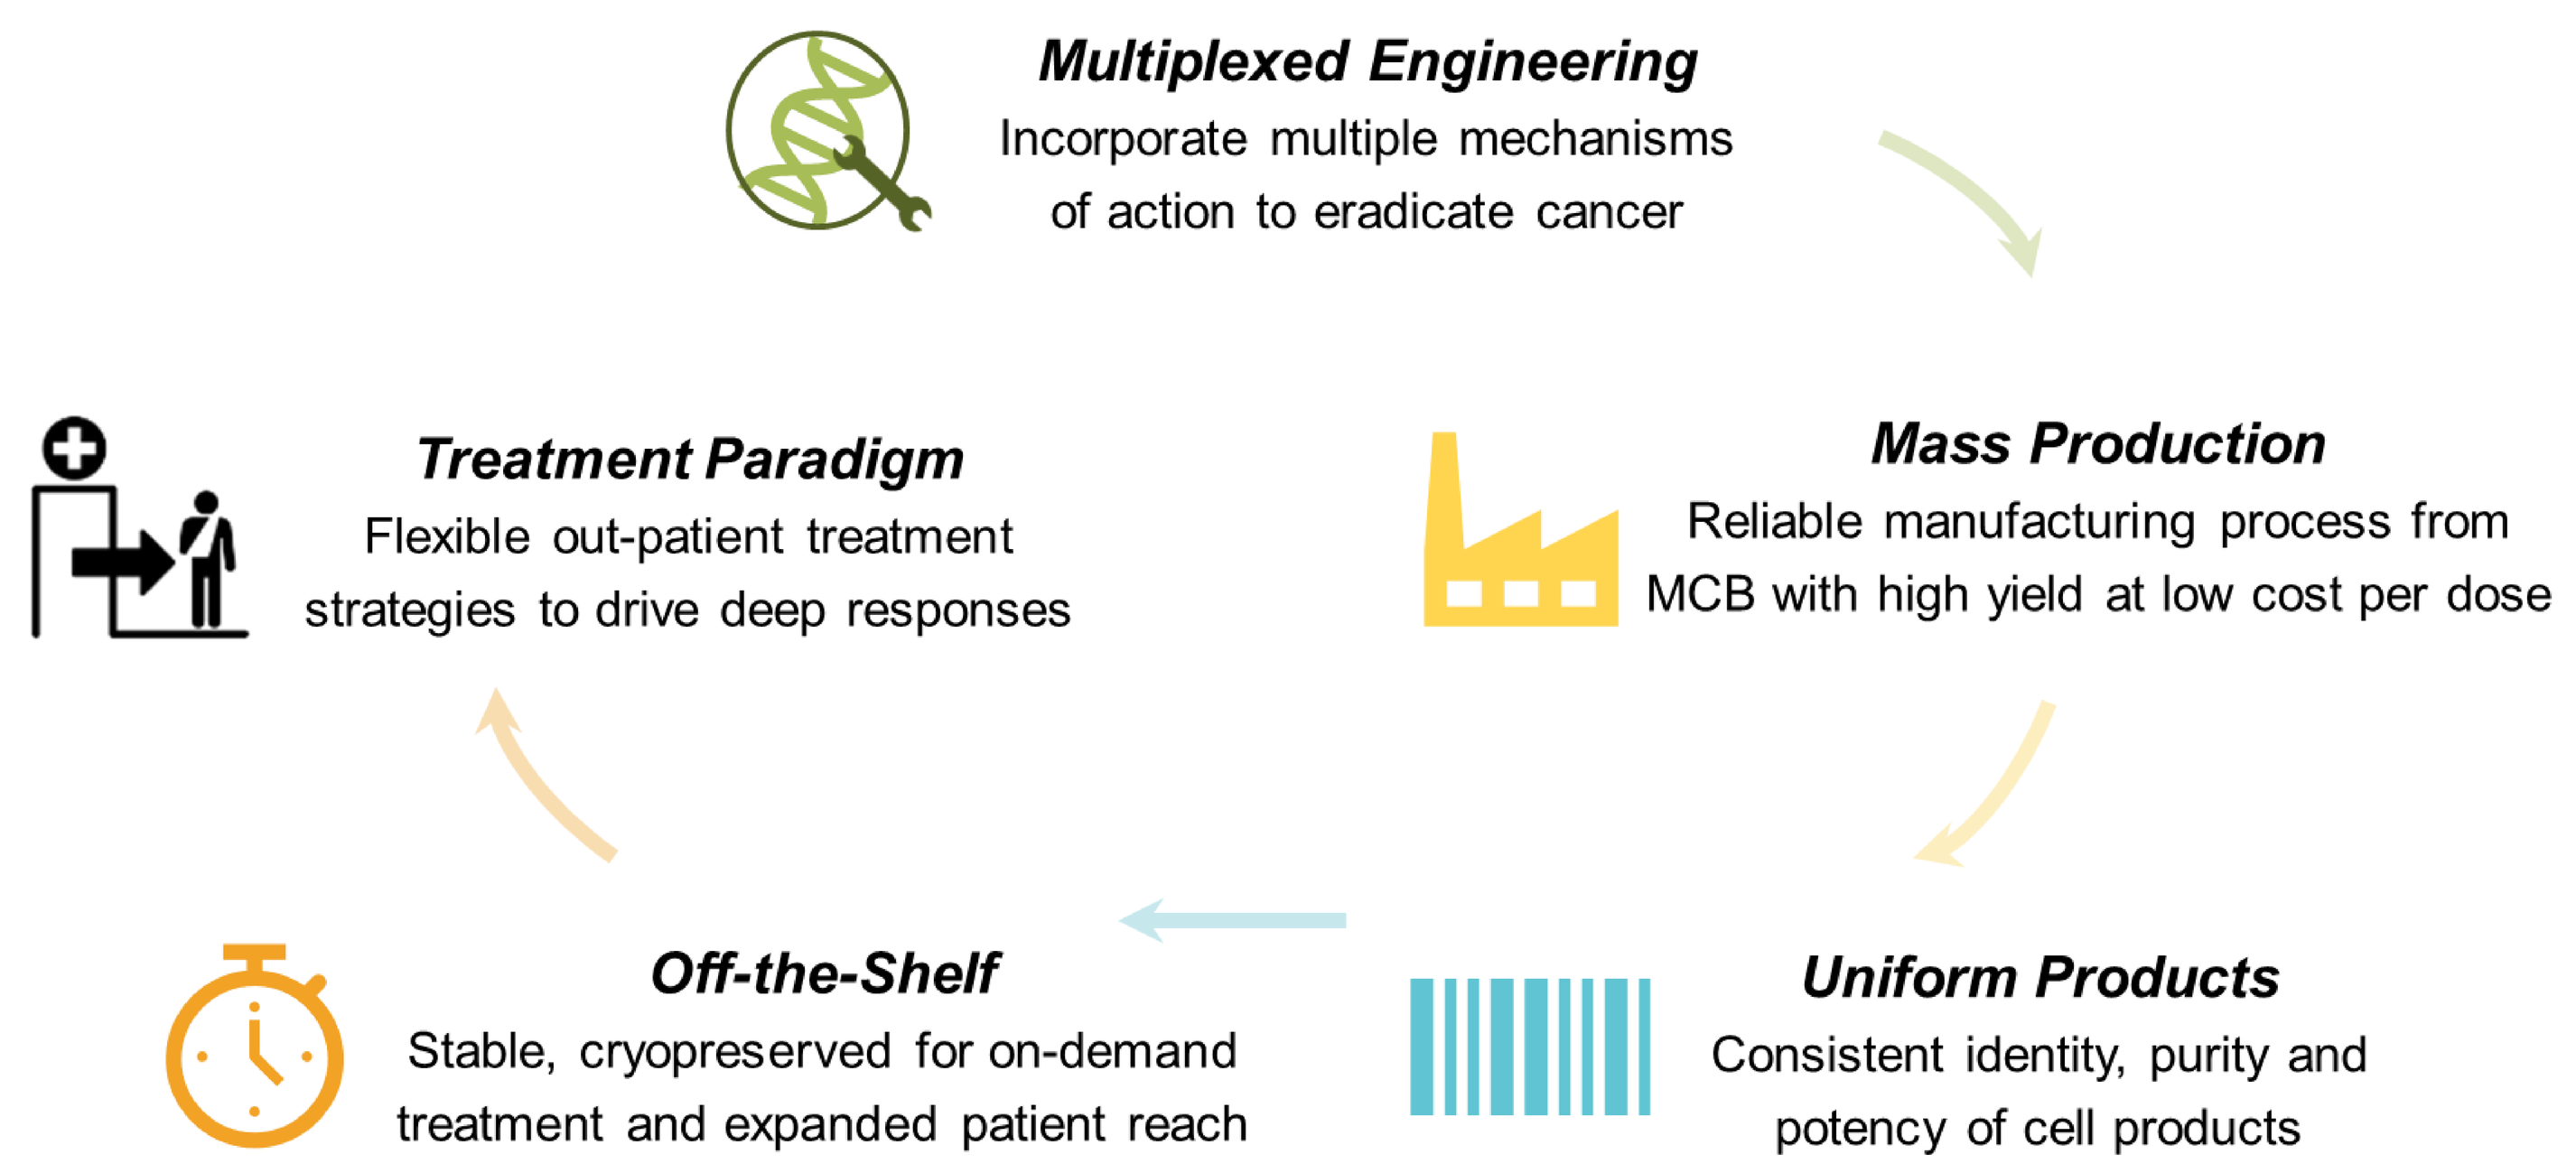

Multiplexed Engineering
Incorporate multiple mechanisms of action to eradicate cancer
Mass Production
Reliable manufacturing process from MCB with high yield at low cost per dose
Uniform Products
Consistent identity, purity and potency of cell products
Off-the-Shelf
Stable, cryopreserved for on-demand treatment and expanded patient reach
Treatment Paradigm
Flexible out-patient treatment strategies to drive deep responses

FT536: Off-the-Shelf Multi-Antigen-Targeting NK Cell Immunotherapy

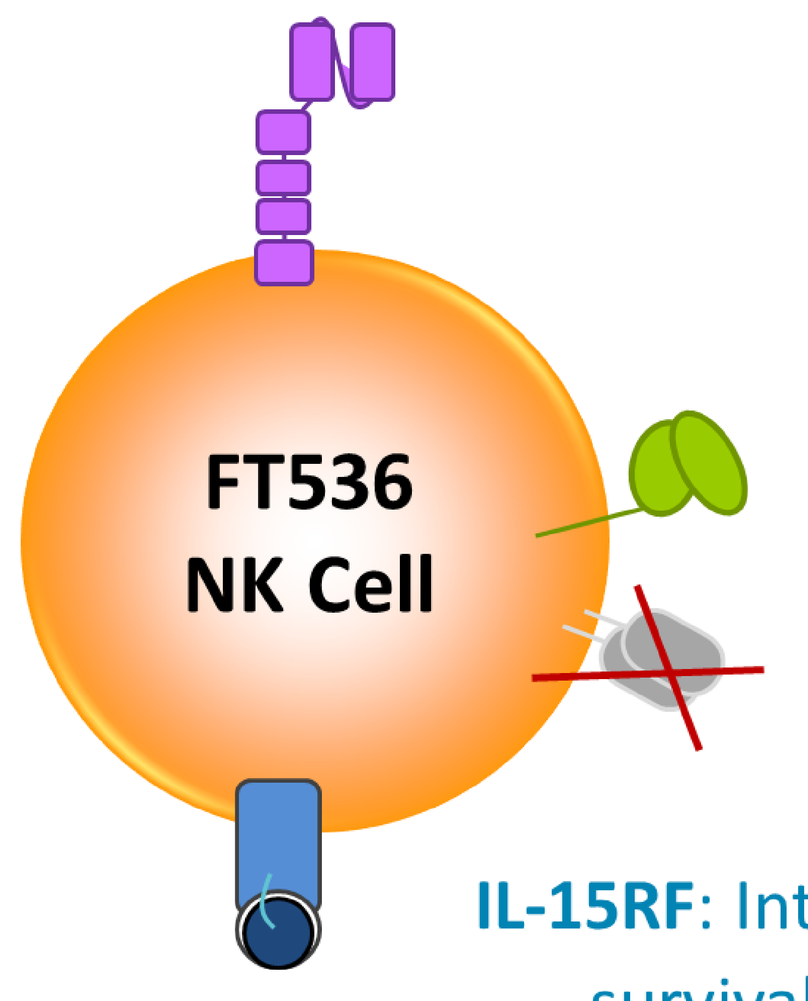

FT536 NK Cell

3MICA/B CAR: Novel Chimeric antigen receptor targeting the membrane proximal α3 domain of MICA/B. *Ferrari de Andrade et al. Science 30 Mar 2018. DOI: 10.1126/science.aao0505*

hnCD16: High-affinity 158V, non-cleavable CD16 Fc receptor modified to augment ADCC. *Zhu et al. Blood 6 Feb 2020. DOI: 10.1182/blood.2019000621*

CD38 KO: Deletion of CD38 to eliminate anti-CD38 antibody mediated NK cell fratricide. *Woan et al. Cell Stem Cell Dec 2021. DOI: 10.1016/j.stem202108013*

IL-15RF: Interleukin-15 receptor fusion; a potent cytokine complex that promotes survival and persistence and reduces the dependency for exogenous cytokine support. *Woan et al. Cell Stem Cell Dec 2021. DOI: 10.1016/j.stem202108013*

FT536: MICA/B CAR Proposed Mechanism of Action

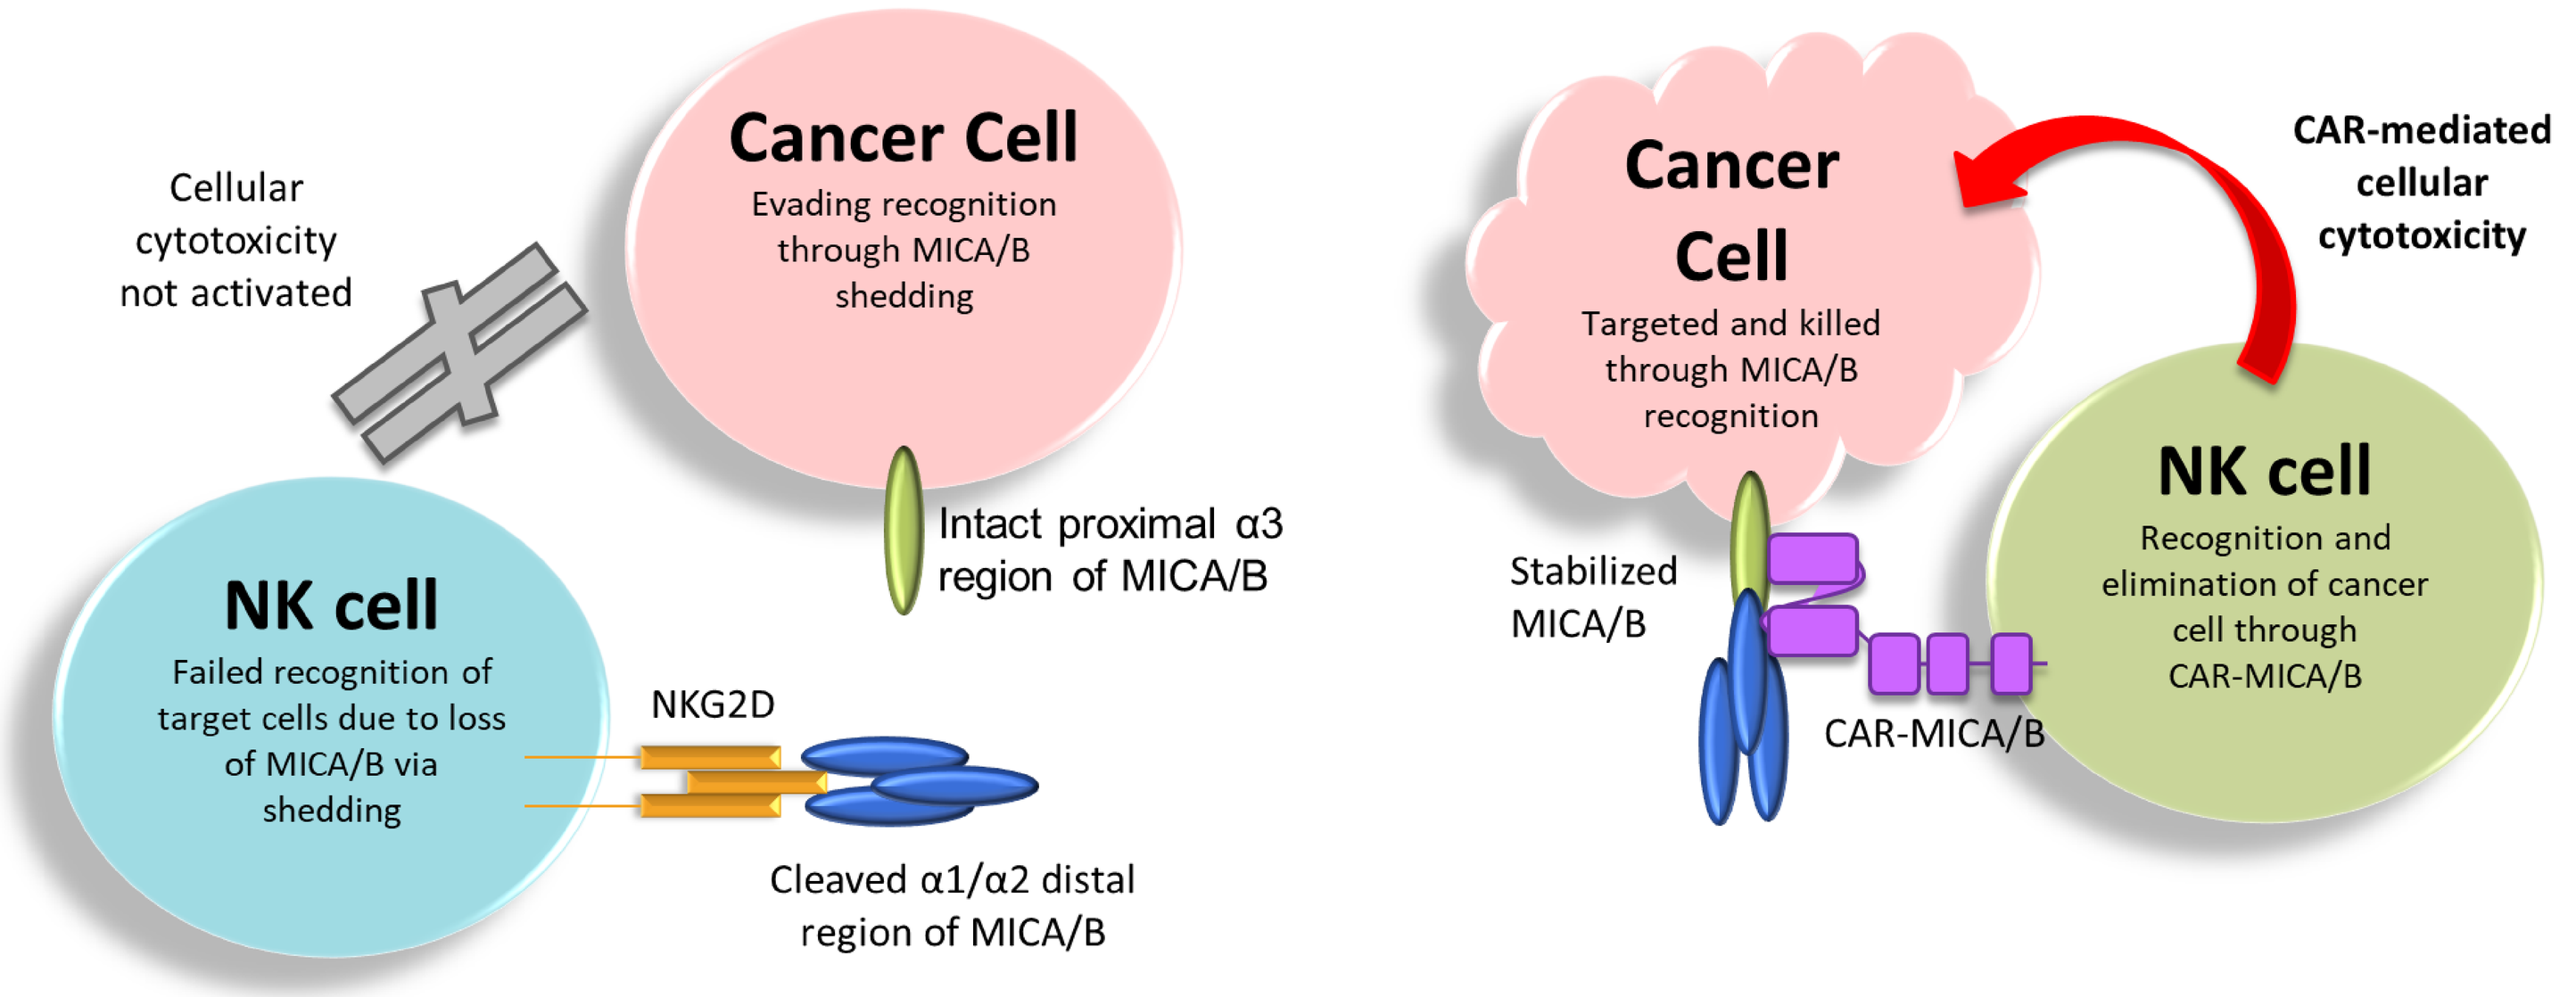

Cellular cytotoxicity not activated
Cancer Cell
Evading recognition through MICA/B shedding
Intact proximal α3 region of MICA/B
NK cell
Failed recognition of target cells due to loss of MICA/B via shedding
NKG2D
Cleaved α1/α2 distal region of MICA/B
Cancer Cell
Targeted and killed through MICA/B recognition
CAR-mediated cellular cytotoxicity
Stabilized MICA/B
CAR-MICA/B
NK cell
Recognition and elimination of cancer cell through CAR-MICA/B

Cancer cells evade immune recognition by proteolytic shedding of MICA/B caused by cleavage within the α3 membrane proximal domain. The released soluble α1/α2 MICA/B peptides have been shown to interfere with NKG2D signaling and further inhibit cancer cell recognition and elimination. An α3 domain-targeting CAR hypothetically avoids soluble α1/α2 MICA/B peptide inhibition and 're-cloaks' evading tumor cells through stabilizing and recognizing surface MICA/B

FT536, a uniformly edited multiplexed engineered CAR iNK cell, demonstrates MICA/B tumor specific activation and cytotoxicity

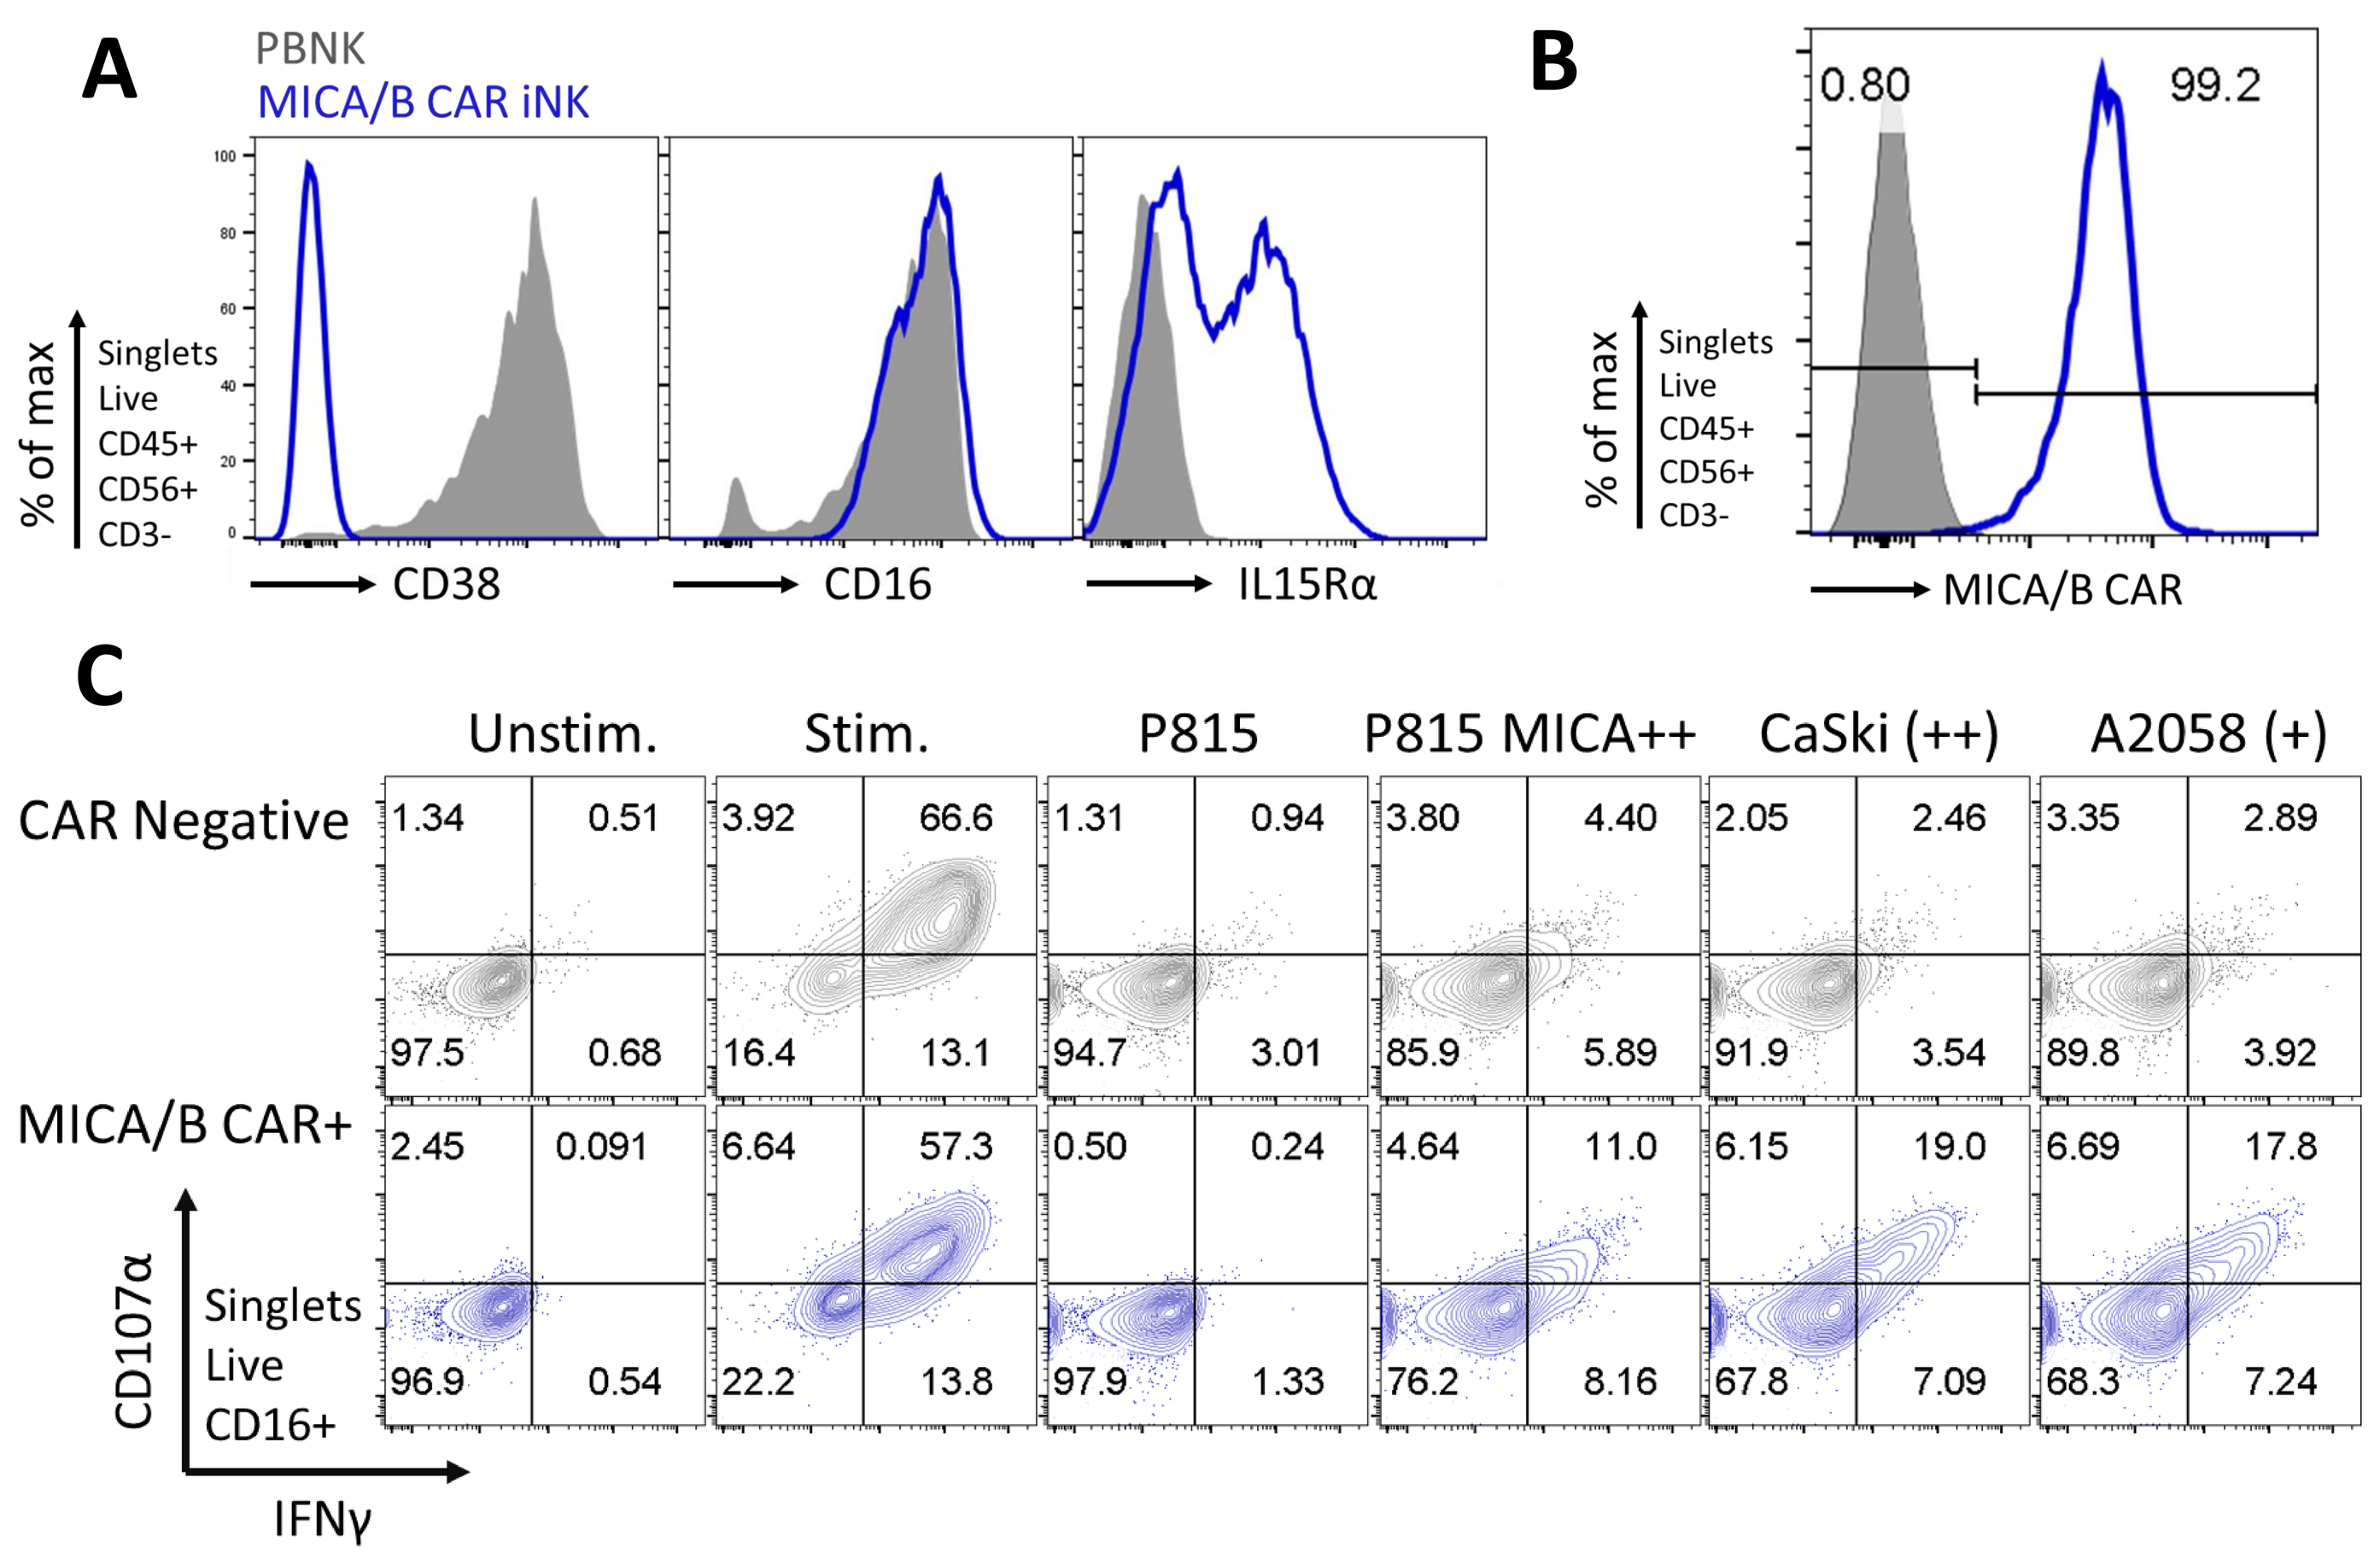

A
PBNK
MICA/B CAR iNK
% of max
Singlets Live CD45+ CD56+ CD3-
CD38
CD16
IL15Rα
B
0.80
99.2
MICA/B CAR
C
Unstim.
Stim.
P815
P815 MICA++
CaSki (++)
A2058 (+)
CAR Negative
MICA/B CAR+
CD107α
Singlets Live CD16+
IFNγ

Figure 1. Multi-edited iPSC derived MICA/B CAR iNK cells demonstrate pan tumor functionality. Directed differentiation of multiplex engineered iPSCs to CD45+/CD56+/CD3- iNK cells show expression of IL15Rα fusion, hnCD16 and a lack of surface CD38 expression **(A)**. CAR-MICA/B iNK cells demonstrate high surface CAR expression and purity **(B)**. MICA/B CAR positive iNK cells show antigen specific activation following incubation at 2:1 effector/target ratio with human MICA over expressing murine mastocytoma P815 cells (high++ MICA expressors), CaSki (cervical epidermoid carcinoma; high++ MICA endogenous expressors) and A2058 (melanoma; medium+ MICA endogenous expressors) as measured by IFNγ production and CD107α+ degranulation **(C)**.

Acute Myelogenous Leukemia cell lines have surface expressed MICB and CD33

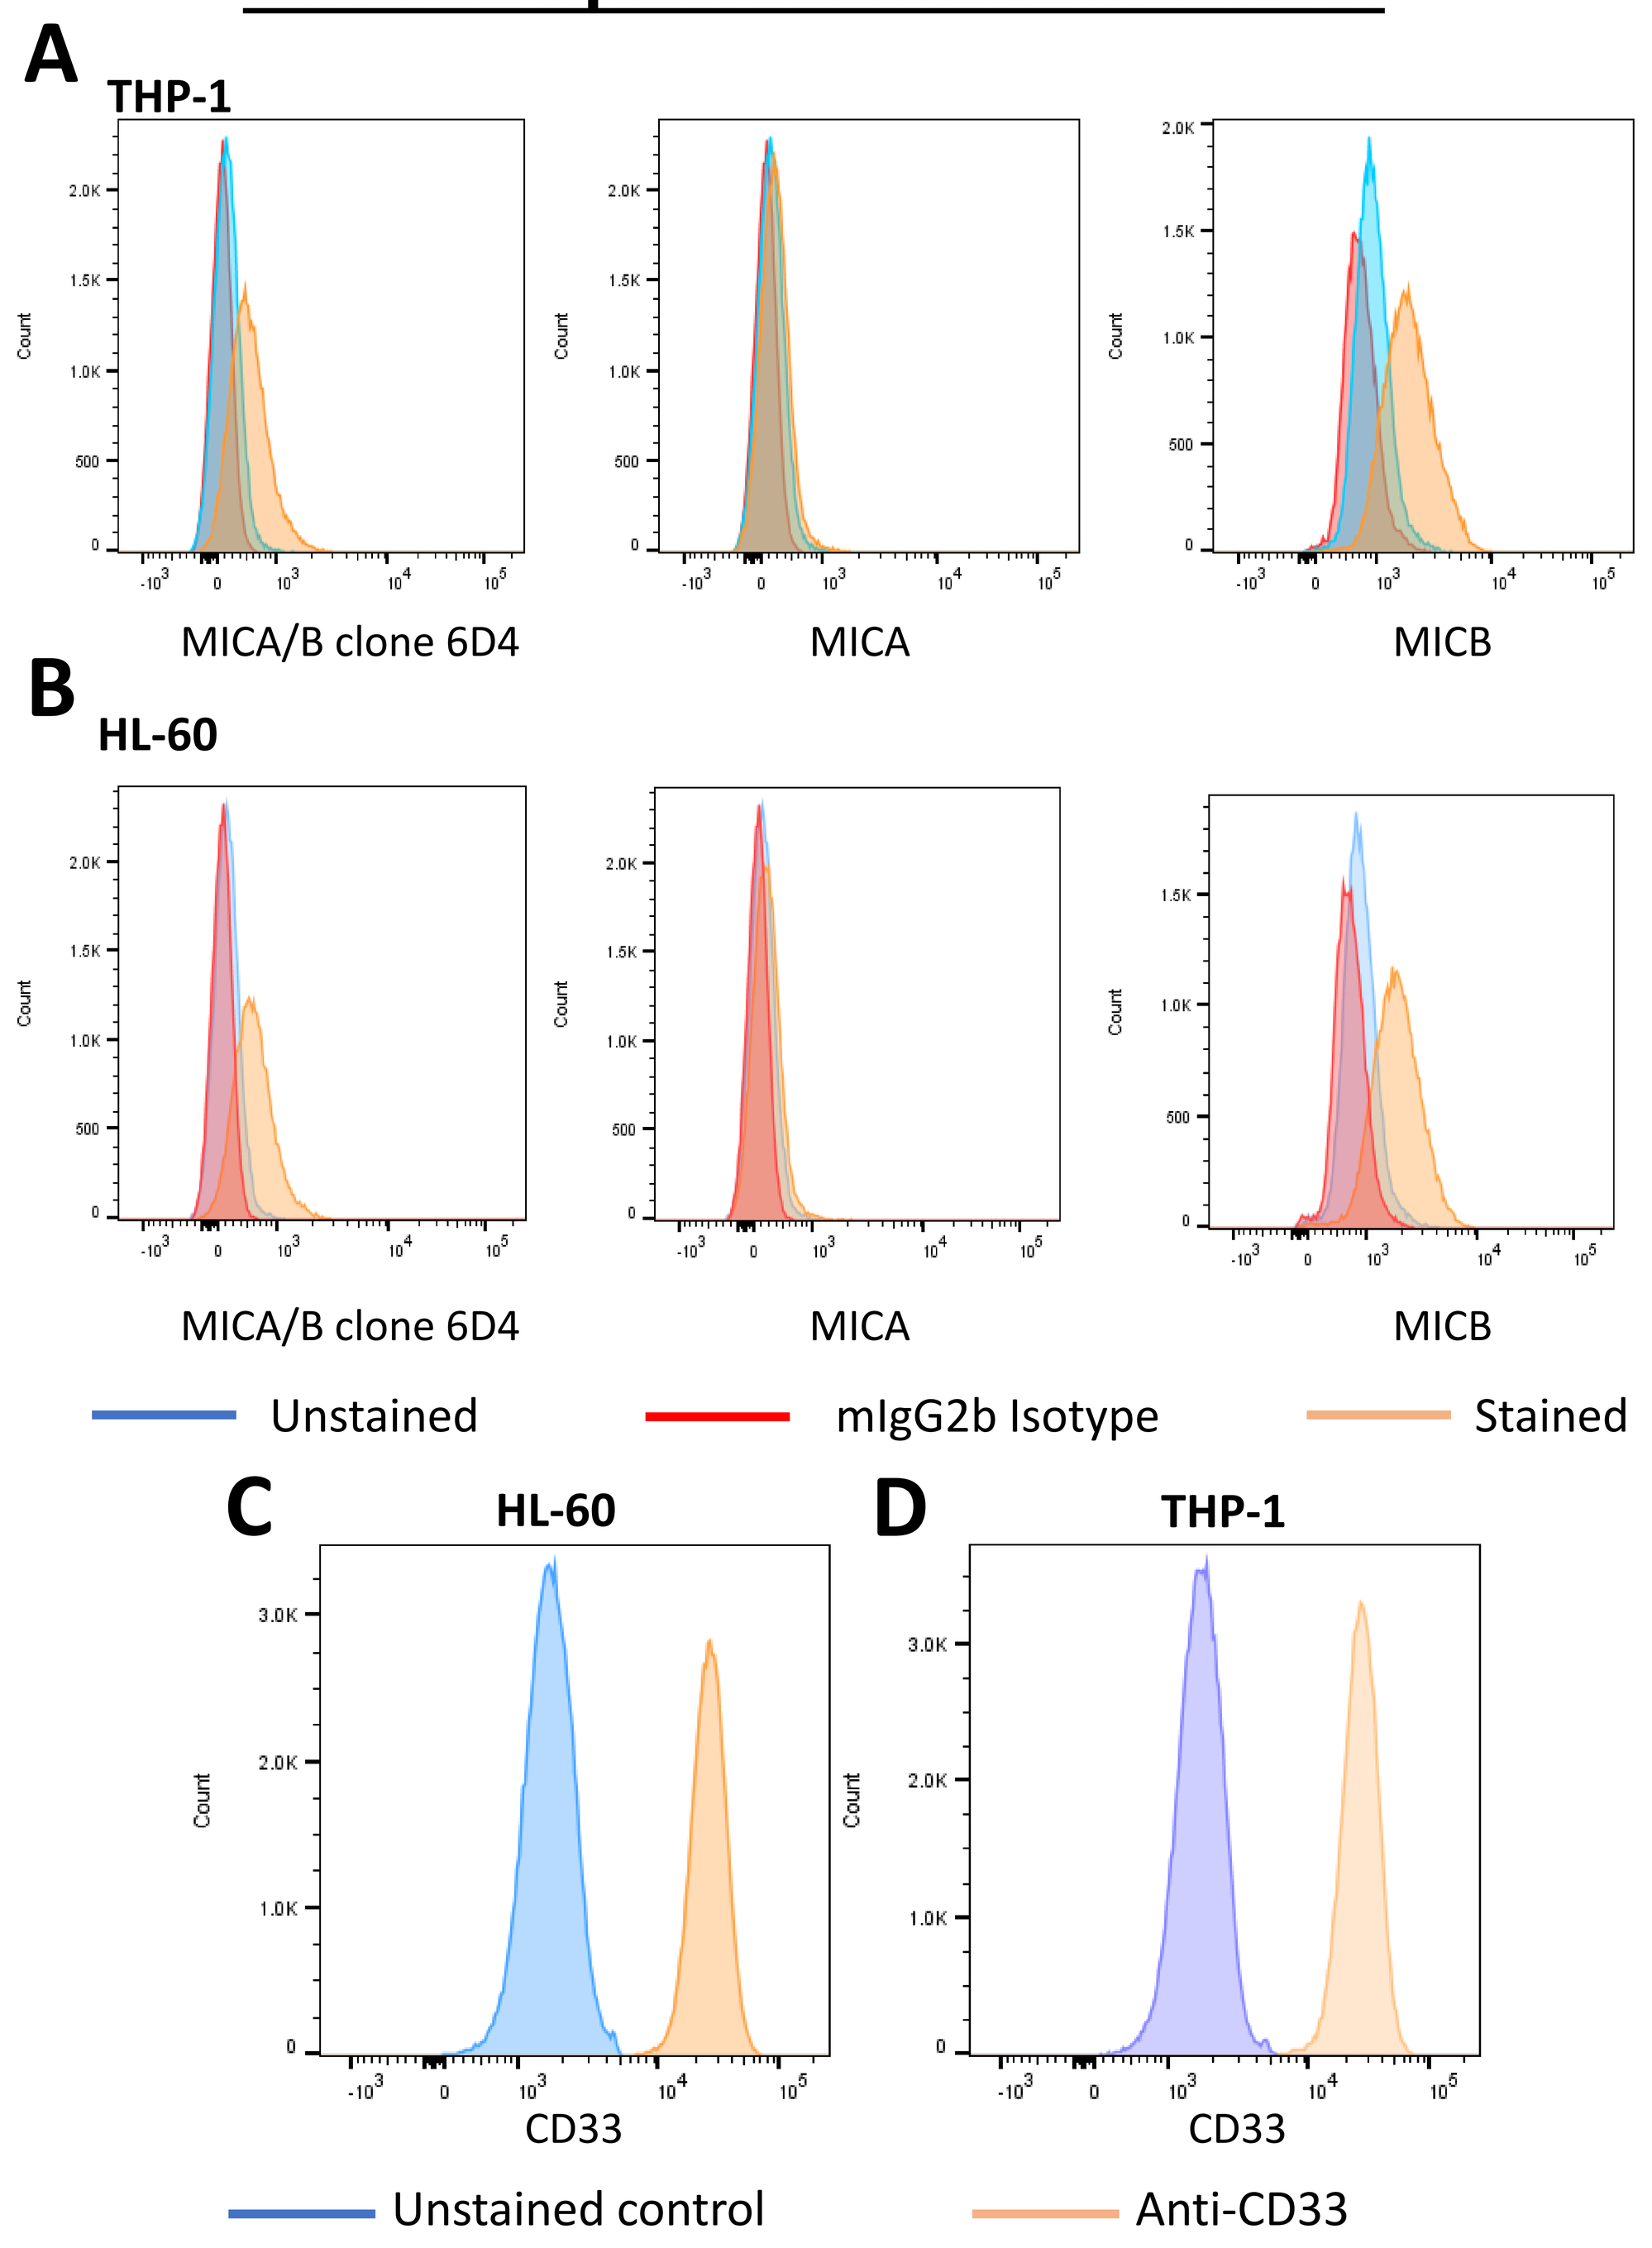

A
THP-1
MICA/B clone 6D4
MICA
MICB
B
HL-60
MICA/B clone 6D4
MICA
MICB
Unstained
mIgG2b Isotype
Stained
C
HL-60
CD33
D
THP-1
CD33
Unstained control
Anti-CD33

Figure 2: MICB but not MICA is expressed on the AML cell lines THP-1 and HL-60. THP-1 **(A)** and HL-60 **(B)** cell lines were stained with a commercial antibody that recognizes a common epitope of the non-cleaved α1 and α2 domains of MICA/B or with antibodies that specifically bind MICA or MICB. THP-1 **(C)** and HL-60 **(D)** were also stained for CD33.

MICA/B CAR binds the α3 domain of MICA/B

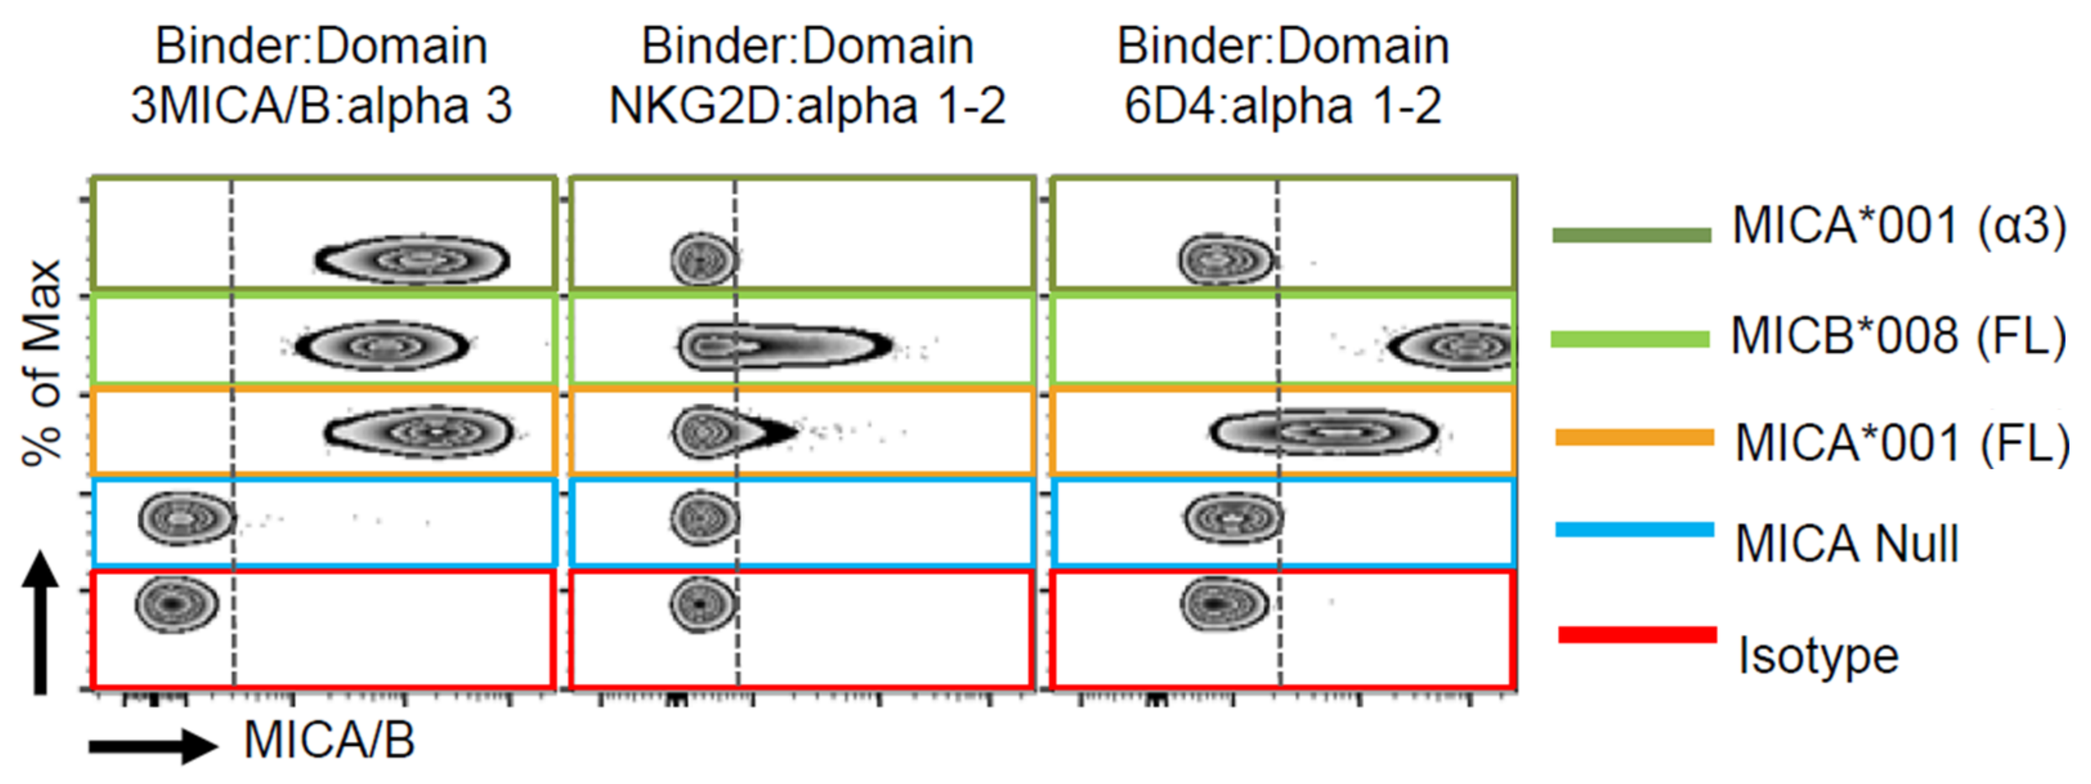

Binder:Domain 3MICA/B:alpha 3
Binder:Domain NKG2D:alpha 1-2
Binder:Domain 6D4:alpha 1-2
% of Max
MICA/B
MICA*001 (α3)
MICB*008 (FL)
MICA*001 (FL)
MICA Null
Isotype

Figure 3: MICA/B CAR binds the α3 domain of MICA/B. MICA and MICB binding specificity were assessed using B16/F10 murine melanoma cells transduced with either full length (FL) MICA*001, MICB*008 or truncated MICA*001 α3 domain proteins.

FT536, uniquely targeting α3 domain of MICA/B, elicits potent ADCC utilizing a CD33 targeted TriKE (GTB-3650)

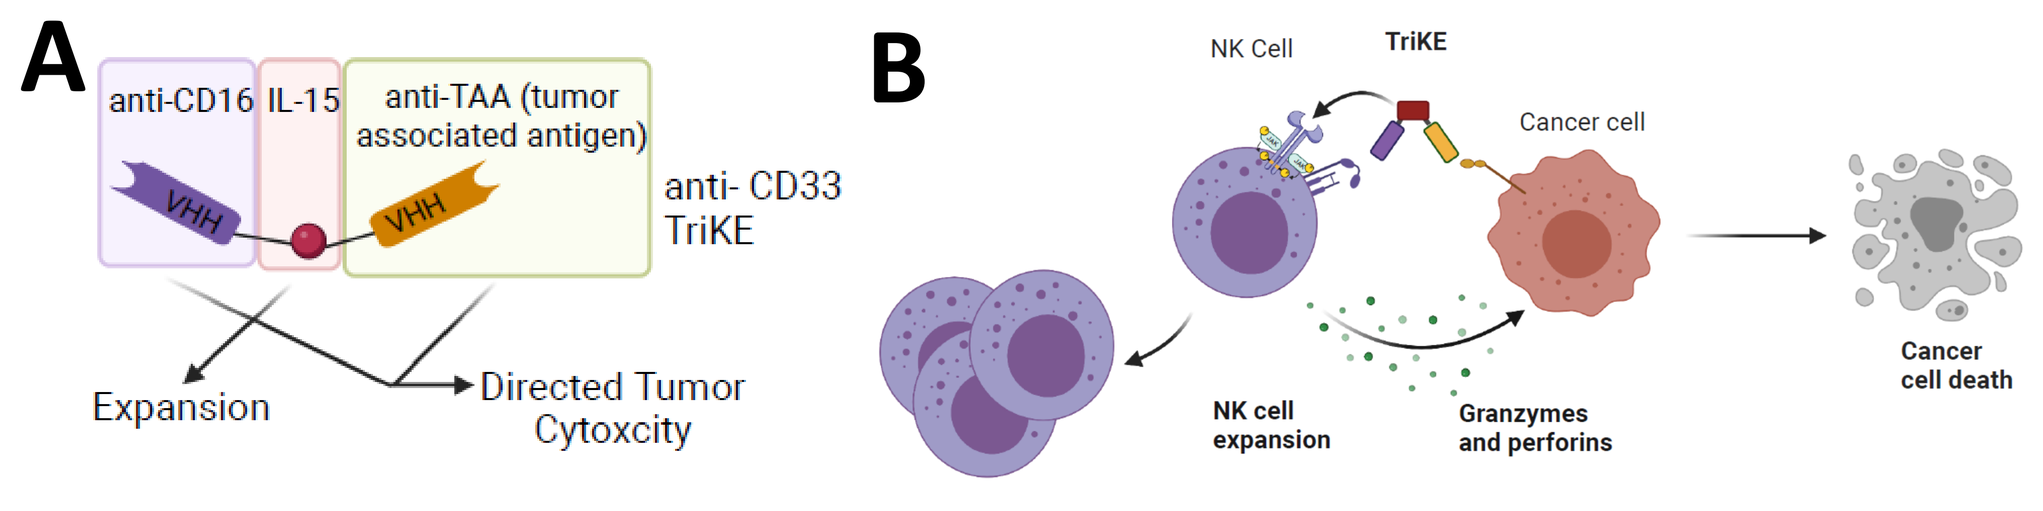

A
anti-CD16
IL-15
anti-TAA (tumor associated antigen)
VHH
VHH
anti- CD33 TriKE
Expansion
Directed Tumor Cytoxcity
B
NK Cell
TriKE
Cancer cell
NK cell expansion
Granzymes and perforins
Cancer cell death

Figure 4: A multi-edited iNK expressing a CAR against the α3 domain of MICA/B can also mediate ADCC utilizing a CD33-targeted TriKE. A CD33 targeted TriKE molecule containing a camelid anti-CD16 component **(A)** can bind CD33 on an AML target and hnCD16 on the iNK inducing NK cell degranulation and cytokine production resulting in the killing of the target cell **(B)**.

FT536 in combination with a CD33 TriKE (GTB-3650) enhances tumor killing of AML

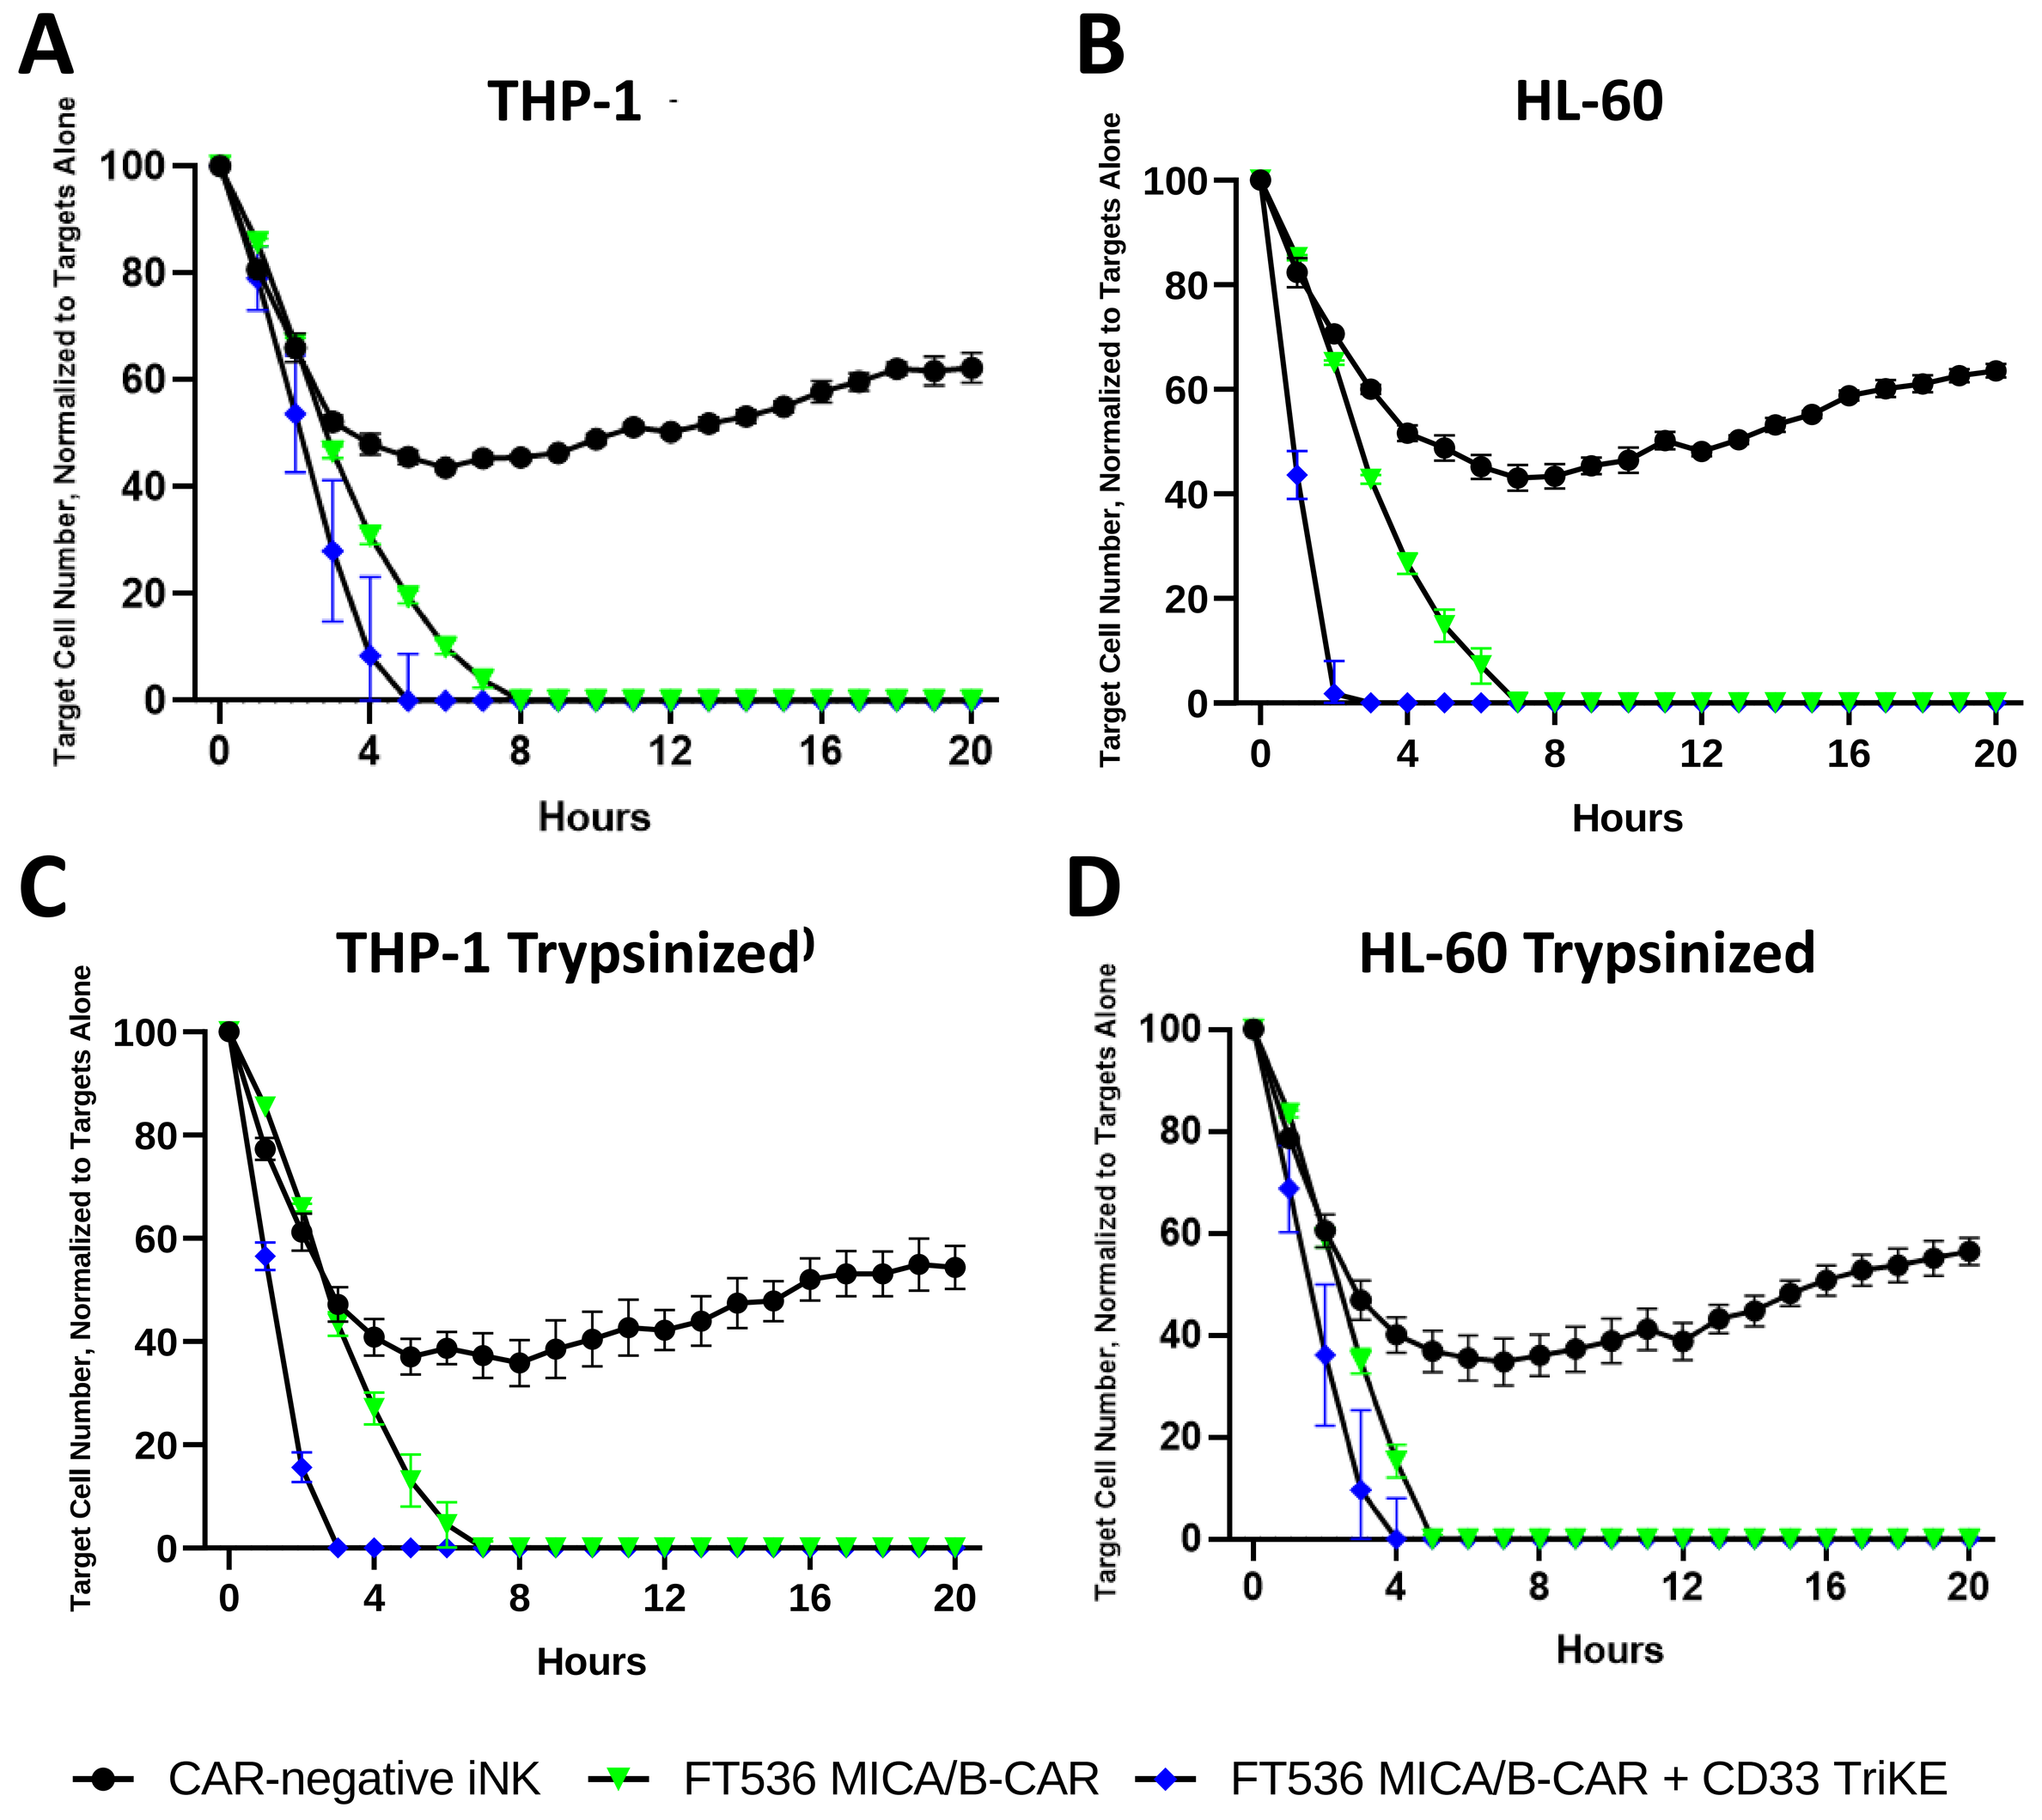

A
THP-1
B
HL-60
C
THP-1 Trypsinized
D
HL-60 Trypsinized
Target Cell Number, Normalized to Targets Alone
Hours
CAR-negative iNK
FT536 MICA/B-CAR
FT536 MICA/B-CAR + CD33 TriKE

Figure 5: FT536 in combination with a CD33 TriKE (GTB-3650) enhances tumor killing compared to FT536 alone. FT536 and a CAR-negative iNK were cultured with the AML lines THP-1**(A)** or HL-60 alone or in combination with GTB-3650 and target cell killing was evaluated by IncuCyte. Trypsinized THP-1 **(C)** or HL-60 **(D)** were also used as targets to evaluate killing against targets with cleaved MICA/B. Effectors were added to targets at an E:T ratio of 2:1.

Primary AML bone marrow aspirates from patients at diagnosis only express the α3 domain of MICA/B

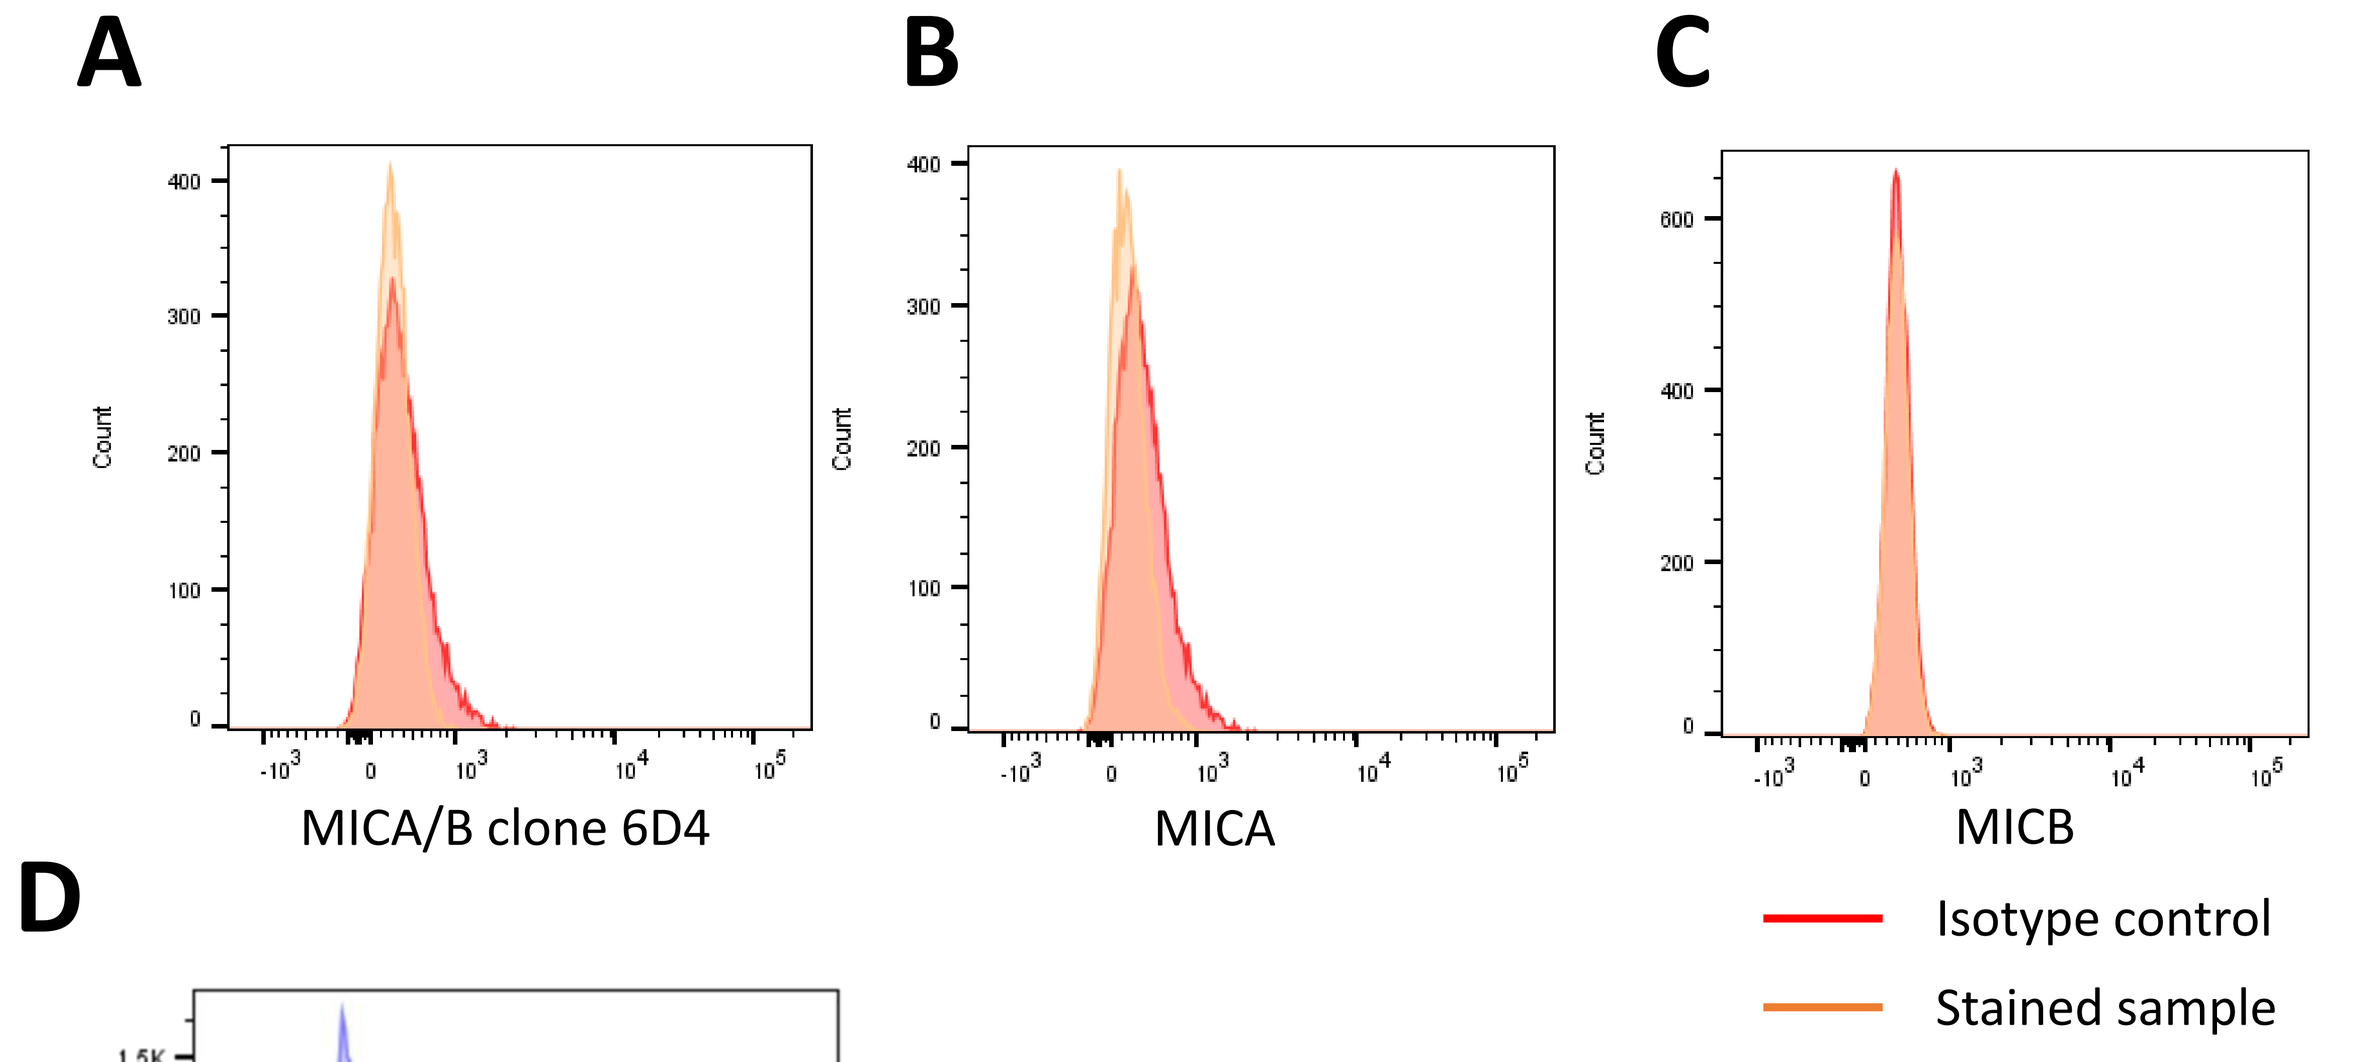

A
MICA/B clone 6D4
B
MICA
C
MICB
Isotype control
Stained sample

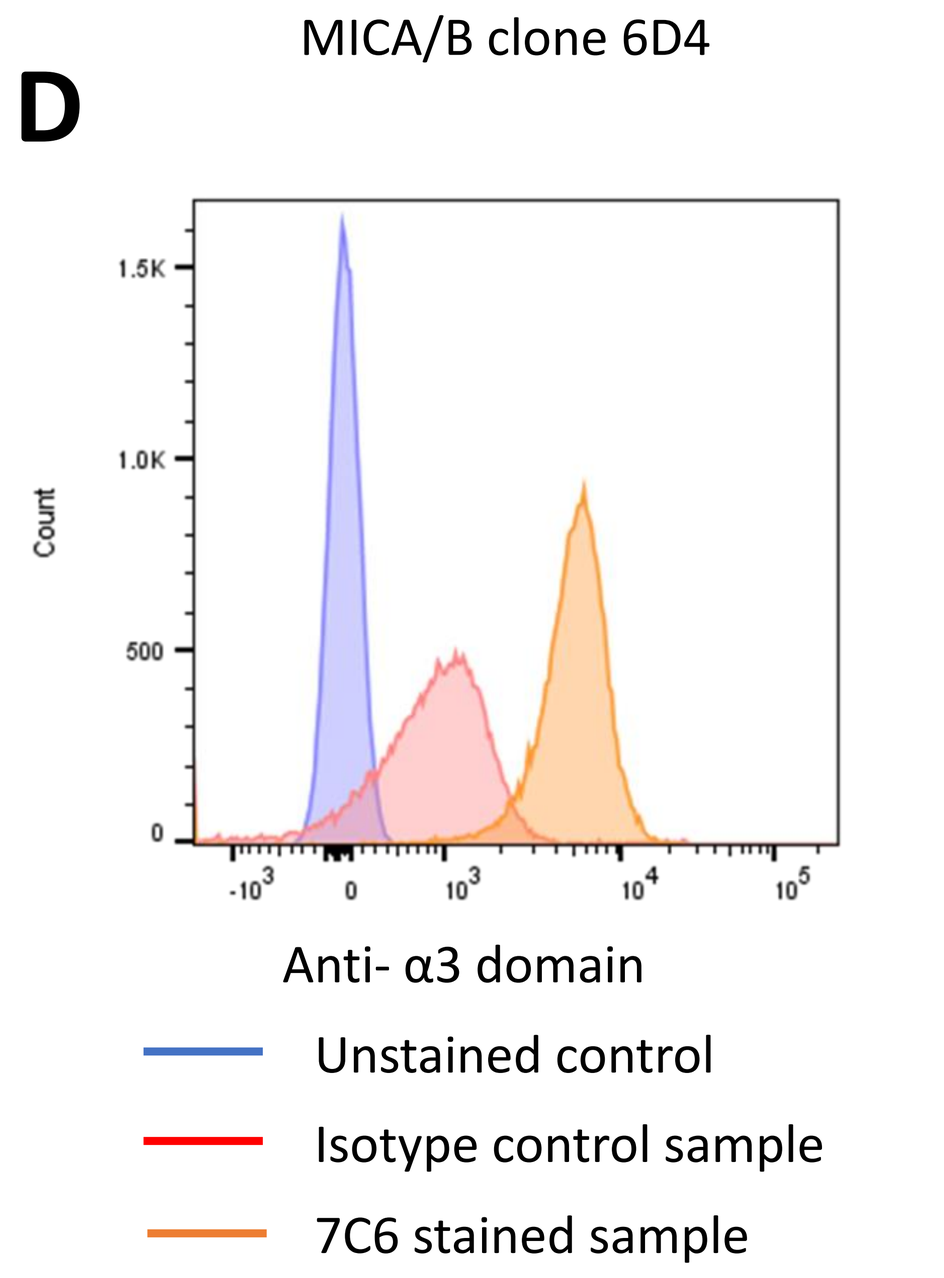

D
Anti- α3 domain
Unstained control
Isotype control sample
7C6 stained sample

Figure 6: Primary AML bone marrow aspirates from patients at diagnosis only express the α3 domain of MICA/B. Bone marrow aspirates were stained for MICA and MICB from an AML patients **(A-C)** using conventional antibodies. A sample from the patient were also stained for the α3 domain of MICA/B, using the antibody clone, 7C6 **(D)**. Figures shown are representative of 4 different patients tested.

Conclusions

- FT536 is a multiplexed-engineered iNK cells expressing a novel MICA/B α3 domain CAR with specific activity against a broad range of tumor cells.
- AML cell lines express MICA/B and CD33, and are killed by FT536 which is further enhanced in combination with GTB-3650.
- Primary AML tumors uniquely express the α3 domain of MICA/B and CD33 suggesting that the combination could be a unique dual targeted therapeutic approach to increase efficacy in AML.