FT819 Drives B cell Compartment Remodeling of Patients with Systemic Lupus Erythematosus Without Conditioning Chemotherapy

Trever Greene[1], Jennifer Medlin[2], Shirley Pang[3], Matthew Lunning [2], Jeffrey S. Miller[4], Kelsea Hubka[1], Seth Peng[1], Cara Bickers[1], Sandeep Kothapally Hanok[1], Bahram Valamehr[1], Vaneet Sandhu[1], Natalie J. Shiff[1], Lilly Wong [1]

[1]*Fate Therapeutics, Inc., San Diego, CA, USA,* [2]*University of Nebraska Medical Center, Omaha, United States of America ,* [3]*Providence Medical Foundation, Fullerton, United States of America* [4]*University of Minnesota, Minneapolis, United States of America*

Background

FT819 Provides Unparalleled CAR T-cell Accessibility to Patients

A

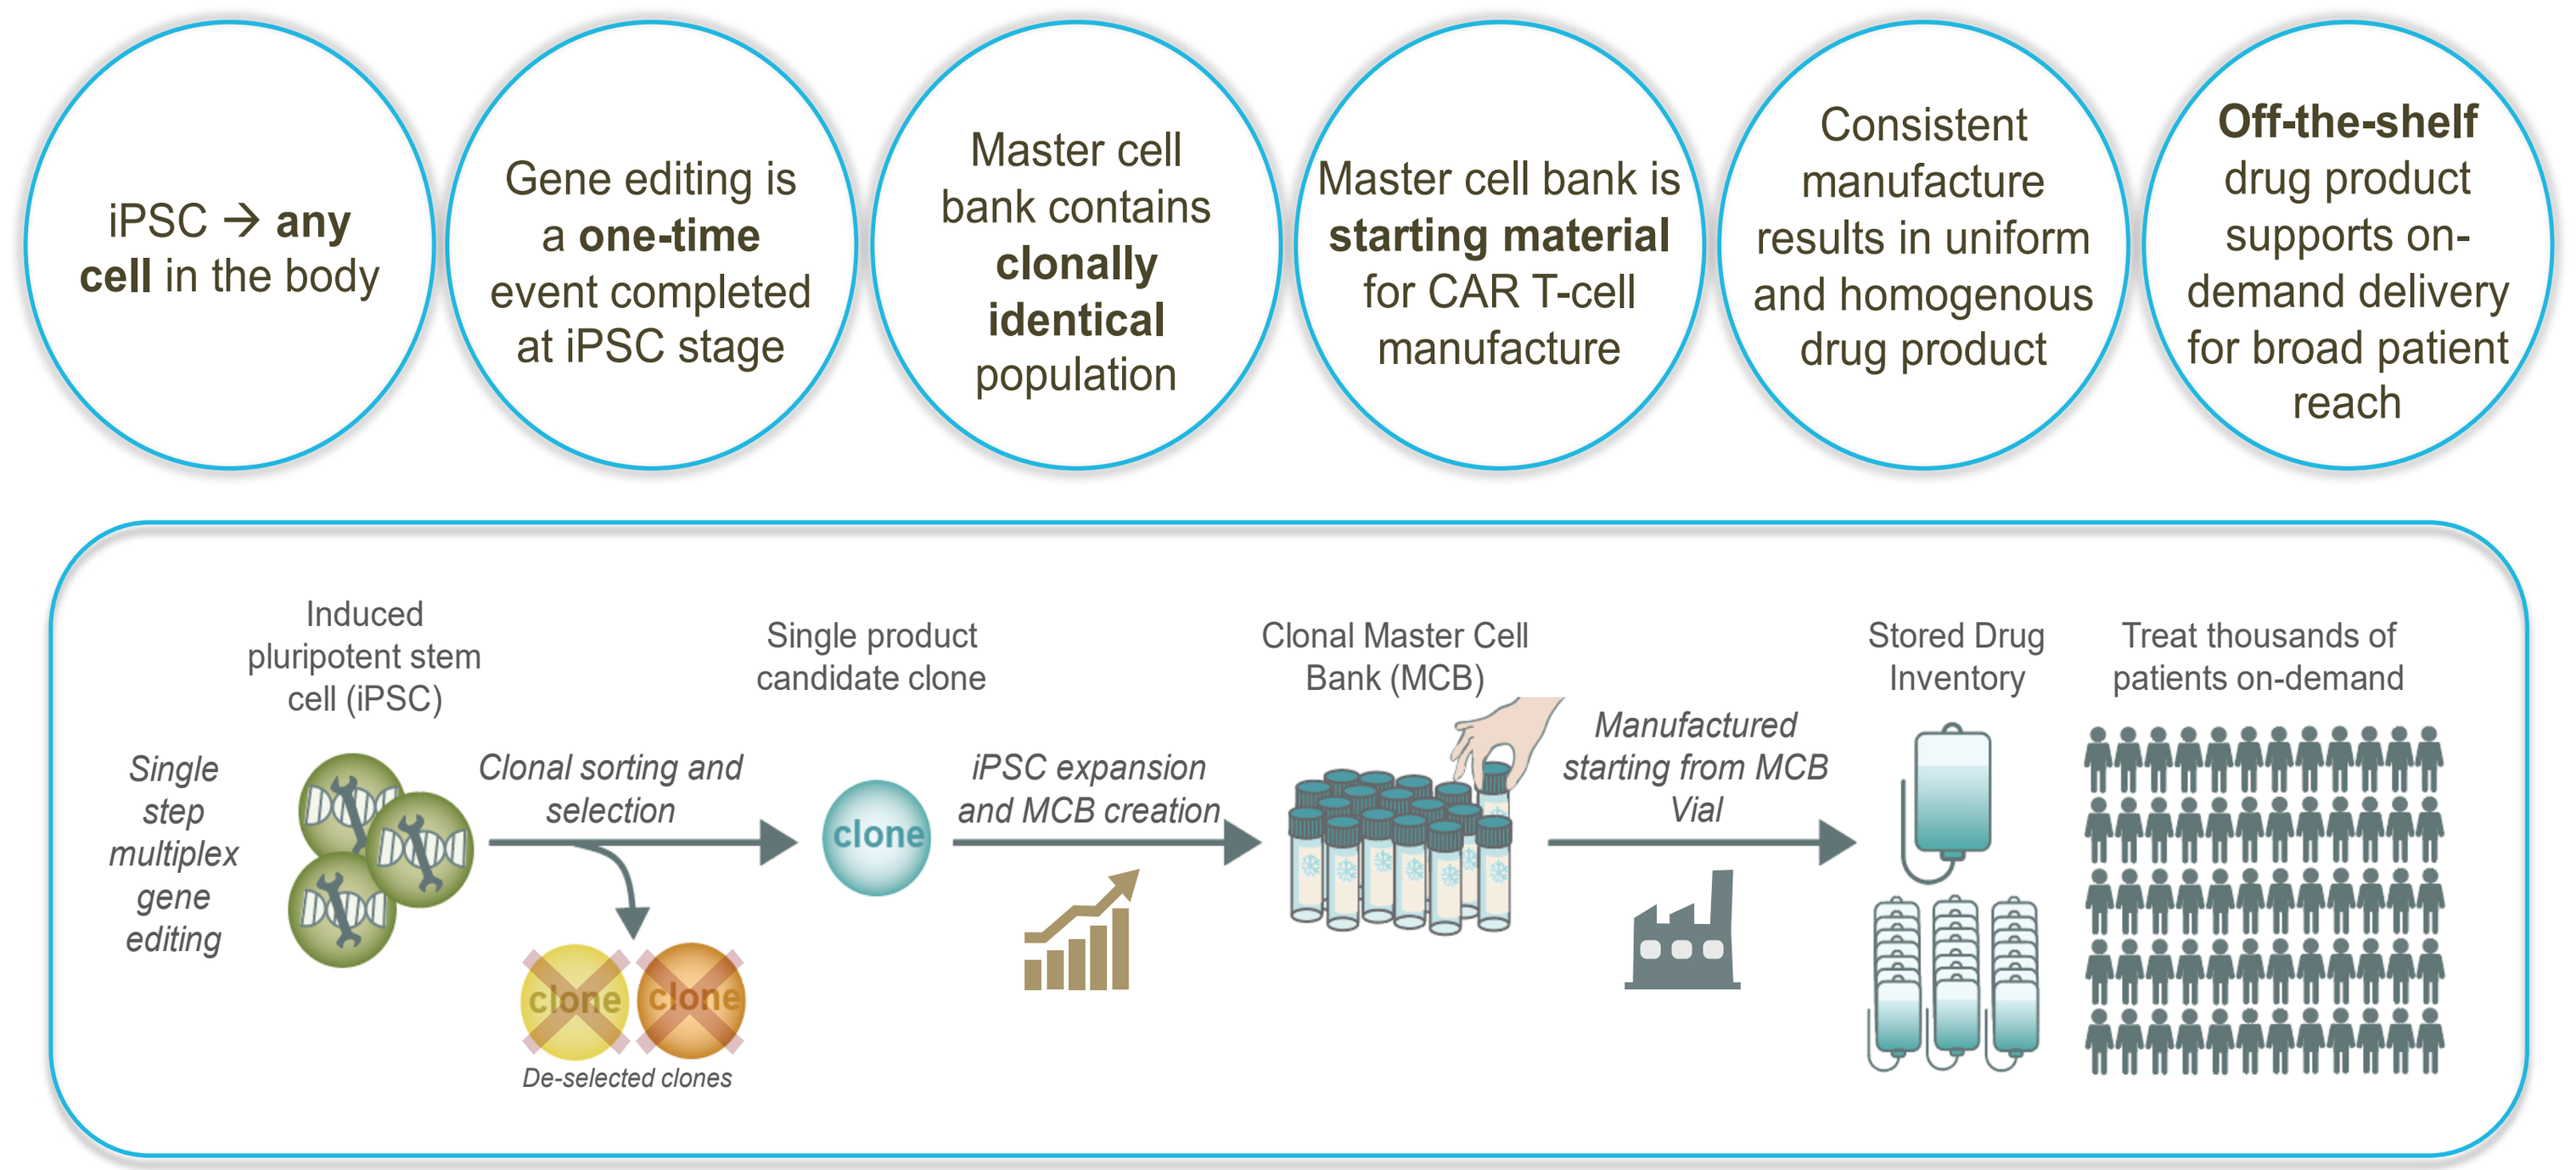
iPSC → **any cell** in the body
Gene editing is a **one-time** event completed at iPSC stage
Master cell bank contains **clonally identical** population
Master cell bank is **starting material** for CAR T-cell manufacture
Consistent manufacture results in uniform and homogenous drug product
Off-the-shelf drug product supports on-demand delivery for broad patient reach
Induced pluripotent stem cell (iPSC)
Single step multiplex gene editing
Clonal sorting and selection
De-selected clones
Single product candidate clone
clone
iPSC expansion and MCB creation
Clonal Master Cell Bank (MCB)
Manufactured starting from MCB Vial
Stored Drug Inventory
Treat thousands of patients on-demand

B *FT819 Regimen B: CAR T-cell treatment without skipping current medication or needing conditioning chemotherapy*

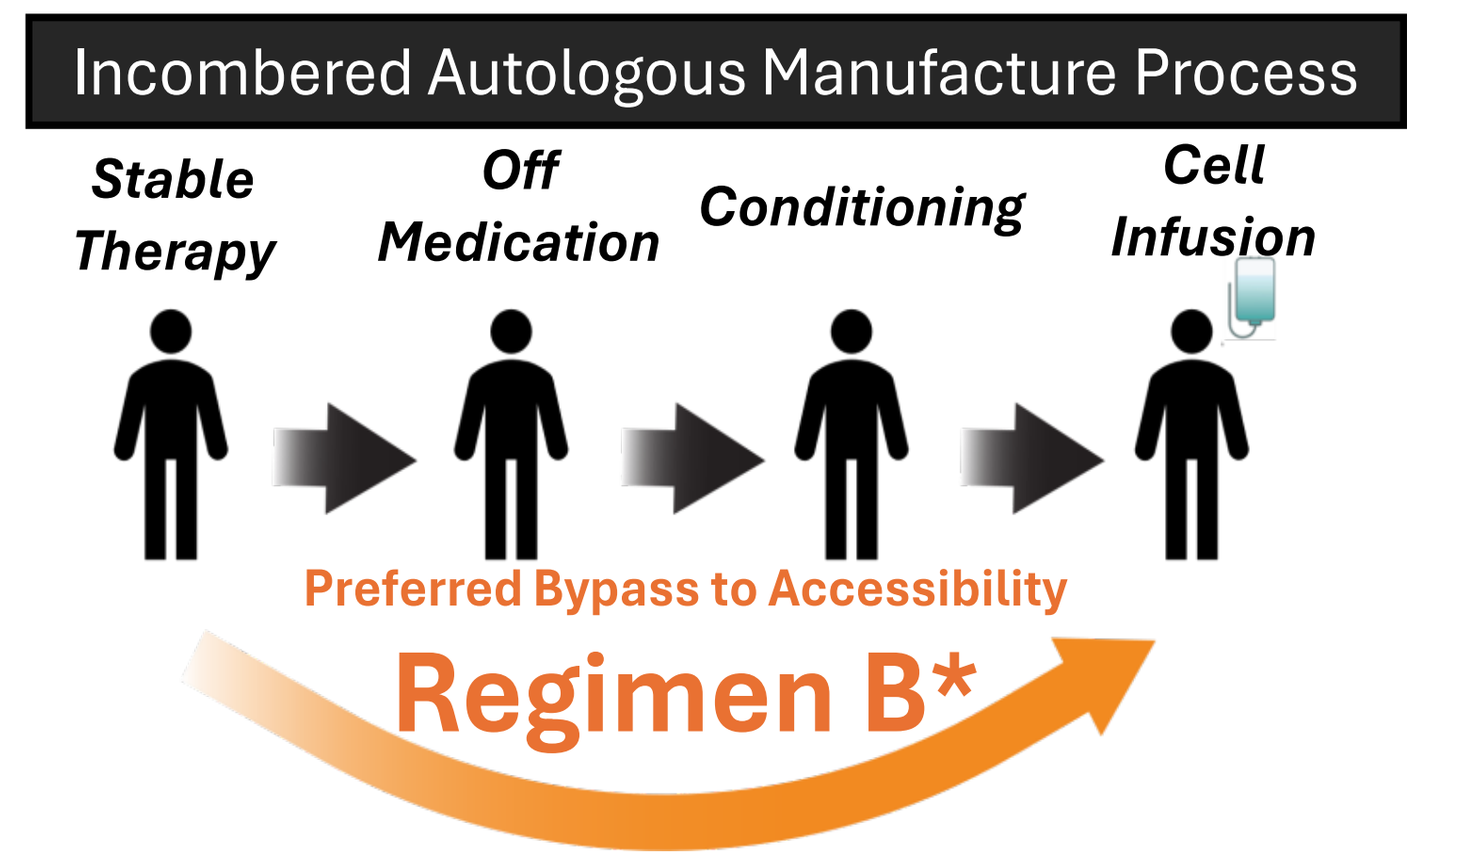
Incombered Autologous Manufacture Process
Stable Therapy
Off Medication
Conditioning
Cell Infusion
Preferred Bypass to Accessibility
Regimen B*

*CCT Free; No Interruption of Background Therapy (Less chance of disease flare); No CCT Toxicity

C *Unique Outpatient CAR T-cell Therapy*

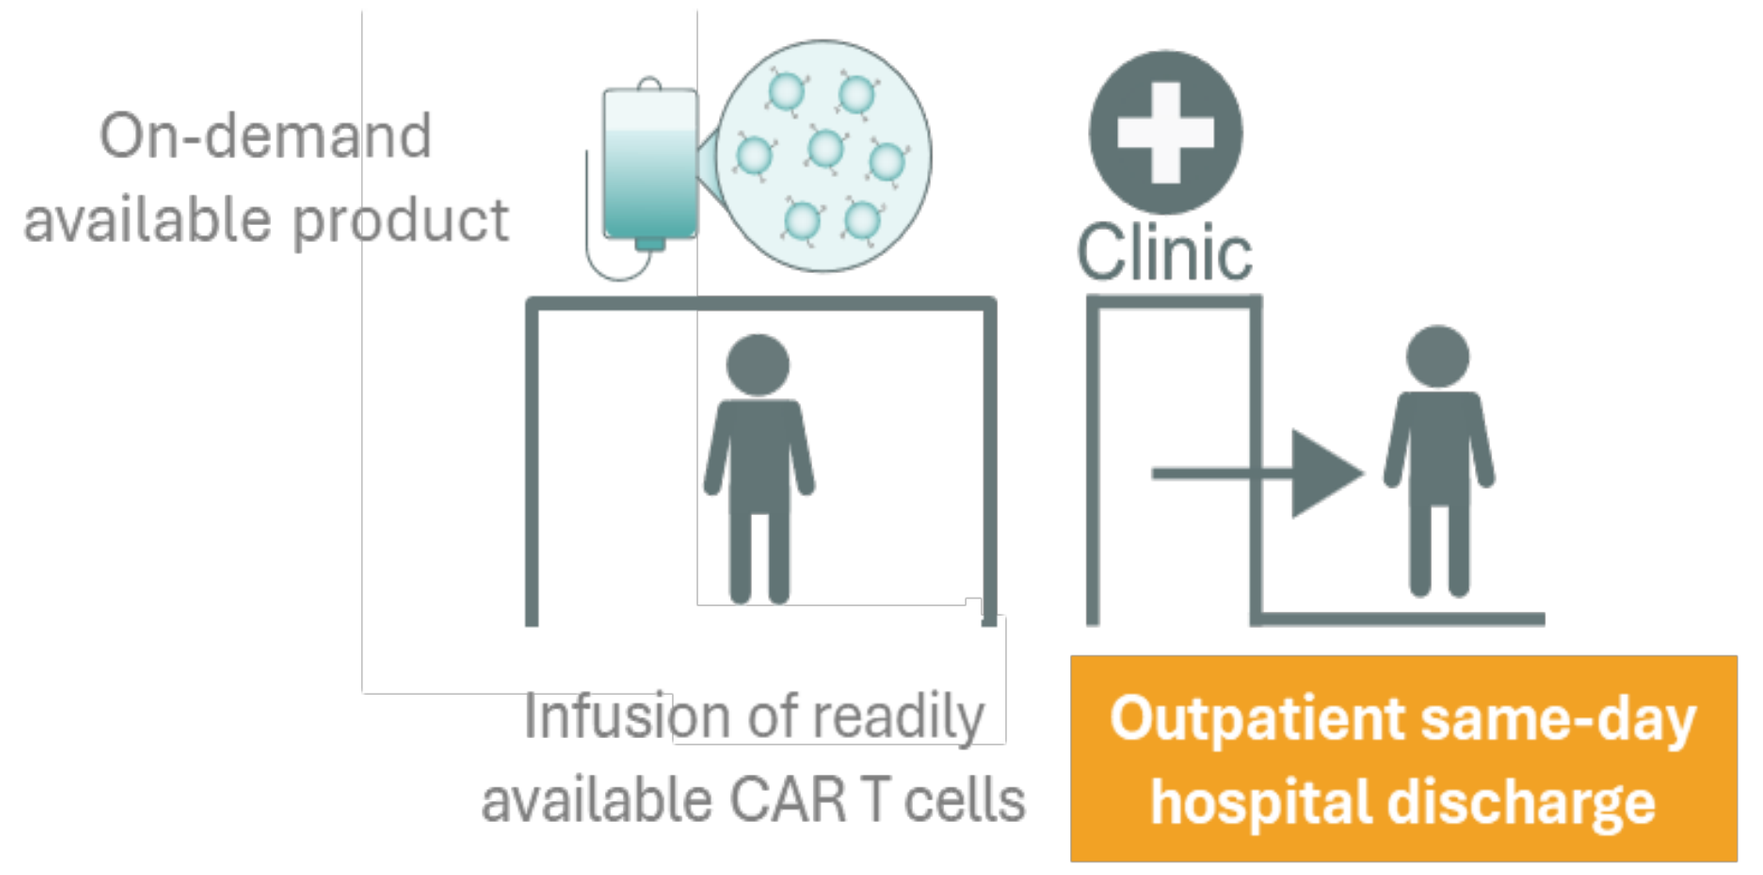
On-demand available product
Clinic
Infusion of readily available CAR T cells
Outpatient same-day hospital discharge

Figure 1. (A) Diagram outlining the iPSC derived CAR T-cell therapy pipeline. **(B)** Regimen B treats with FT819 without drug holiday or conditioning. **(C)** FT819 is being given in outpatient (community) setting.

Results

FT819 Without Conditioning Chemotherapy Shows Clinical Benefit for Treatment of SLE at Single Dose of Dose Level 1

Patient	Alloreactivity	PK After Infusion	SRI-4*	LLDAS†
1	None	✓	✓	✓
2	None	✓	✓	-
3	None	✓	✓	✓

Table 1. Data cut April 9, 2026. SRI-4 and LLDAS are shown as best overall response. Patient 1 achieved LLDAS at 3 months and maintained. Patient 3 achieved LLDAS at day 29, but reverted at 3 months due to leukopenia, 6 month data pending.

**SRI-4, SLE Responder Index 4: ≥4-point reduction in SLEDAI, no new BILAG A or ≤2 new BILAG B score. No worsening in PGA by ≥0.3*
†LLDAS, Lupus Low Disease Activity State: SLEDAI of ≤4, no new disease activity or activity in major organ systems, PGA ≤ 1, steroids ≤7.5mg daily, and standard maintenance doses of immunosuppressive drugs and approved biological agents.

Results

FT819 Demonstrates anti-CD19 activity in SLE Without the Need for Conditioning Chemotherapy Resulting in Meaningful Clinical Outcomes

A *FT819 Drives B Cell Depletion Comparable to treatment with CCT*

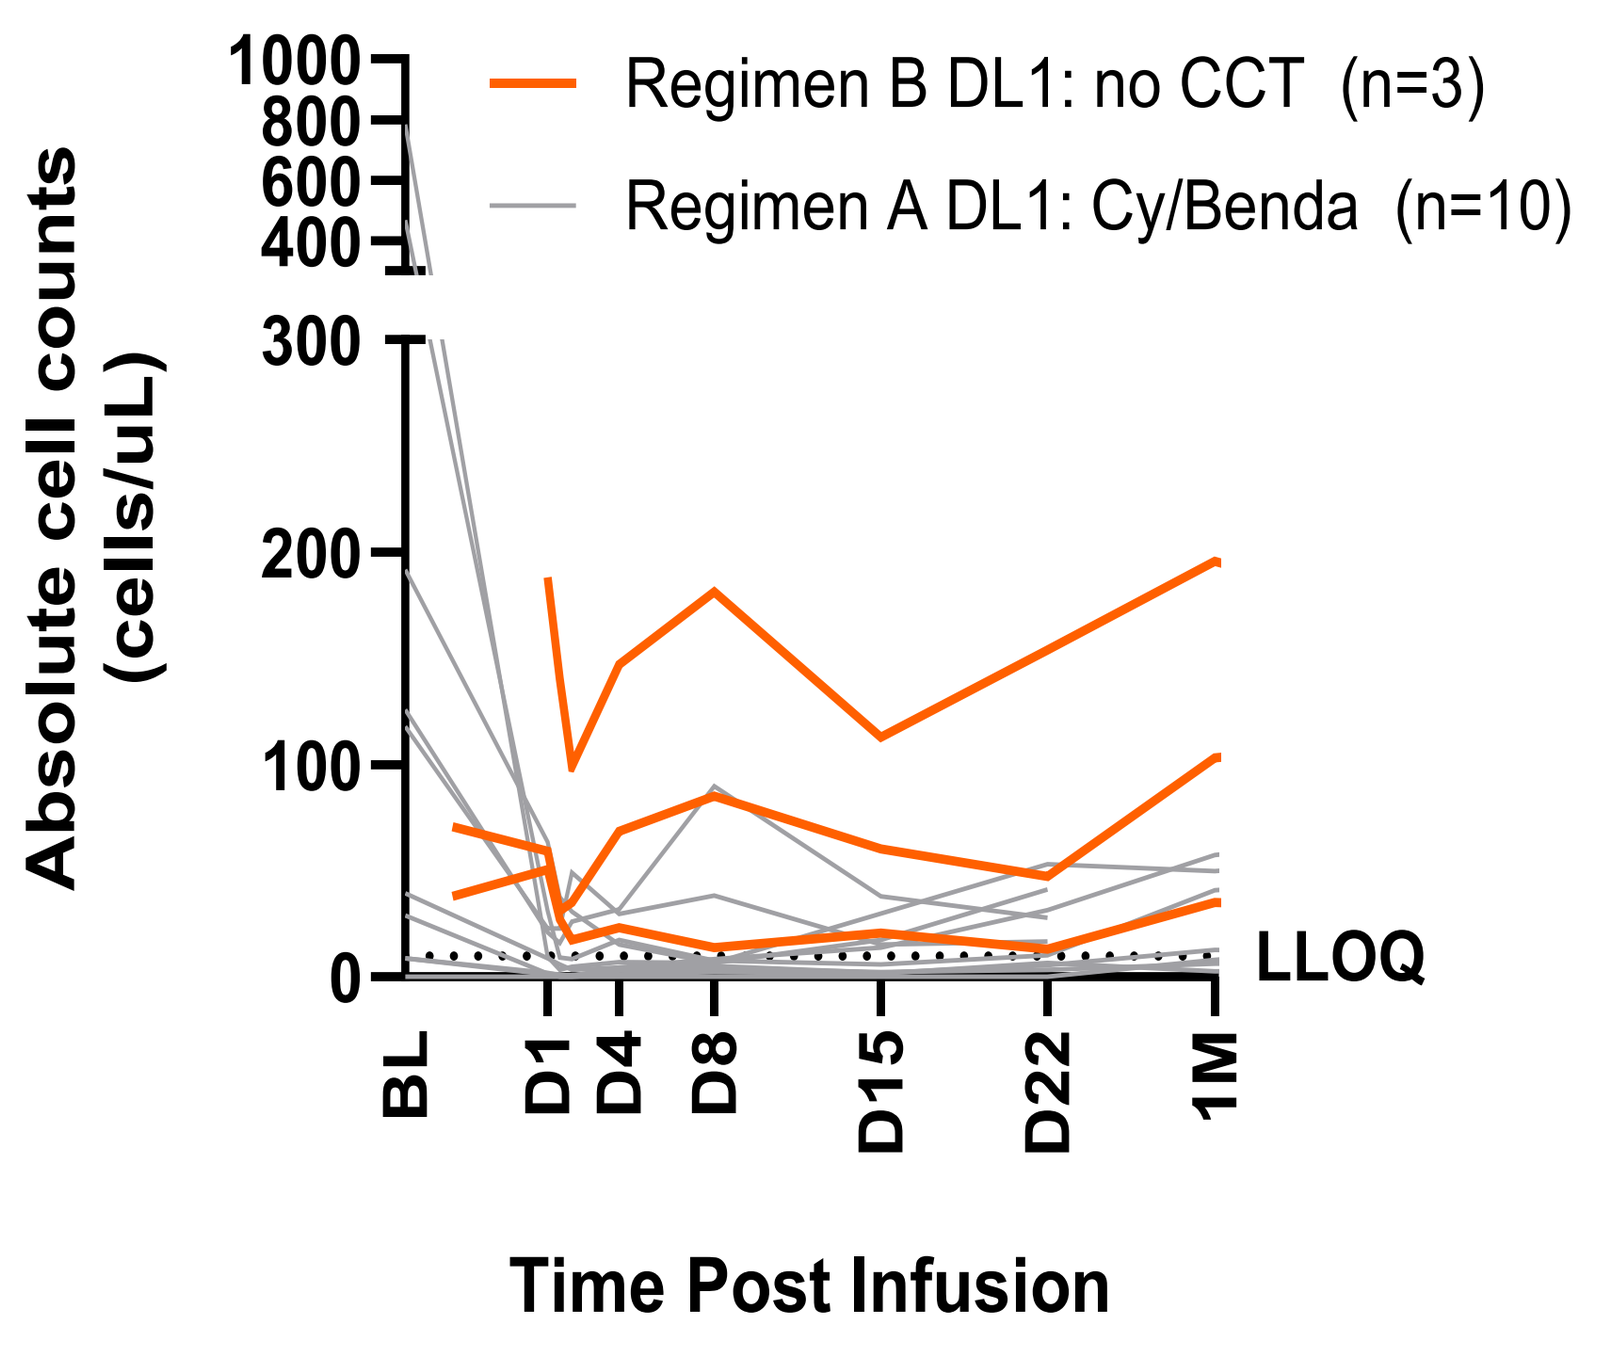
Regimen B DL1: no CCT (n=3)
Regimen A DL1: Cy/Benda (n=10)
Absolute cell counts (cells/uL)
1000
800
600
400
300
200
100
0
LLOQ
BL
D1
D4
D8
D15
D22
1M
Time Post Infusion

B *FT819 Depletes B Cells In Lymphoid Tissues*

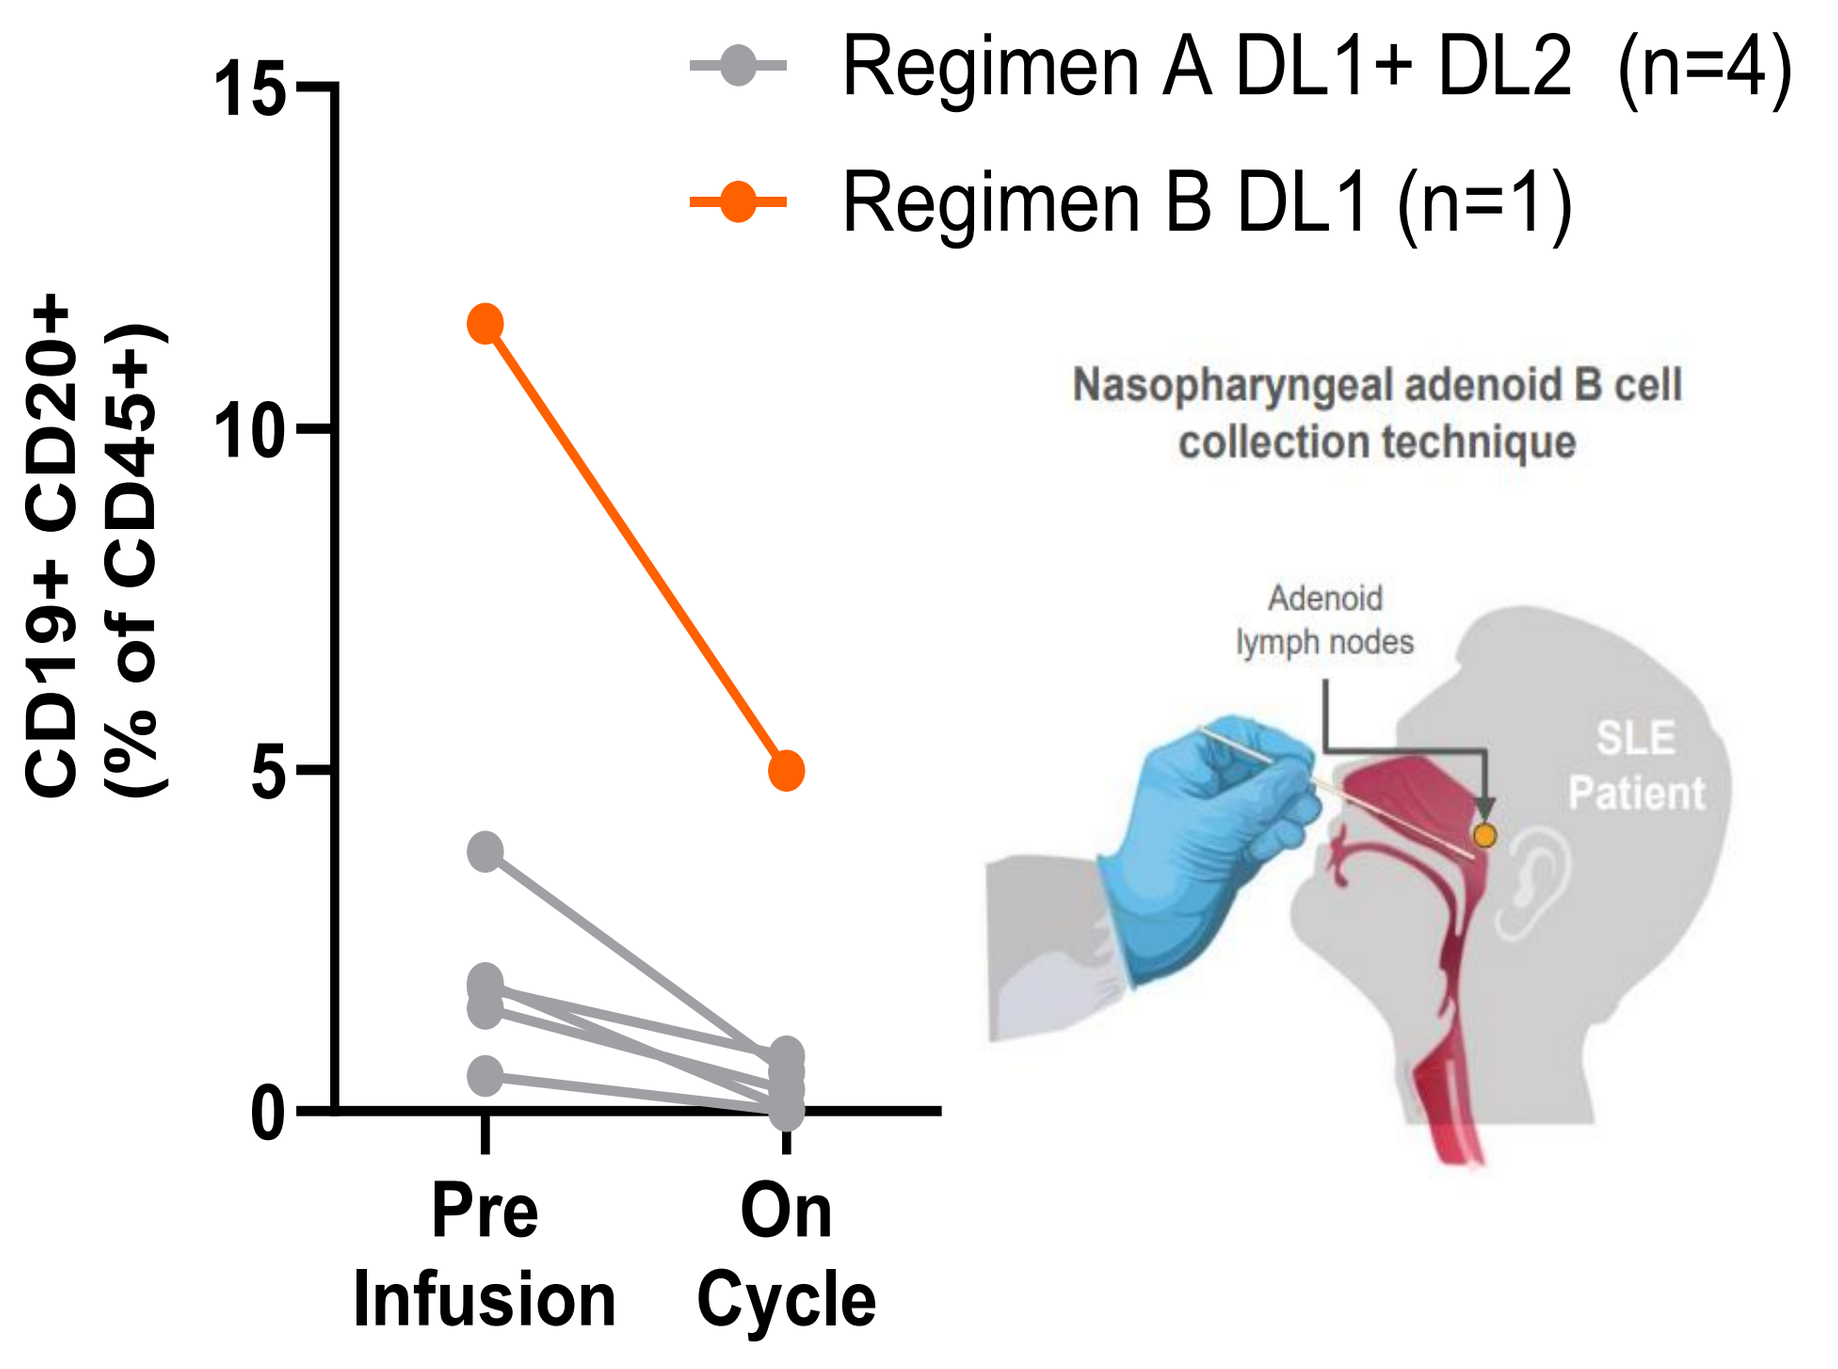
Regimen A DL1+ DL2 (n=4)
Regimen B DL1 (n=1)
CD19+ CD20+ (% of CD45+)
15
10
5
0
Pre Infusion
On Cycle
Nasopharyngeal adenoid B cell collection technique
Adenoid lymph nodes
SLE Patient

C *Detection of Live FT819 Cells After Infusion*

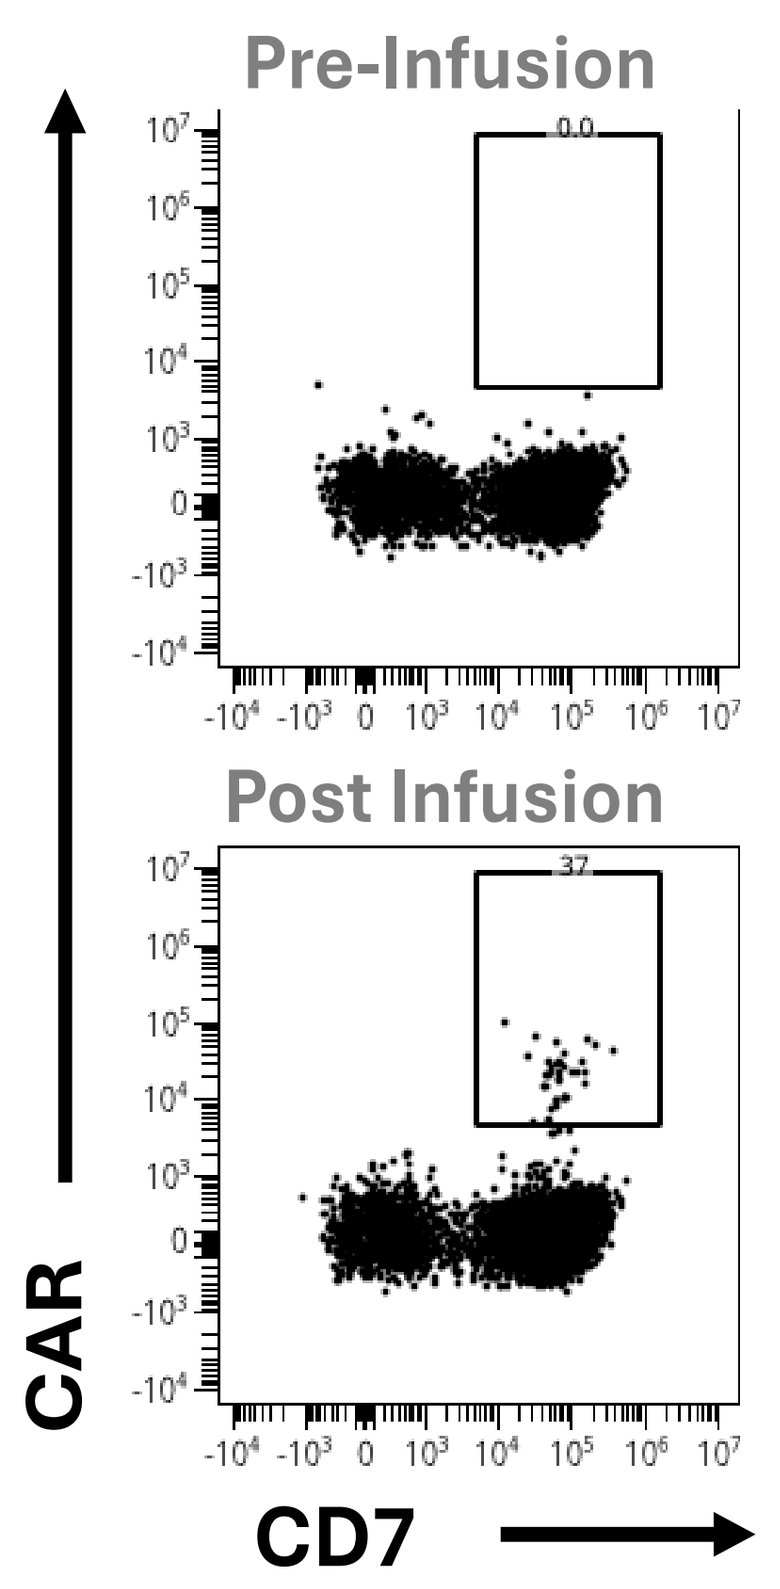
Pre-Infusion
0.0
Post Infusion
37
CAR
CD7

D *Improved Clinical Outcome Following Single Dose of FT819 Infusion without Conditioning Chemotherapy up to 12 Months Post treatment*

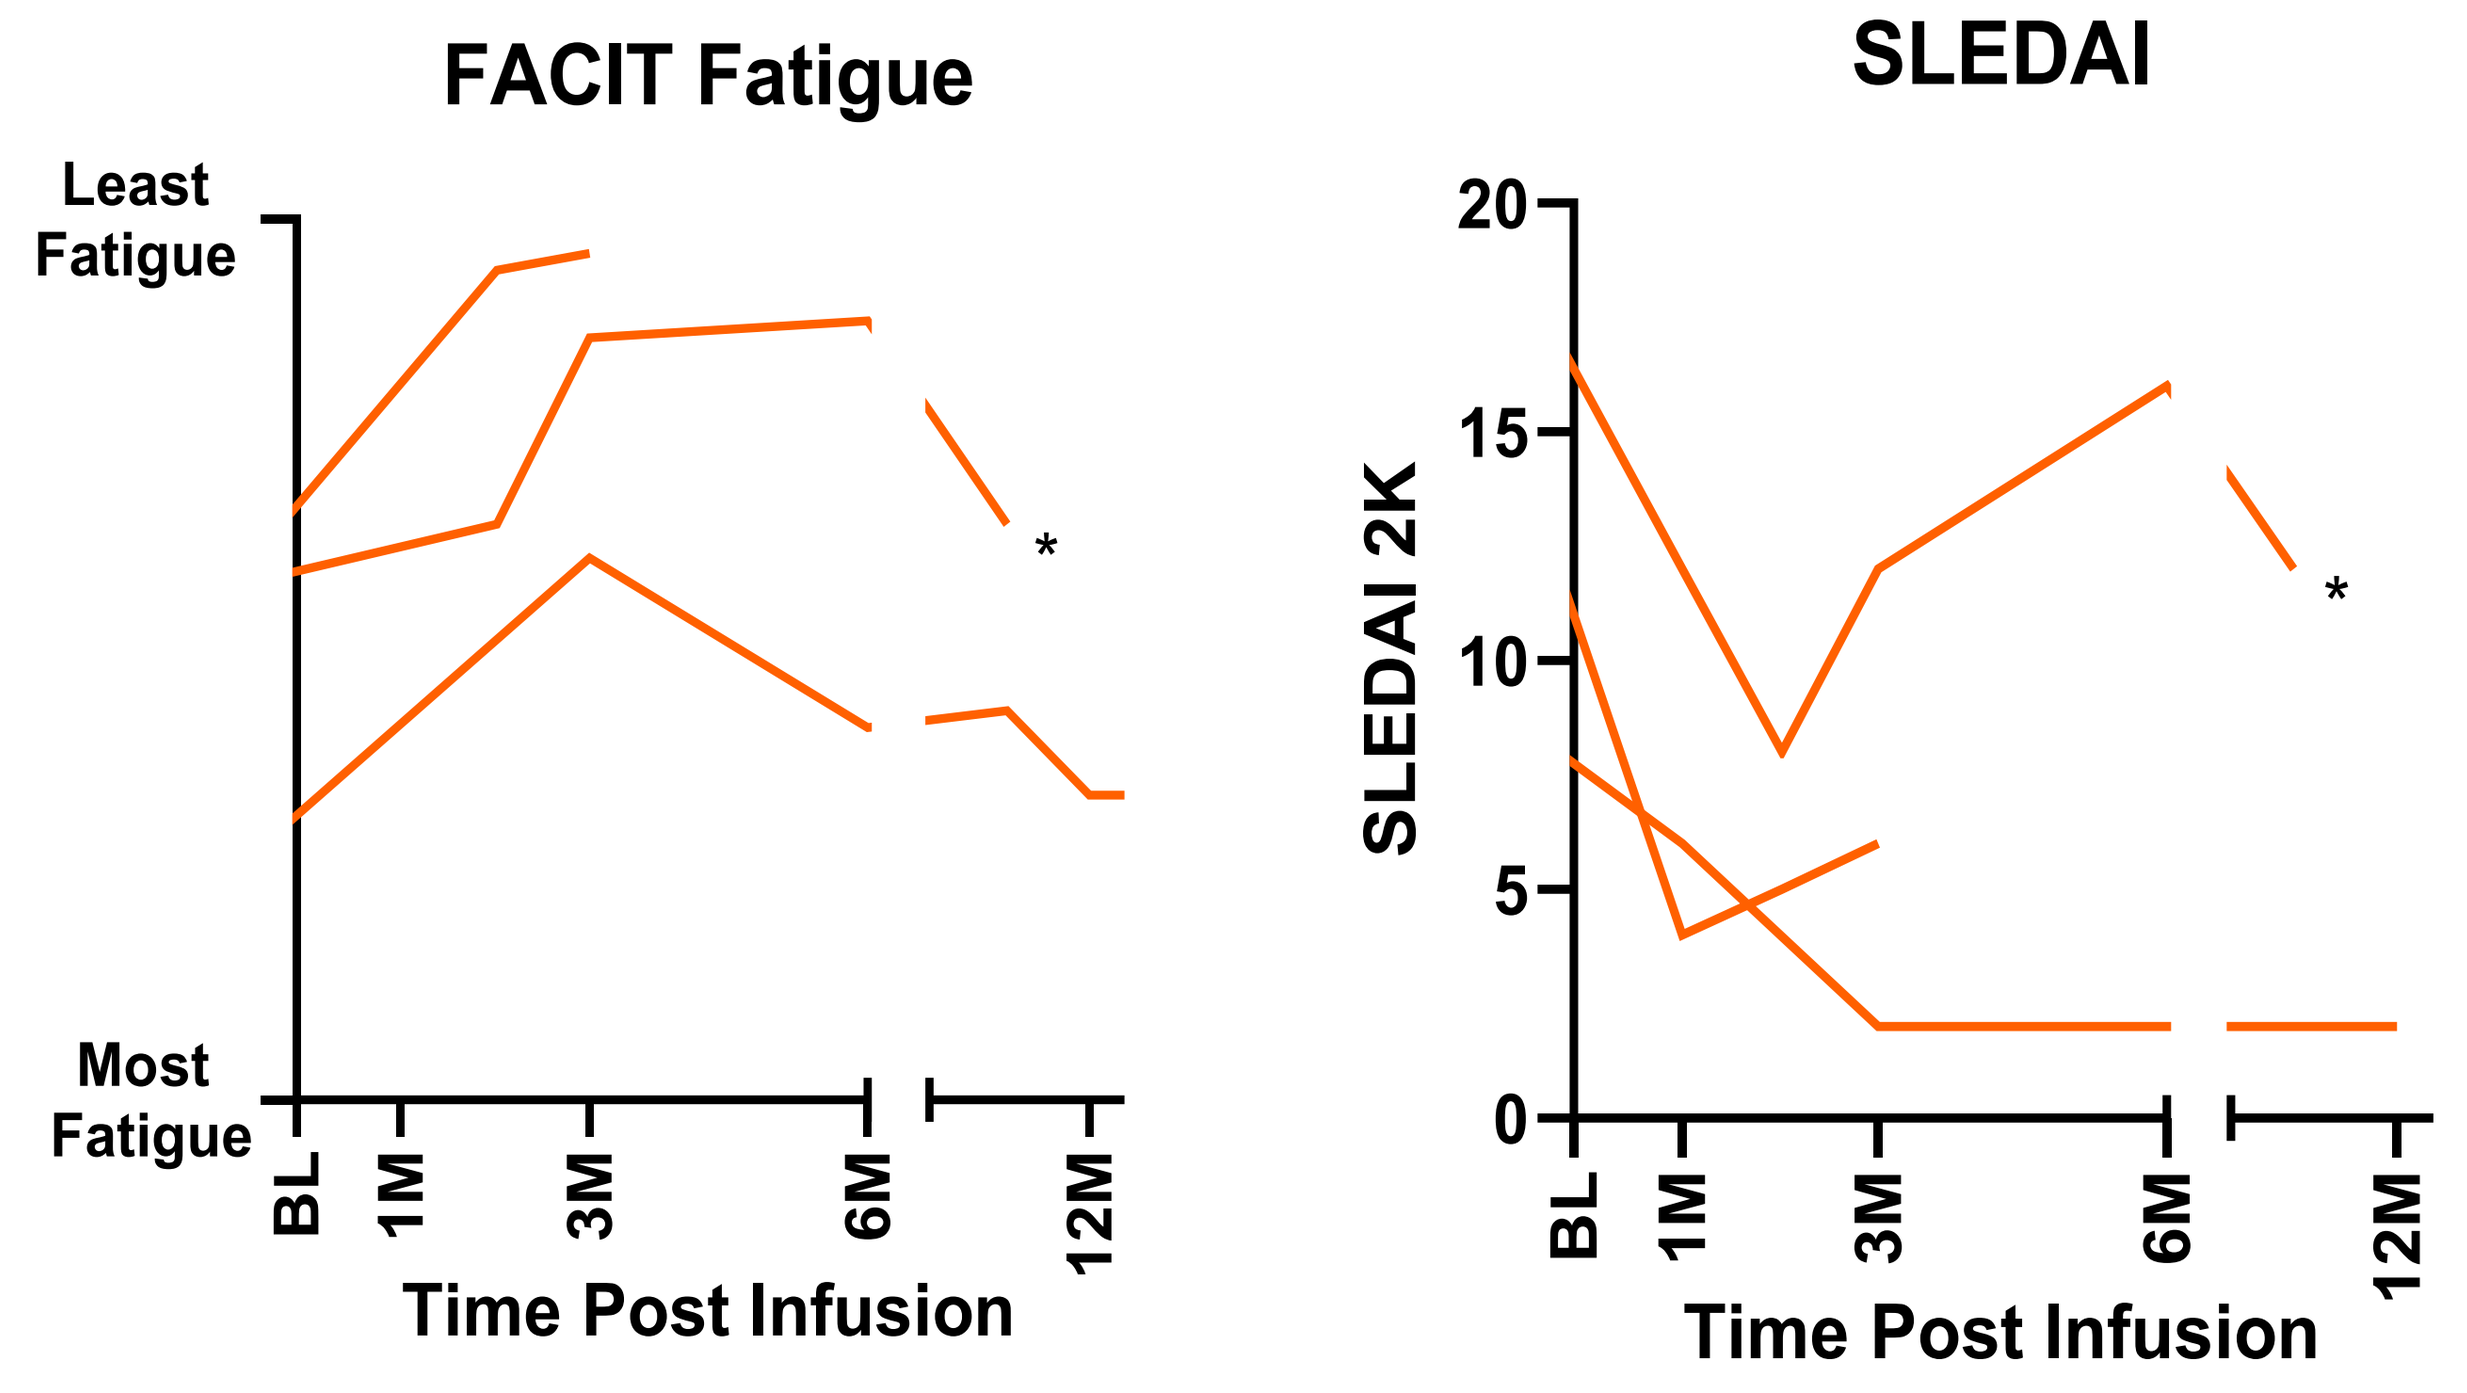
FACIT Fatigue
Least Fatigue
Most Fatigue
BL
1M
3M
6M
12M
Time Post Infusion
*
SLEDAI
SLEDAI 2K
20
15
10
5
0
BL
1M
3M
6M
12M
Time Post Infusion
*

Figure 2. FT819 functions in the absence of CCT. (A,B) Without CCT low dose FT819 shows significant, but transient B cell depletion in the peripheral blood with depth similar to that seen with conditioning chemotherapy **(A)**, as well as in secondary lymphoid tissue (nasopharyngeal swabs) **(B)**. **(C)** Following infusion, live FT819 can be detected in the periphery of patients without conditioning chemotherapy. **(D)** FACIT Fatigue **(left)** and SLEDAI **(right)** scores of patients in regimen B (orange). All Regimen B patients demonstrate improvement from their baseline, both immediately after treatment and out to the latest timepoint analyzed. Data cut for regimen A (Dec 23, 2025), Data cut for regimen B (April 9, 2026). *Due to disease flare, patient received cycle 2, last data point shown is screening for cycle 2 (response data pending).

Single Dose Treatment of FT819 without Conditioning Chemotherapy Drives Long-term Changes of up to 12 months in the B Cell Repertoire

A *FT819 Alone Eliminates Major B Cell Clones Long Term*

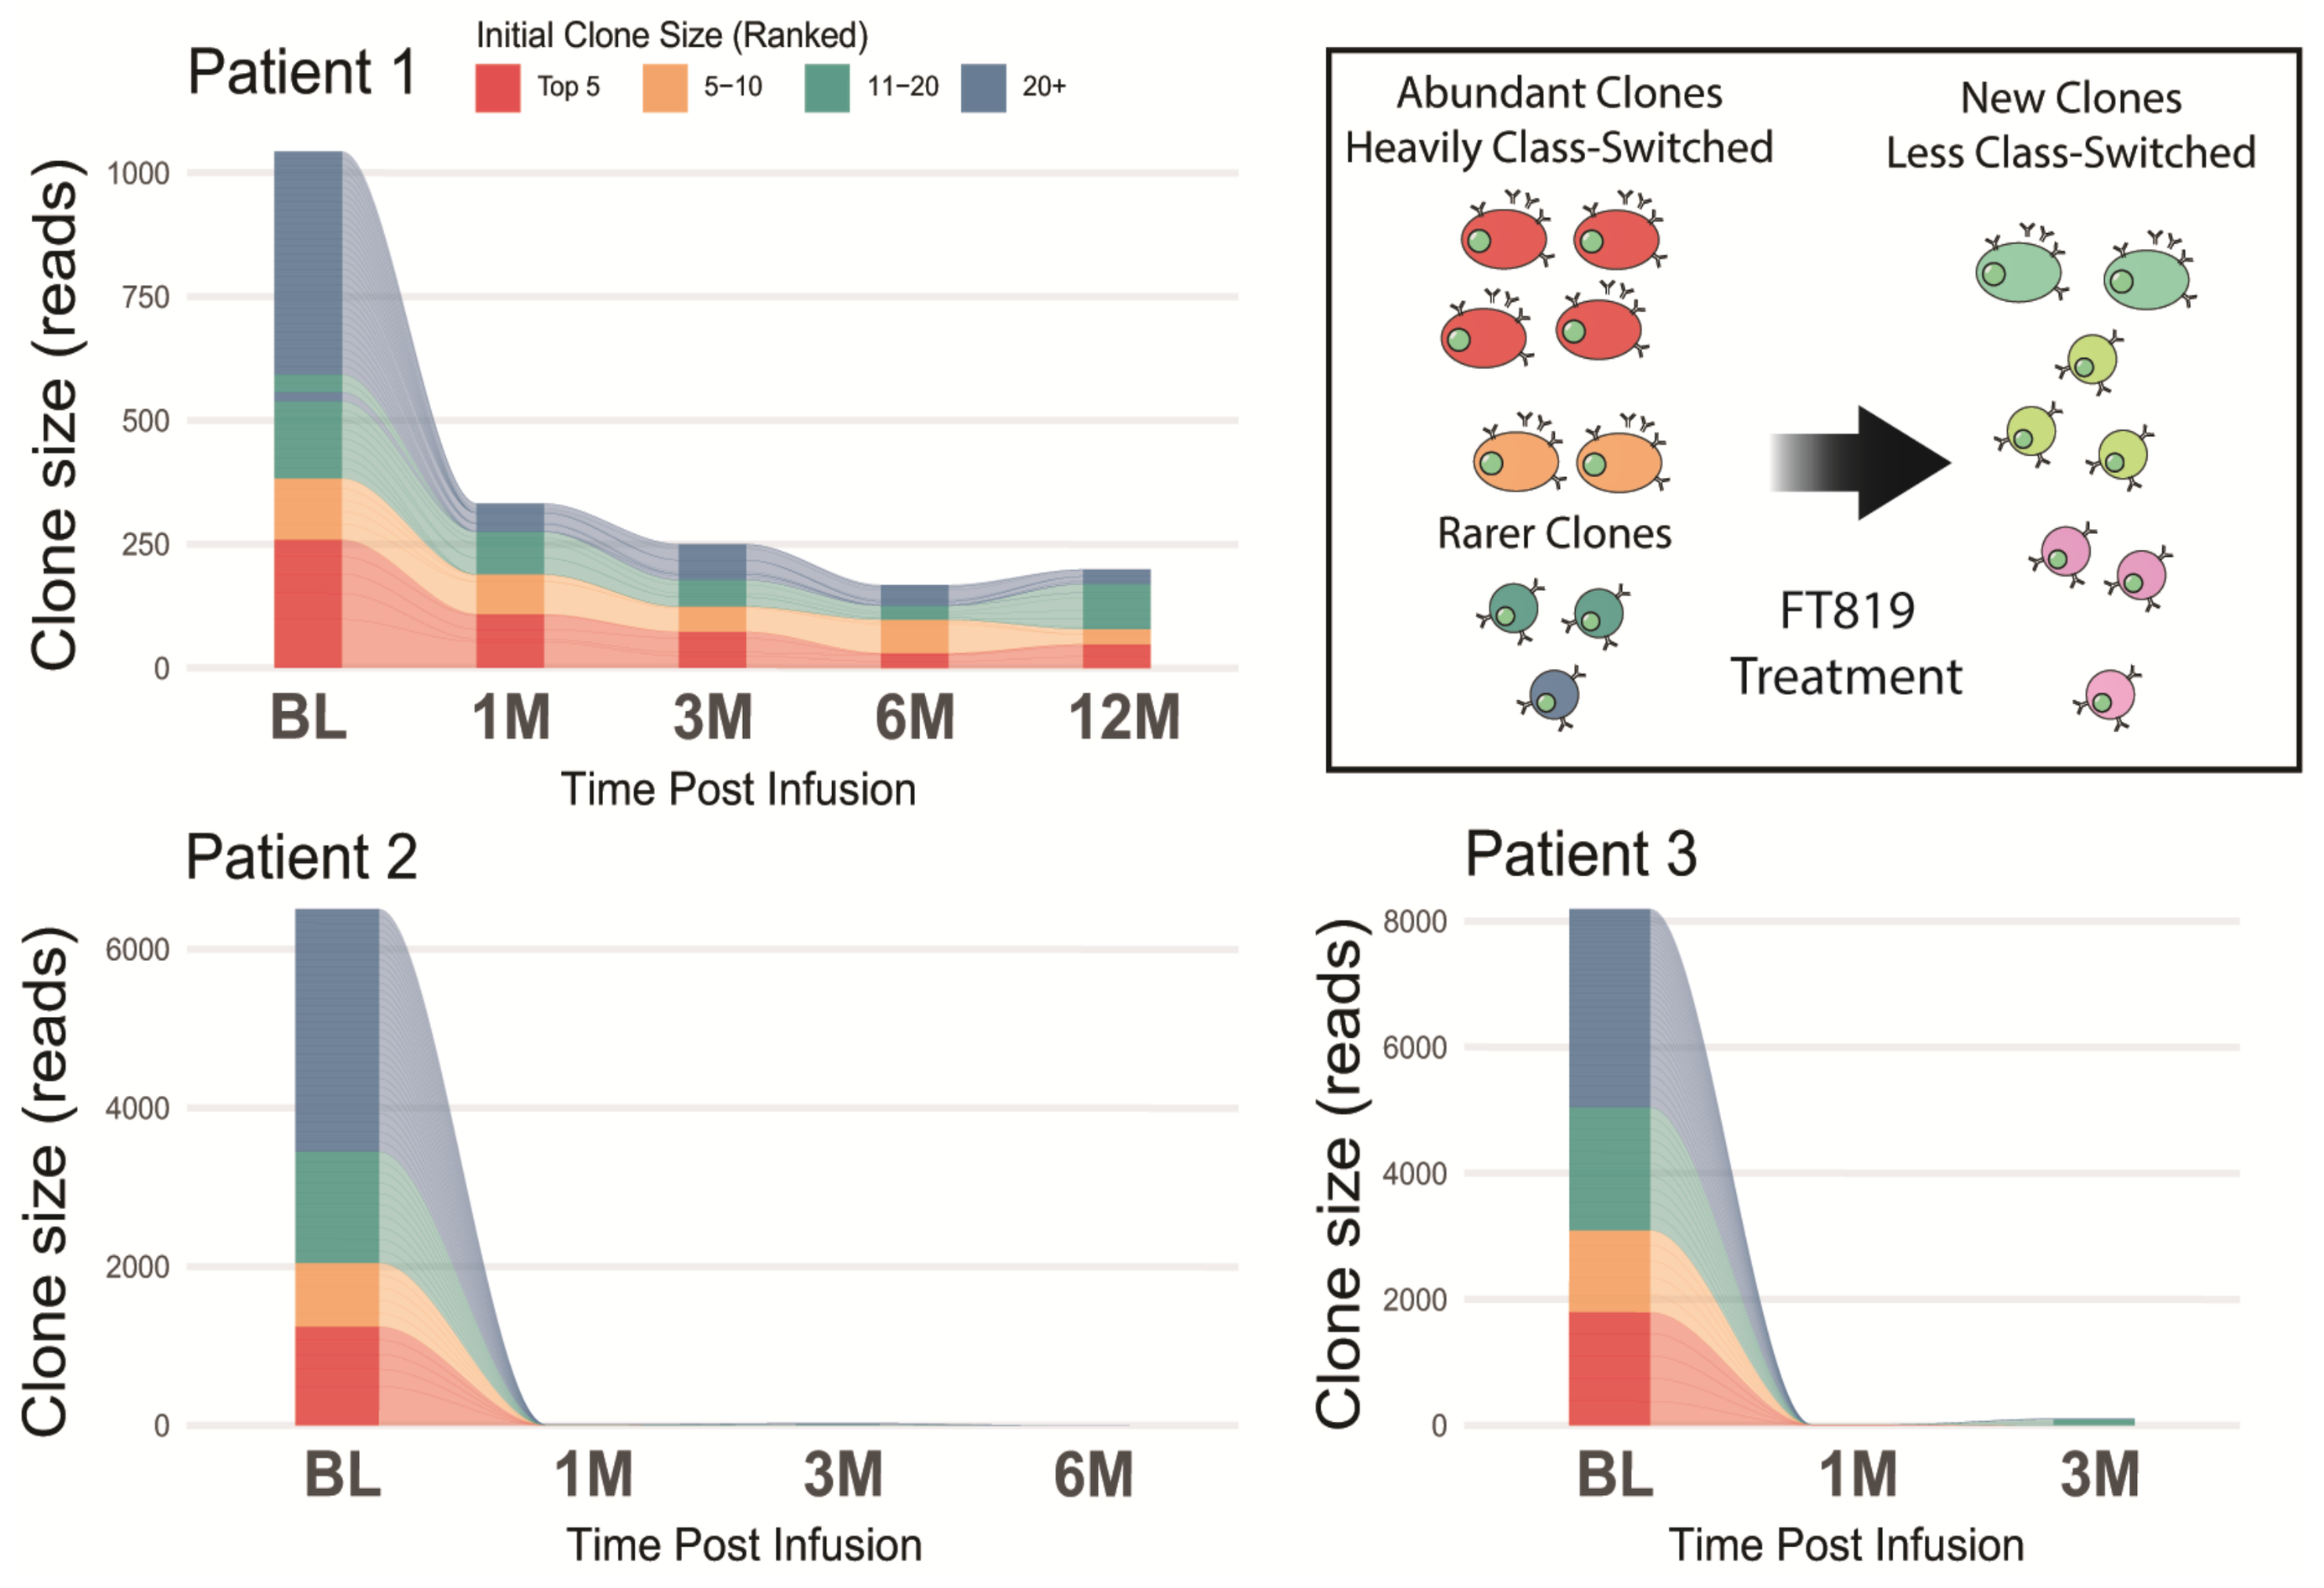
Patient 1
Initial Clone Size (Ranked)
Top 5
5–10
11–20
20+
Clone size (reads)
1000
750
500
250
0
BL
1M
3M
6M
12M
Time Post Infusion
Patient 2
6000
4000
2000
0
BL
1M
3M
6M
Time Post Infusion
Abundant Clones Heavily Class-Switched
New Clones Less Class-Switched
Rarer Clones
FT819 Treatment
Patient 3
8000
6000
4000
2000
0
BL
1M
3M
Time Post Infusion

B *FT819 Alone Shifts BCR Repertoire Towards Naïve*

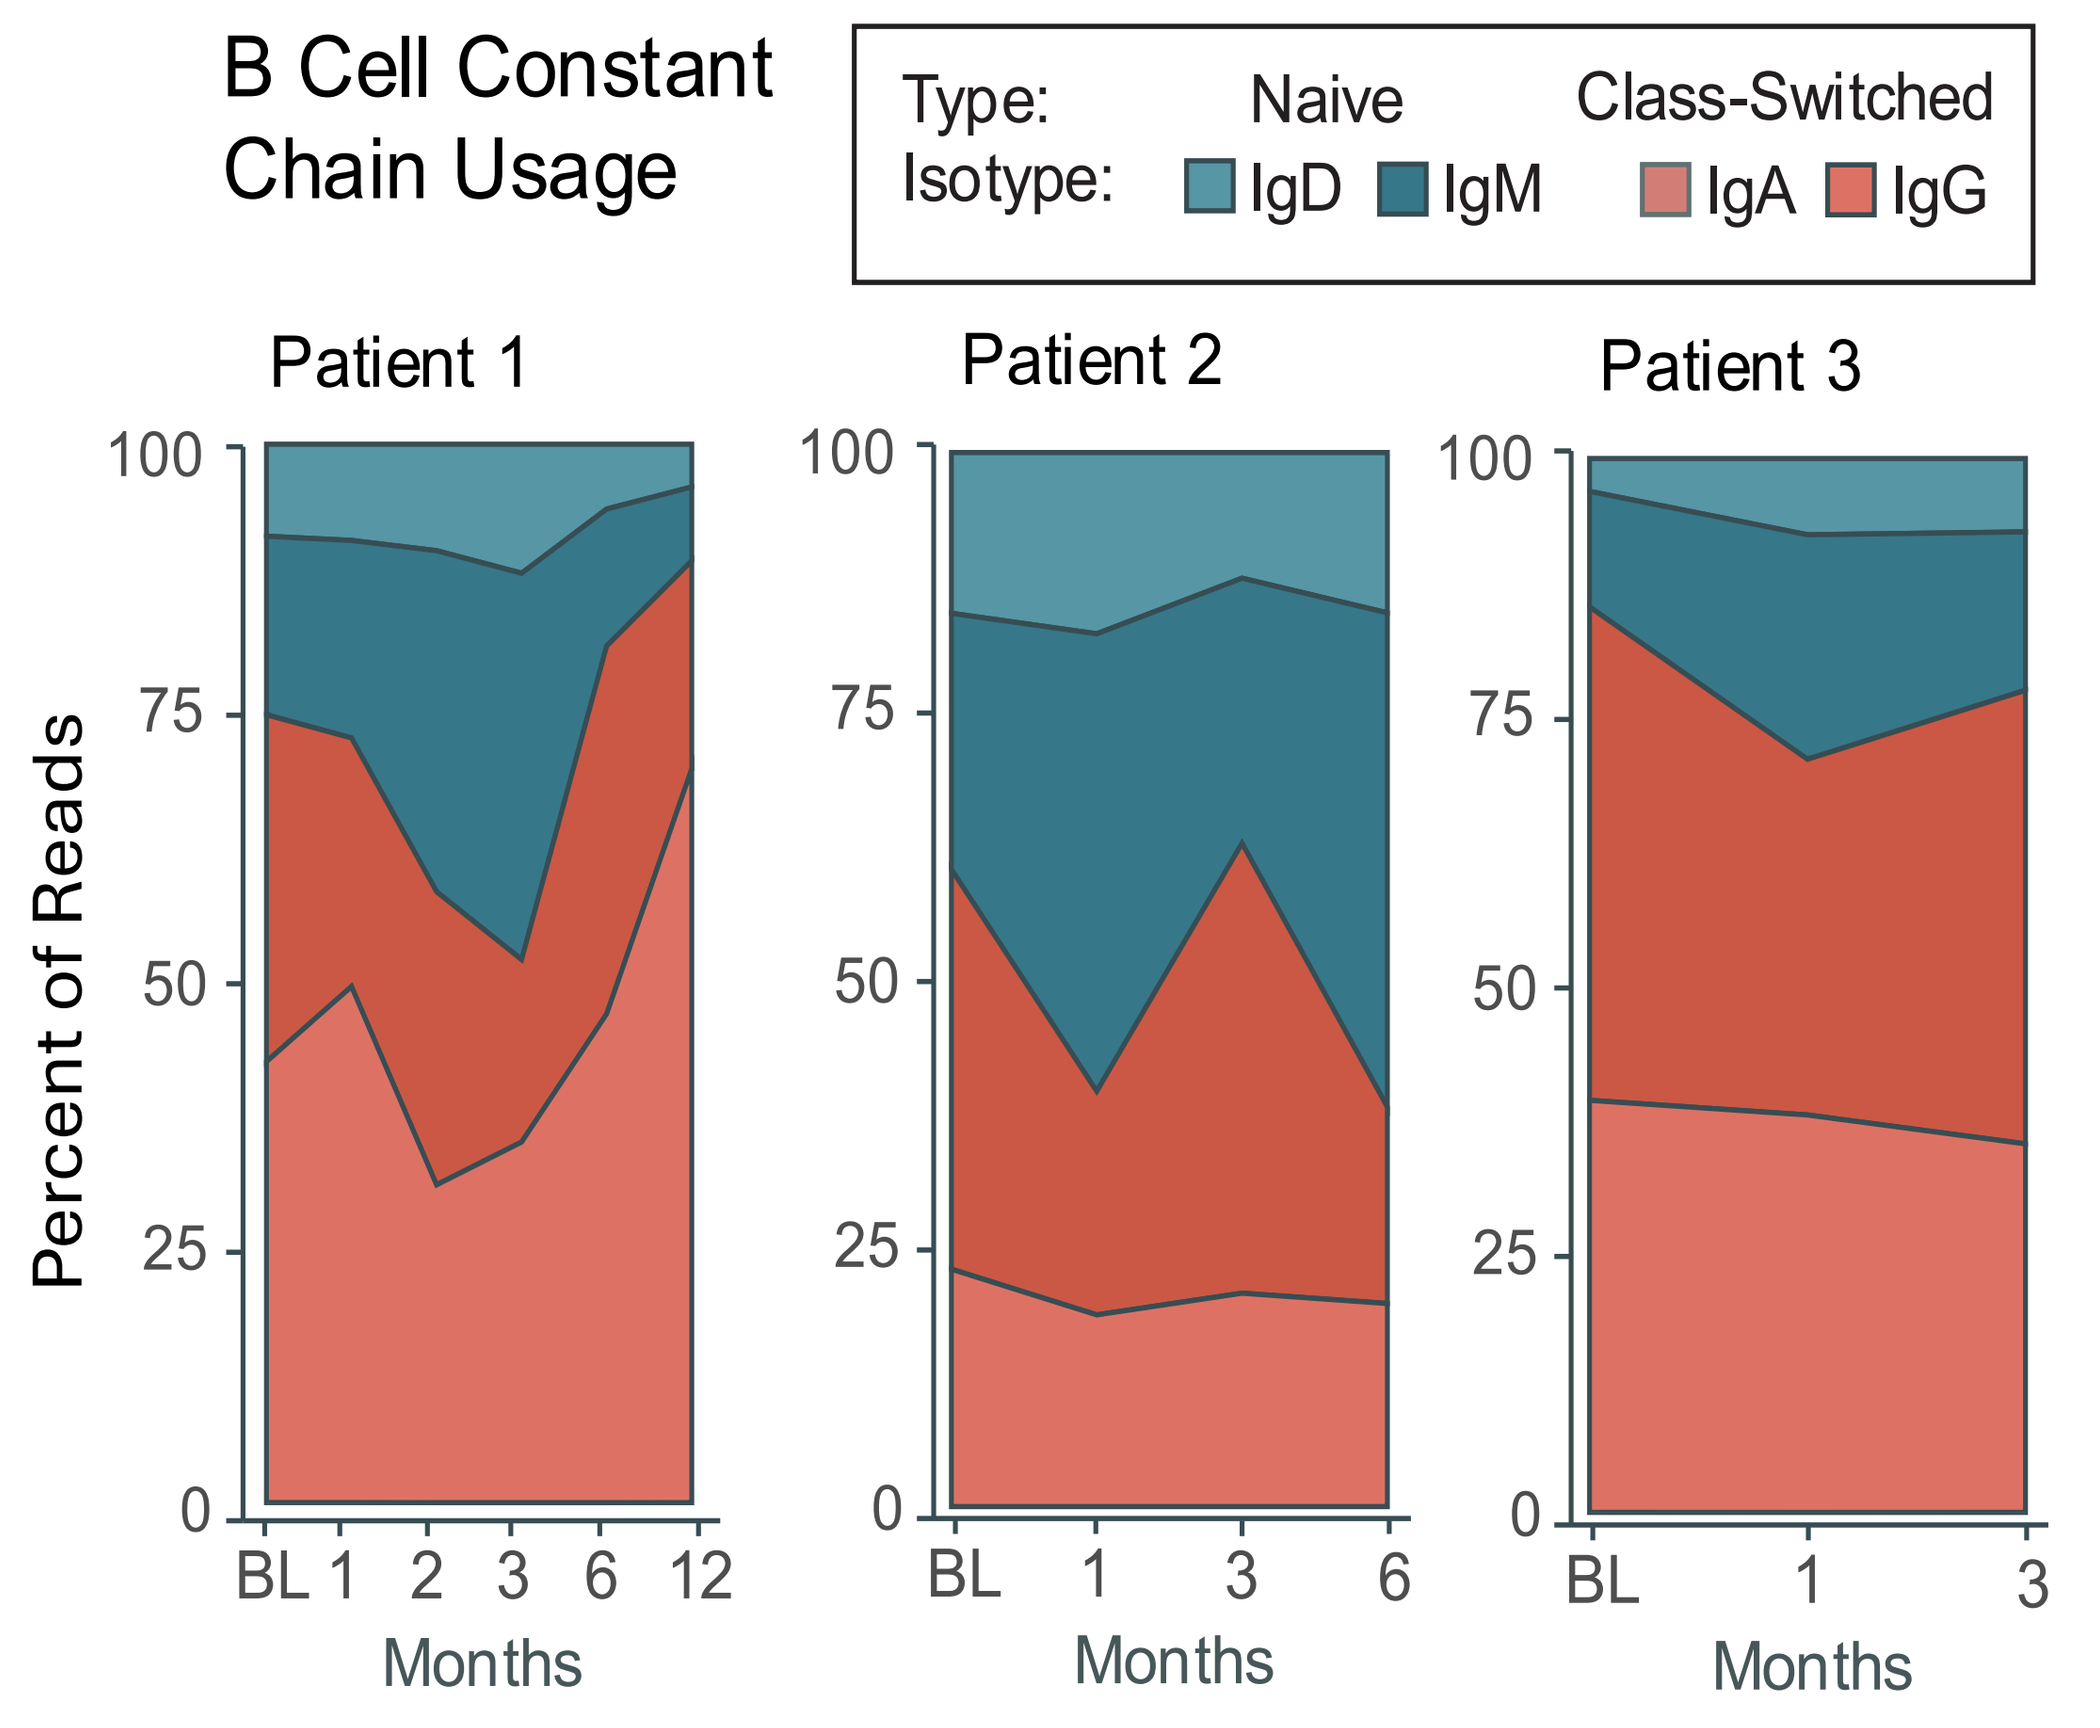
B Cell Constant Chain Usage
Type: Naive Class-Switched
Isotype: IgD IgM IgA IgG
Patient 1
Patient 2
Patient 3
Percent of Reads
100
75
50
25
0
BL 1 2 3 6 12
BL 1 3 6
BL 1 3
Months

Figure 3. FT819 Alone Drives Long-term alterations to the B Cell Repertoire. (A) Clonal deletion without significant recovery of baseline clones in for patients 1-3 and diagram of clonal deletion in CAR T-cell therapy, initially skewed repertoires are expected to be replaced by new B cells (immune reset). **(B)** The balance of Constant Chain usage shifts towards naïve (IgD/IgM) in 3/3 patients following treatment with FT819 alone.

Conclusions

- FT819 treatment without CCT, and in the presence of background therapy shows:
 - ✓ Meaningful clinical benefit at dose level 1; **SRI-4 in 3/3 patients, with 2/3 also achieving LLDAS**
 - ✓ Depletion of B cells in the periphery, and in secondary lymphoid tissues (nasopharyngeal swab)
 - ✓ Depletion of major B cell clones up to 12 months following treatment; Shift towards less class-switched BCR repertoire
 - ✓ Preferable safety profile; Patient treatment at higher dose of 900 million cells has been initiated (dose level 2)
- FT819 overcomes numerous limitations associated with patient- and donor-sourced CAR T-cell therapies with the unique ability to be mass produced in a uniform and consistent manner from a master cell bank as the starting material for off-the-shelf and on-demand accessibility. Low cost of goods to make, at less than $3000 a dose.
- Sequencing FT819 treatment with and w/o conditioning can be a strategic clinical treatment strategy to support long-term durability; Less frequent treatment with meaningful outcomes will be highly competitive to current biologics

For more information on next generation iPSC derived CAR T-Cell Therapy: Oral Presentation ID: 307, Thursday 8am, Westin Seaport Grand Ballroom AB

View Online