Preliminary Phase 1 Results of a MICA/B-targeted CAR T cell Designed to Overcome Solid Tumor Escape Mechanisms and Avoid the Requirement for Conditioning Chemotherapy

Manish Patel[1], Ecaterina Dumbrava[2], Heinz-Josef Lenz[3], Peter Vu[4], Kyla D. Omilusik[5], Samad Ibitokou[5], Cara Bickers[5], Olena Kolesnichenko[5], Beatrice Ferguson[5], Seth Peng[5], Deepa Patel[5], Bertha Villa[5], Martin Hosking[5], Bahram Valamehr[5], Rebecca Elstrom[5], Lilly Wong[5], Ida Micaily[6]

[1]Masonic Cancer Center, Minneapolis, MN; [2]MD Anderson Cancer Center, Houston, TX ; [3]USC-Norris Comprehensive Cancer Center, Los Angeles, CA; [4]UCSD, San Diego, CA; [5]Fate Therapeutics, Inc., San Diego, CA; [6]Sidney Kimmel Cancer Center, Philadelphia, PA

Introduction

Cancer immunotherapy is a rapidly evolving field that has transformed the treatment of many tumor types, including solid tumors. Barriers to the clinical success of CAR T-cell therapy in solid tumors include the heterogeneity of putative solid tumor target antigen expression, trafficking of the T-cell product into tumors and overcoming the suppressive tumor immune microenvironment.

FT836 is a multipoint engineered iPSC-derived allogeneic CAR T cell, targeting the conserved α3 domain of MICA/B stress ligands, enabling recognition of diverse solid tumors. The MICA/B-CAR has the potential to mitigate MICA/B shedding by tumor cells and is resistant to suppression via soluble MICA/B (Goulding et al., 2023).

This Phase 1 study is evaluating FT836 with or without chemotherapy and/or mAb, in patients with solid tumors.

Figure 1. Unique Platform for Delivering Off-the-Shelf Cellular Therapies

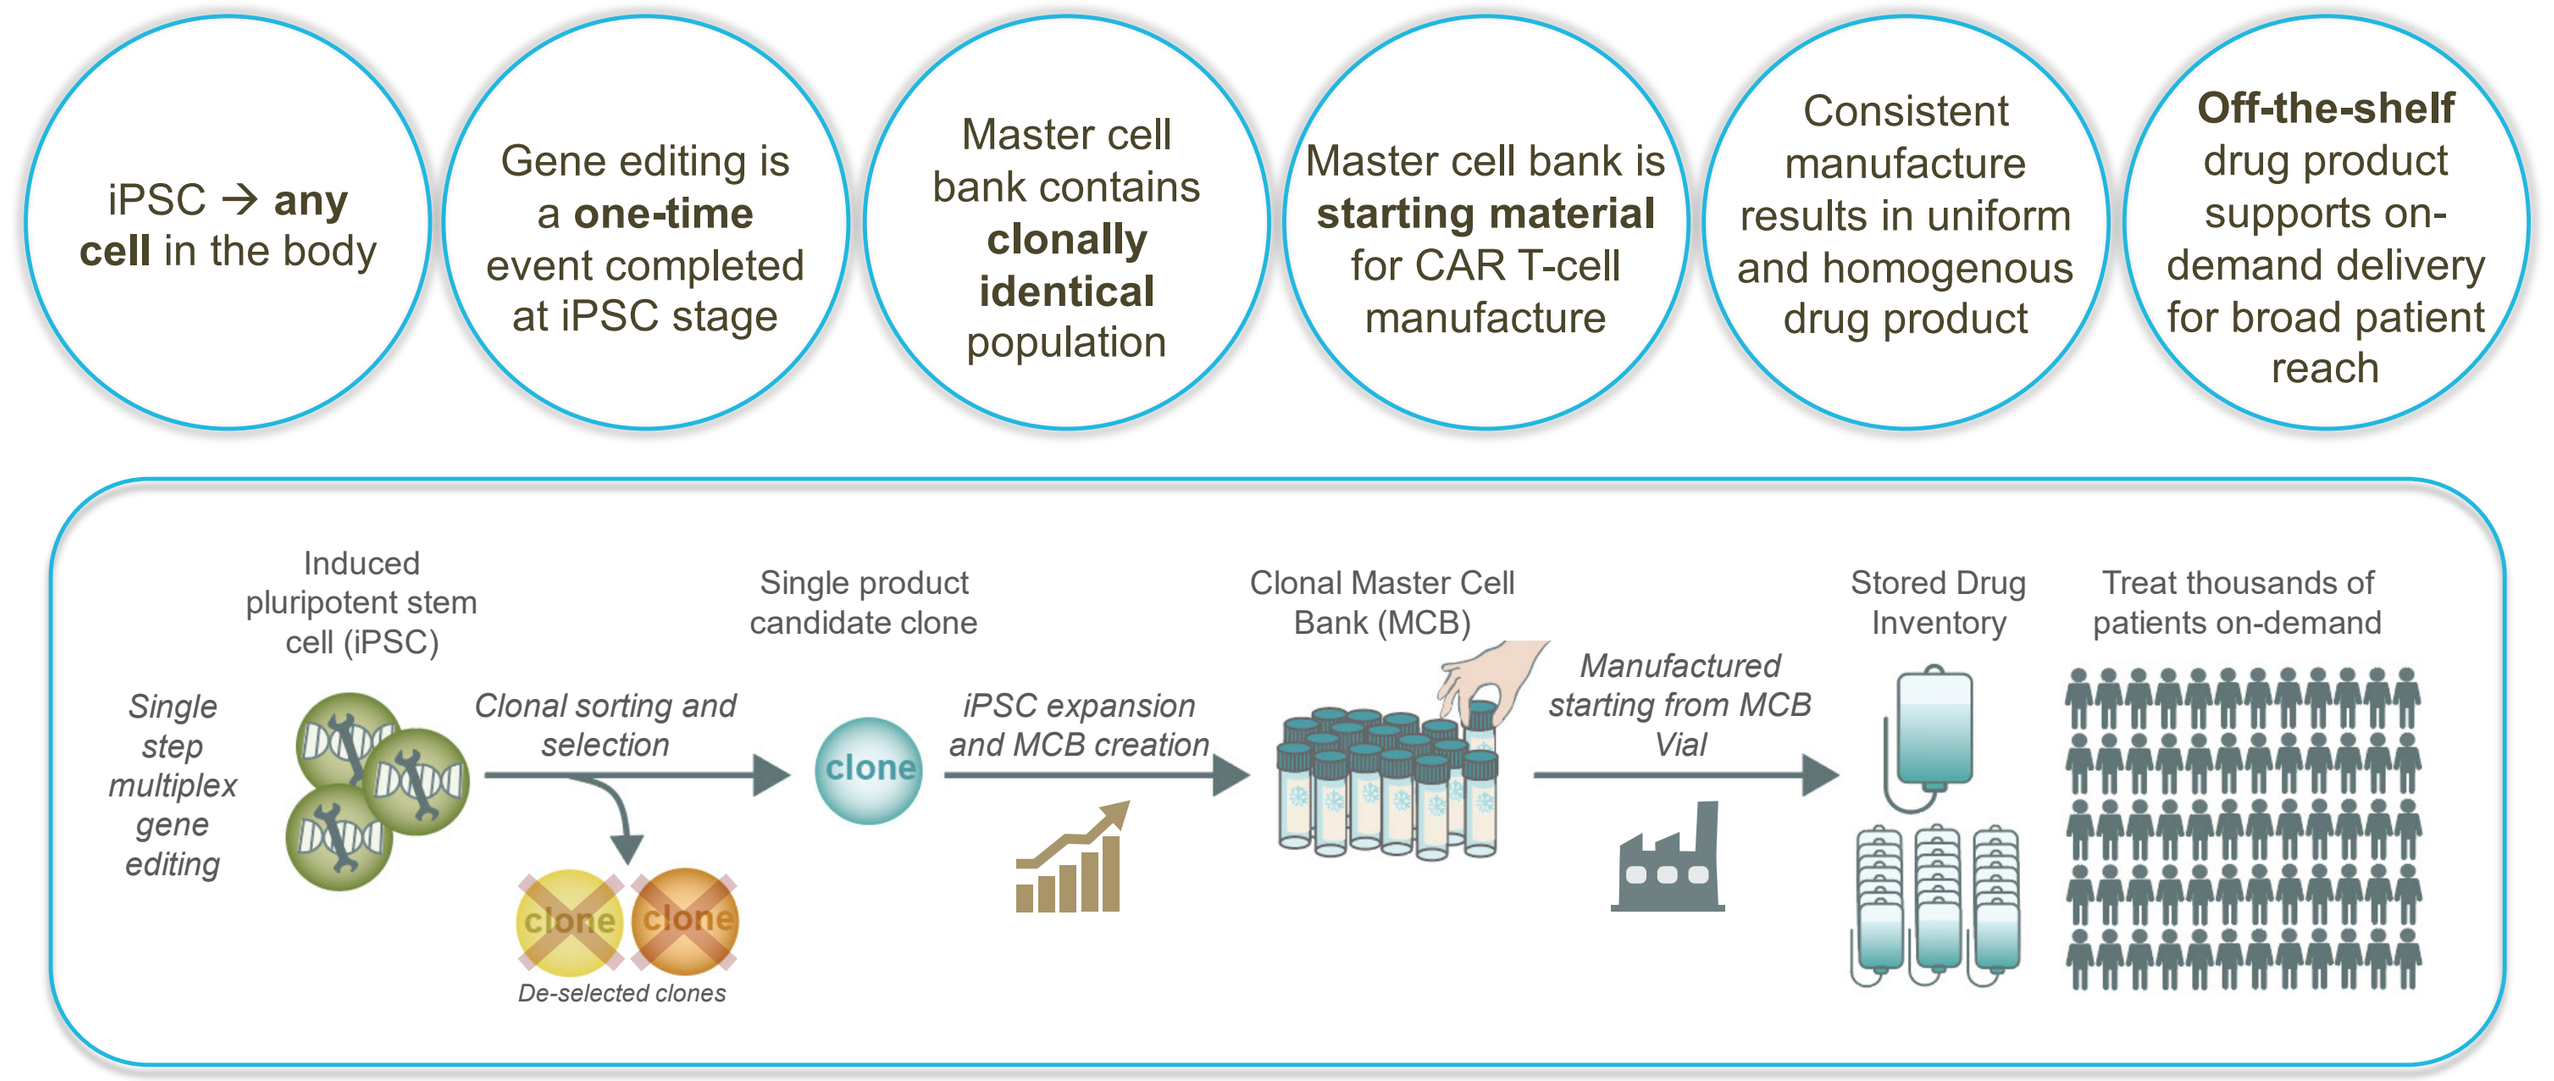
iPSC → any cell in the body
Gene editing is a one-time event completed at iPSC stage
Master cell bank contains clonally identical population
Master cell bank is starting material for CAR T-cell manufacture
Consistent manufacture results in uniform and homogenous drug product
Off-the-shelf drug product supports on-demand delivery for broad patient reach
Single step multiplex gene editing
Induced pluripotent stem cell (iPSC)
Clonal sorting and selection
De-selected clones
Single product candidate clone
clone
iPSC expansion and MCB creation
Clonal Master Cell Bank (MCB)
Manufactured starting from MCB Vial
Stored Drug Inventory
Treat thousands of patients on-demand

Figure 2. FT836, an Off-the-Shelf Next-Generation Multi-Antigen-Targeting CAR T-Cell Drug Product Candidate

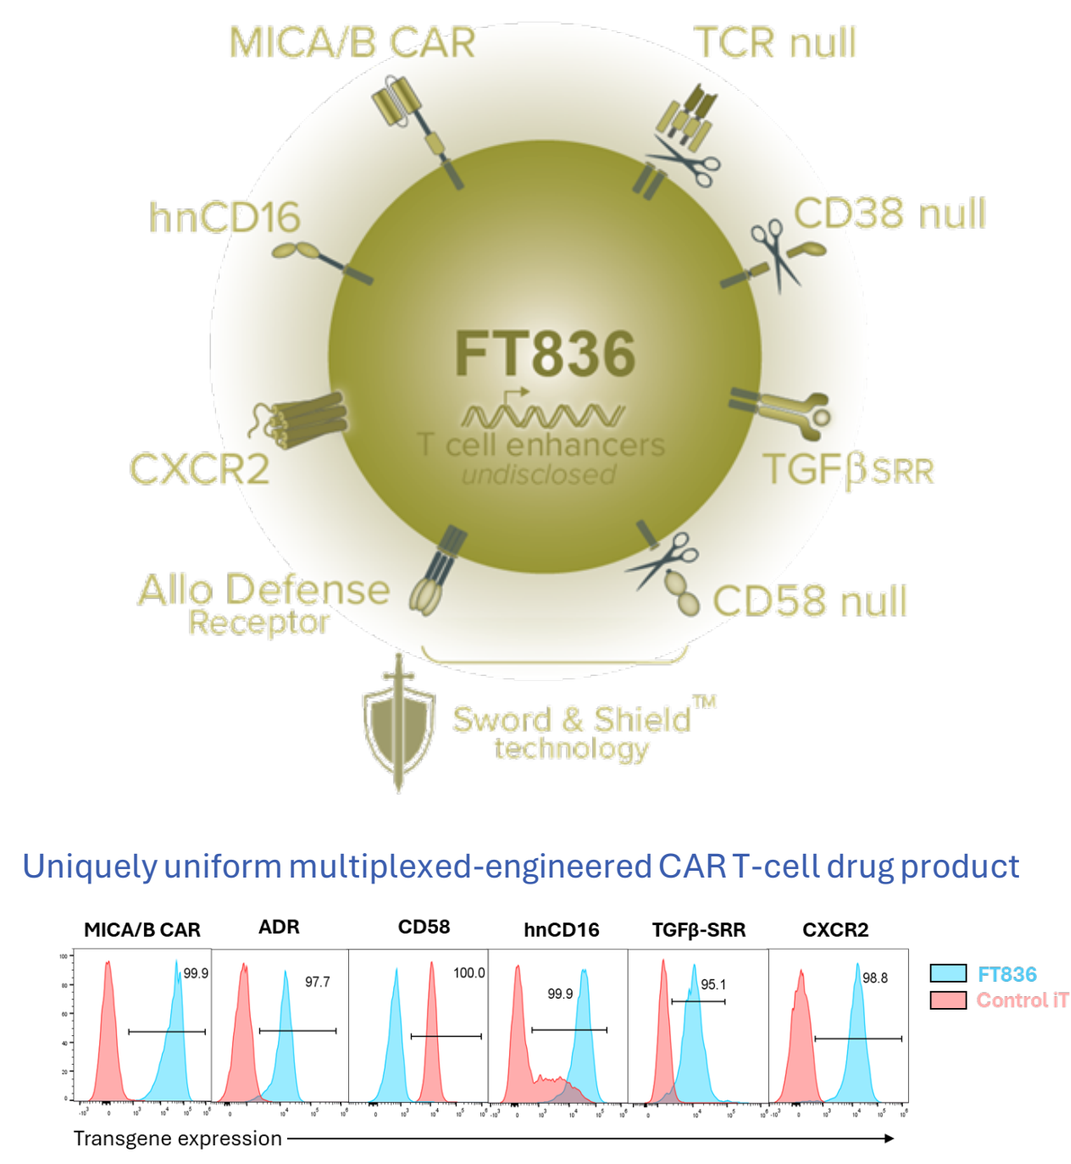
MICA/B CAR
TCR null
hnCD16
CD38 null
FT836
T cell enhancers undisclosed
CXCR2
TGFβSRR
Allo Defense Receptor
CD58 null
Sword & Shield™ technology
Uniquely uniform multiplexed-engineered CAR T-cell drug product
MICA/B CAR
ADR
CD58
hnCD16
TGFβ-SRR
CXCR2
FT836
Control IT
Transgene expression

- **3MICA/B CAR:** Innovative strategy to broadly target tumors while minimizing healthy tissue recognition
- **hnCD16:** Enables ADCC when combined with therapeutic antibodies to expand antigen targeting
- **Sword & Shield™ (ADR/CD58 null):** Designed to reduce or eliminate the need for conditioning chemotherapy
- **T-Cell Enhancer(s):** Optimized support for persistence and potency
- **CXCR2/TGFβ SRR:** Engineered for enhanced tissue trafficking and function within inhibitory tumor microenvironments
- **TCR Null:** Complete bi-allelic disruption of TRAC ablates TCR expression and eliminates the possibility of GvHD
- **CD38 Null:** Potential to enhance metabolic cell fitness

Figure 3. Phase1 Study Design (NCT07216105)

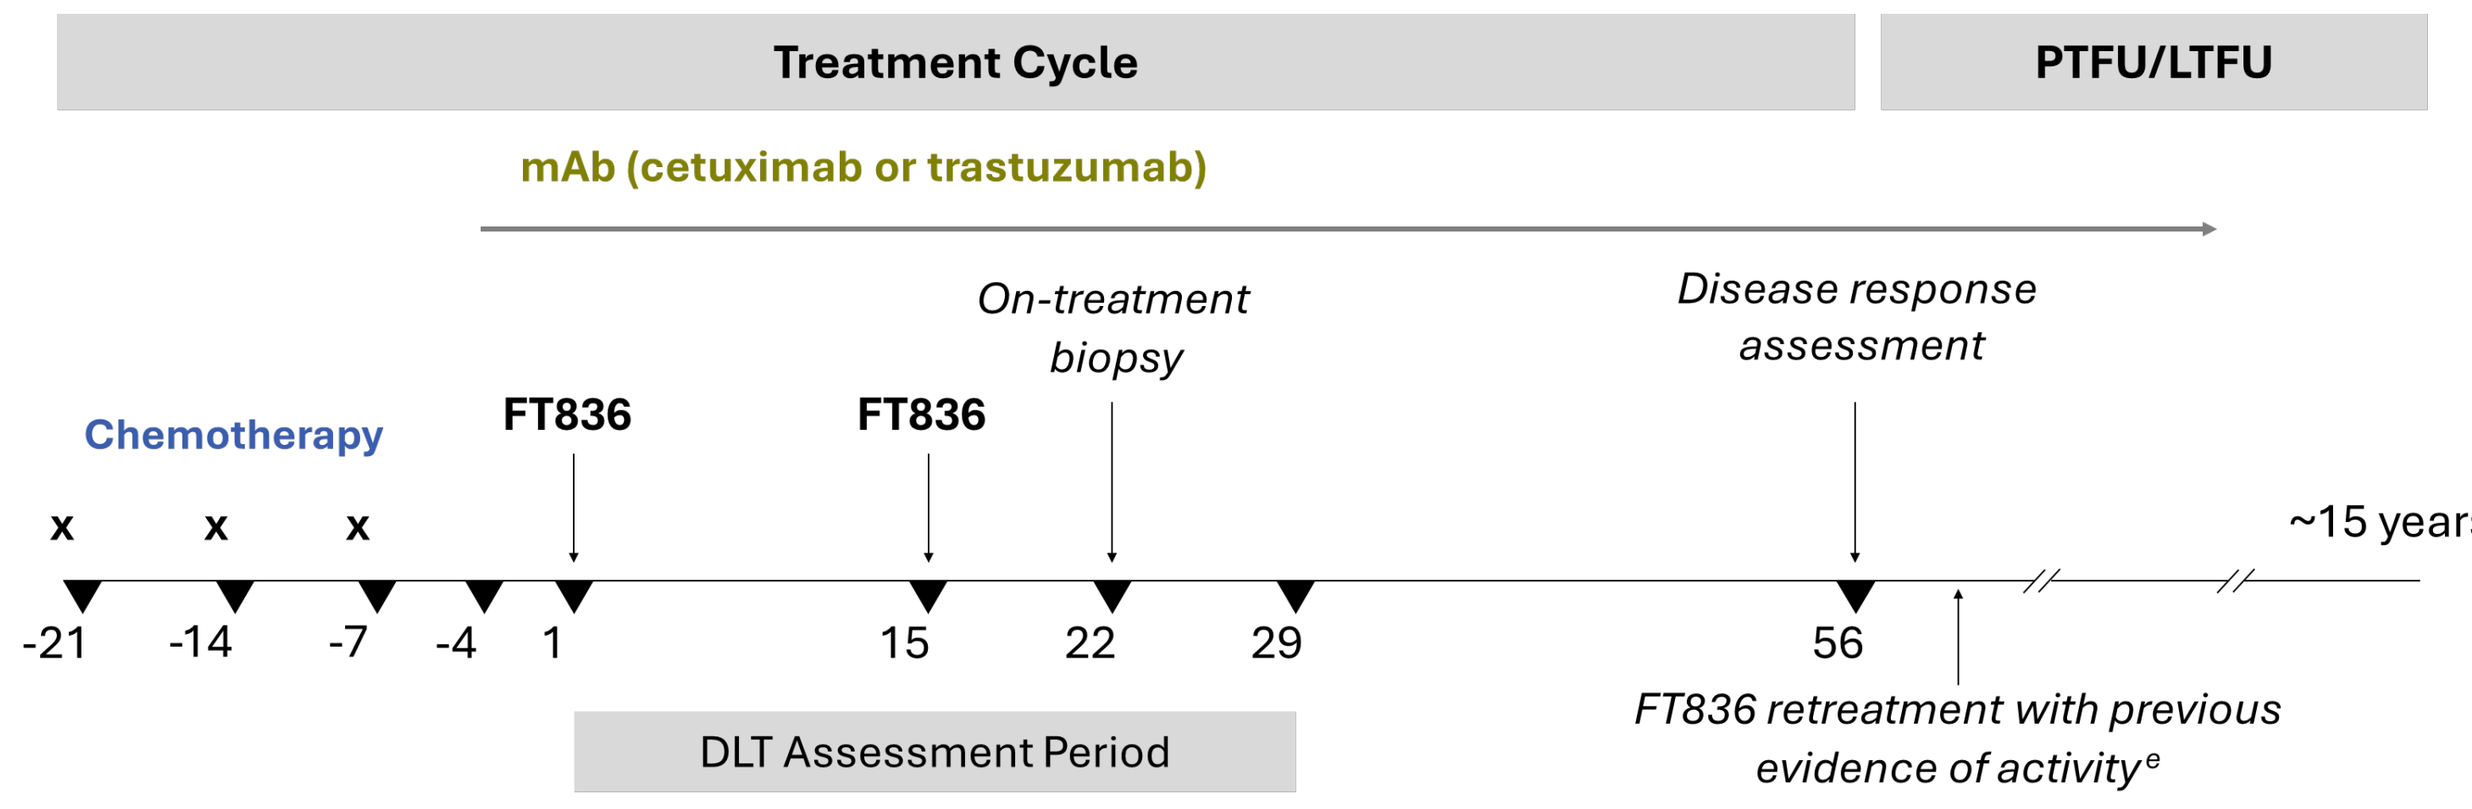
Treatment Cycle
PTFU/LTFU
mAb (cetuximab or trastuzumab)
On-treatment biopsy
Disease response assessment
Chemotherapy
FT836
FT836
x x x
~15 years
-21 -14 -7 -4 1 15 22 29 56
DLT Assessment Period
FT836 retreatment with previous evidence of activity[e]

*D8 FT836 in DL2 only

Regimen A: FT836 Monotherapy
Regimen C: FT836 + Cetuximab
Regimen E: FT836 + Trastuzumab
Regimen B: Paclitaxel Chemotherapy Followed by FT836
Regimen D: Paclitaxel Chemotherapy Followed by FT836 + Cetuximab
Regimen F: Paclitaxel Chemotherapy Followed by FT836 + Trastuzumab

Methods

Key Eligibility Criteria

- **Regimen C:** HNSCC, NSCLC, and CRC with or without prior EGFR-directed therapy, depending on driver mutation status
- **Regimen E:** HER2-expressing cancers defined as IHC 3+ and/or IHC 2+ in ≥50% of tumor cells
- Received at least 1 prior systemic therapy with no curative treatment option
- Anticipated life expectancy of at least 3 months
- No active CNS involvement by malignancy
- No prior CAR T-cell or other cellular therapy

Patient Enrollment

Table 1. Baseline Characteristics

Characteristic		Regimen C: FT836 + cetuximab (N=6)	Regimen E: FT836 + trastuzumab (N=3)
Median Age		46.0 (30, 65)	54.0 (39, 63)
Sex	Male	4 (67)	0
	Female	2 (33)	3 (100)
ECOG	0	4 (67)	1 (33)
	1	2 (33)	2 (67)
Tumor Type	Colorectal	6 (100)	0
	Endometrial	0	1 (33)
	GEJ	0	1 (33)
	Ovarian	0	1 (33)*
HER2 IHC 3+		0	3 (100)
BRAF	Wild-type	4 (67)	2 (67)
	BRAF-V600E	1 (17)	0
	Mutated-other	1 (17)	0
KRAS	Wild-type	4 (67)	1 (33)
	KRAS-G12V	2 (33)	0
	Not assessed	0	2 (67)
Prior EGFR Therapy		4 (67)	0
Prior HER2 Therapy		0	3 (100)*
Median Prior Lines of Therapy		4 (2,7)	3 (2,7)

Data cutoff 20 Apr 2026; * Includes data point entered post data cut.

Safety

Table 2. Adverse Events of Interest by Regimen, Any Grade and Grade >3

Adverse Event of Interest	Regimen C (N=6) n (%)		Regimen E (N=3) n (%)	
	All Grades	Grade 3+	All Grades	Grade 3+
CRS	0	0	0	0
ICANS	0	0	0	0
DLT	0	0	0	0
Infections*	1 (17)	1 (17)	0	0

Data cutoff 20 Apr 2026; * Sepsis

Overall Adverse Events

- Most common any grade TEAEs across both regimens included dermatitis acneiform (n=5) and blood LDH increased (n=4)
- Grade ≥3 TEAEs in Regimen C included abdominal pain, large intestinal obstruction, sepsis, alk phos increase, and seizure (n=1 for all)
- One Grade ≥3 TEAE of hyponatremia was observed in Regimen E
- FT836-related TEAEs included chills, fatigue, anemia, and hyperhidrosis (n=1 for all)

Results

Preliminary Anti-Tumor Activity Observed in KRAS Wild-Type Colorectal Cancer

CLINICAL CASE 1

- 45-year-old male with metastatic CRC, KRAS wild-type
- 7 prior lines of therapy, including cetuximab
- At study entry, multiple metastatic sites including lung, bone and nodal disease
- Enrolled on Regimen C, DL1, receiving 3 x 10^8 cells/dose on Days 1 and 15
- FT836-related adverse events were not observed
- CEA decreased from 487 at screening to 185 on Day 56
- All target lesions reduced in size (overall 70 mm → 55 mm); new hepatic lesions

Uniform expression of MICA/B in biopsy | Early and sustained reduction in CEA | Meaningful tumor volume decrease in target lesions

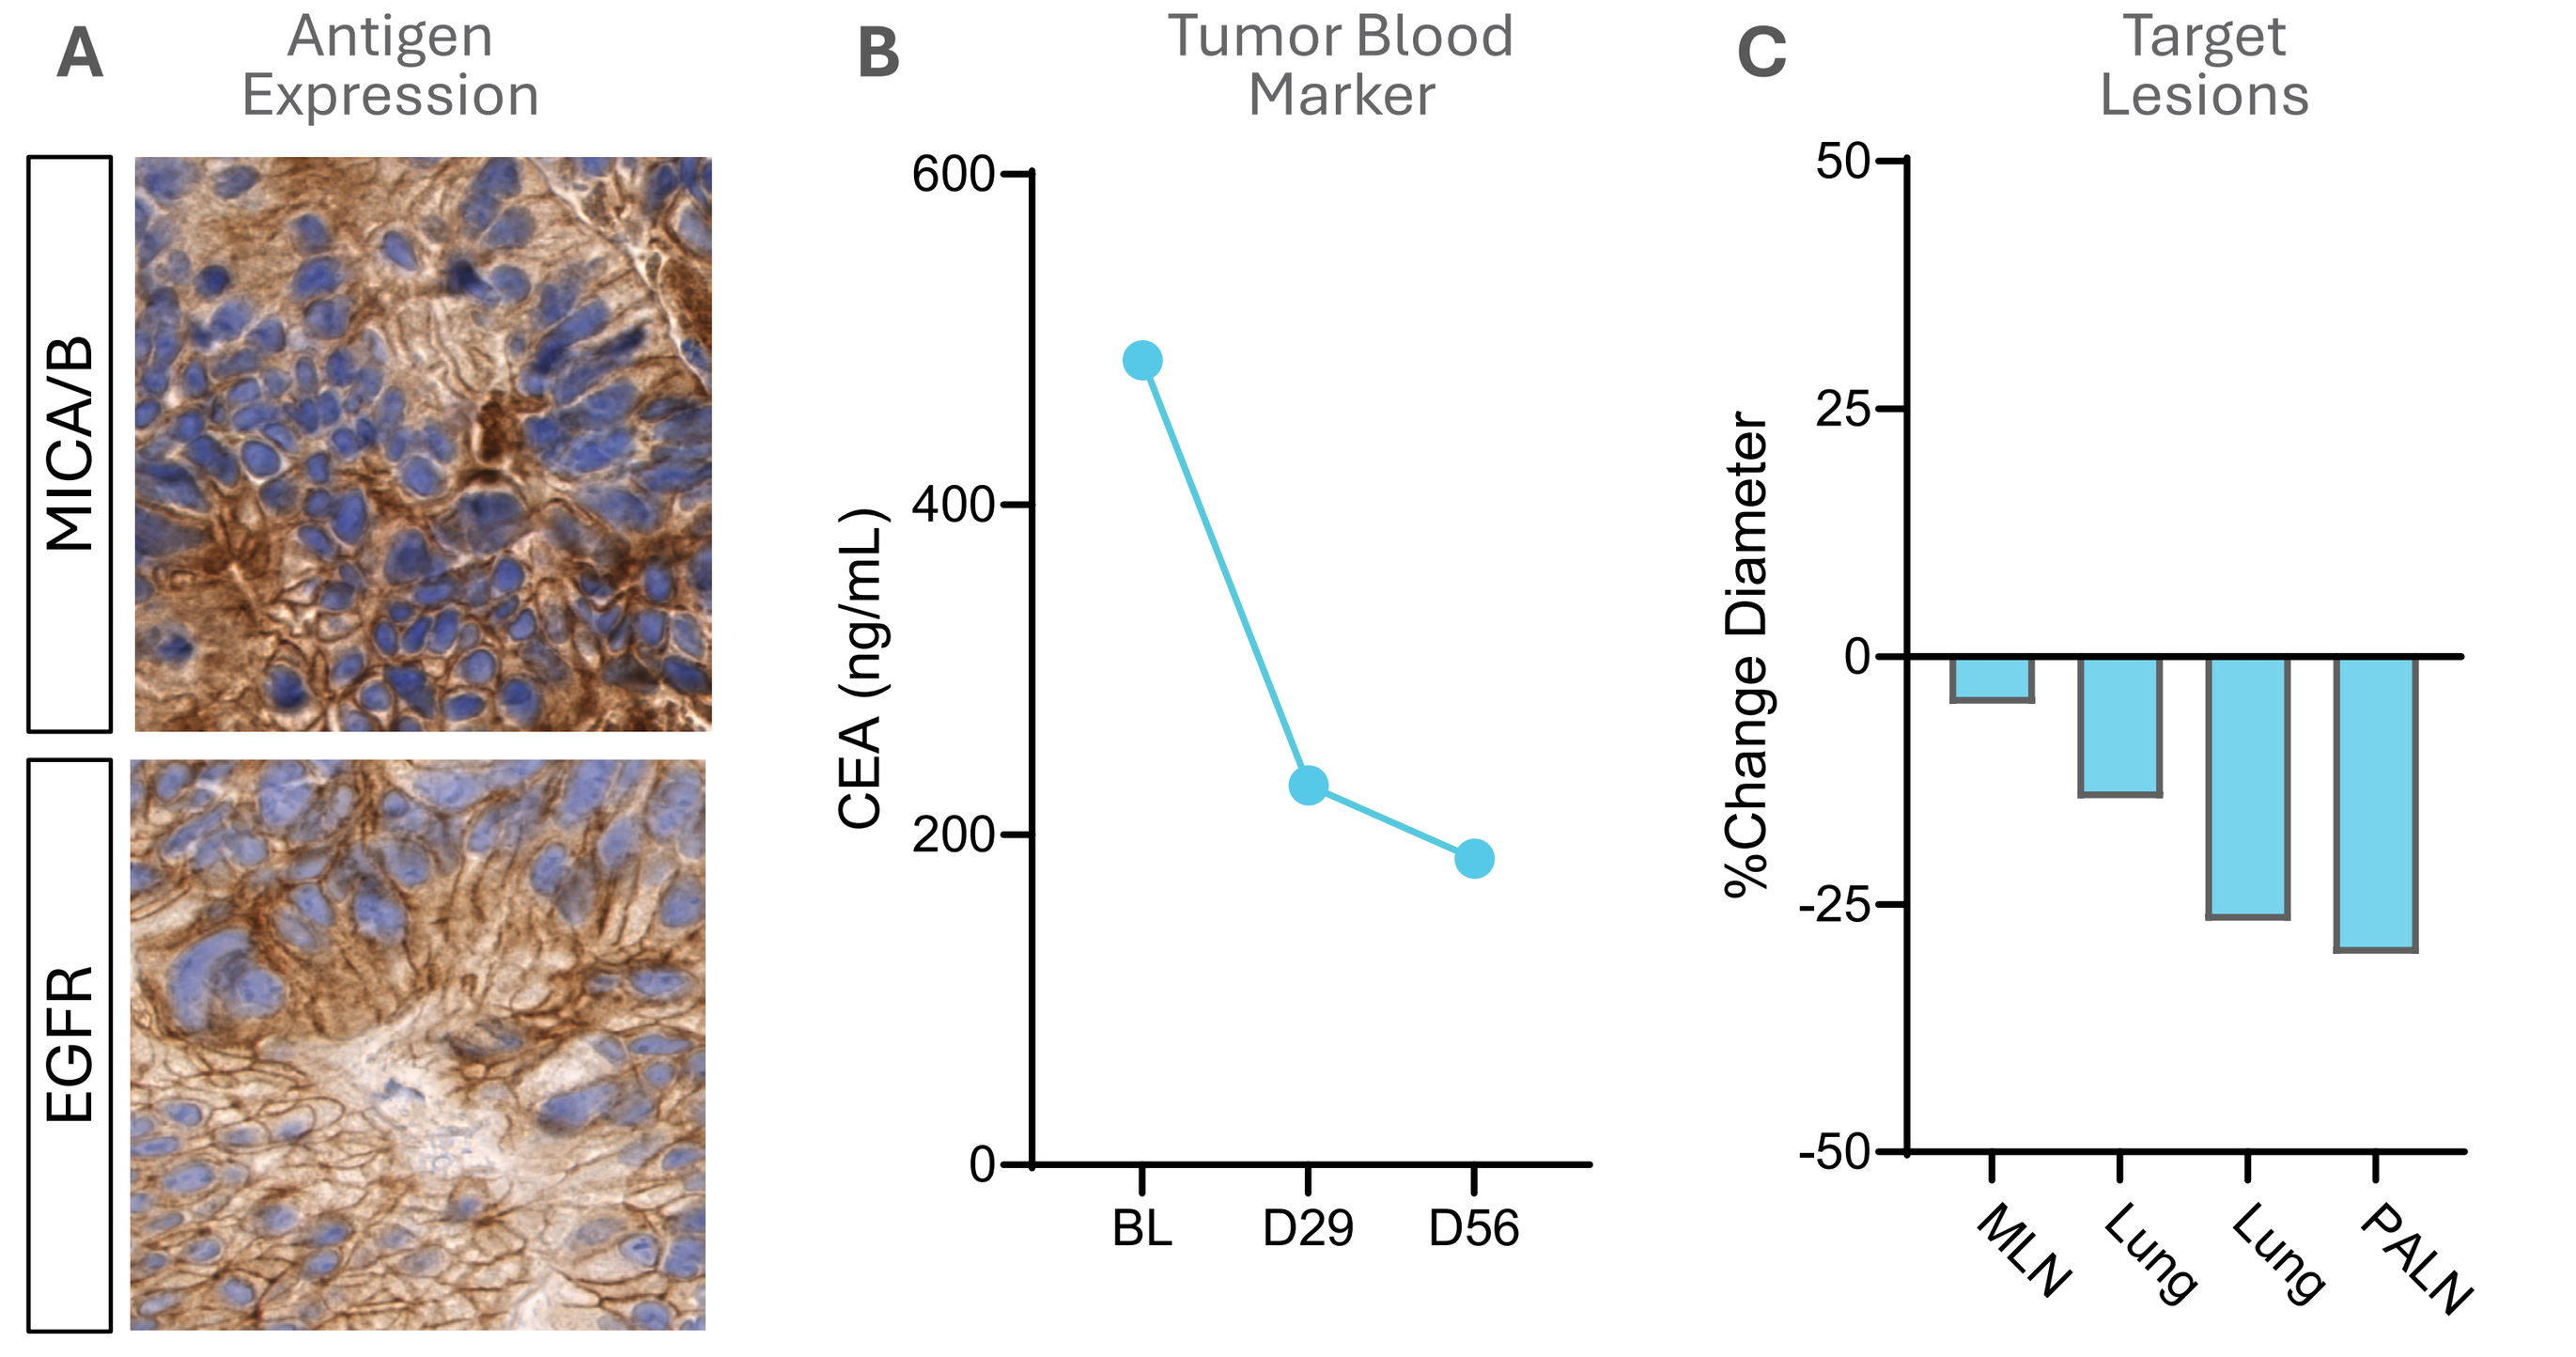
A
Antigen Expression
MICA/B
EGFR
B
Tumor Blood Marker
CEA (ng/mL)
600
400
200
0
BL
D29
D56
C
Target Lesions
%Change Diameter
50
25
0
-25
-50
MLN
Lung
Lung
PALN

Figure 4. Case Study of patient with CRC, KRASwt; Regimen C DL1. **(A)** Robust co-expression of MICA/B and EGFR target antigen in tumor-bearing lymph node biopsy as assessed by IHC. Images shown at 35x magnification. **(B)** Marked decrease in CEA tumor blood marker through Cycle 1. **(C)** Diameter changes for individual tumor target lesions assessed by RECIST analysis, with an overall 19% reduction in sum of diameters. LN=lymph node, MLN=mesenteric lymph node, PALN=paraaortic lymph node.

CLINICAL CASE 2

- 53-year-old male with metastatic CRC, KRAS wild-type
- 7 prior lines of therapy, including multiple cetuximab-containing
- At study entry, multiple metastatic sites including lung, hepatic, adrenal, and nodal disease
- Enrolled in Regimen C DL2, receiving 9 x 10^8 cells/dose on Days 1, 8, and 15
- FT836-related adverse events were not observed
- CEA decreased from 350 to a nadir of 333; CA19-9 decreased from 258 to 144 on Day 56
- Tumor-related pain resolved
- Target lesion size stable overall with RECIST-assessed stable disease (post data cut-off)

Uniform expression of MICA/B in biopsy | Reduction in CEA and CA19-9 | Meaningful tumor volume decrease in target lesion

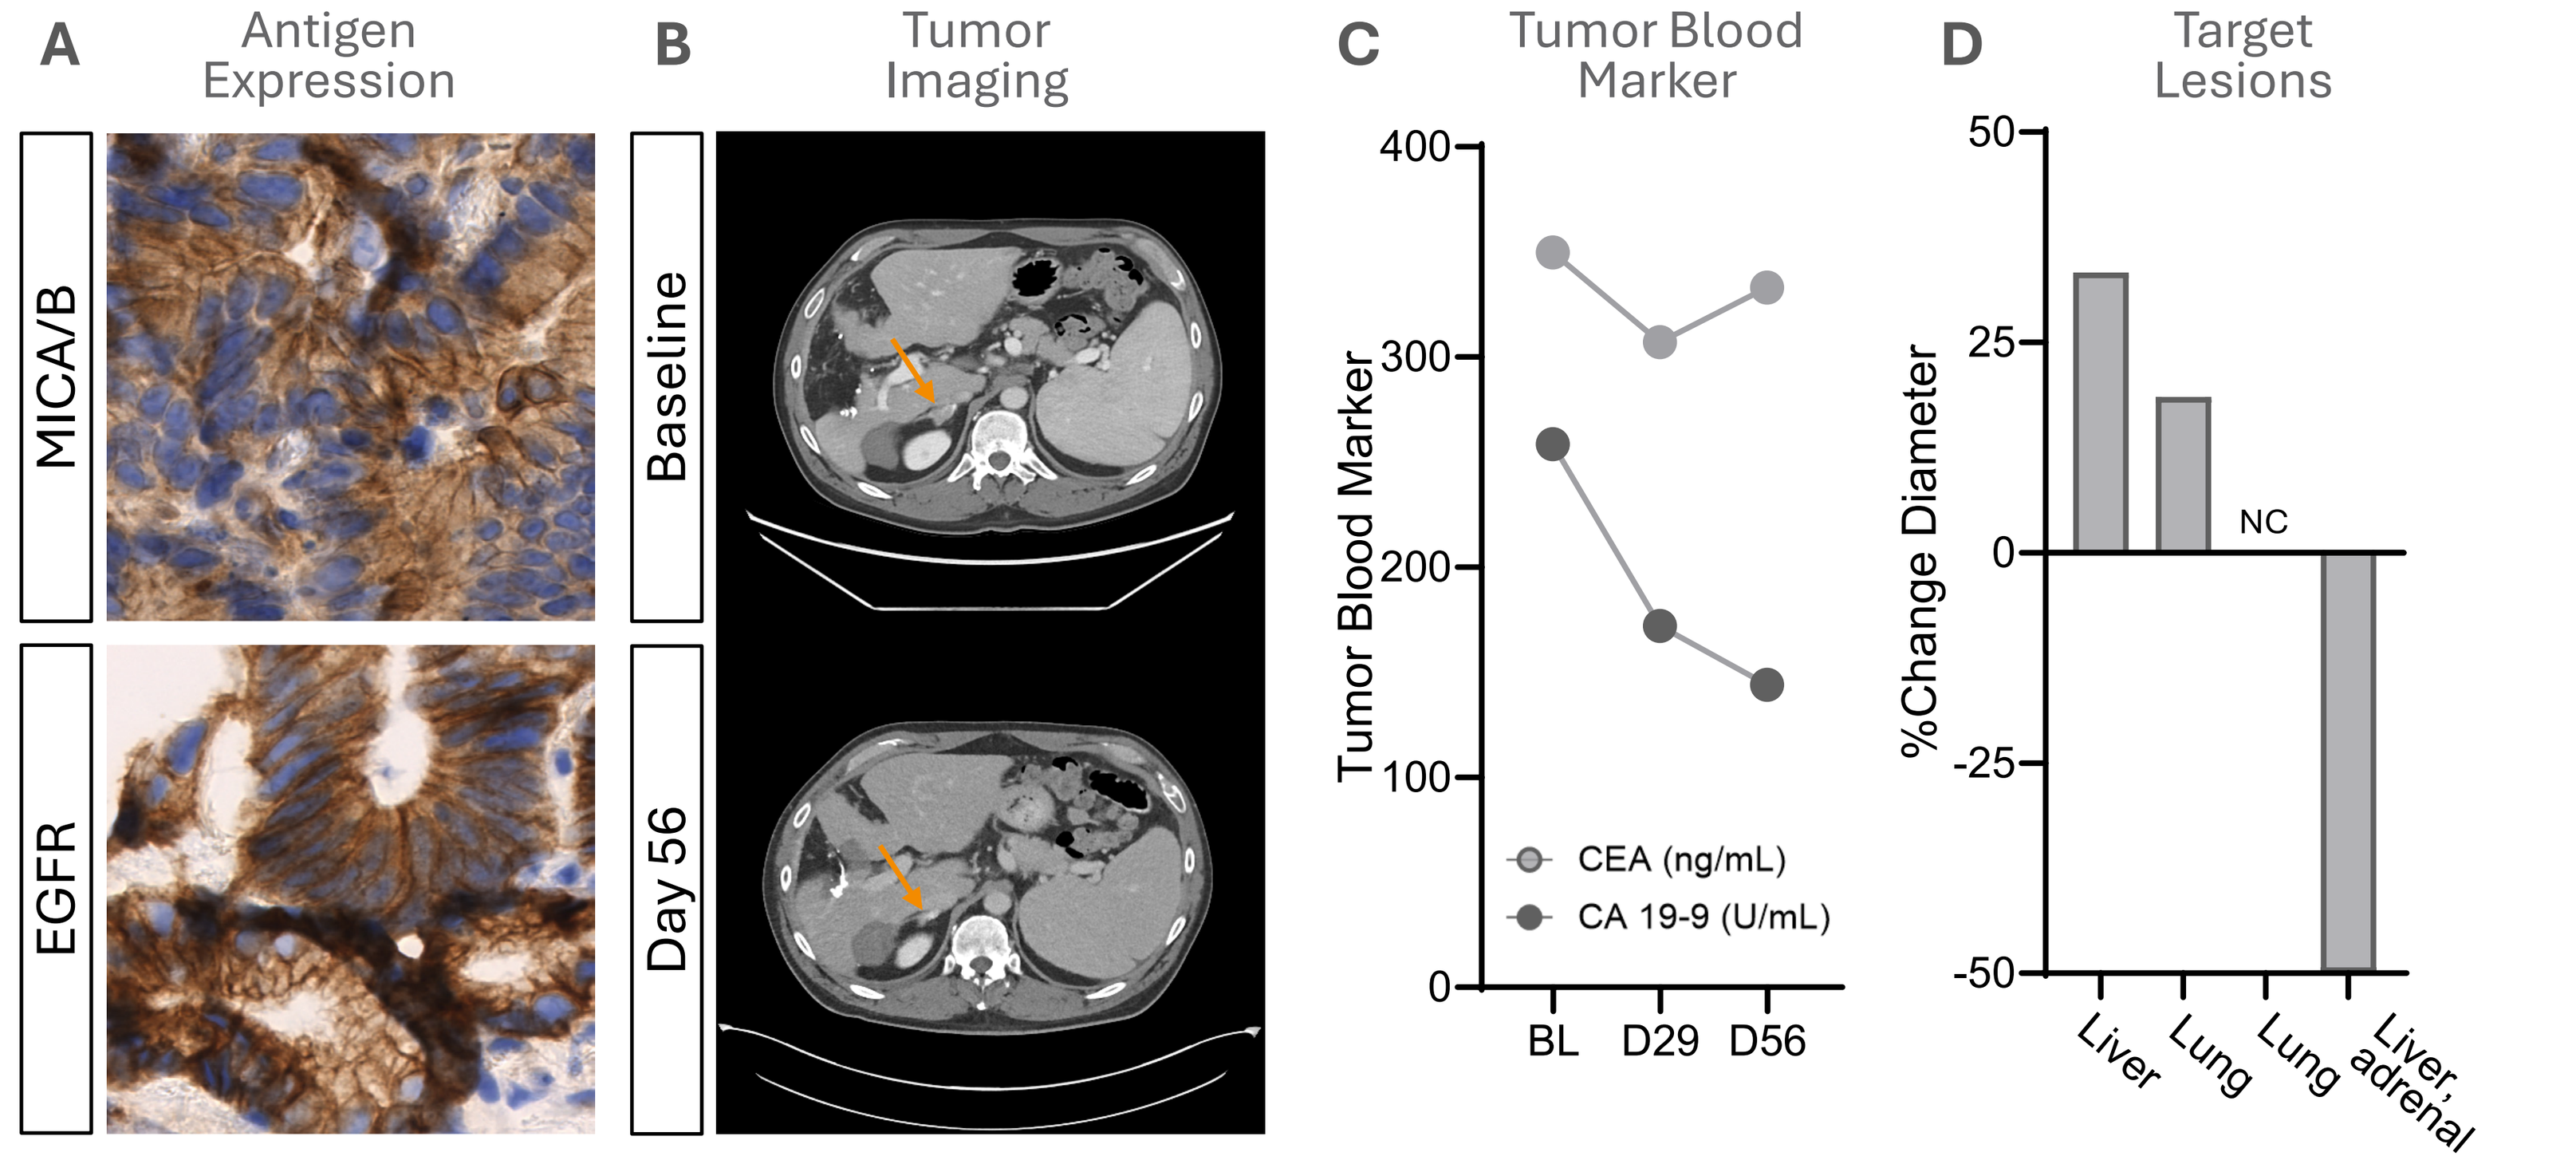
A
Antigen Expression
MICA/B
EGFR
B
Tumor Imaging
Baseline
Day 56
C
Tumor Blood Marker
Tumor Blood Marker
400
300
200
100
0
CEA (ng/mL)
CA 19-9 (U/mL)
BL
D29
D56
D
Target Lesions
%Change Diameter
50
25
0
-25
-50
NC
Liver
Lung
Lung
Liver, adrenal

Figure 5. Case Study of patient with CRC, KRASwt; Regimen C DL2. **(A)** Strong co-expression of MICA/B and EGFR target antigen in lung biopsy as assessed by IHC. Images shown at 35x magnification. **(B)** Baseline and D56 CT imaging demonstrating changes in target burden. **(C)** Decreases in CEA and CA19-9 tumor blood marker levels in Cycle 1. **(D)** Diameter changes for individual tumor target lesions measured by RECIST analysis, including a 52% reduction in a liver lesion. NC=no change.

Of 5 patients evaluable for response (Regimen C: DL1 n=3, DL2 n=1; Regimen E: DL1 n=1), one patient showed SD.

Observations of Persistence, Trafficking, and Activity

FT836 persistence in the periphery and tumor tissue post infusion

Redistribution of CD8 cells in tumor tissue post infusion

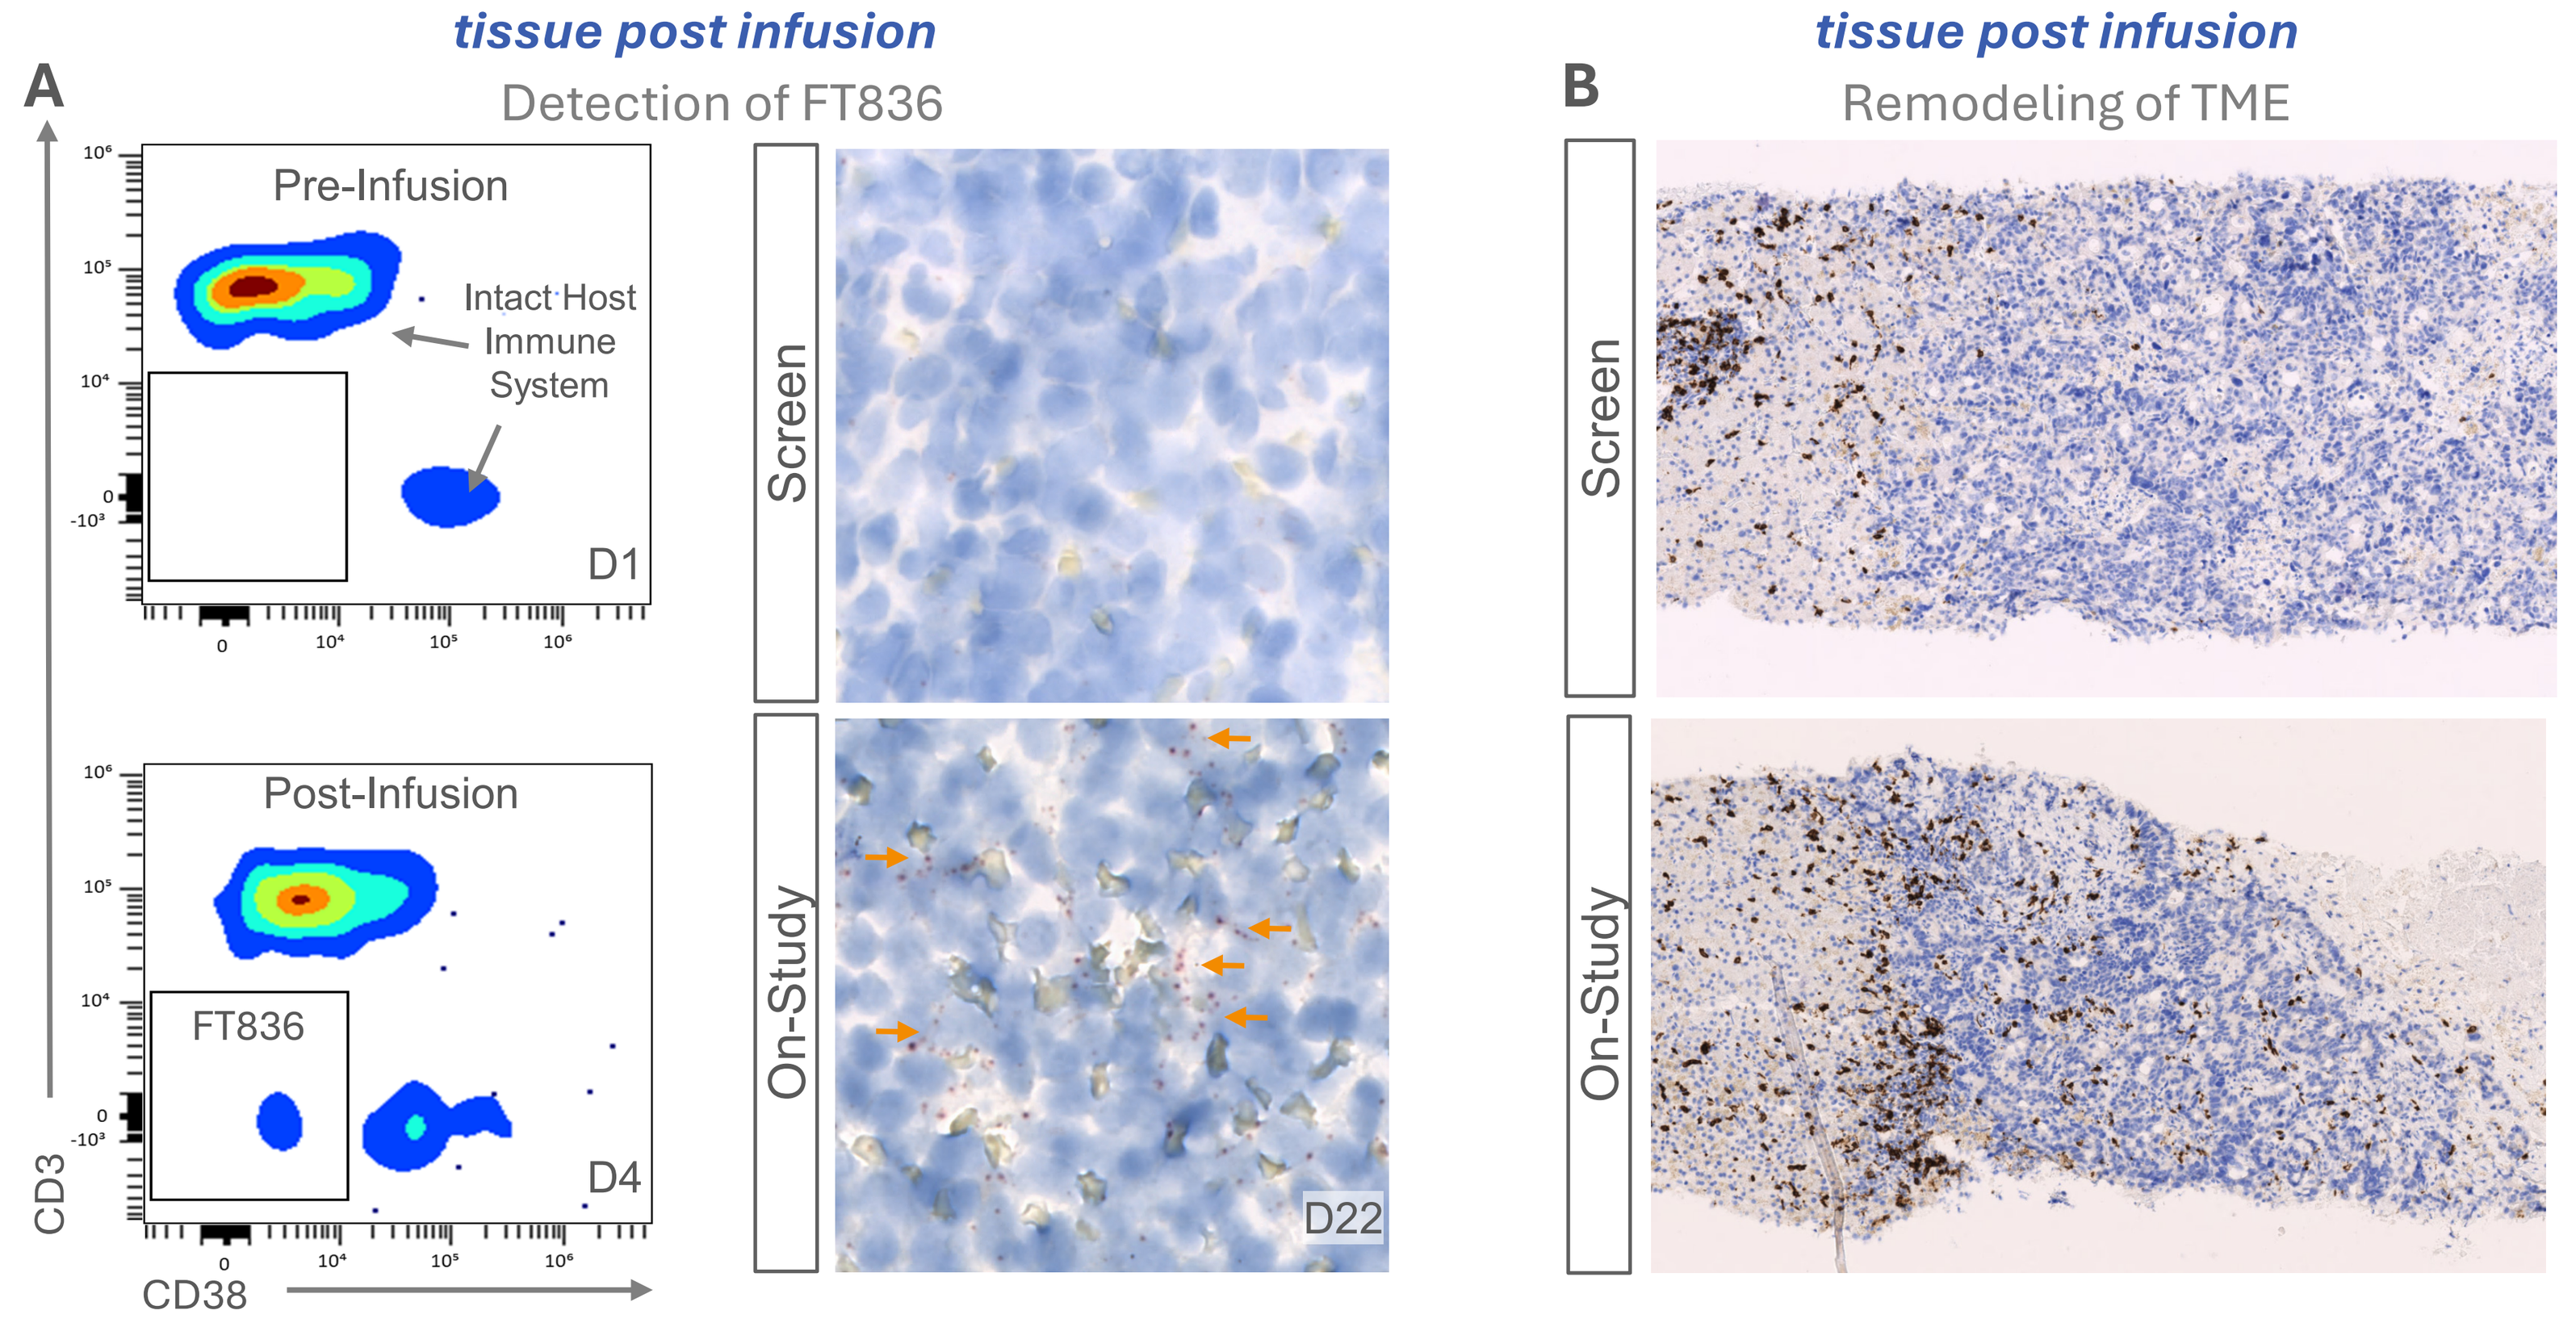
A
Detection of FT836
Pre-Infusion
Intact Host Immune System
D1
Post-Infusion
FT836
D4
CD3
CD38
Screen
On-Study
D22
B
Remodeling of TME
Screen
On-Study

Figure 6. (A) FT836 is detected in the periphery and tissue. FT836 (CD7+TGFβRIII+,CD3-,CD38-) is detected within the intact host immune system in peripheral blood by flow cytometry up to a week following first dose with a peak at Day 4 (left). FT836 (orange arrow) is observed in biopsy tissue by hnCD16-targeted RNAscope at Day 22 (right; 40x magnification) suggesting efficient trafficking to tumor-bearing tissue and prolonged persistence in the tissue relative to blood. Data shown are from Regimen C DL1 patient samples. **(B)** At Screen (top), CD8+ cells detected by IHC imaging are spatially separated from the tumor. On-study imaging analysis demonstrates CD8+ cells adjacent to and in direct contact with tumor cells, supportive of treatment-associated activation and recruitment of endogenous immune cells. Images are shown at 4x magnification from Regimen C DL2 patient liver biopsies. Data collected post data cut-off.

Conclusion

- FT836 is a 9-point edited CAR T-cell drug product developed to overcome key challenges associated with treatment of solid tumors
- Manufactured from a clonal MCB starting material, FT836 is uniquely uniform and consistent with its incorporated genetic edits and manufactured at large scale for true off-the-shelf availability
- FT836 administered in combination with cetuximab or trastuzumab without conditioning chemotherapy is well-tolerated with no DLT, CRS, or ICANS observed
- Preliminary evidence of anti-tumor activity was observed in the two refractory and metastatic KRASwt CRC patients, with each having 7 prior lines of therapy
 - Meaningful reduction in target lesions size observed for both patients; significant reduction in CEA tumor biomarker seen in both patients
 - Detection of FT836 in the periphery and tumor tissue illustrates the unique ability to traffic and functionally persist without conditioning chemotherapy
- Preliminary data supports further evaluation of FT836 in KRASwt CRC

Abbreviations: ADCC = antibody-dependent cellular cytotoxicity; ADR = allo-defense receptor; BRAF = B-Raf proto-oncogene; CAR = chimeric antigen receptor; CEA = carcinoembryonic antigen; CNS = central nervous system; CRC = colorectal cancer; CRS = cytokine-release syndrome; CXCR2 = C-X-C motif chemokine receptor 2; D = Day; DL = dose level; DLT = dose-limiting toxicity; ECOG = Eastern Cooperative Oncology Group; EGFR = epidermal growth factor receptor; GEJ = gastroesophageal junction; GvHD = graft-versus-host disease; HER2 = human epidermal growth factor receptor 2; HNSCC = head and neck squamous cell carcinoma; ICANS = immune effector cell-associated neurotoxicity syndrome; IHC = immunohistochemistry; iPSC = induced pluripotent stem cell; KRAS = Kirsten rat sarcoma viral oncogene homolog; LDH = lactate dehydrogenase; LTFU = long-term follow-up; mAb = monoclonal antibody; MIC A/B = MHC class I chain-related proteins A and B; NSCLC = non-small cell lung cancer; PTFU = post-treatment follow-up; RECIST = Response Evaluation Criteria in Solid Tumors; SD = stable disease; TCR = T-cell receptor; TEAE = treatment emergent adverse event; TGFβ SRR = transforming growth factor-beta signal redirect receptor; TRAC = T-cell receptor alpha constant. **Reference:** Goulding J, Yeh WI, Hancock B, et al. Med. 2023;4(7):457-477.e8.

Sponsored by Fate Therapeutics, Inc. **Abstract 2560.**
Presented at the ASCO Annual Meeting. May 29–June 2, 2026. Chicago, IL.
Contact Information: Manish Patel patel069@umn.edu